Abstract 586

Multiomic Profiling Identifies Pharmacodynamic and Response Biomarkers for Casdozokitug with PD-L1/VEGF Blockade in Unresectable Hepatocellular Carcinoma from SRF-388-201 Phase 2 Study

Xiao Wang, Sruthi Ravindranathan, Helen Tang, Koho Iizuka, Narendiran Rajasekaran, Rosh Dias, Theresa LaVallee, Varun N Kapoor

Coherus Oncology, Inc., Redwood City, CA

Background

- **Interleukin (IL)-27, a heterodimeric IL-12/IL-23 family cytokine, is expressed by activated macrophages and dendritic cells (DC) and acts as an immunoregulatory cytokine that dampens effector T and natural killer (NK) cell functions. IL-27 is highly expressed by tumor-associated macrophages in several cancers, including hepatocellular carcinoma (HCC), making it a potential target for therapy.**
- Casdozokitug (casdozo) is a first-in-class and only clinical-stage IL-27 antagonistic antibody. IL-27 neutralization by casdozokitug stimulates immune activation and promotes anti-tumor response in cancer patients.
- In preclinical models, inhibition of IL-27 specifically results in antitumor activity in orthotopic HCC models and requires NK cells for antitumor activity.
- In the Phase 1 study SRF-388-101 (NCT04374877), casdozokitug demonstrated a favorable safety profile and anti-tumor activity (PR) as a single agent and in combination with PD-1 inhibitors in indications known to have high levels of IL-27 pathway activation (NSCLC, RCC, and HCC).[1]
- In the Phase 2 study SRF-388-201 (NCT05359861), casdozokitug in combination with atezolizumab/bevacizumab demonstrated a manageable safety profile and a CR rate of 17.2% in unresectable metastatic HCC.[2]
- Biomarker data from this open-label Phase 2 study (SRF-388-201) are presented.

IL-27 Suppresses Antitumor Immunity in the Tumor Microenvironment (TME)

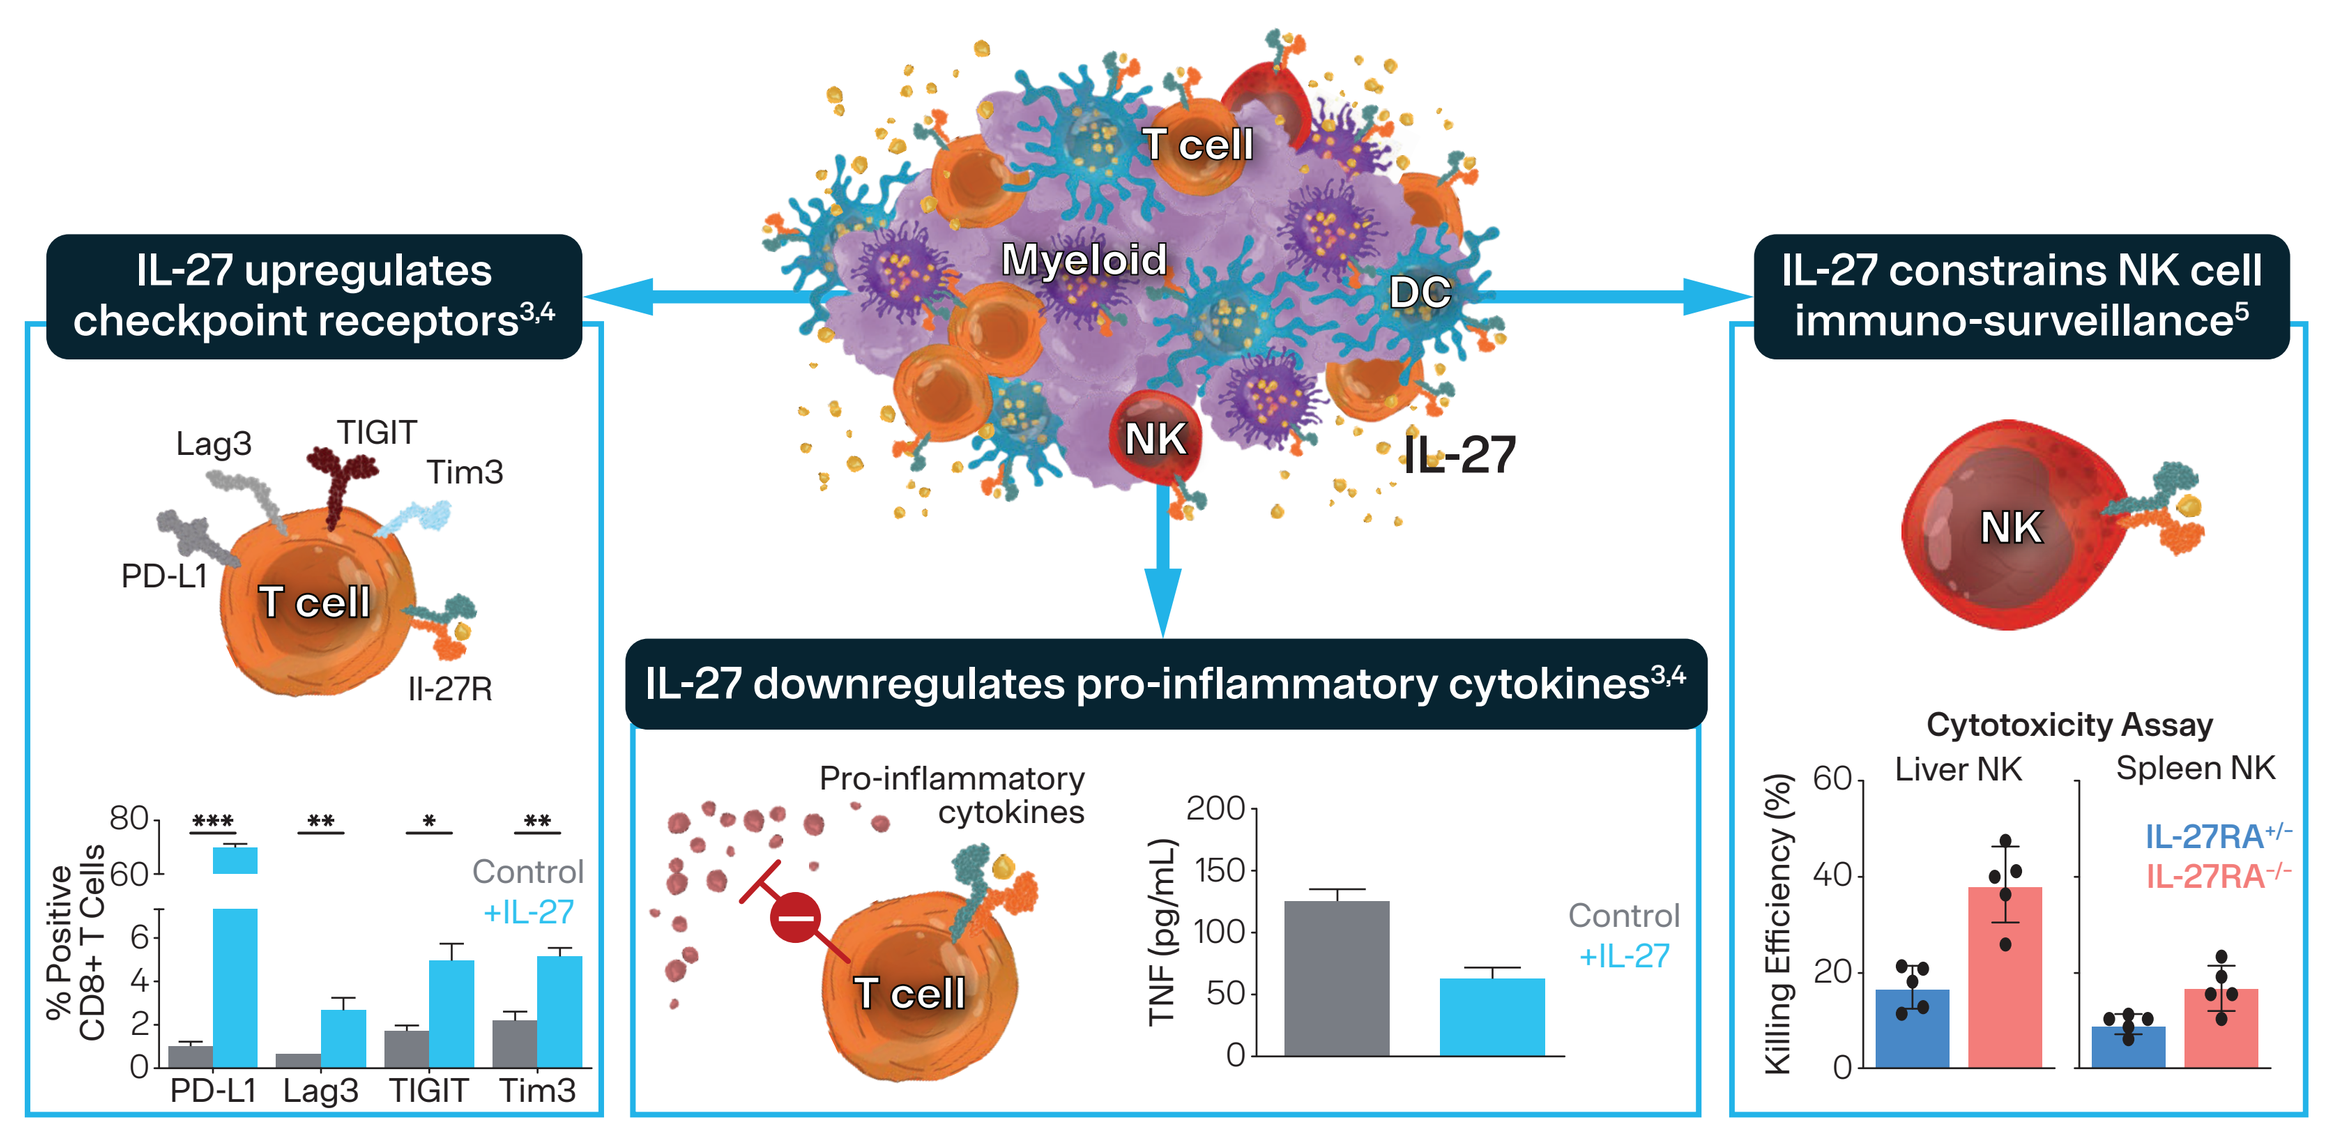
T cell
Myeloid
DC
NK
IL-27
IL-27 upregulates checkpoint receptors3,4
IL-27 downregulates pro-inflammatory cytokines3,4
IL-27 constrains NK cell immuno-surveillance5
Lag3
TIGIT
Tim3
PD-L1
Il-27R
% Positive CD8+ T Cells
Control
+IL-27
Pro-inflammatory cytokines
TNF (pg/mL)
Cytotoxicity Assay
Liver NK
Spleen NK
Killing Efficiency (%)
IL-27RA+/-
IL-27RA-/-

Rationale for Blocking IL-27 in Hepatocellular Carcinoma

High Levels of IL-27 Expression in HCC Tumors

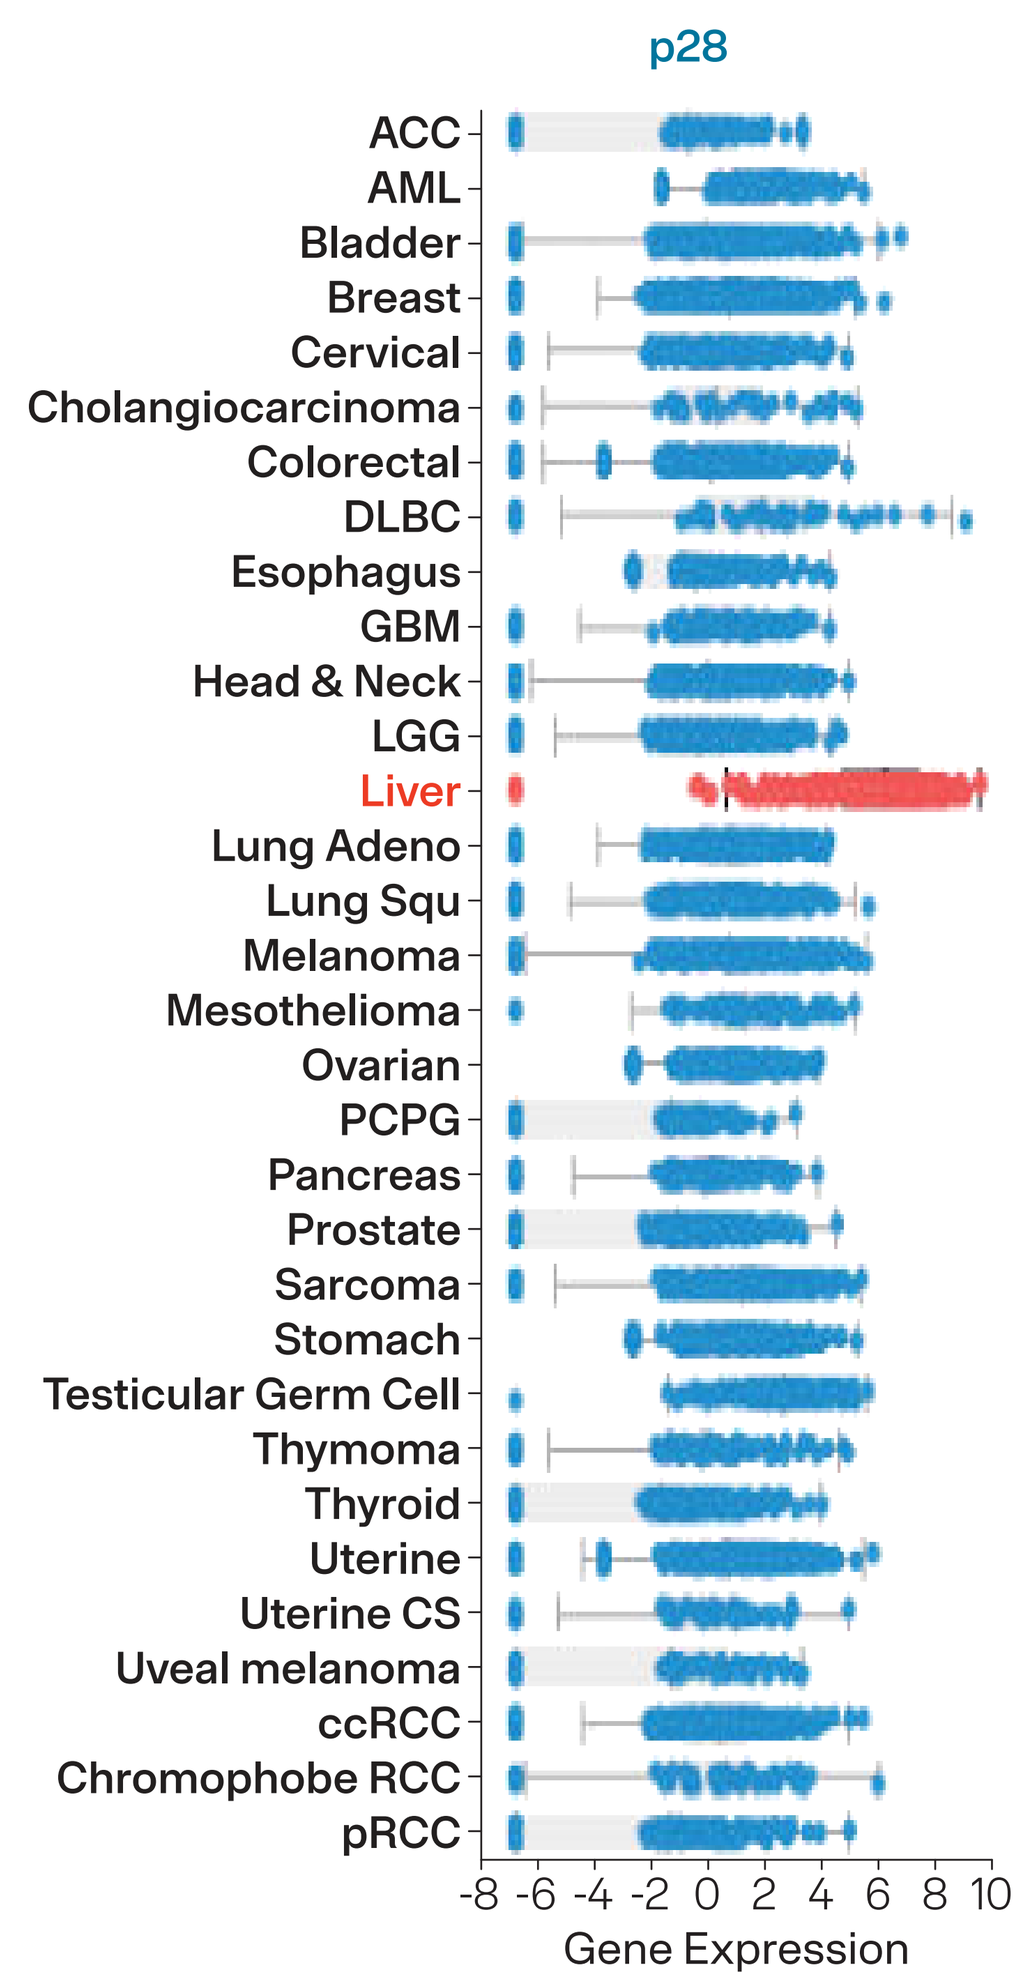
p28
ACC
AML
Bladder
Breast
Cervical
Cholangiocarcinoma
Colorectal
DLBC
Esophagus
GBM
Head & Neck
LGG
Liver
Lung Adeno
Lung Squ
Melanoma
Mesothelioma
Ovarian
PCPG
Pancreas
Prostate
Sarcoma
Stomach
Testicular Germ Cell
Thymoma
Thyroid
Uterine
Uterine CS
Uveal melanoma
ccRCC
Chromophobe RCC
pRCC
Gene Expression

Elevated Levels of EBI3 in Patient Serum

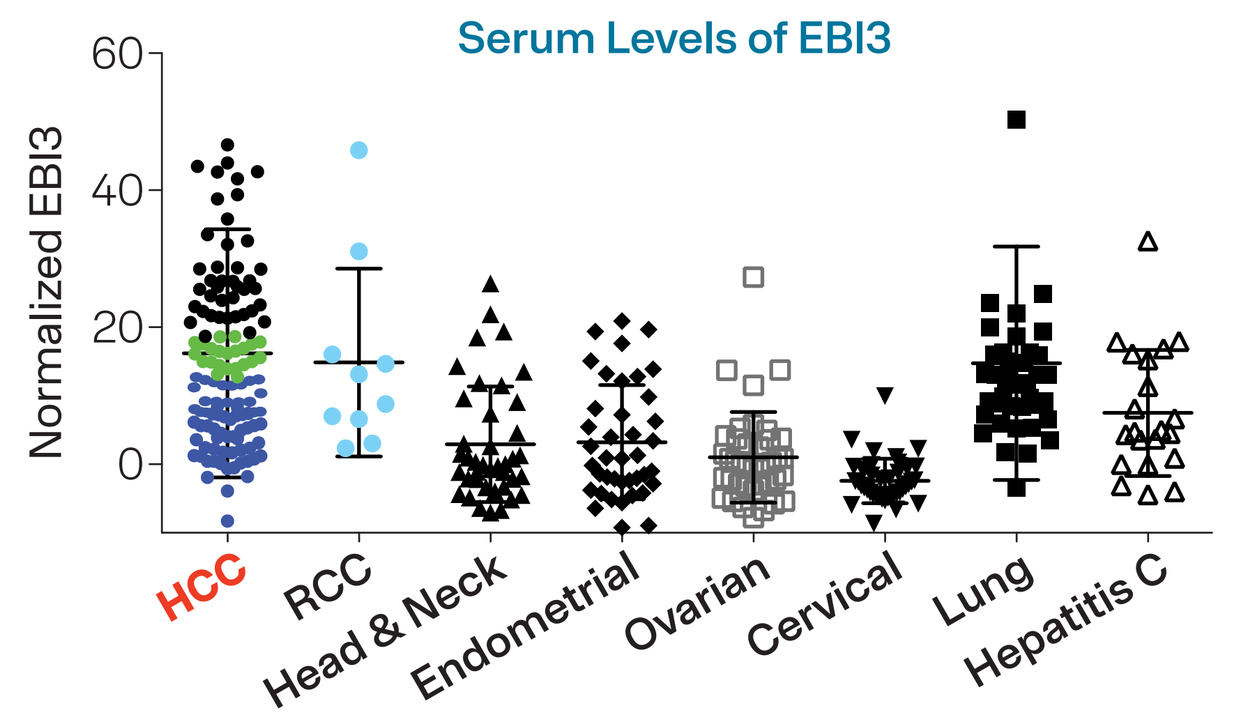
Serum Levels of EBI3
Normalized EBI3
HCC
RCC
Head & Neck
Endometrial
Ovarian
Cervical
Lung
Hepatitis C

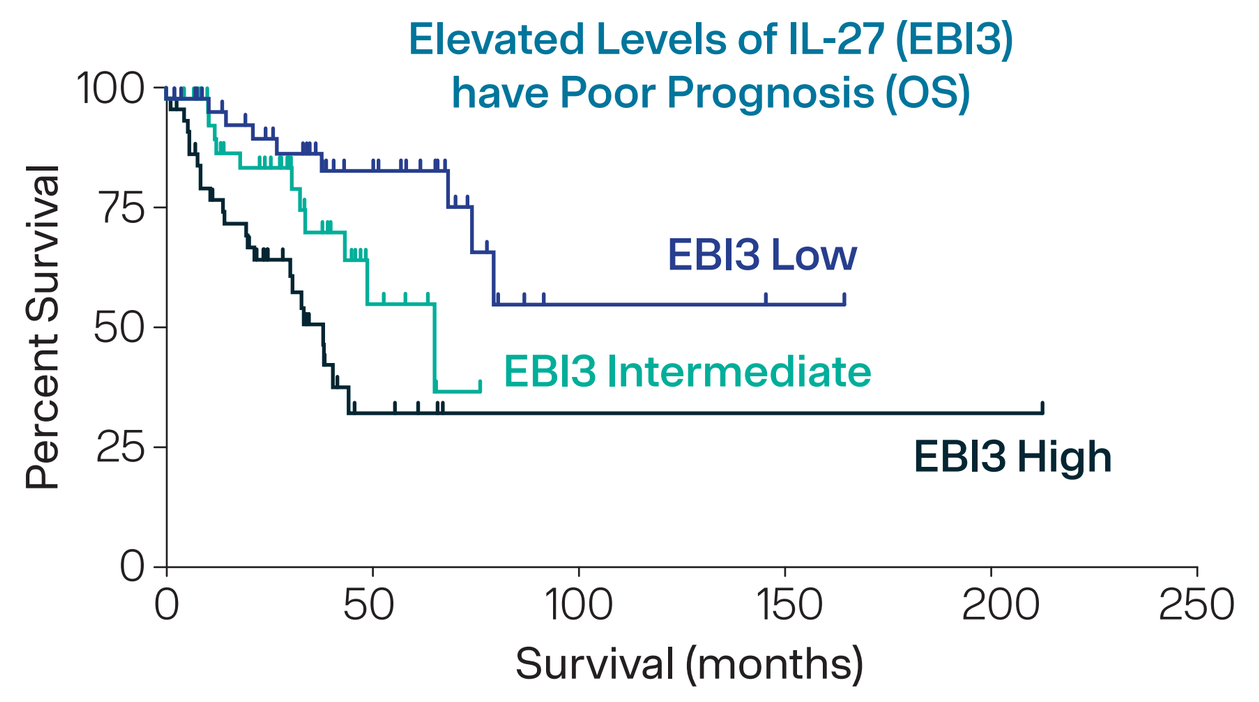
Elevated Levels of IL-27 (EBI3) have Poor Prognosis (OS)
Percent Survival
EBI3 Low
EBI3 Intermediate
EBI3 High
Survival (months)

IL-27 Inhibition Suppresses HCC Tumor Growth in Multiple Murine Models[5]

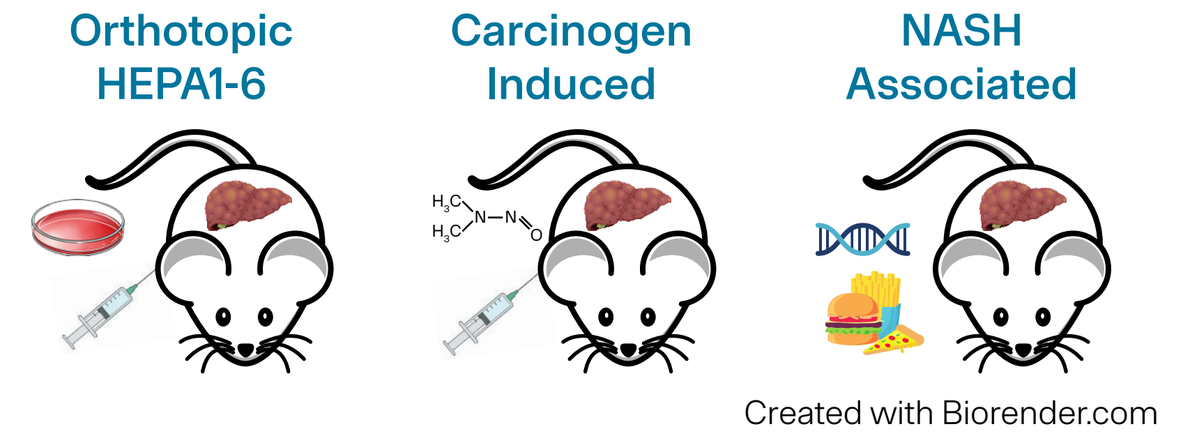
Orthotopic HEPA1-6
Carcinogen Induced
NASH Associated
Created with Biorender.com

Clinical Study Design and Response Outcomes

SRF-388-201: A Phase 2 Trial Evaluating the Efficacy and Safety of Casdozokitug in Combination with Atezolizumab/Bevacizumab in Patients with 1L Advanced or Metastatic HCC

Open-Label (N = 30)
1L unresectable/metastatic HCC
aPD-(L)1 naïve
≥1 measurable lesion
BCLC B or C
Child-Pugh A
ECOG PS 0-1
Controlled HBV or Cured HCV

→ Atezolizumab (atezo) 1200 mg / Bevacizumab (bev) 15 mg/kg / Casdozokitug (casdozo) 10 mg/kg — IV q3w →

Primary endpoint: Safety
Key secondary endpoints: PFS* ORR*, DCR

*RECIST v1.1 (primary) and mRECIST (secondary)

Best overall response (RECIST v1.1), n (%)	Response evaluable (n = 29)
Overall response rate (ORR)	11 (37.9)
Complete response (CR)	5 (17.2)
Partial response (confirmed) (PR)	6 (20.7)
Stable disease (SD)†	8 (27.6)
Progressive disease (PD)	9 (31.0)
Not evaluable (NE)	1 (3.4)

†SD includes one participant with an unconfirmed PR

Casdozokitug in combination with atezolizumab/bevacizumab demonstrated:

- Clinical response irrespective of viral status (eg, HBV, HCV, none)
- A complete response (CR) rate of 17.2%, which is higher than the 8% or 3% CR rate reported in prior Phase 3 studies in HCC (IMbrave150[6] and HIMALAYA[7]), respectively

The combination had an acceptable safety profile with AEs consistent with the known profiles of the individual agents and no new safety signals observed.

Casdozokitug in Combination with Atezolizumab and Bevacizumab (SOC) in HCC Demonstrated Improved Antitumor Activity, Including Depth of Response (CR Rate)

11 Objective Responses per RECIST v1.1, Including 5 CRs

Best % Change from Baseline in Sum of Target Lesions (n = 28)

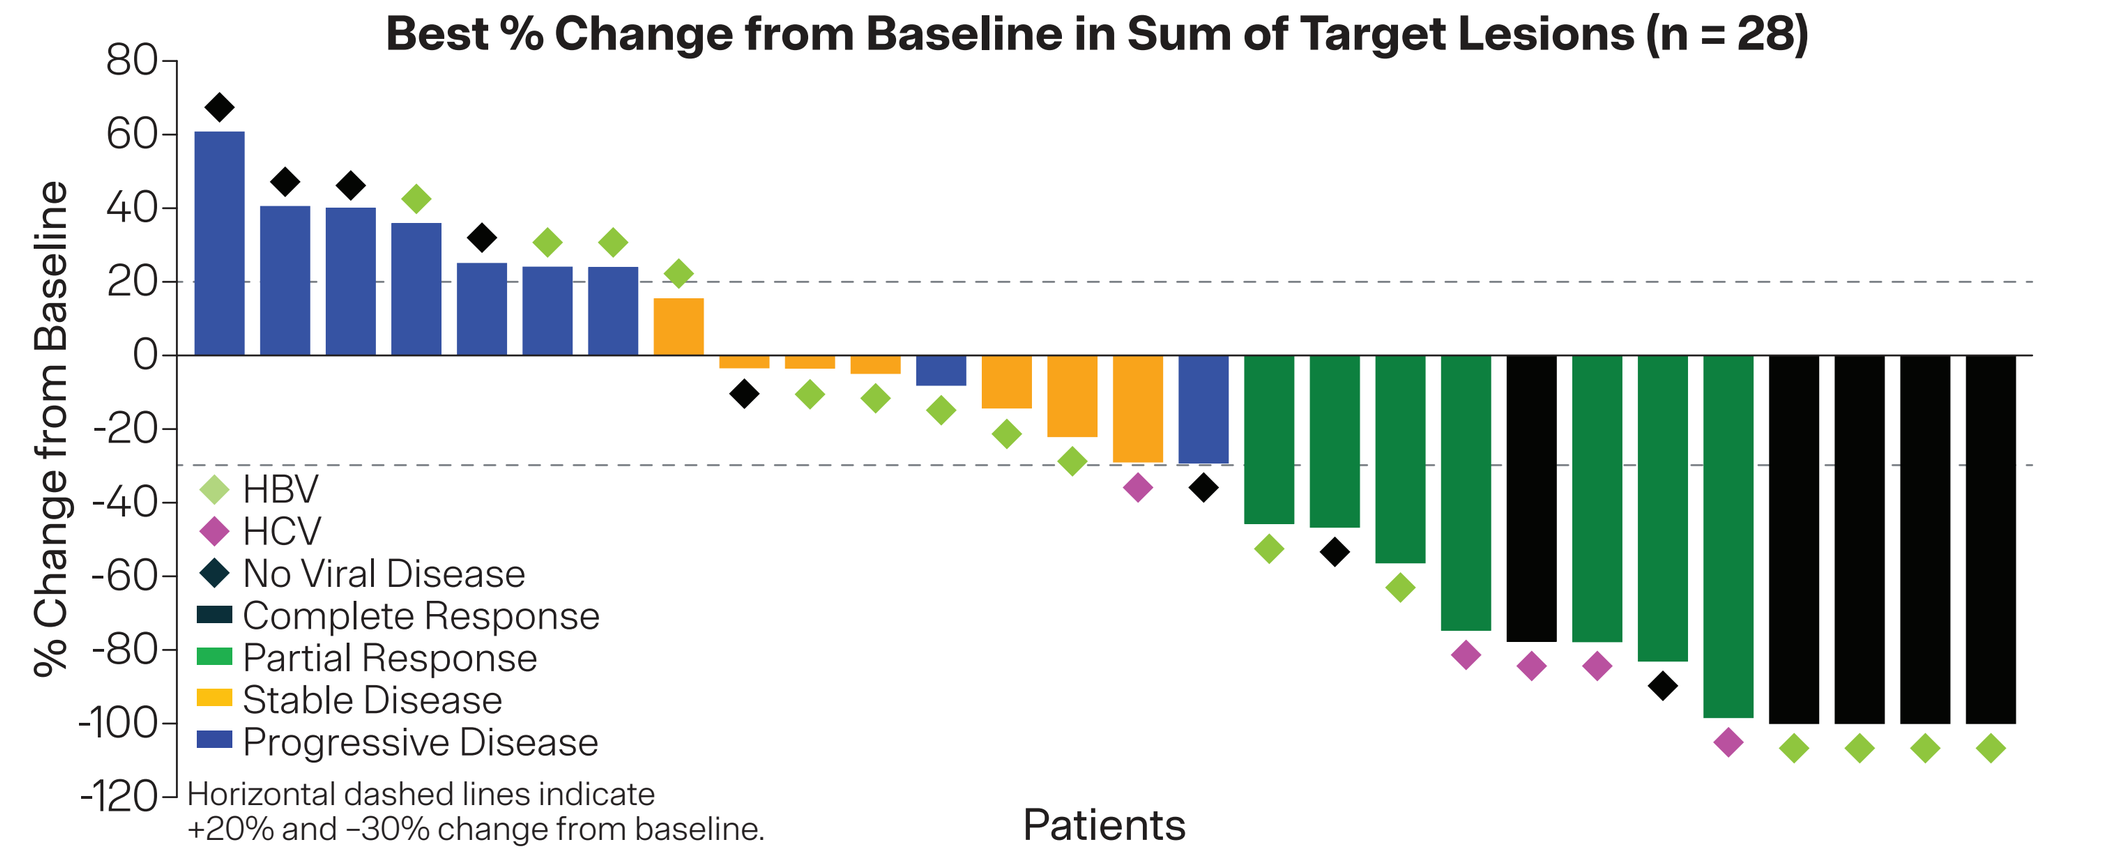
% Change from Baseline
HBV
HCV
No Viral Disease
Complete Response
Partial Response
Stable Disease
Progressive Disease
Horizontal dashed lines indicate +20% and -30% change from baseline.
Patients

Biomarker Samples and Assays Used in SRF-388-201

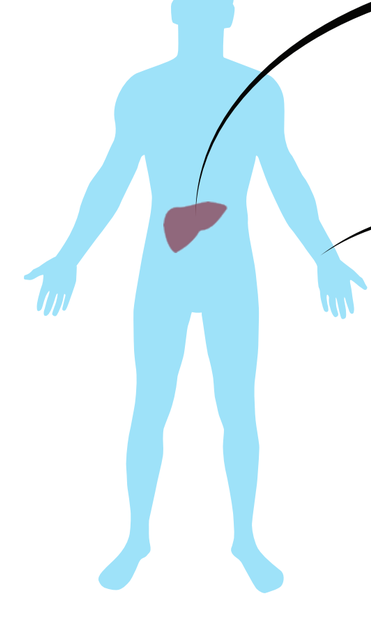

Tissue-Based Biomarkers
- Archival Tissue (Target expression: IL-27 IHC)

Peripheral Blood Biomarkers (Baseline and C2D1)
- PBMC (Immune profiling: Cytek Flow cytometry; Transcriptomics: Bulk RNAseq)
- Serum (Proteomics: Olink and MSD)

Abbreviations: 1L = first line; atezo = atezolizumab; AE = adverse event; BCLC = Barcelona clinic liver cancer; bev = bevucizumab; casdozo = Casdozokitug; CR = complete response; CxDy = Cycle x Day y; DC = dendritic cells; DCR = disease control rate; EBI3 = Epstein-Barr virus-induced gene 3/IL27B subunit; ECOG PS = Eastern Cooperative Oncology Group Performance Status; GSEA = gene set enrichment analysis; HBV = hepatitis B virus; HCC = hepatocellular carcinoma; HCV = hepatitis C virus; IFNγ = interferon gamma; IHC = immunohistochemistry; IL = interleukin; IV = intravenous; LLOQ = lower limit of quantification; mRECIST = modified Response Evaluation Criteria in Solid Tumors; MSD = Meso Scale Discovery; NES = normalized enrichment score; NE = not evaluable; NK = natural killer; NR = non-responder; NSCLC = non-small cell lung cancer; ORR = overall response rate; p28 = IL27A subunit; OS = overall survival; PBMC = peripheral blood mononuclear cells; PD = progressive disease; PD-(L)1 = programmed death (ligand)-1; PFS = progression-free survival; PR = partial response; q3w = every 3 weeks; R = responder; RCC = renal cell carcinoma; RECIST v1.1 = Response Evaluation Criteria in Solid Tumors version 1.1; SD = stable disease; SOC = standard of care; TAM = tumor-associated macrophages, TME = tumor microenvironment; uHCC = unresectable HCC; ULOQ = upper limit of quantification; VEGF = vascular endothelial growth factor.

References: 1) Daneng Li et al. *J Clin Oncol* 43, TPS4217-TPS4217(2025). 2) Rajasekaran et al. Cytokines 2025 Poster PB-020. 3) Chihara et al. *Nature* 558, 2018. 4) DeLong et al. *Immunohorizons* 3, 2019. 5) Aghayev et al. *Cancer Discov*, 12, 2022. 6) Finn RS et al. *N Engl J Med*. 2020;382:1894-1905. 7) Abou-Alfa GK et al. *NEJM Evid*. 2022;1(8):EVIDoa2100070.

Acknowledgments: The authors would like to thank the participants and their families, caregivers, and investigators involved in this study. Medical writing, editorial assistance, and poster production support were provided by Acumen Medical Communications, funded by Coherus Oncology, Inc.

Statistics: * = p < 0.05; ** = p < 0.01; *** = p < 0.001; **** = p < 0.0001; ns = not significant.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster. Contact Varun Kapoor (vkapoor@coherus.com) with any questions or comments.

Pharmacodynamic Activity of Casdozokitug/Atezolizumab/Bevacizumab

Casdozokitug in Combination with Atezolizumab/Bevacizumab Promotes Robust Peripheral Immune Activation in HCC (Serum Protein)

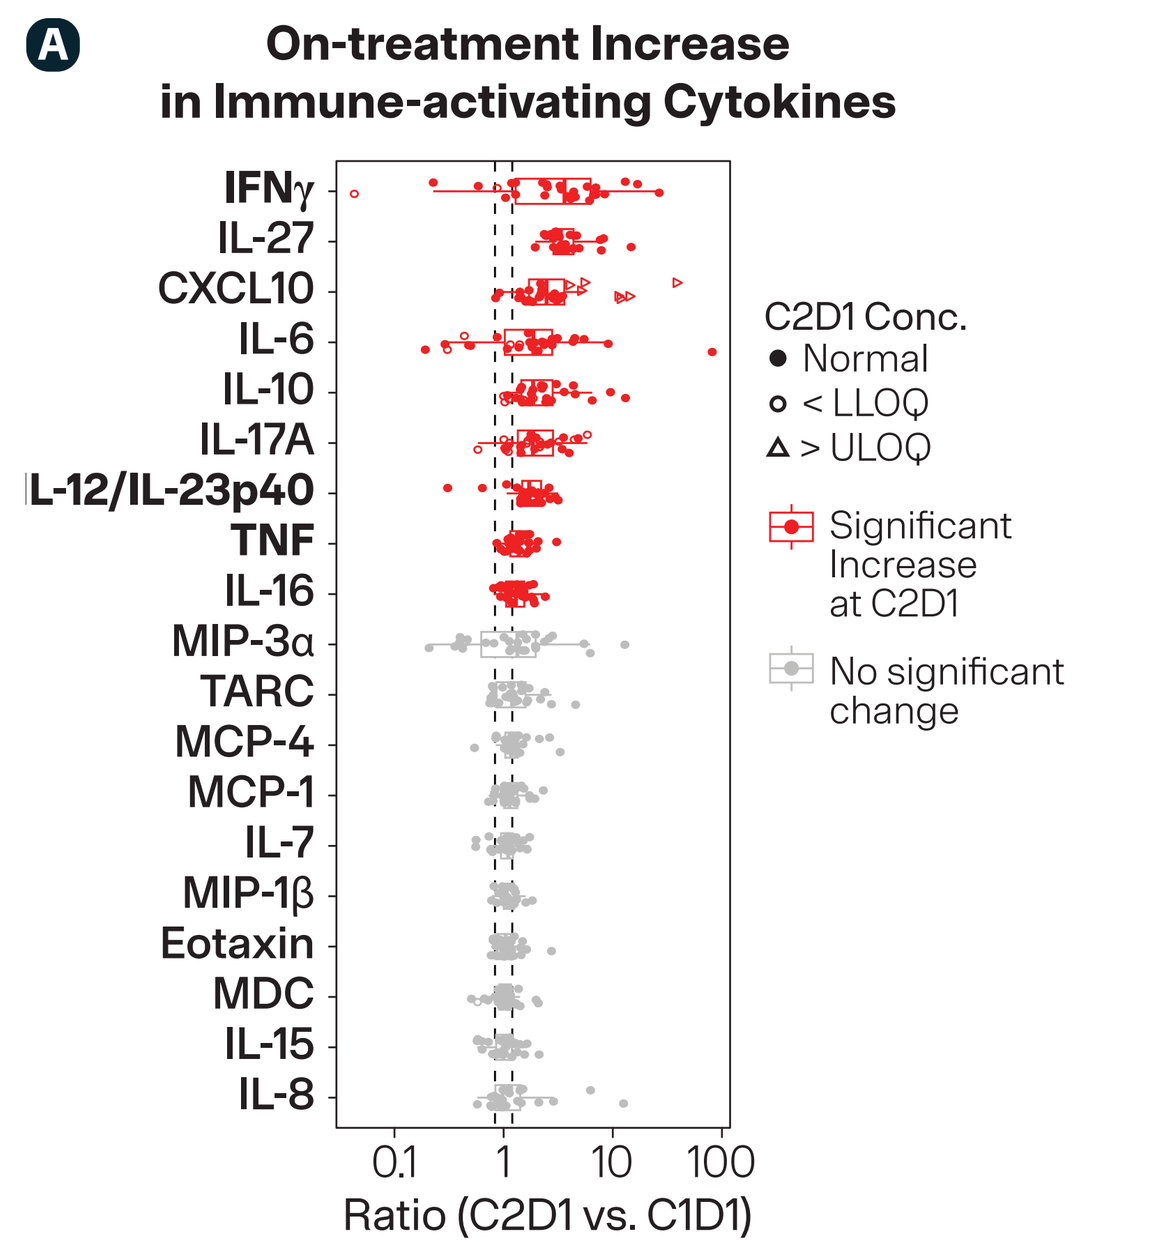
A
On-treatment Increase in Immune-activating Cytokines
IFNγ
IL-27
CXCL10
IL-6
IL-10
IL-17A
IL-12/IL-23p40
TNF
IL-16
MIP-3α
TARC
MCP-4
MCP-1
IL-7
MIP-1β
Eotaxin
MDC
IL-15
IL-8
Ratio (C2D1 vs. C1D1)
C2D1 Conc.
Normal
< LLOQ
> ULOQ
Significant Increase at C2D1
No significant change

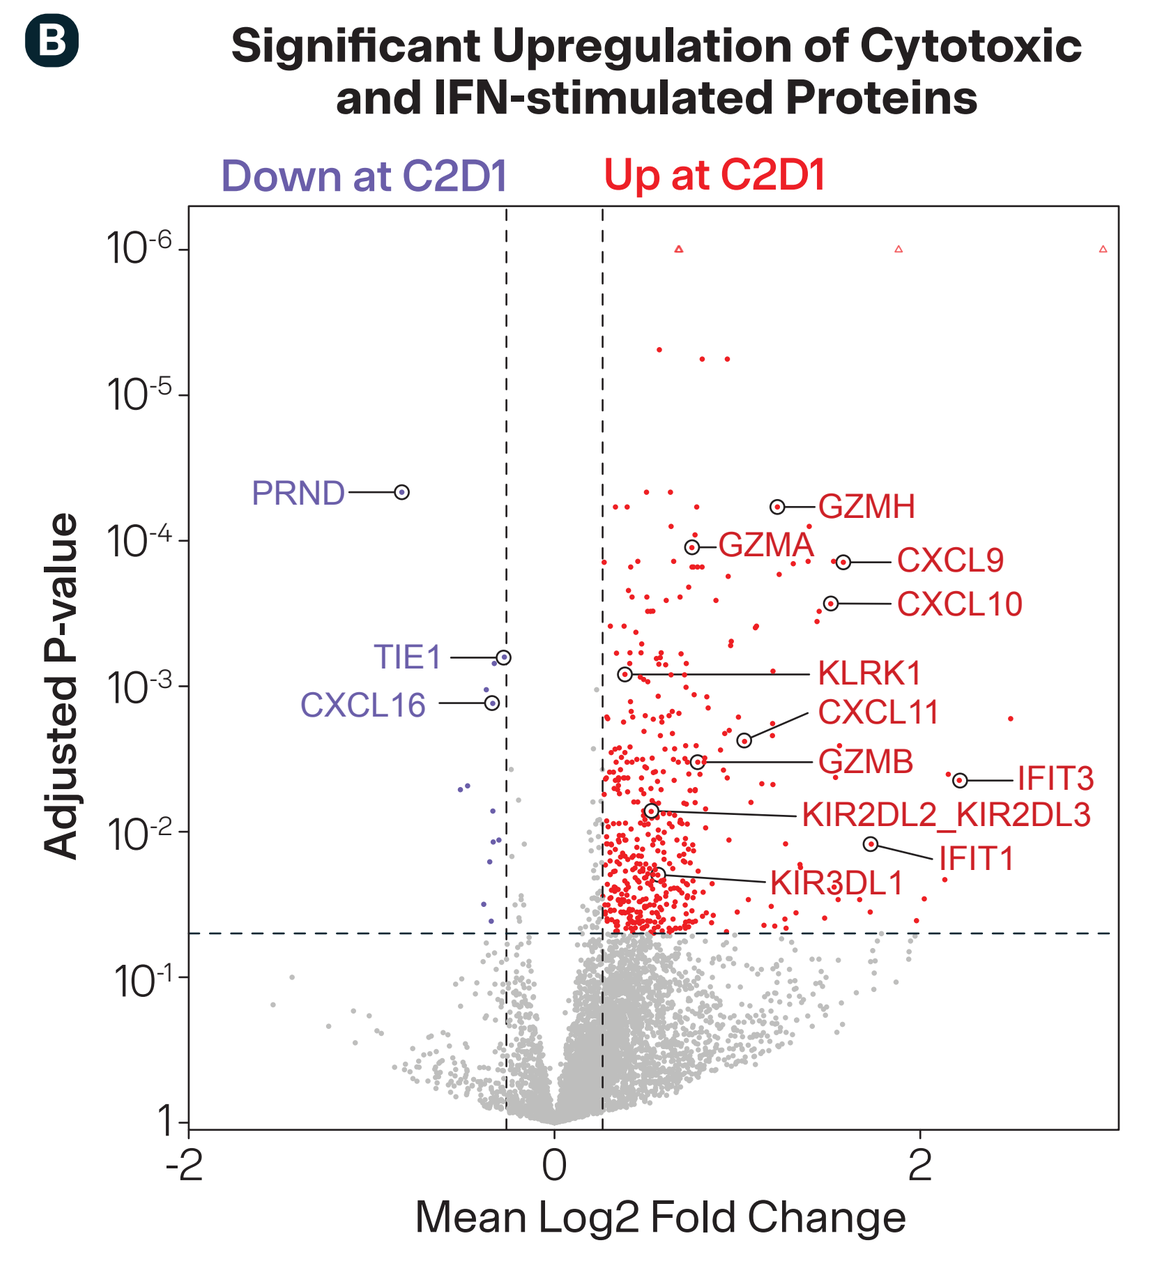
B
Significant Upregulation of Cytotoxic and IFN-stimulated Proteins
Down at C2D1
Up at C2D1
Adjusted P-value
PRND
TIE1
CXCL16
GZMH
GZMA
CXCL9
CXCL10
KLRK1
CXCL11
GZMB
IFIT3
KIR2DL2_KIR2DL3
IFIT1
KIR3DL1
Mean Log2 Fold Change

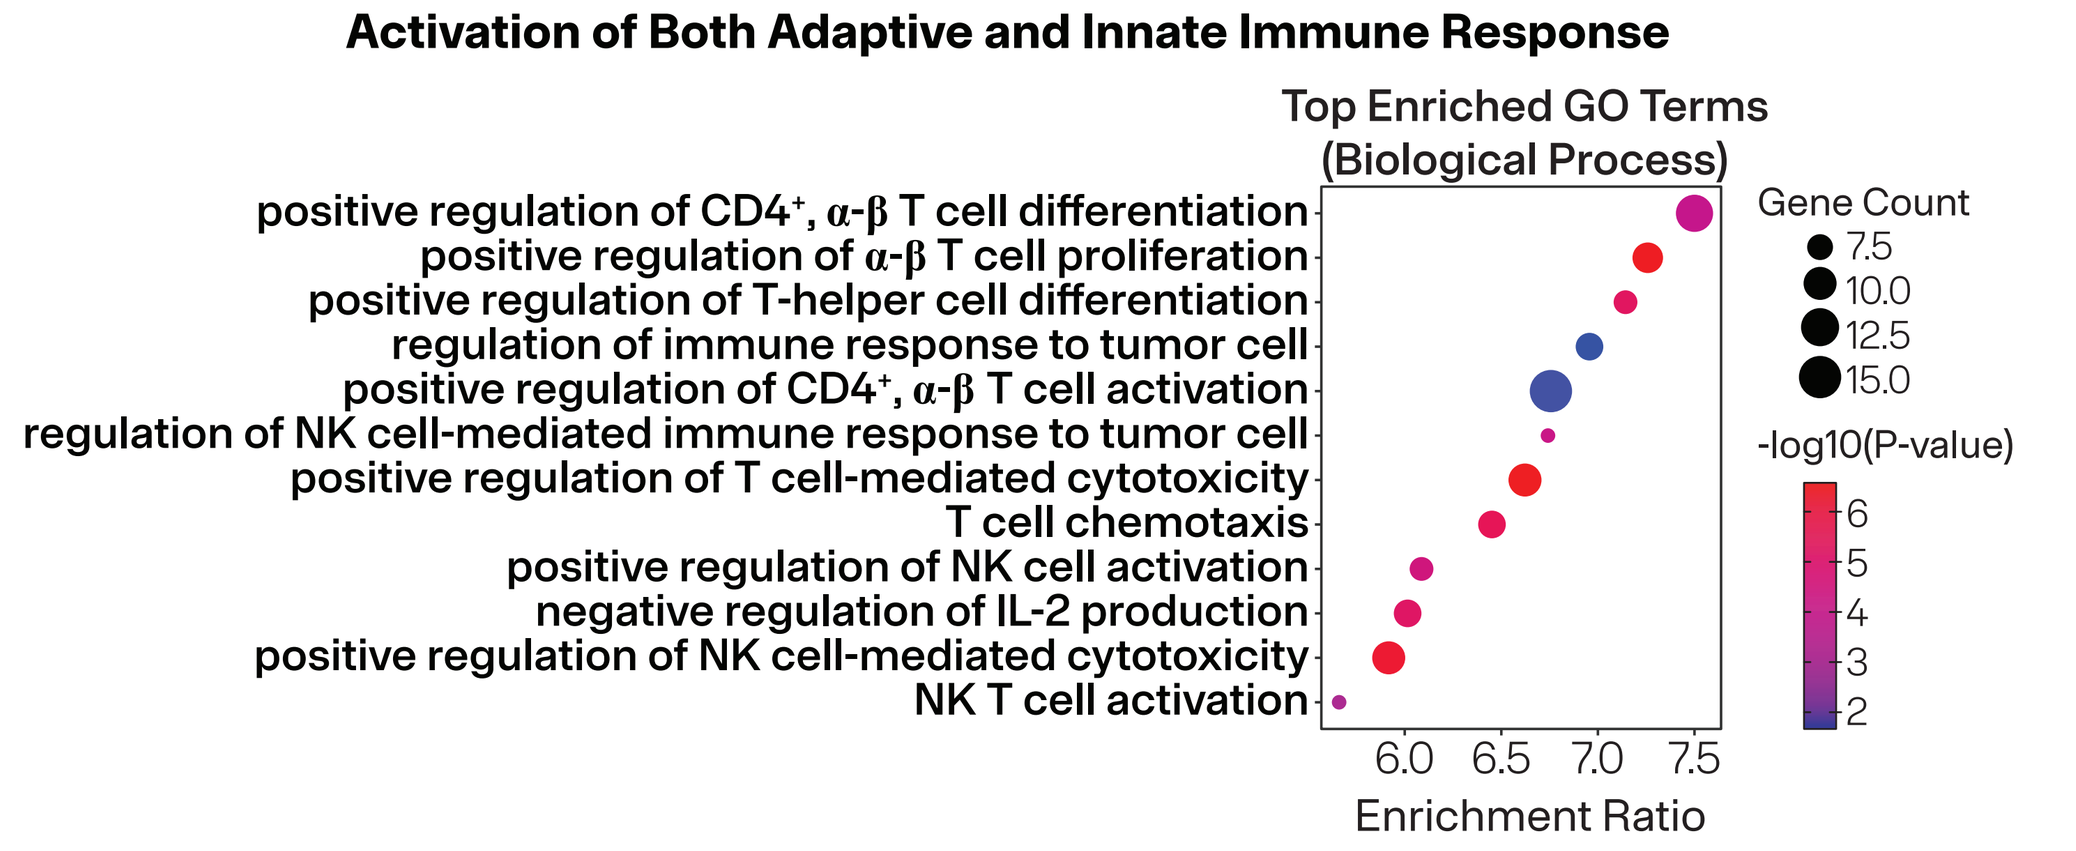
C
Activation of Both Adaptive and Innate Immune Response
Top Enriched GO Terms (Biological Process)
positive regulation of CD4+, α-β T cell differentiation
positive regulation of α-β T cell proliferation
positive regulation of T-helper cell differentiation
regulation of immune response to tumor cell
positive regulation of CD4+, α-β T cell activation
regulation of NK cell-mediated immune response to tumor cell
positive regulation of T cell-mediated cytotoxicity
T cell chemotaxis
positive regulation of NK cell activation
negative regulation of IL-2 production
positive regulation of NK cell-mediated cytotoxicity
NK T cell activation
Gene Count
-log10(P-value)
Enrichment Ratio

Proteomic analysis of paired C1D1 and C2D1 serum samples from HCC participants with casdozokitug/atezolizumab/bevacizumab treatment (n = 28) by MSD assay (A) and Olink Explore HT platform (B, C). On-treatment (C2D1) samples show **(A)** Significant increase of immune-activating cytokines including TNF, IL-12, and IFNγ. **(B)** Upregulated interferon-stimulated proteins (IFIT1, IFIT3), chemokines (CXCL9, CXCL10, CXCL11), and downregulated angiogenic factors (PRND, TIE1, CXCL16). **(C)** Significant enrichment of T cell proliferation/activation and NK cell activation pathways, identified by topGO on significantly upregulated proteins. Significant proteins, shown in red and blue in (A, B), were defined with p < 0.05 and median fold-change > 1.2. Statistical test: paired Wilcoxon test (A); paired t-test (B); horizontal and vertical dotted lines indicate p-value = 0.05 and fold change = 1.2, respectively.

Casdozokitug in Combination with Atezolizumab/Bevacizumab Promotes NK and T Cell Activation (PBMC RNAseq)

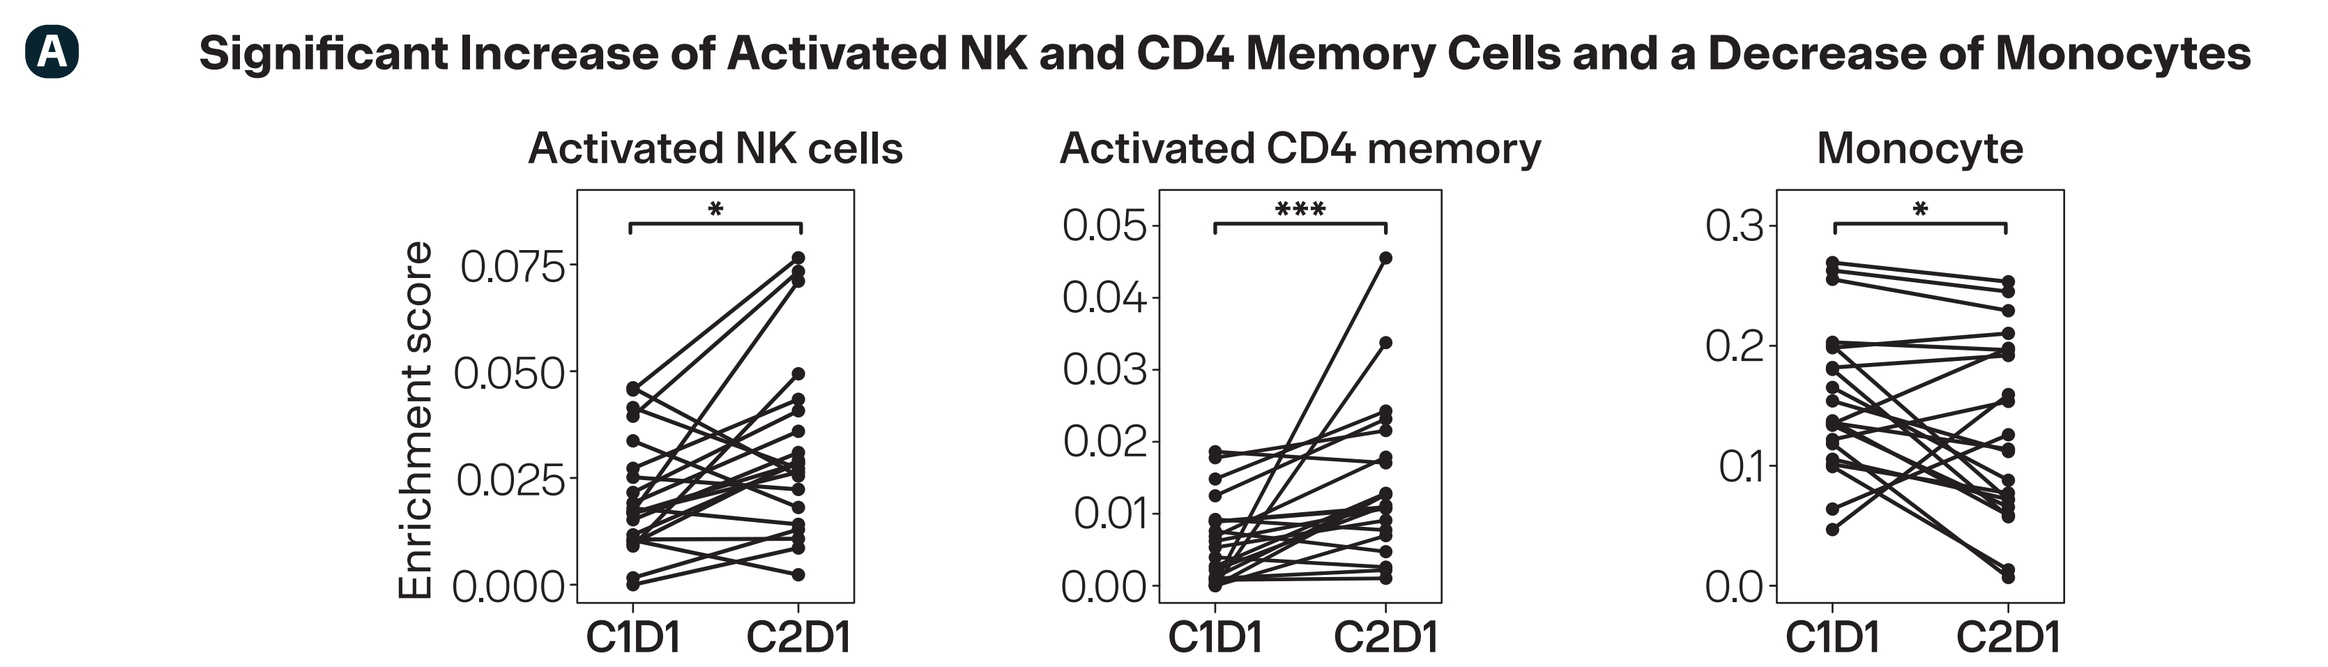
A
Significant Increase of Activated NK and CD4 Memory Cells and a Decrease of Monocytes
Activated NK cells
Activated CD4 memory
Monocyte
Enrichment score
C1D1
C2D1

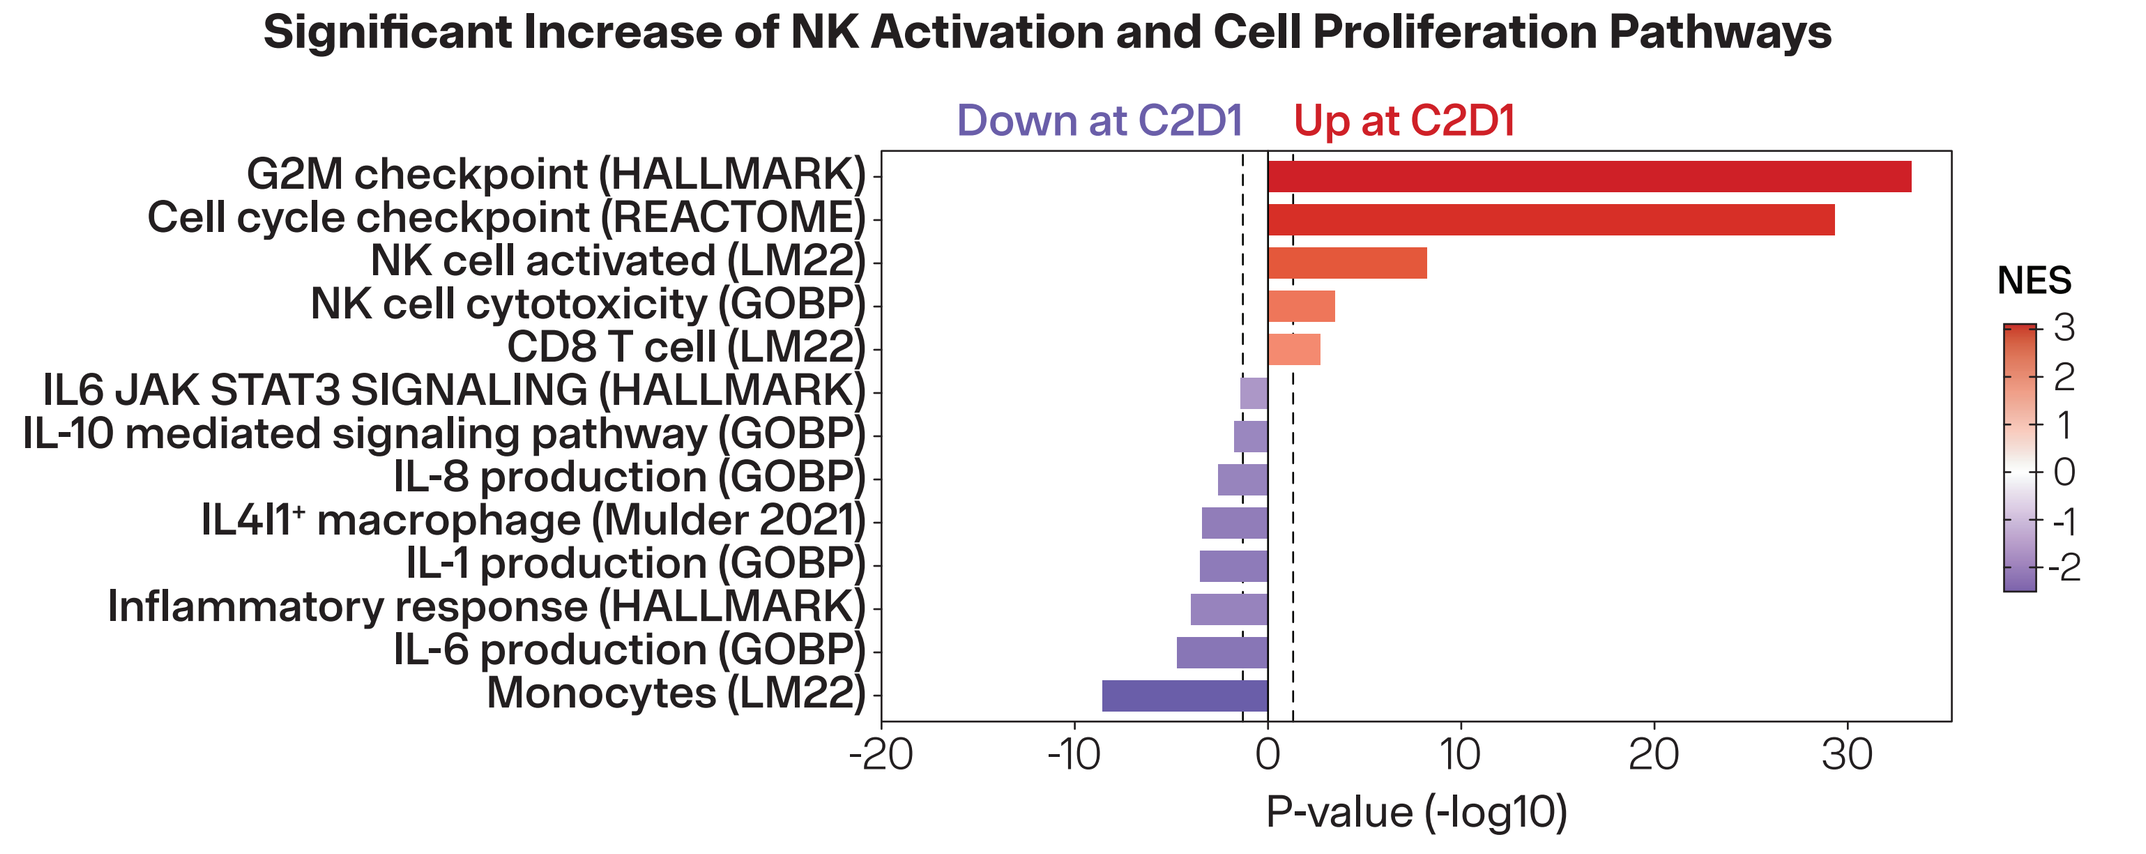
B
Significant Increase of NK Activation and Cell Proliferation Pathways
Down at C2D1
Up at C2D1
G2M checkpoint (HALLMARK)
Cell cycle checkpoint (REACTOME)
NK cell activated (LM22)
NK cell cytotoxicity (GOBP)
CD8 T cell (LM22)
IL6 JAK STAT3 SIGNALING (HALLMARK)
IL-10 mediated signaling pathway (GOBP)
IL-8 production (GOBP)
IL4I1+ macrophage (Mulder 2021)
IL-1 production (GOBP)
Inflammatory response (HALLMARK)
IL-6 production (GOBP)
Monocytes (LM22)
NES
P-value (-log10)

Bulk RNAseq of paired PBMC samples at C1D1 and C2D1 from HCC participants with casdozokitug/atezolizumab/bevacizumab treatment (n = 21). (A) Cell-type enrichment analysis (xCell, LM22) identified significantly altered cell populations at C2D1. Statistical test: paired Wilcoxon test. **(B)** Gene set enrichment analysis (GSEA) comparing C2D1 vs C1D1 (n = 21) shows significant upregulation of NK / T activation and cell proliferation, and downregulation of inflammatory and myeloid-associated gene sets. Vertical dotted lines indicate a p-value = 0.05.

Biomarkers Associated with Clinical Response

Early On-treatment Increase in IFNγ Is Associated with Clinical Benefit

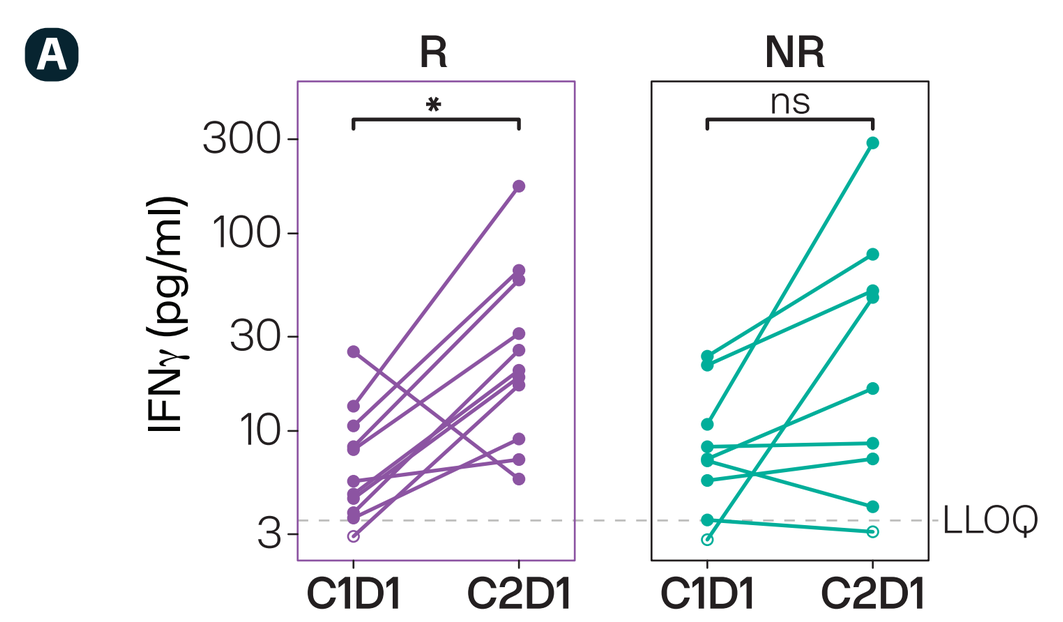
A
R
NR
IFNγ (pg/ml)
LLOQ
C1D1
C2D1

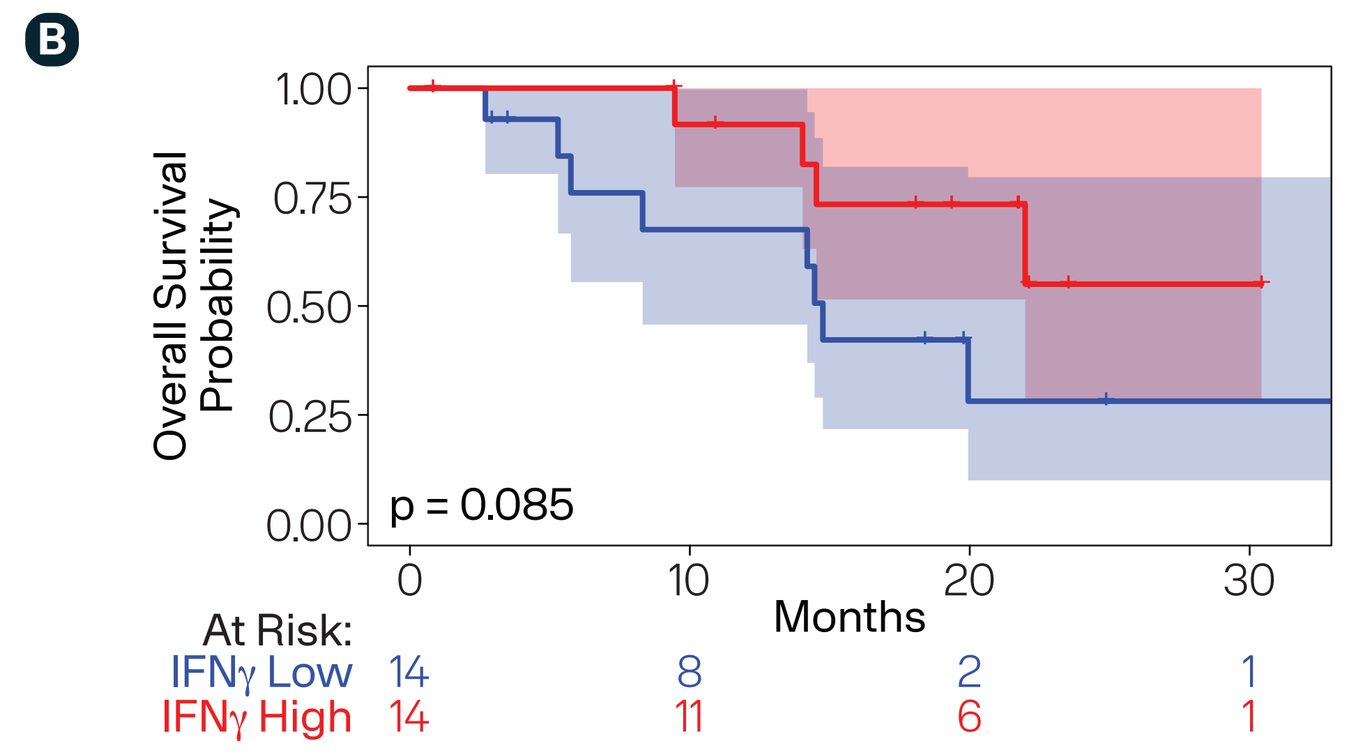
B
Overall Survival Probability
p = 0.085
Months
At Risk:
IFNγ Low 14 8 2 1
IFNγ High 14 11 6 1

Distinct On-treatment Proteomic Signatures Reveal Potential Response and Resistance Mechanisms

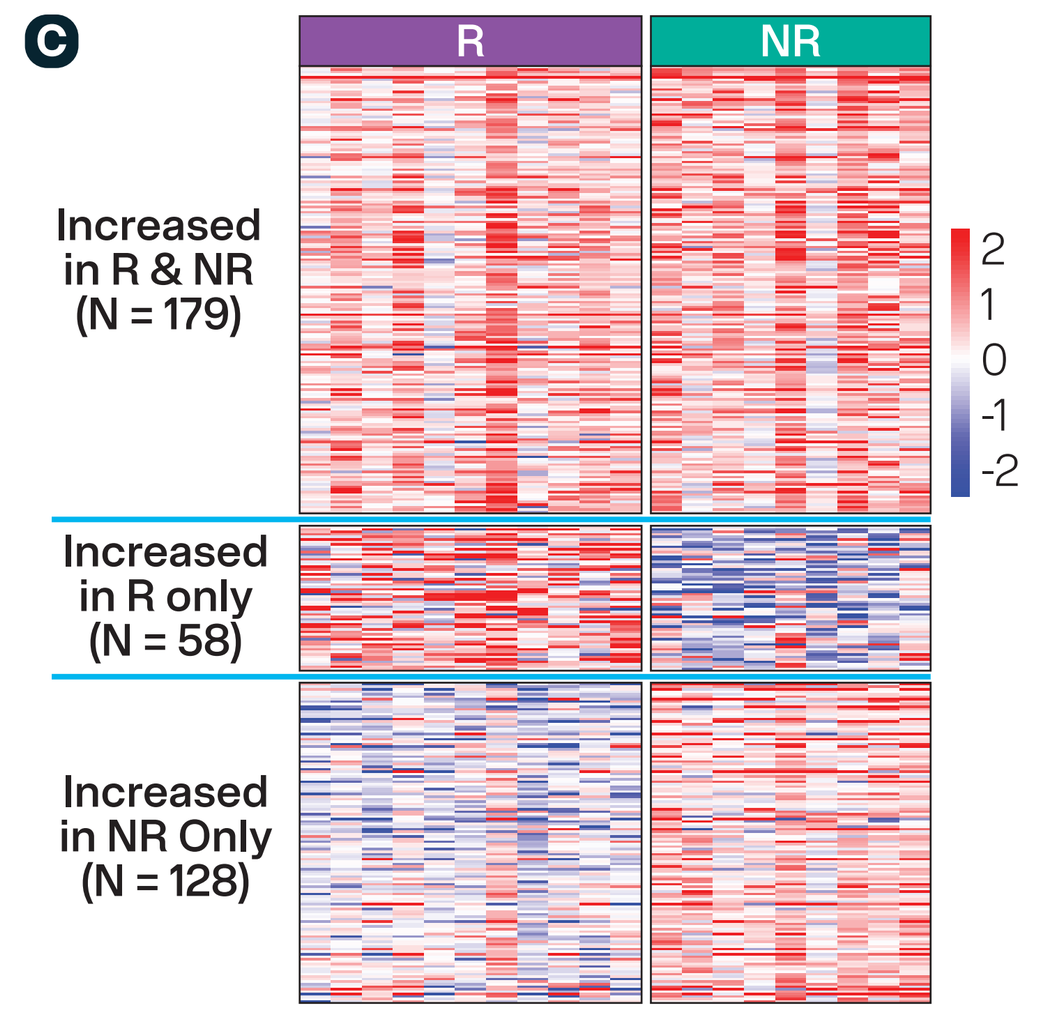
C
R
NR
Increased in R & NR (N = 179)
Increased in R only (N = 58)
Increased in NR Only (N = 128)

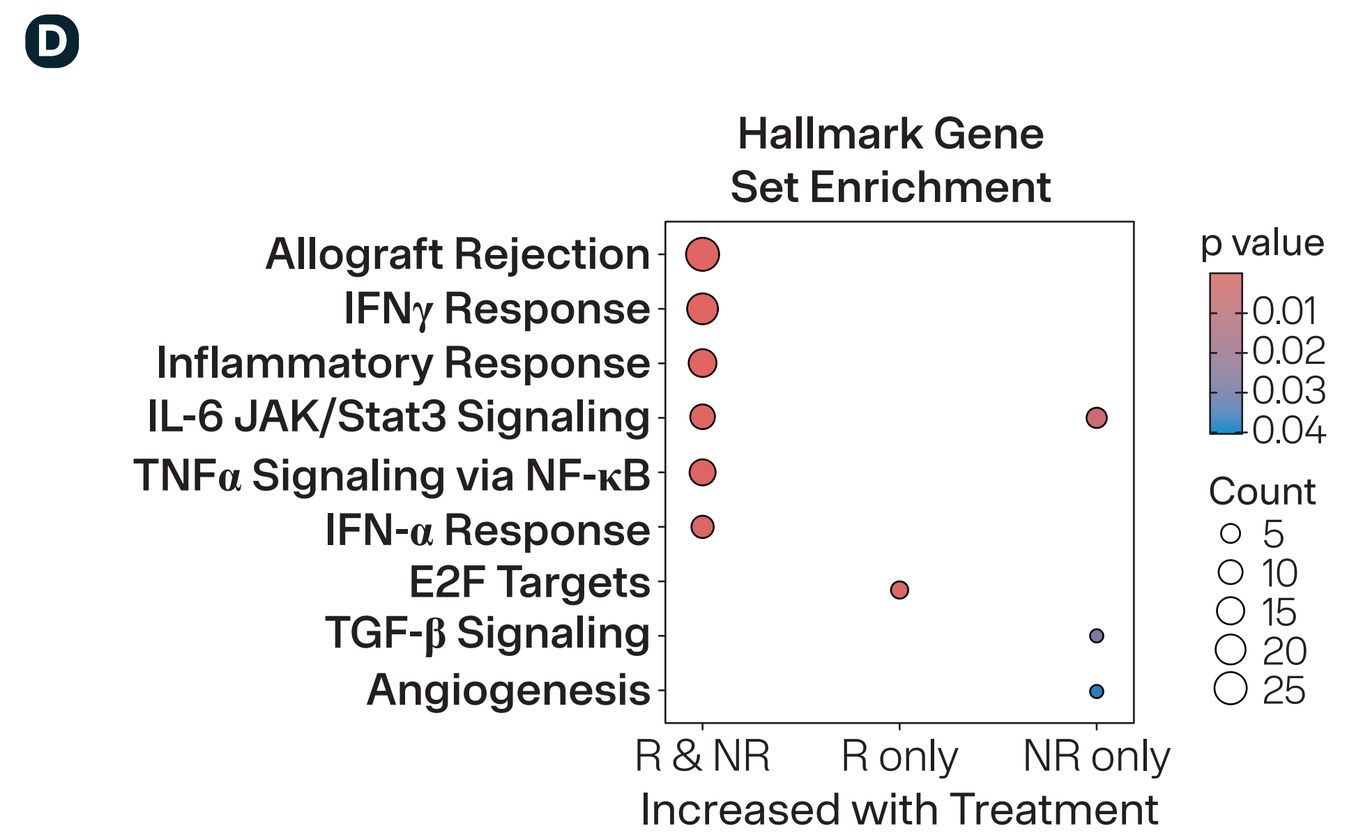
D
Hallmark Gene Set Enrichment
Allograft Rejection
IFNγ Response
Inflammatory Response
IL-6 JAK/Stat3 Signaling
TNFα Signaling via NF-κB
IFN-α Response
E2F Targets
TGF-β Signaling
Angiogenesis
R & NR
R only
NR only
Increased with Treatment
p value
Count

Response-associated on-treatment changes in circulating protein biomarkers. (A) Significant increase in circulating IFNγ at C2D1 compared to baseline in responders (CR/PR, n = 11) but not in non-responders (PD, N = 9). **(B)** On-treatment increase of IFNγ trended to be associated with longer Overall Survival (OS). Kaplan-Meier survival curves compare OS between patients stratified by the median on-treatment change of IFNγ. The p-value was calculated using the Log-Rank Test (N = 28) **(C)** Heatmap of proteins with distinct patterns of on-treatment change in responders and non-responders. **(D)** HALLMARK gene sets enriched in sets of proteins identified in (C) using hypergeometric test. Responder-specific proteins were enriched in HALLMARK_E2F_TARGETS (proliferation). Non-responder-specific proteins were enriched in IL-6/JAK/STAT3, TGF-β signaling, and angiogenesis, consistent with HCC resistance.

Stronger Peripheral T and NK Cell Activation Signatures in Responders (PBMC RNAseq)

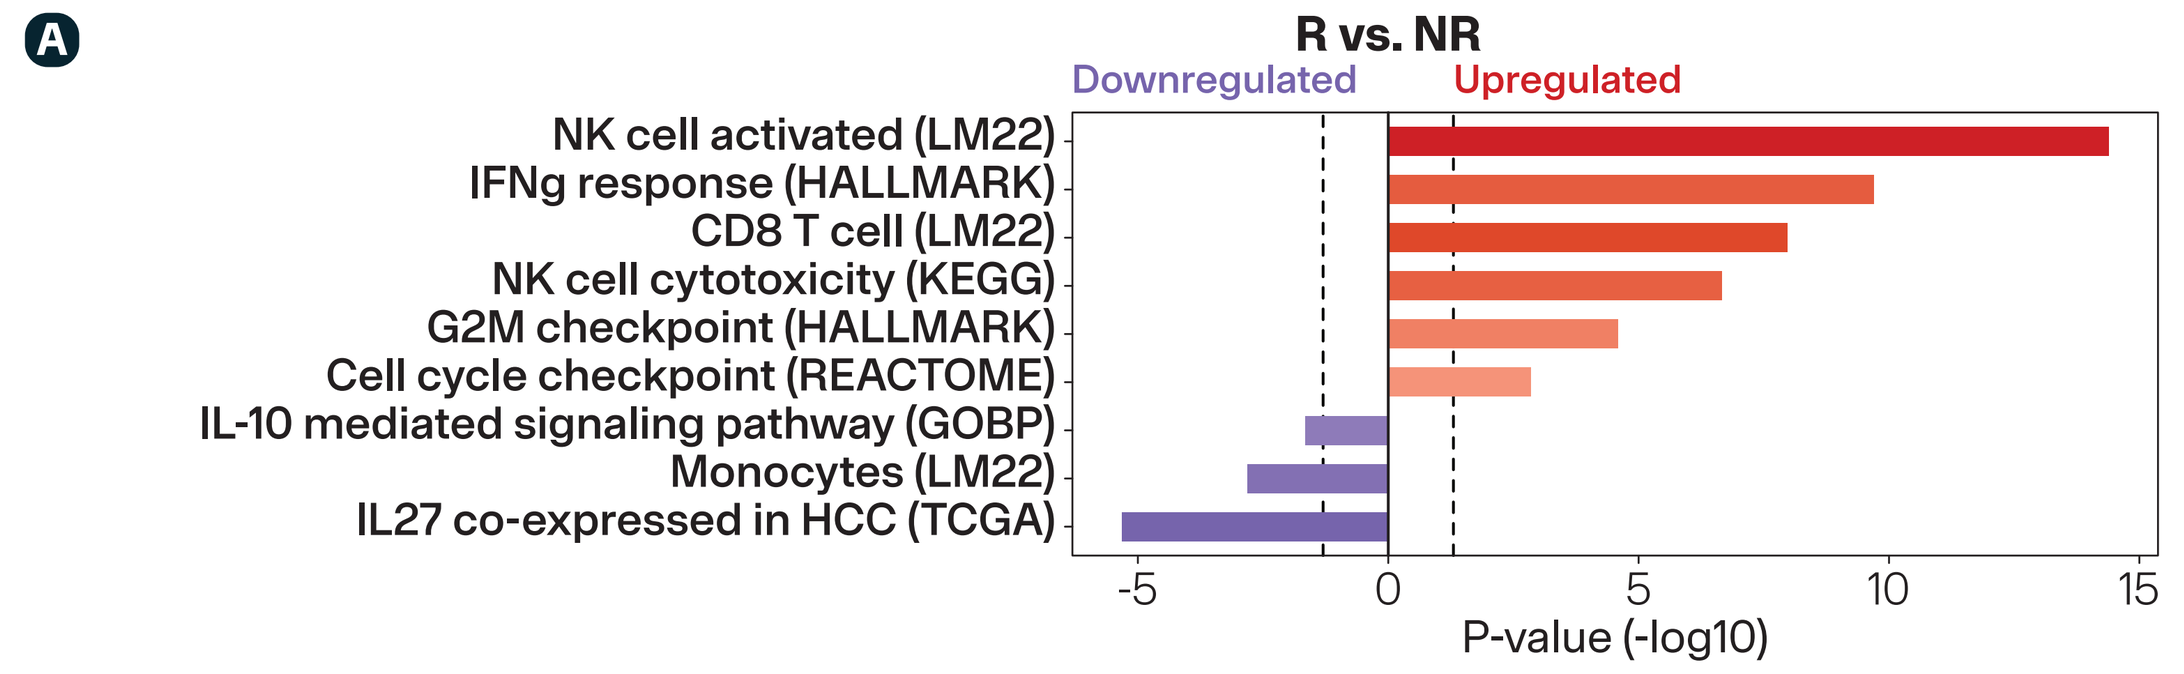
A
R vs. NR
Downregulated
Upregulated
NK cell activated (LM22)
IFNg response (HALLMARK)
CD8 T cell (LM22)
NK cell cytotoxicity (KEGG)
G2M checkpoint (HALLMARK)
Cell cycle checkpoint (REACTOME)
IL-10 mediated signaling pathway (GOBP)
Monocytes (LM22)
IL27 co-expressed in HCC (TCGA)
NES
P-value (-log10)

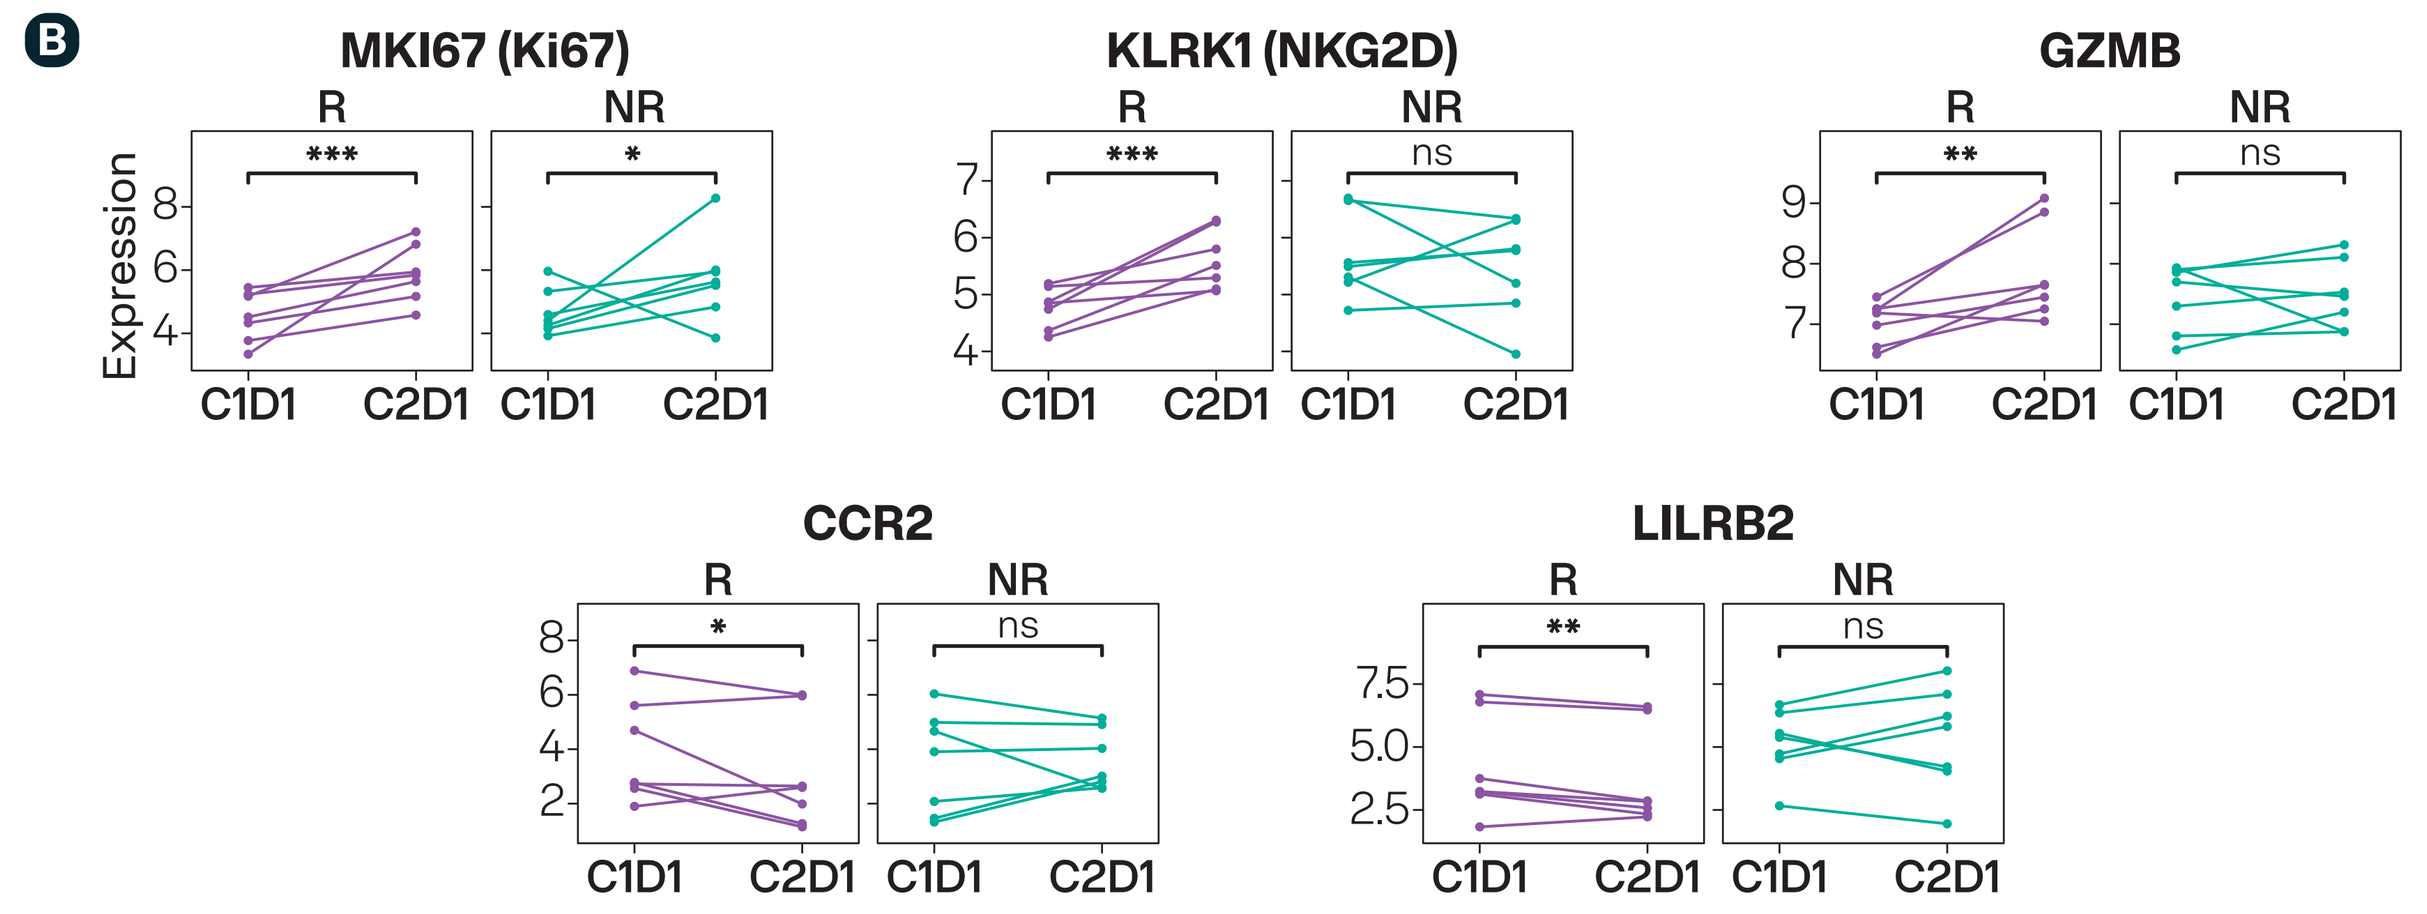
B
MKI67 (Ki67)
KLRK1 (NKG2D)
GZMB
CCR2
LILRB2
R
NR
Expression
C1D1
C2D1

On-treatment transcriptomic changes (C2D1 compared to pre-treatment, C1D1) correlate with clinical response. (A) Pre-ranked GSEA on R vs NR (n = 7 per group) in C2D1-C1D1 differences in PBMCs. Responders (R, defined as PR or CR), compared to Non-responders (NR, defined as PD), show a clear enrichment of pro-inflammatory/anti-tumorigenic pathways (including NK cell activation, IFNγ response, CD8 T-cell cytotoxicity, and cell cycle) and suppression of signatures associated with monocyte and IL-10, as well as IL-27 co-expression genes (HCC-TCGA). Vertical dotted lines indicate a p-value = 0.05. **(B)** Responders uniquely exhibit on-treatment increases in genes associated with proliferation (MKI67), cytotoxicity (KLRK1, GZMB), and on-treatment decrease in genes regulating monocyte recruitment and macrophage differentiation (CCR2, LILRB2). Statistical test: Limma

Preliminary Association of Higher Level of IL-27+ Associated Tumor Macrophages in Archival Tissue Samples with Clinical Response (CR/PR), Small N

A

Complete Responder (RECIST v1.1 and mRECIST) with HBV+HCC with scattered IL-27+ macrophages with Kupffer cell-like pattern

Complete Responder (mRECIST) with non-viral HCC with clusters of IL-27+ TAMs

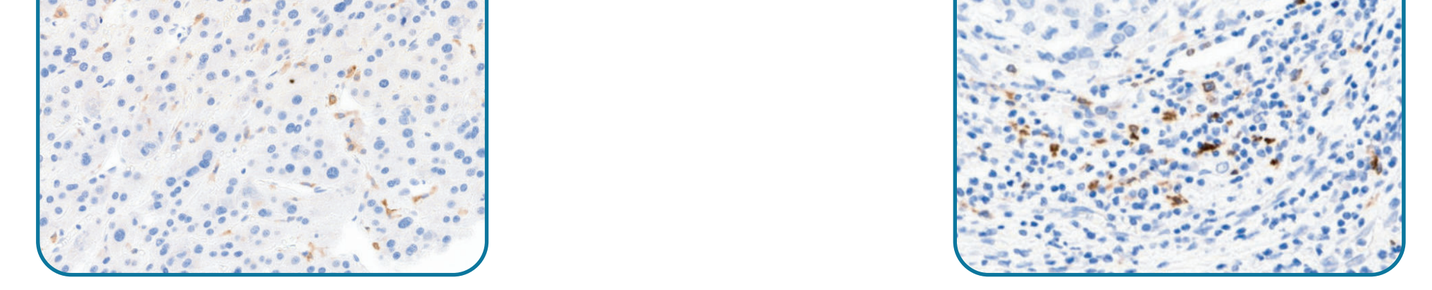

B

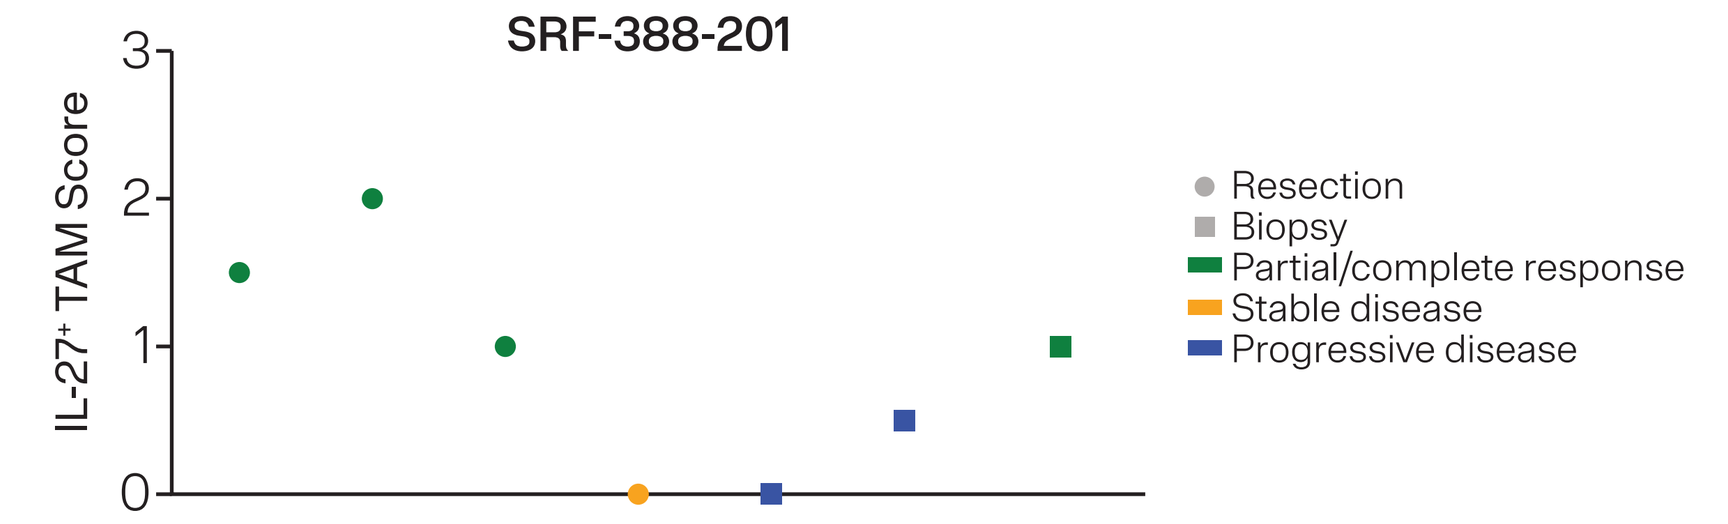
SRF-388-201
IL-27+ TAM Score
Resection
Biopsy
Partial/complete response
Stable disease
Progressive disease

Patient	#1	#2	#3	#4	#5	#6	#7
Site	Liver	Liver	Liver	Liver	Liver	Liver	LN Met
Viral status	Neg	HBV	HBV	HCV	Neg	Neg	HBV
IL-27 score	1.5	2	1	0	0	0.5	1

(A) Immunohistochemical staining for IL-27 was performed on archival resection and biopsy samples using an independently validated assay (B) Samples were scored blindly based on semi-quantitative scoring system (0-3+) evaluating the abundance of IL-27+ tumor-associated macrophages (TAM)

NK Cell Activation in HCC Following Casdozokitug Single Agent or Combination Treatment

On-treatment Response-associated Biomarkers in Casdozokitug/Atezolizumab/Bevacizumab Study Were Consistent with Casdozokitug Monotherapy

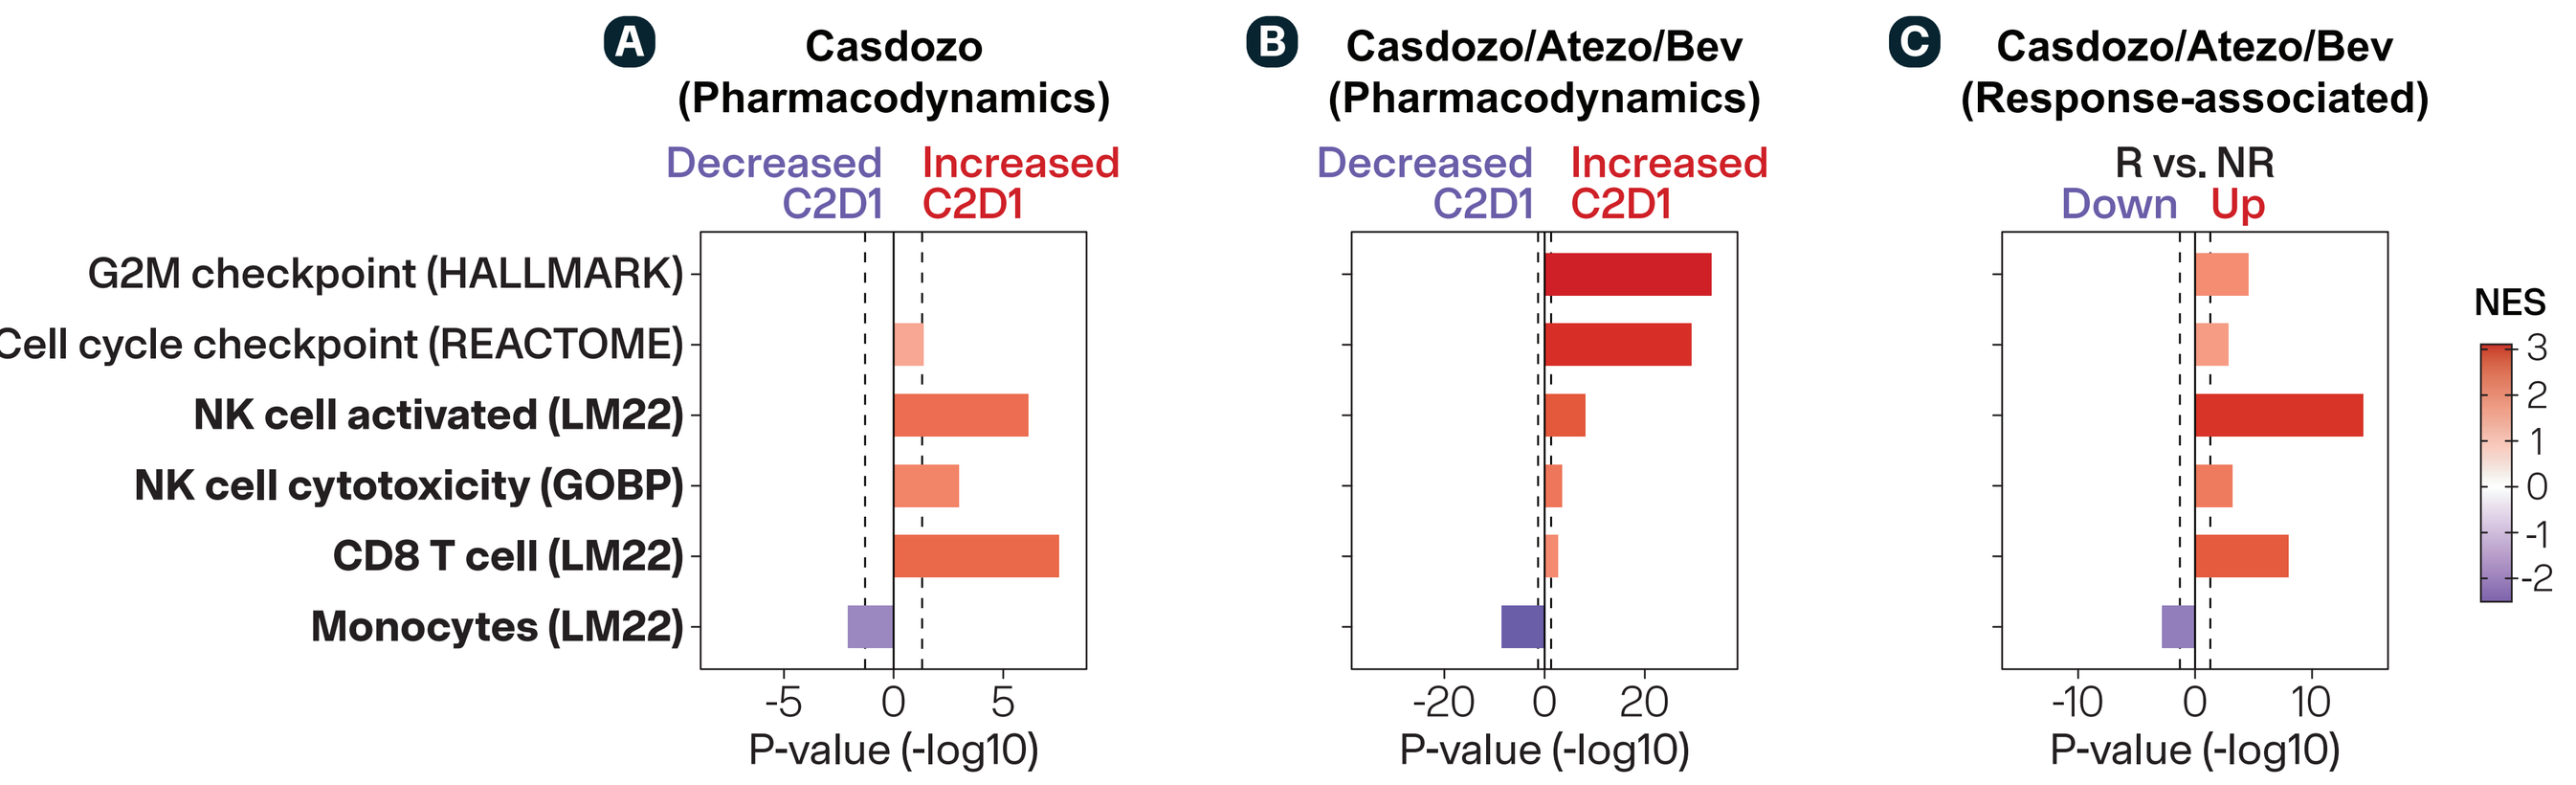
A
Casdozo (Pharmacodynamics)
B
Casdozo/Atezo/Bev (Pharmacodynamics)
C
Casdozo/Atezo/Bev (Response-associated)
Decreased C2D1
Increased C2D1
R vs. NR
Down
Up
G2M checkpoint (HALLMARK)
Cell cycle checkpoint (REACTOME)
NK cell activated (LM22)
NK cell cytotoxicity (GOBP)
CD8 T cell (LM22)
Monocytes (LM22)
NES
P-value (-log10)

On-treatment transcriptomic changes in PBMC from HCC participants: casdozokitug monotherapy (SRF-388-101) vs combination treatment and response stratification (SRF-388-201). (A) GSEA for casdozokitug monotherapy shows increased NK and T cell activity and decreased monocyte signatures. **(B)** GSEA for casdozokitug/atezolizumab/bevacizumab treatment shows similar, and in some cases more pronounced, increases in cell cycle and immune activation, with reduced monocyte signatures (C2D1 vs C1D1). **(C)** Comparison of on-treatment changes (C2D1 vs C1D1) between R and NR to casdozokitug/atezolizumab/bevacizumab treatment. Responders demonstrate a pronounced enrichment of immune activation (NK cell, CD8 T cell) and cell cycle pathways, suggesting a proliferative and active immune response associated with clinical benefit. (A-C) Vertical dotted lines indicate a p-value = 0.05.

Conclusions

- Triplet blockade of IL-27, PD-(L)1, and VEGF pathways with casdozokitug/atezolizumab/bevacizumab shows manageable safety profile and adverse events were consistent with the known profiles of atezolizumab and bevacizumab, with no new safety signals identified.
- Casdozokitug in combination with atezolizumab/bevacizumab demonstrated a CR rate of 17.2%, which is higher than the 3-6% CR rate reported in prior Phase 3 studies in HCC (IMbrave150 and HIMALAYA).[6,7]
- Pharmacodynamic biomarkers demonstrate immune activation in all peripheral biomarker assays.
- Response-associated on-treatment biomarkers in casdozokitug/atezolizumab/bevacizumab study were consistent with casdozokitug single agent treatment. Enhanced immune activation, particularly NK-cell activation, correlated with clinical response, supporting a casdozokitug-driven effect.
- IL-27 expression level may be associated with casdozokitug clinical activity and will be further explored in future clinical studies.[1,5]
- The randomized Phase 2 study CHS-388-202 (NCT06679985) is evaluating casdozokitug plus toripalimab (anti-PD-1)/bevacizumab vs toripalimab/bevacizumab in 1L uHCC to assess treatment benefit and response biomarkers.