Depletion of CCR8⁺ Tumor Treg cells with SRF114 or Anti-CCR8 Therapy Promotes Robust Antitumor Activity and Reshapes the Tumor Microenvironment Toward a More Pro-inflammatory Milieu

Marisella Panduro, Yue Ren, Ricard Masia, Yu Yang, Andrew C. Lake, Vito J. Palombella, Jonathan A. Hill, James F. Mohan

Surface Oncology, Cambridge, MA, USA

SURFACE ONCOLOGY®

#5125

Background

- Foxp3⁺ regulatory T (Treg) cells play a crucial role in orchestrating immune responses across several tissues, including the tumor microenvironment (TME).[1,2]
- The dominant immune regulatory activity of Treg cells is evidenced, both clinically and preclinically, by the profound inflammatory dysregulation that arises in their absence.[1,2]
- CCR8 is a seven transmembrane G-coupled chemokine receptor protein. Its expression is predominantly upregulated on Treg cells in several types of tumors compared with peripheral blood and other lymphoid tissues.[3,4]
- CCR8⁺ tumor Tregs have been shown to have an activated phenotype and are highly suppressive *in vitro*.[3,4]
- Therapeutic antibodies targeting CCR8 have been developed to deplete tumor Treg cells. This represents an attractive strategy to reshape the local TME and enhance anti-tumor immune responses.[7-10]
- SRF114 is an afucosylated fully human IgG1 antibody that selectively targets human CCR8 (hCCR8) and induces depletion of CCR8⁺ Treg cells. SRF114 is currently in Phase 1 Clinical Studies (NCT05635643).

CCR8⁺ Tumor Tregs Are Highly Activated Cells with Potent Immune Suppressive Function

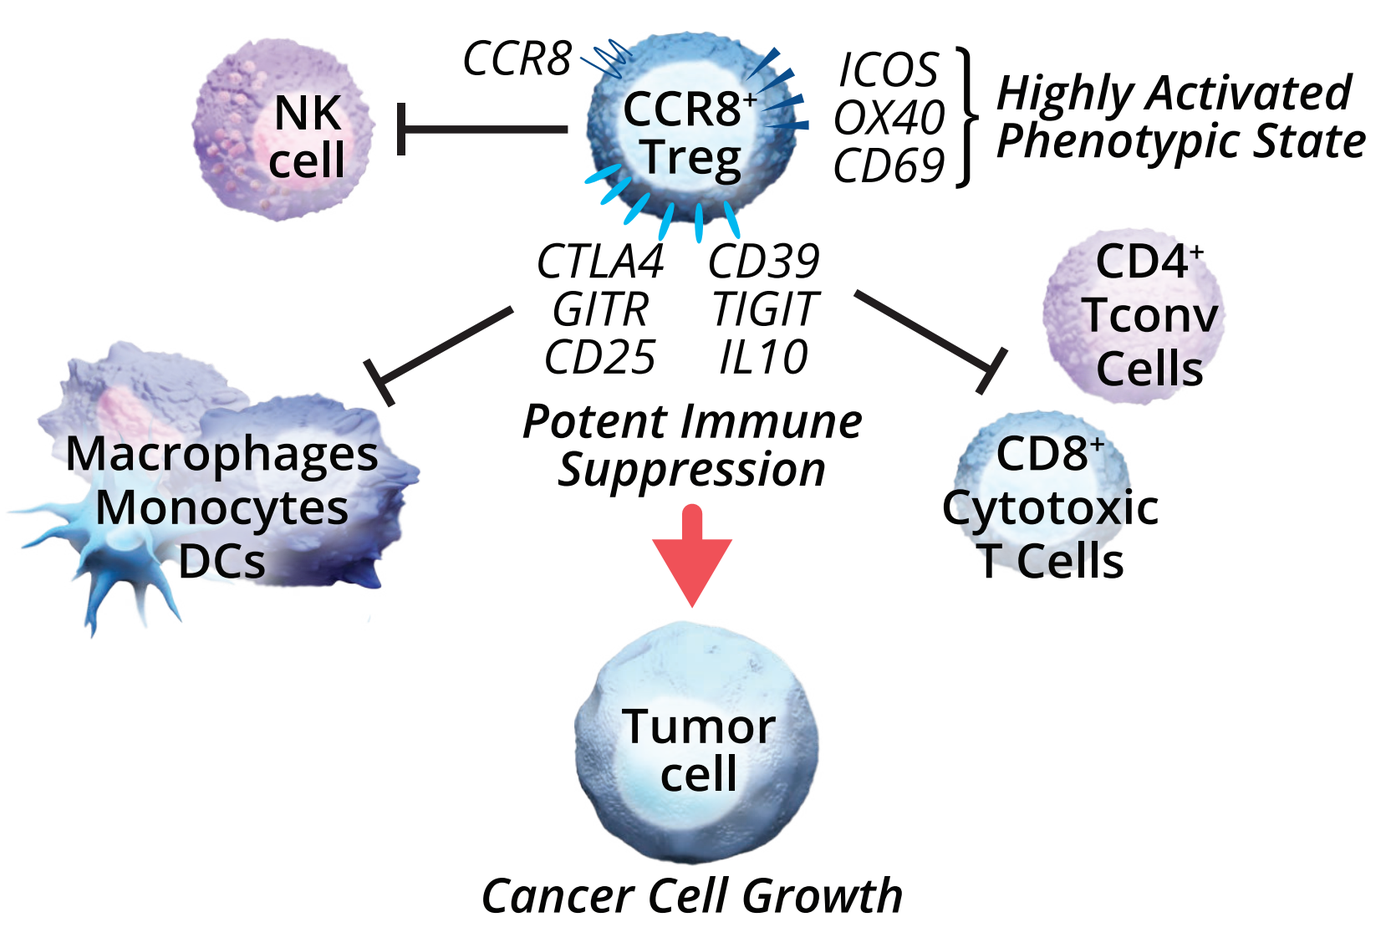

CCR8
NK cell
CCR8⁺ Treg
ICOS
OX40
CD69
Highly Activated Phenotypic State
CTLA4 CD39
GITR TIGIT
CD25 IL10
Potent Immune Suppression
CD4⁺ Tconv Cells
CD8⁺ Cytotoxic T Cells
Macrophages Monocytes DCs
Tumor cell
Cancer Cell Growth

Anti-CCR8 Therapy Results in Robust Anti-Tumor Activity and Depletion of Tumor Tregs, While Preserving Peripheral Treg Cells

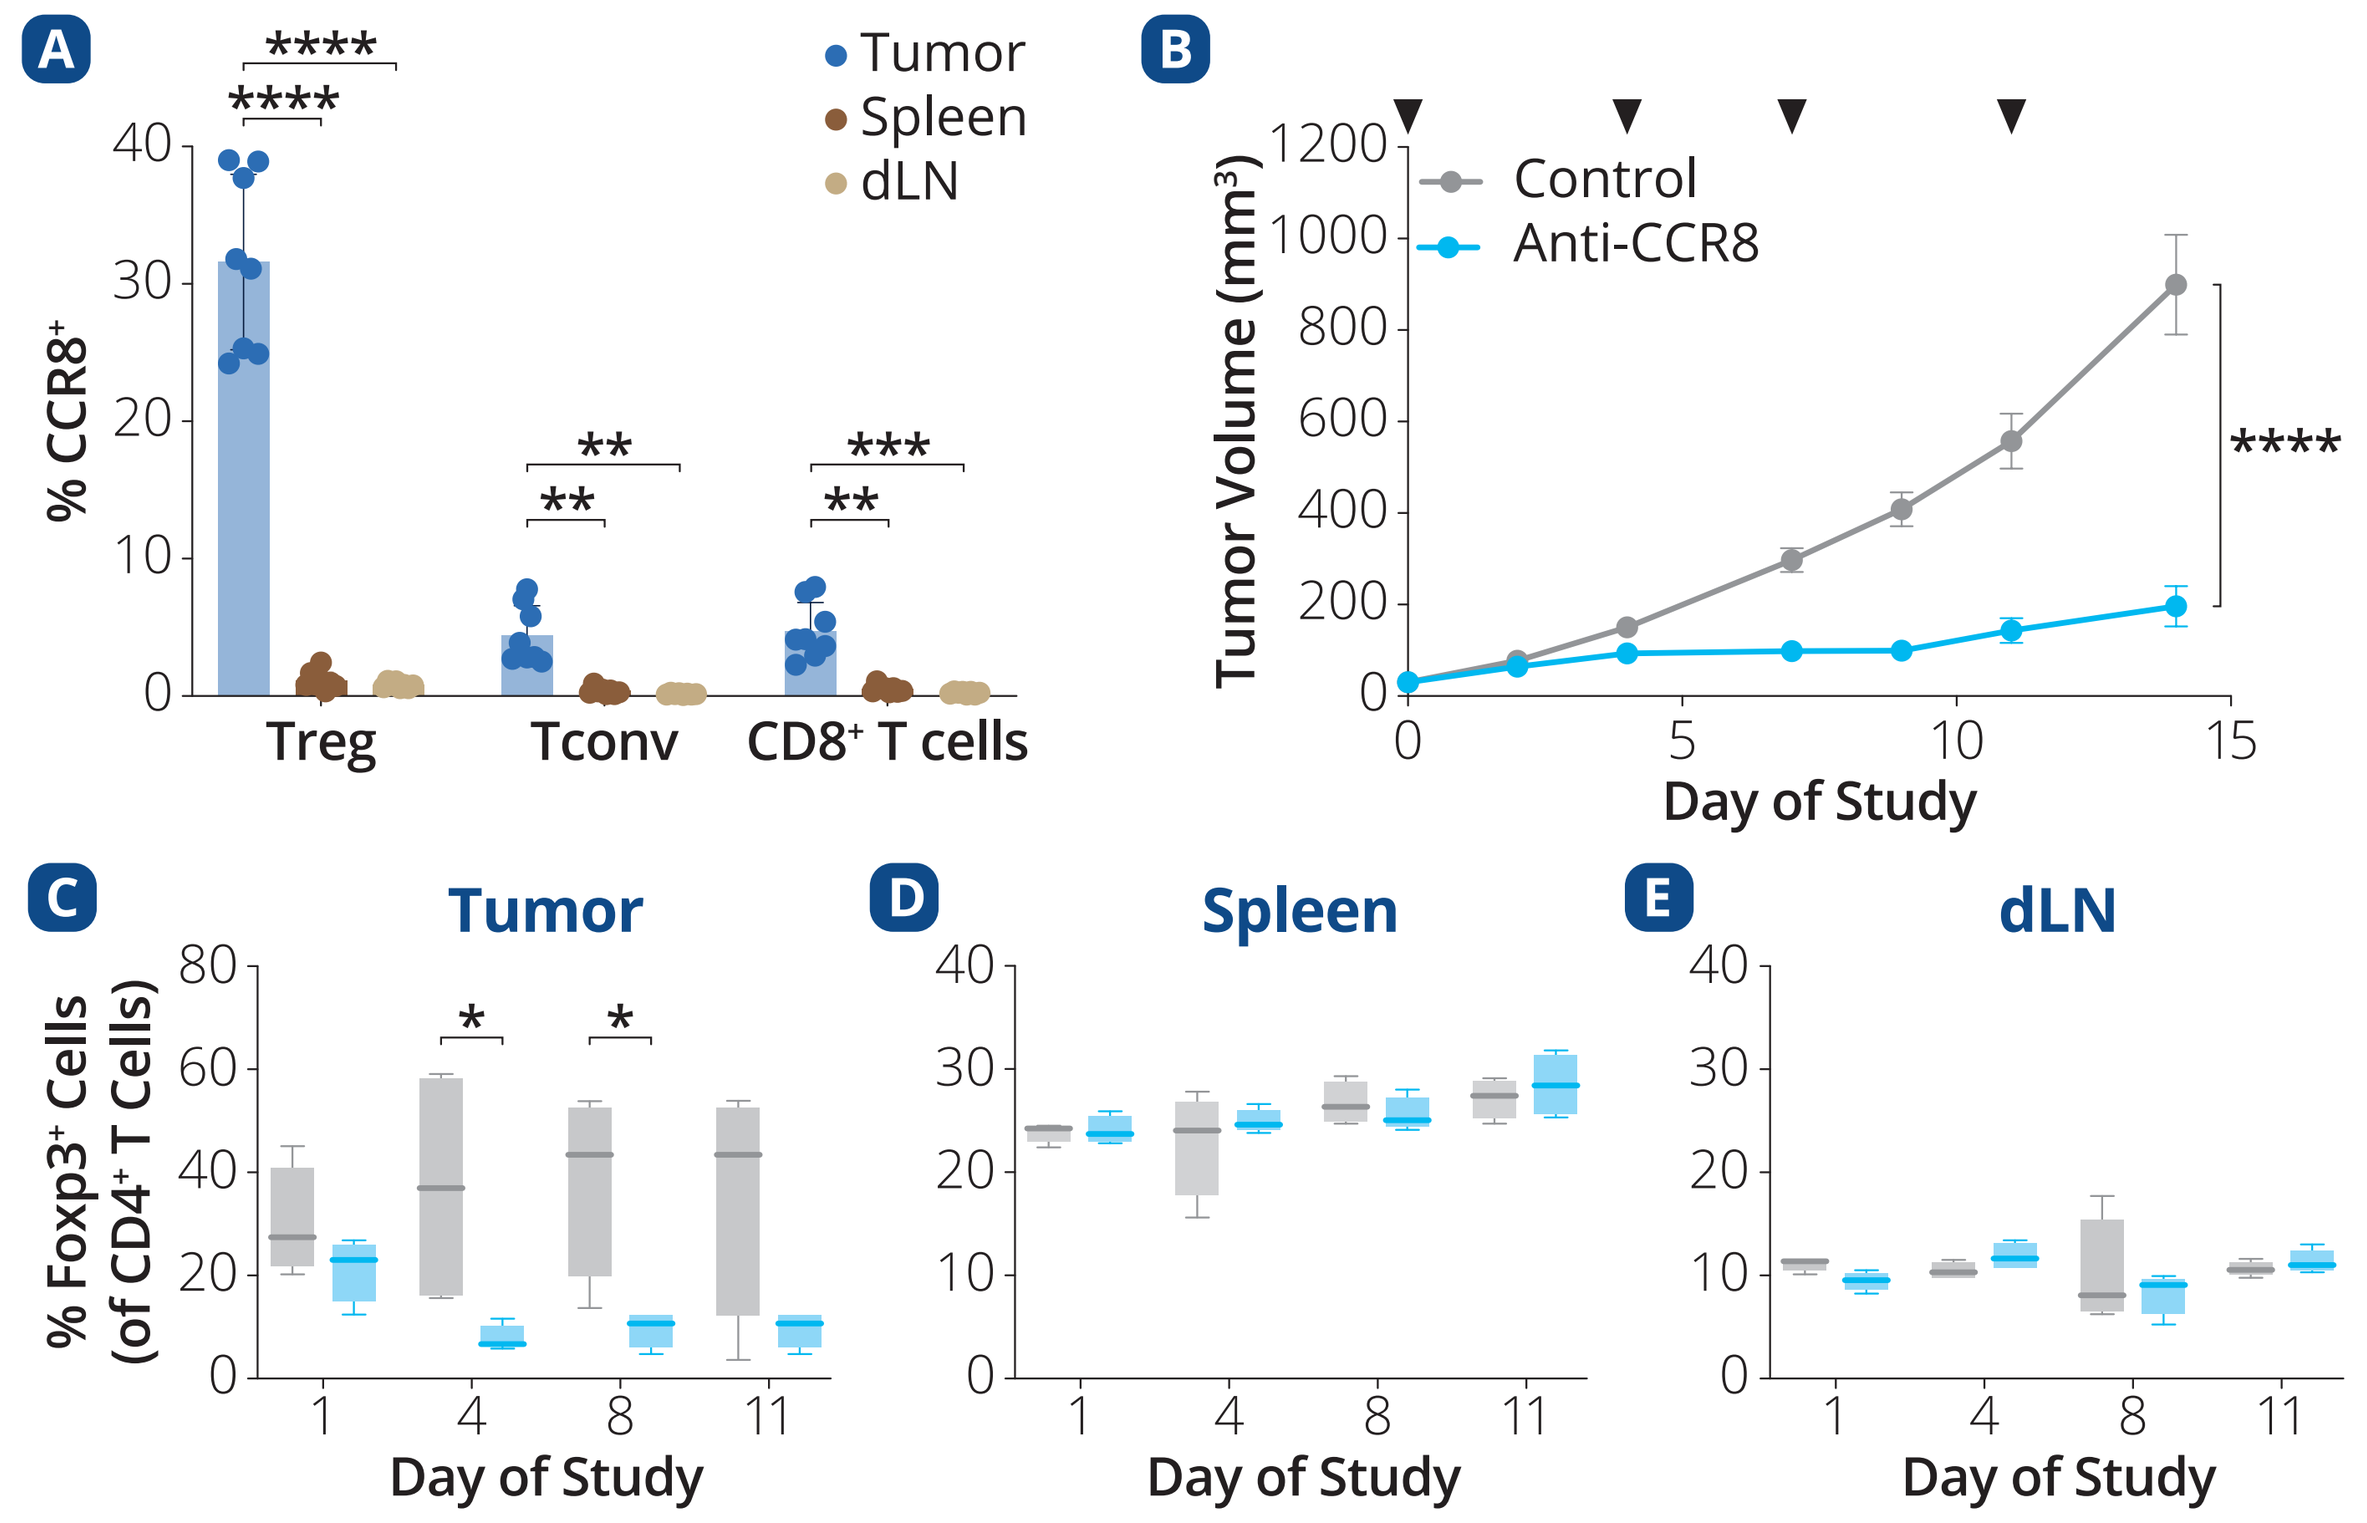

A
% CCR8⁺
Tumor
Spleen
dLN
Treg
Tconv
CD8⁺ T cells
B
Tumor Volume (mm³)
Control
Anti-CCR8
Day of Study
C Tumor
D Spleen
E dLN
% Foxp3⁺ Cells (of CD4⁺ T Cells)
Day of Study

A) CCR8 expression in T cell subsets in dissociated CT26 tumors, spleen, and draining lymph nodes (dLN) on Day 14 post-implantation. **B)** Efficacy study in CT26 tumor model comparing anti-CCR8 to control antibody treatment. **C-E)** Cytometry analysis of Treg cell frequency in tumor, spleen, and dLN, respectively.

A) Dissociated tissues were collected from untreated mice and analyzed by flow cytometry (n = 8). B) Tumor volume was measured after dosing with anti-CCR8 or control antibodies (n = 10). C-E) Tissues were dissociated and analyzed by flow cytometry. Treg cells were gated as live CD45⁺, TCRβ⁺, CD4⁺, Foxp3⁺ cells, (n = 4).

Anti-CCR8 Therapy Impacts the Myeloid Cell Compartment and Increases its Maturation State

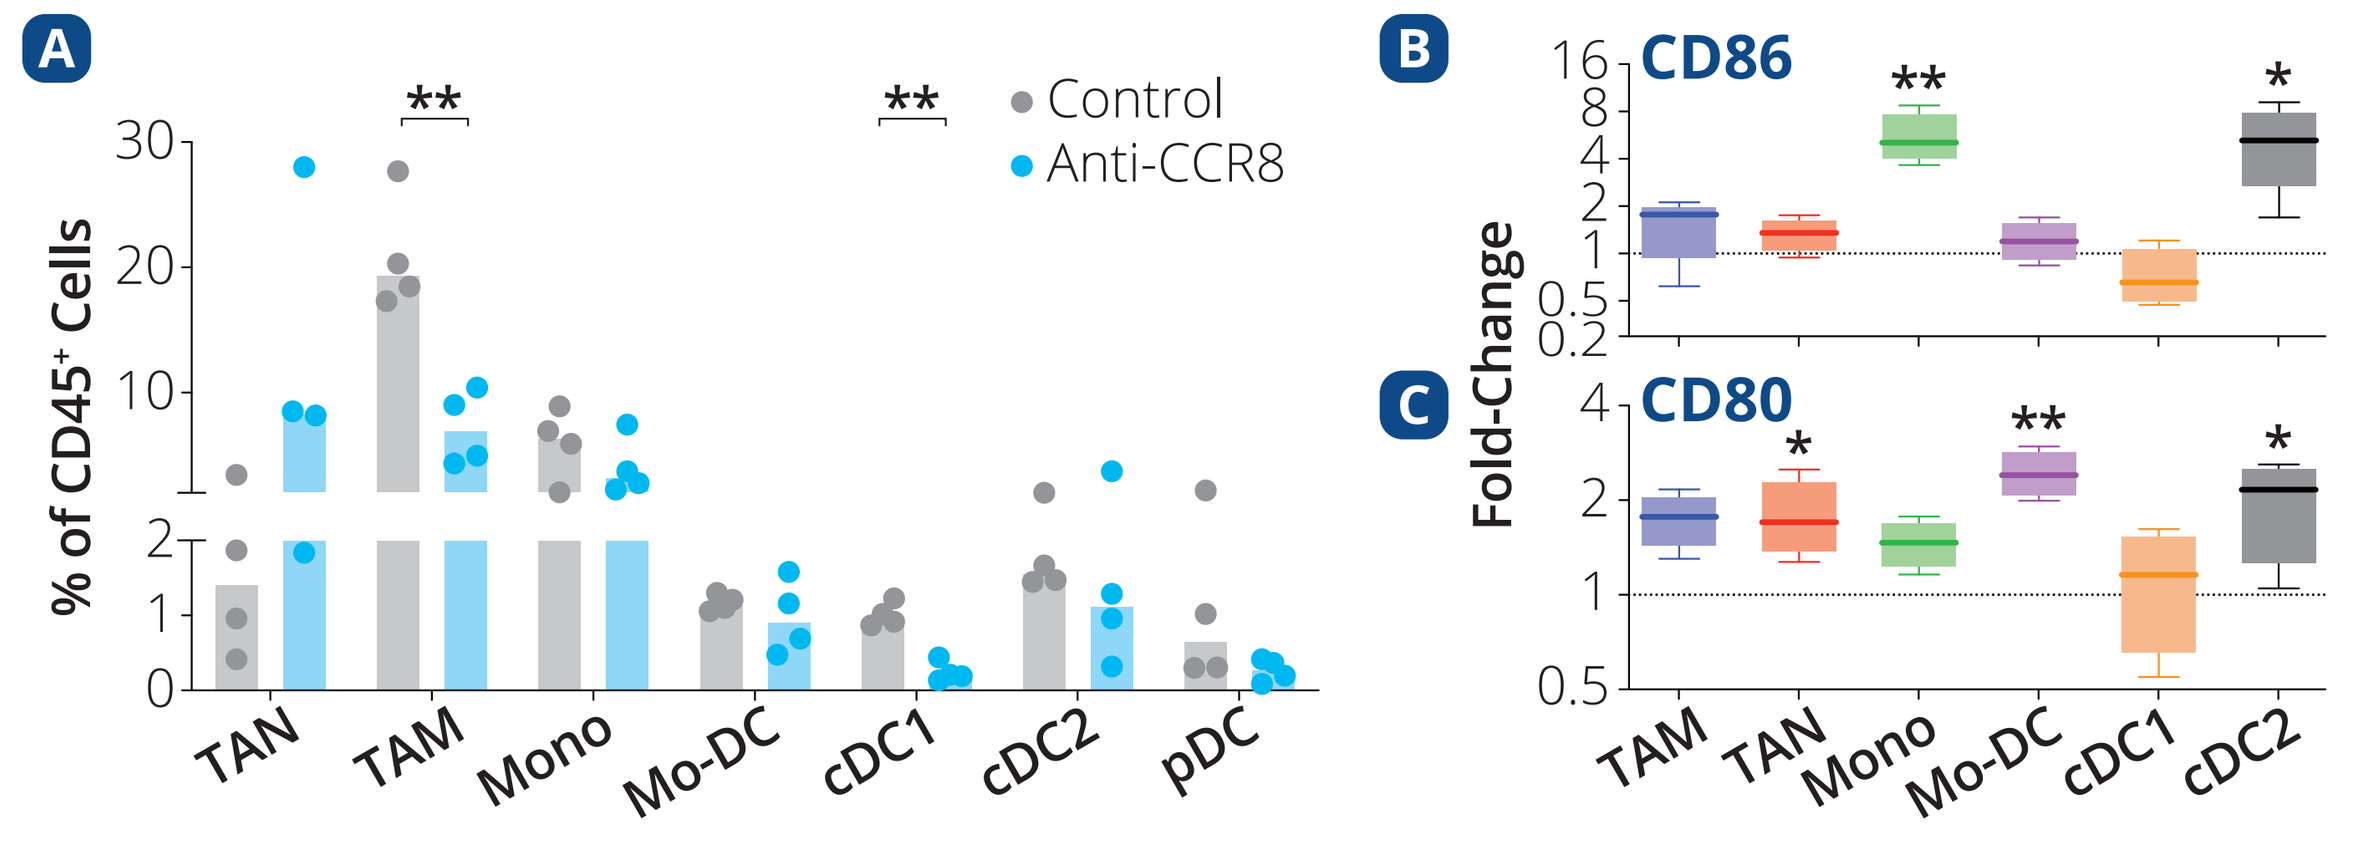

A
% of CD45⁺ Cells
Control
Anti-CCR8
TAN
TAM
Mono
Mo-DC
cDC1
cDC2
pDC
B CD86
C CD80
Fold-Change

Cytometric analysis of the myeloid cell compartment in dissociated CT26 tumors on Day 11 after treatment with anti-CCR8 or control antibody. **A)** Cell frequency of each subset and **B-C)** changes in the expression of co-stimulatory molecules CD86 and CD80 in each myeloid cell subset, expressed as Fold-Change (anti-CCR8/Control, n = 4).

Anti-CCR8 Therapy has Robust Anti-Tumor Activity in a Checkpoint-Resistant Melanoma Tumor Model

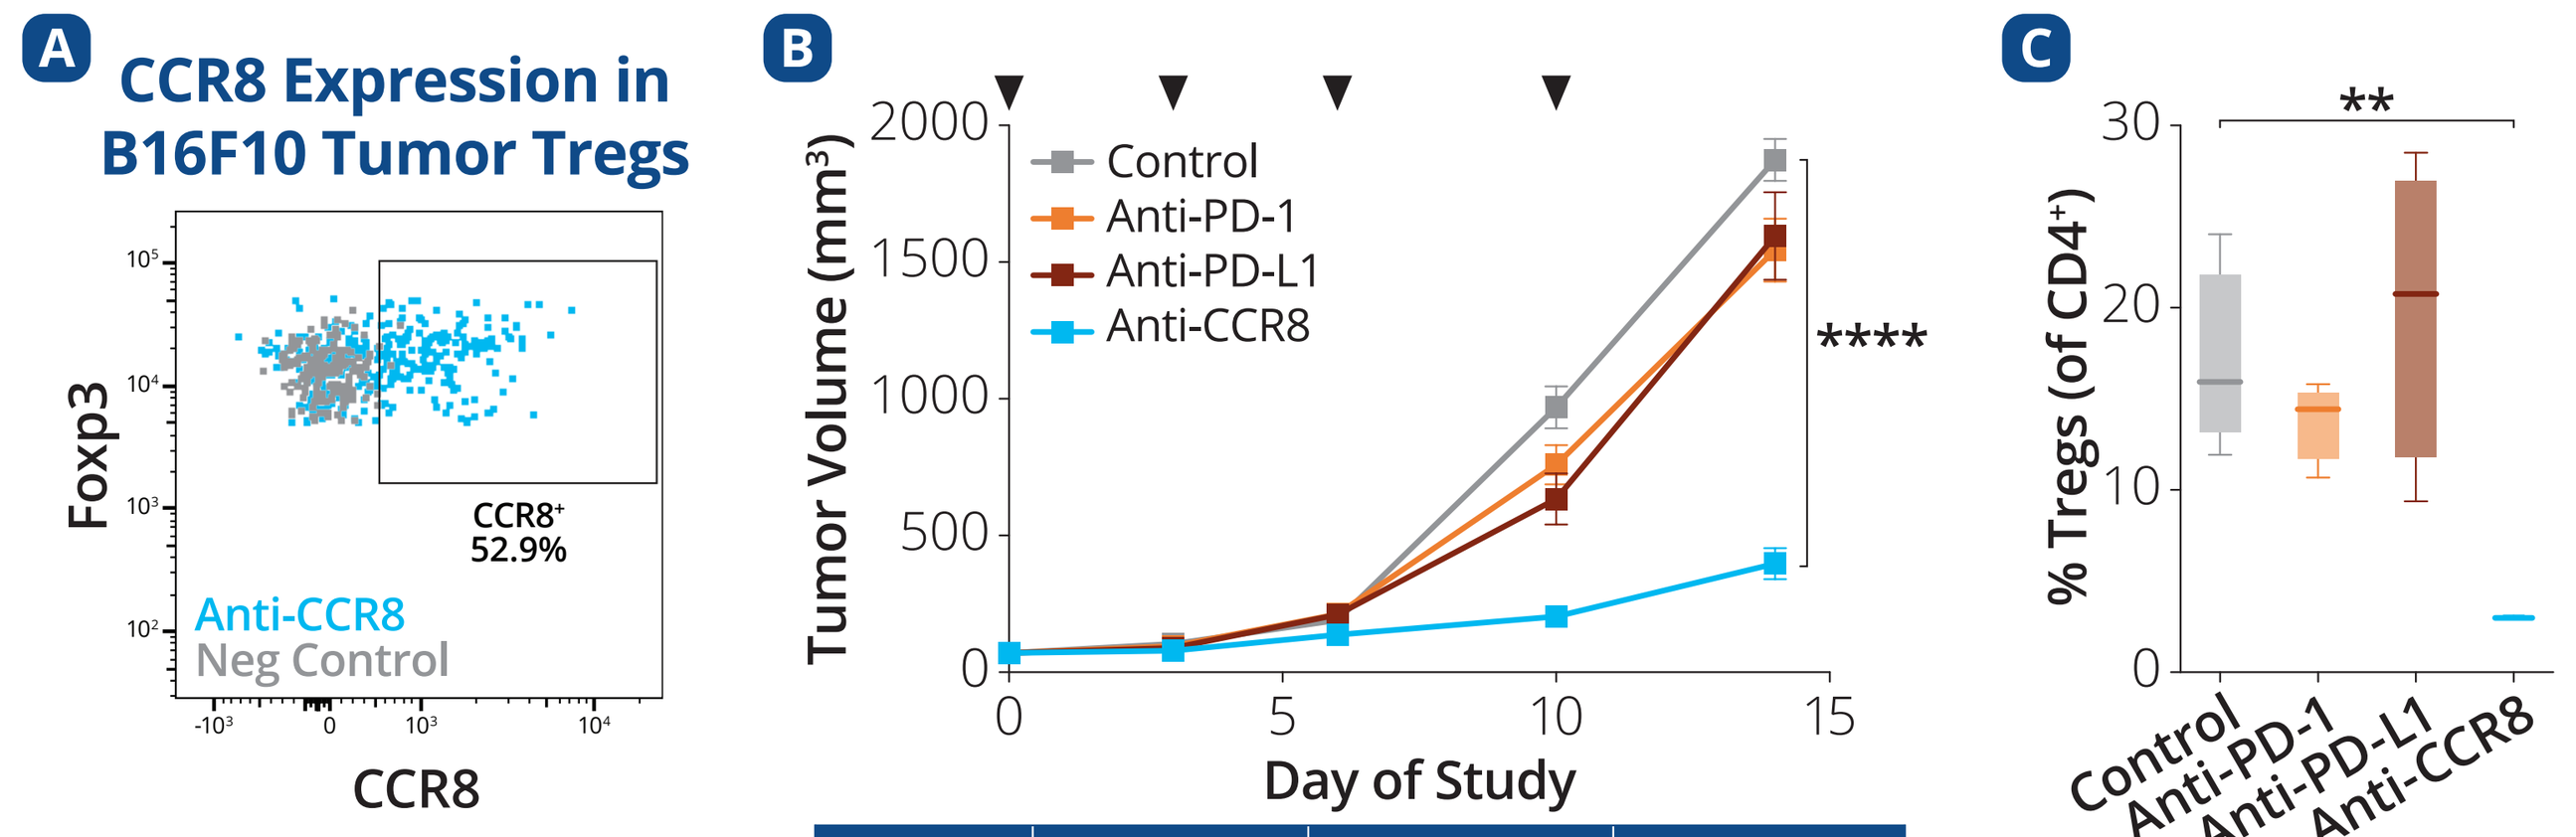

A
CCR8 Expression in B16F10 Tumor Tregs
Foxp3
CCR8
CCR8⁺ 52.9%
Anti-CCR8
Neg Control
B
Tumor Volume (mm³)
Control
Anti-PD-1
Anti-PD-L1
Anti-CCR8
Day of Study
C
% Tregs (of CD4⁺)

	Anti-PD-1	Anti-PD-L1	Anti-CCR8
TGI (%)	18	15	79

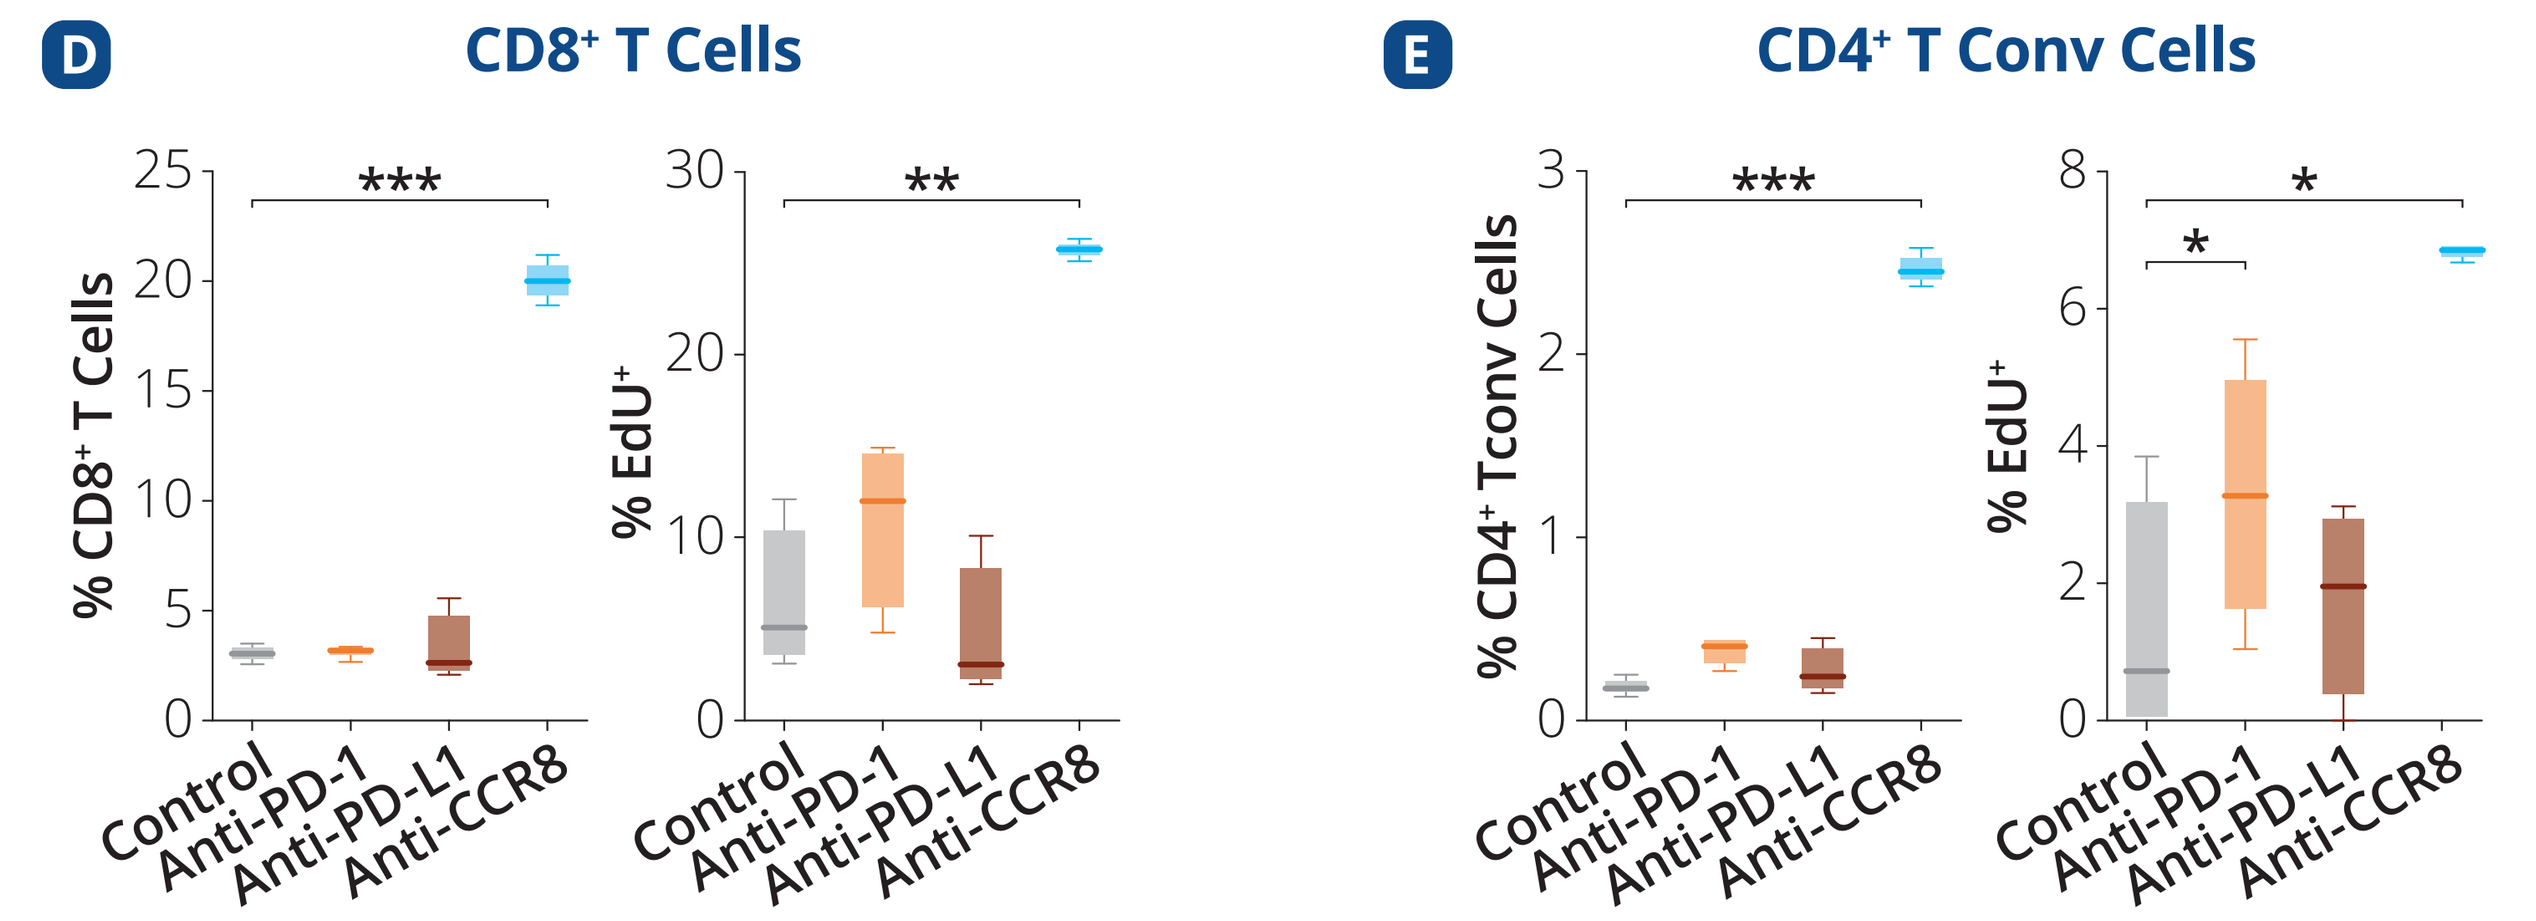

D CD8⁺ T Cells
% CD8⁺ T Cells
% EdU⁺
E CD4⁺ T Conv Cells
% CD4⁺ Tconv Cells
% EdU⁺
Control
Anti-PD-1
Anti-PD-L1
Anti-CCR8

A) Representative graph of cytometric analysis of CCR8 expression on tumor Tregs measured in dissociated B16F10 tumors. **B)** Efficacy study evaluating anti-tumor activity of anti-CCR8, anti-PD-1, or anti-PD-L1 monotherapy treatments in the B16F10 tumor model. **C-E)** Cytometric analyses of tumor Treg cells and proliferating effector T cells from dissociated B16F10 tumors collected on Day 9 of study.

A) B16F10 tumors were collected on Day 13 after implantation and CCR8 expression was detected on Foxp3⁺ Treg cells by flow cytometry. B-E) Mice bearing subcutaneous B16F10 tumors were treated with anti-CCR8, anti-PD-1, anti-PD-L1, or control antibodies. B) Percent tumor growth inhibition (TGI, %) was calculated on Day 14 of study, (n = 10). C) Tumors were collected and the fraction of Foxp3⁺ Treg cells was determined (n=4). D-E) Mice were dosed with 1 mg 5-Ethynyl-2'-deoxyuridine (EdU) 24 hours prior to sample collection and fraction of EdU⁺ CD8⁺ effector T cells and EdU⁺ CD4⁺ Tconv cells were measured, respectively, (n = 4).

Anti-CCR8 Therapy Promotes Production of Proinflammatory Cytokines & Chemokines

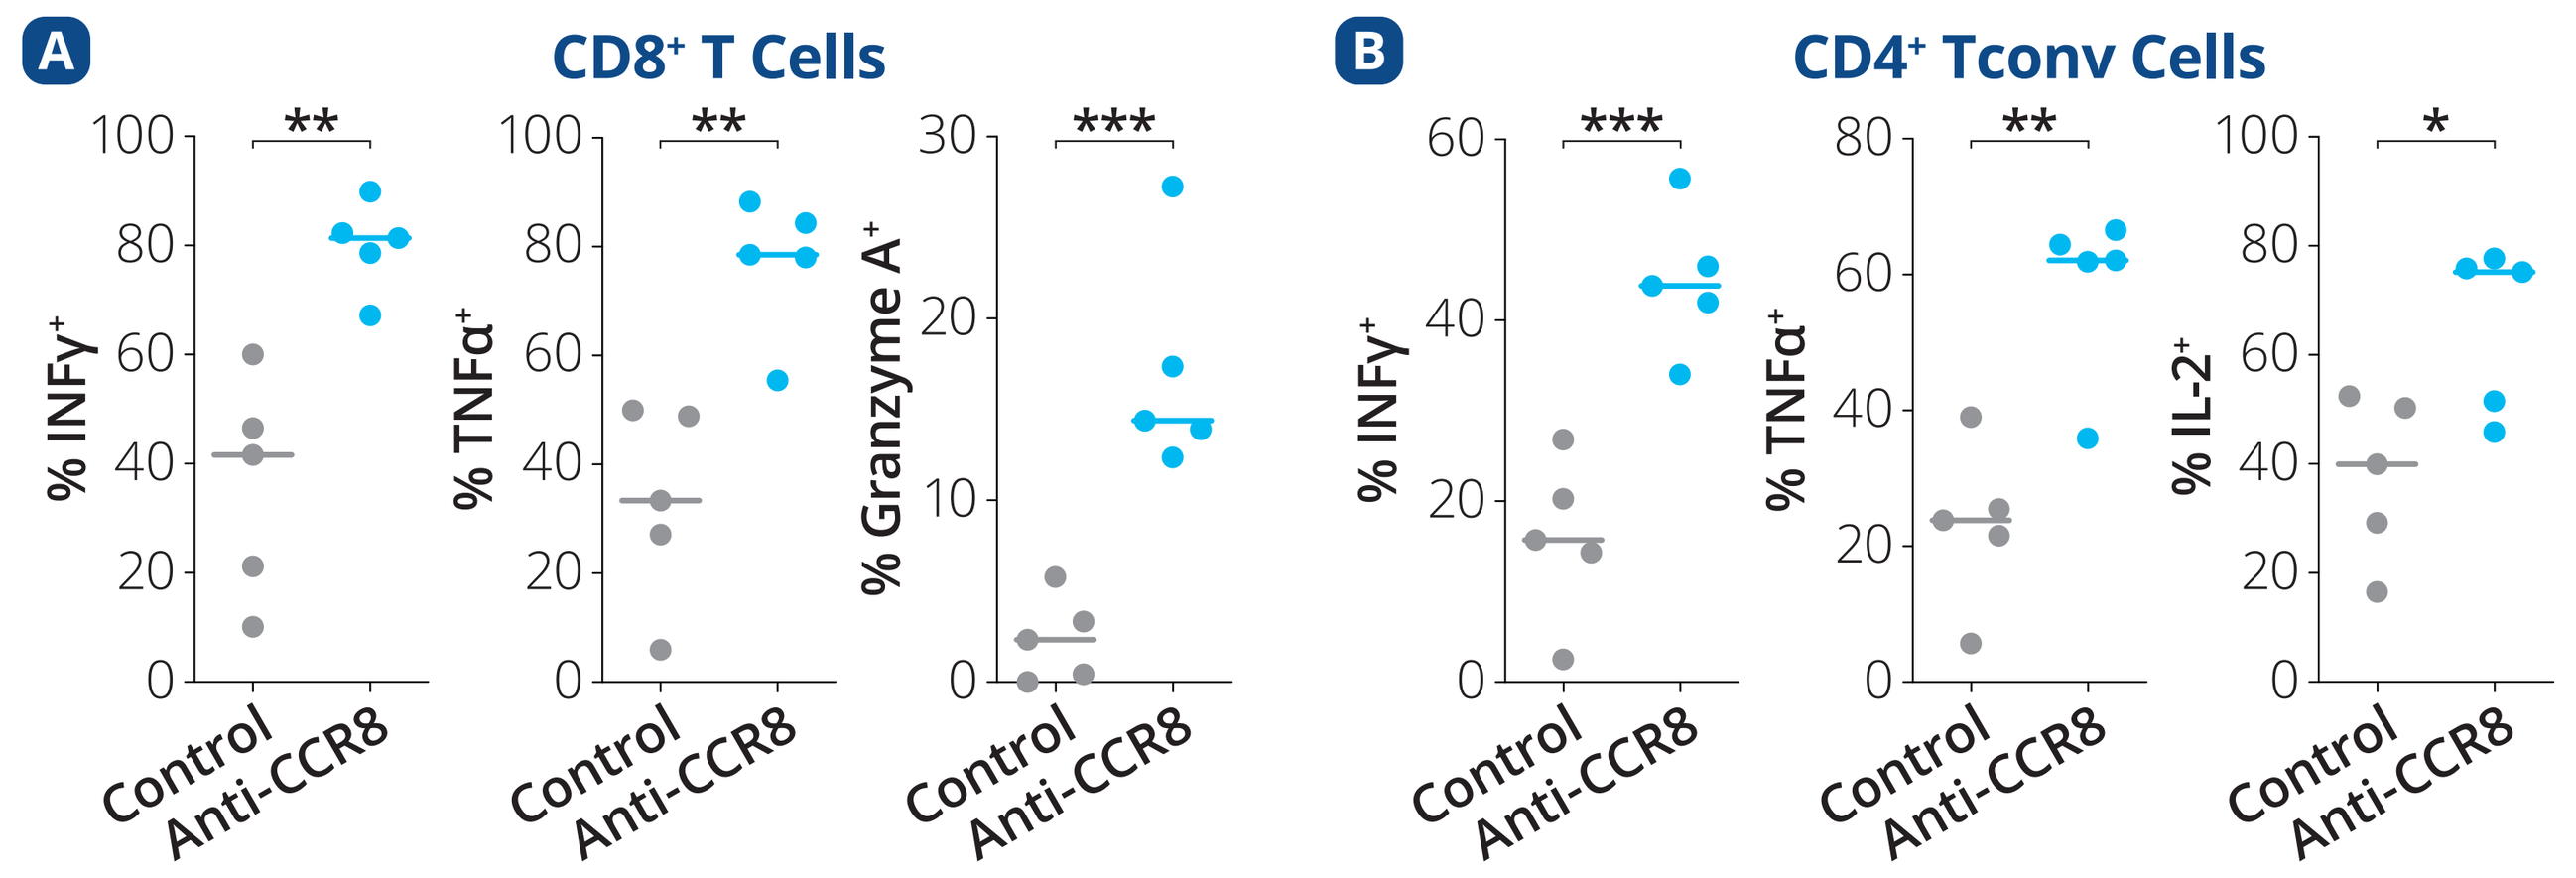

A CD8⁺ T Cells
% INFγ⁺
% TNFα⁺
% Granzyme A⁺
B CD4⁺ Tconv Cells
% INFγ⁺
% TNFα⁺
% IL-2⁺
Control
Anti-CCR8

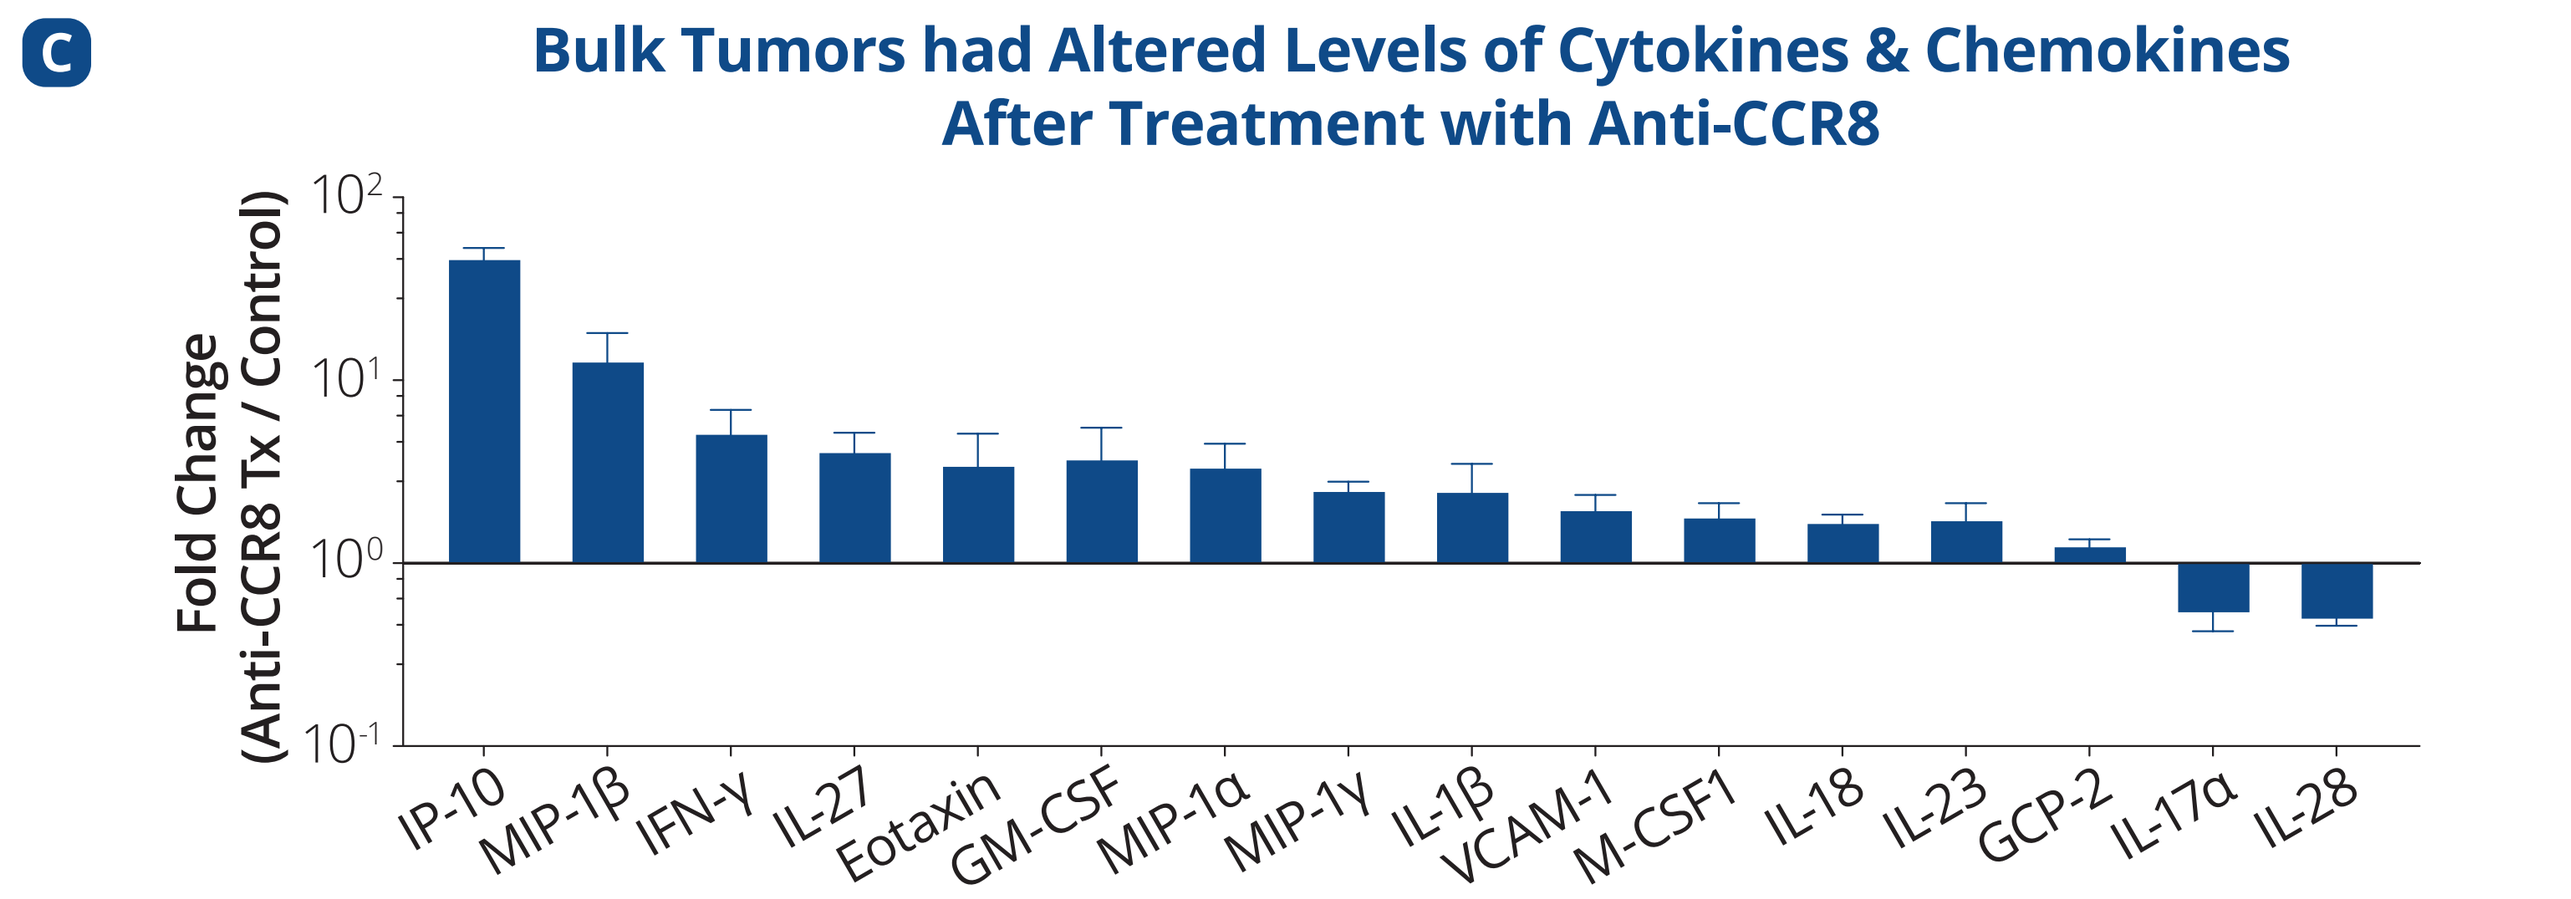

C
Bulk Tumors had Altered Levels of Cytokines & Chemokines After Treatment with Anti-CCR8
Fold Change (Anti-CCR8 Tx / Control)
IP-10
MIP-1β
IFN-γ
IL-27
Eotaxin
GM-CSF
MIP-1α
MIP-1γ
IL-1β
VCAM-1
M-CSF1
IL-18
IL-23
GCP-2
IL-17α
IL-28

A-B) Resected tumor samples on Day 8 post-treatment initiation were restimulated *ex vivo* with PMA and ionomycin to evaluate intracellular cytokine levels of CD8⁺ T and CD4⁺ Tconv cells, respectively. **C)** Fold-change in cytokine levels evaluated in bulk tumor lysates.

Mice bearing B16F10 tumors were dosed with anti-CCR8 or control antibodies. A-B) Flow cytometric analyses of intracellular cytokine levels of TILs after *ex vivo* restimulation of dissociated tumors (n = 5). C) Levels detected by Luminex in snap-frozen and lysed bulk B16F10 tumors 4 days after treatment initiation with anti-CCR8 or control antibodies, (n = 4).

Abbreviations: ▼ = dosing day; APCs = antigen-presenting cells; cDC = conventional dendritic cell; CCR8 = C-C Chemokine Receptor 8; DC = dendritic cells; DF = diffusion dimension; dLN = draining lymph nodes; FFPE = formalin-fixed paraffin-embedded; Foxp3 = forkhead box P3 protein; hCCR8 = human CCR8; hCCR8KI = human CCR8 knock-in; IFNγ = interferon-gamma; IHC = immunohistochemistry; IL = interleukin; MO-DC = monocyte-derived dendritic cells; Mono = monocytes; NK = natural killer; PD-1 = programmed death 1; pDC = plasmacytoid dendritic cells; PMA = phorbol 12-myristate 13-acetate; TAM = tumor-associated macrophages; TAN = tumor-associated neutrophils; Tconv = conventional T cells; TGI = tumor growth inhibition; TIL = tumor-infiltrating lymphocytes; TME = tumor microenvironment; TNFα = tumor necrosis factor alpha; Tregs = regulatory T cells.

Statistics: * = p < 0.05; ** = p < 0.01; *** = p < 0.001; **** = p < 0.0001

References: **1)** Plitas et al, *Ann Rev Can Biol* 2020. **2)** Sakaguchi et al, *Ann Rev Imm* 2020. **3)** Plitas et al, *Immunity* 2016. **4)** Wang et al, *Nat. Imm* 2019. **5)** Alvisi et al, *JCI* 2020. **6)** Whiteside et al, *Immunology* 2021. **7)** Campbell et al, *Can Res* 2021. **8)** Weaver et al, *OncoImm* 2022. **9)** VanDamme et al, *JITC* 2021. **10)** Kidani et al, *PNAS* 2022.

Anti-CCR8 and anti-PD1 Combination Therapy Enhances Overall Survival and Promotes Expansion of Effector T cells in B16F10 Melanoma

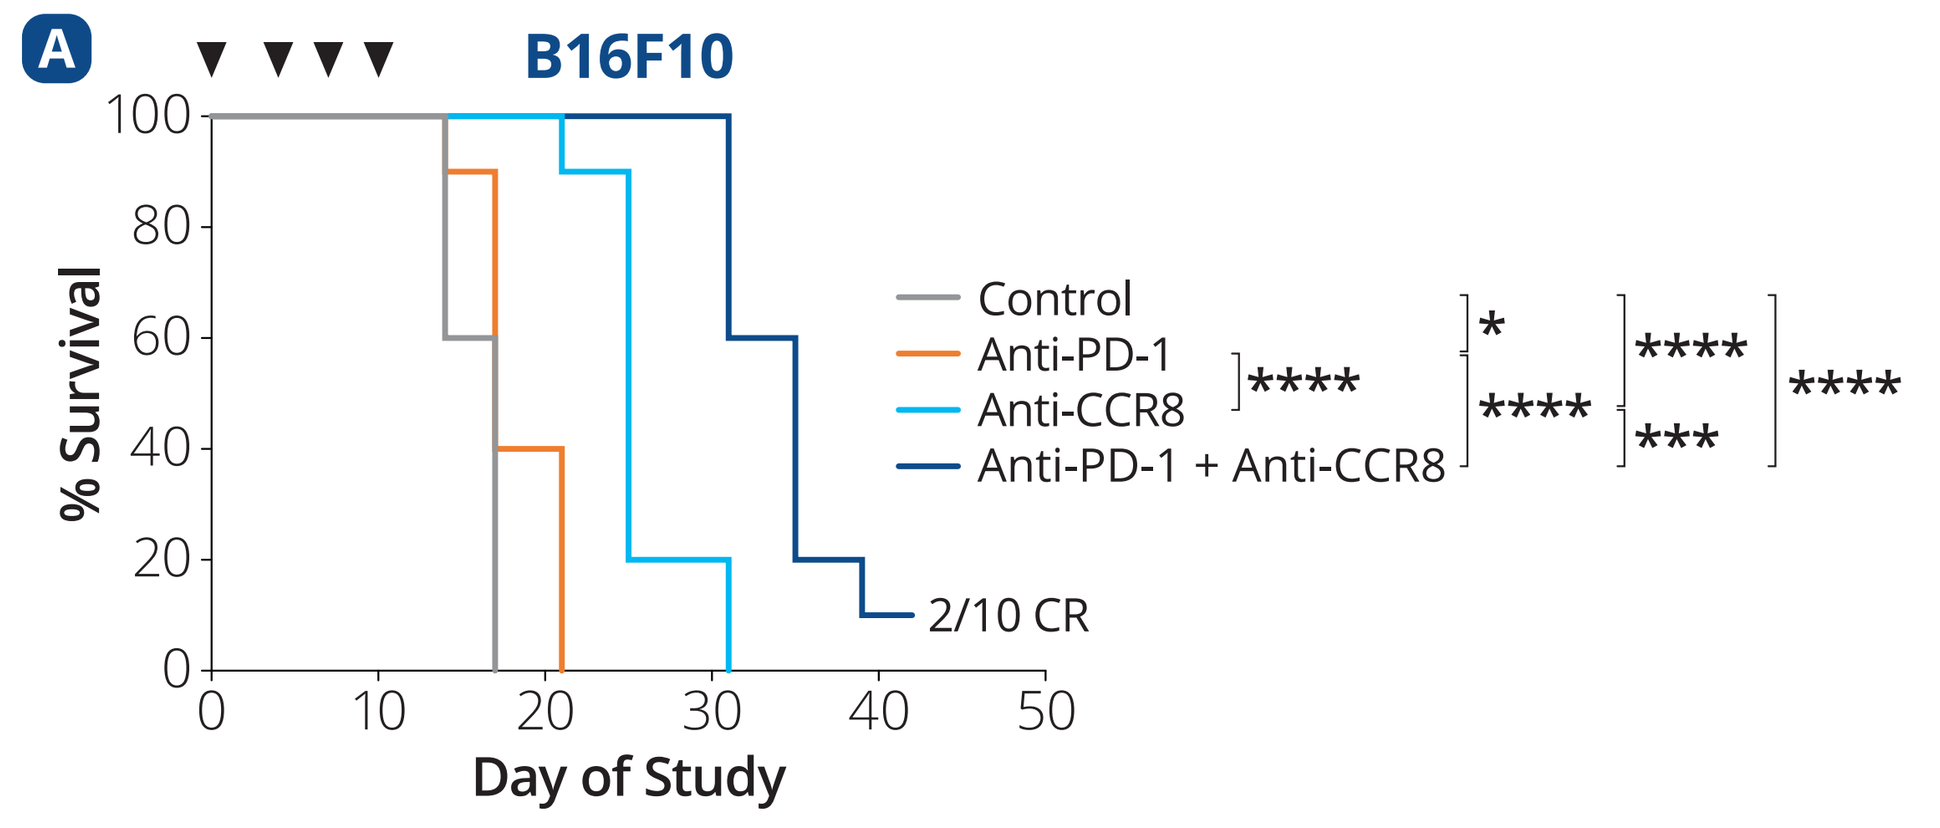

A
B16F10
% Survival
Control
Anti-PD-1
Anti-CCR8
Anti-PD-1 + Anti-CCR8
2/10 CR
Day of Study

	Median Survival (Days)
Control	17
Anti-PD-1	17
Anti-CCR8	25
Anti-PD-1 + Anti-CCR8	35

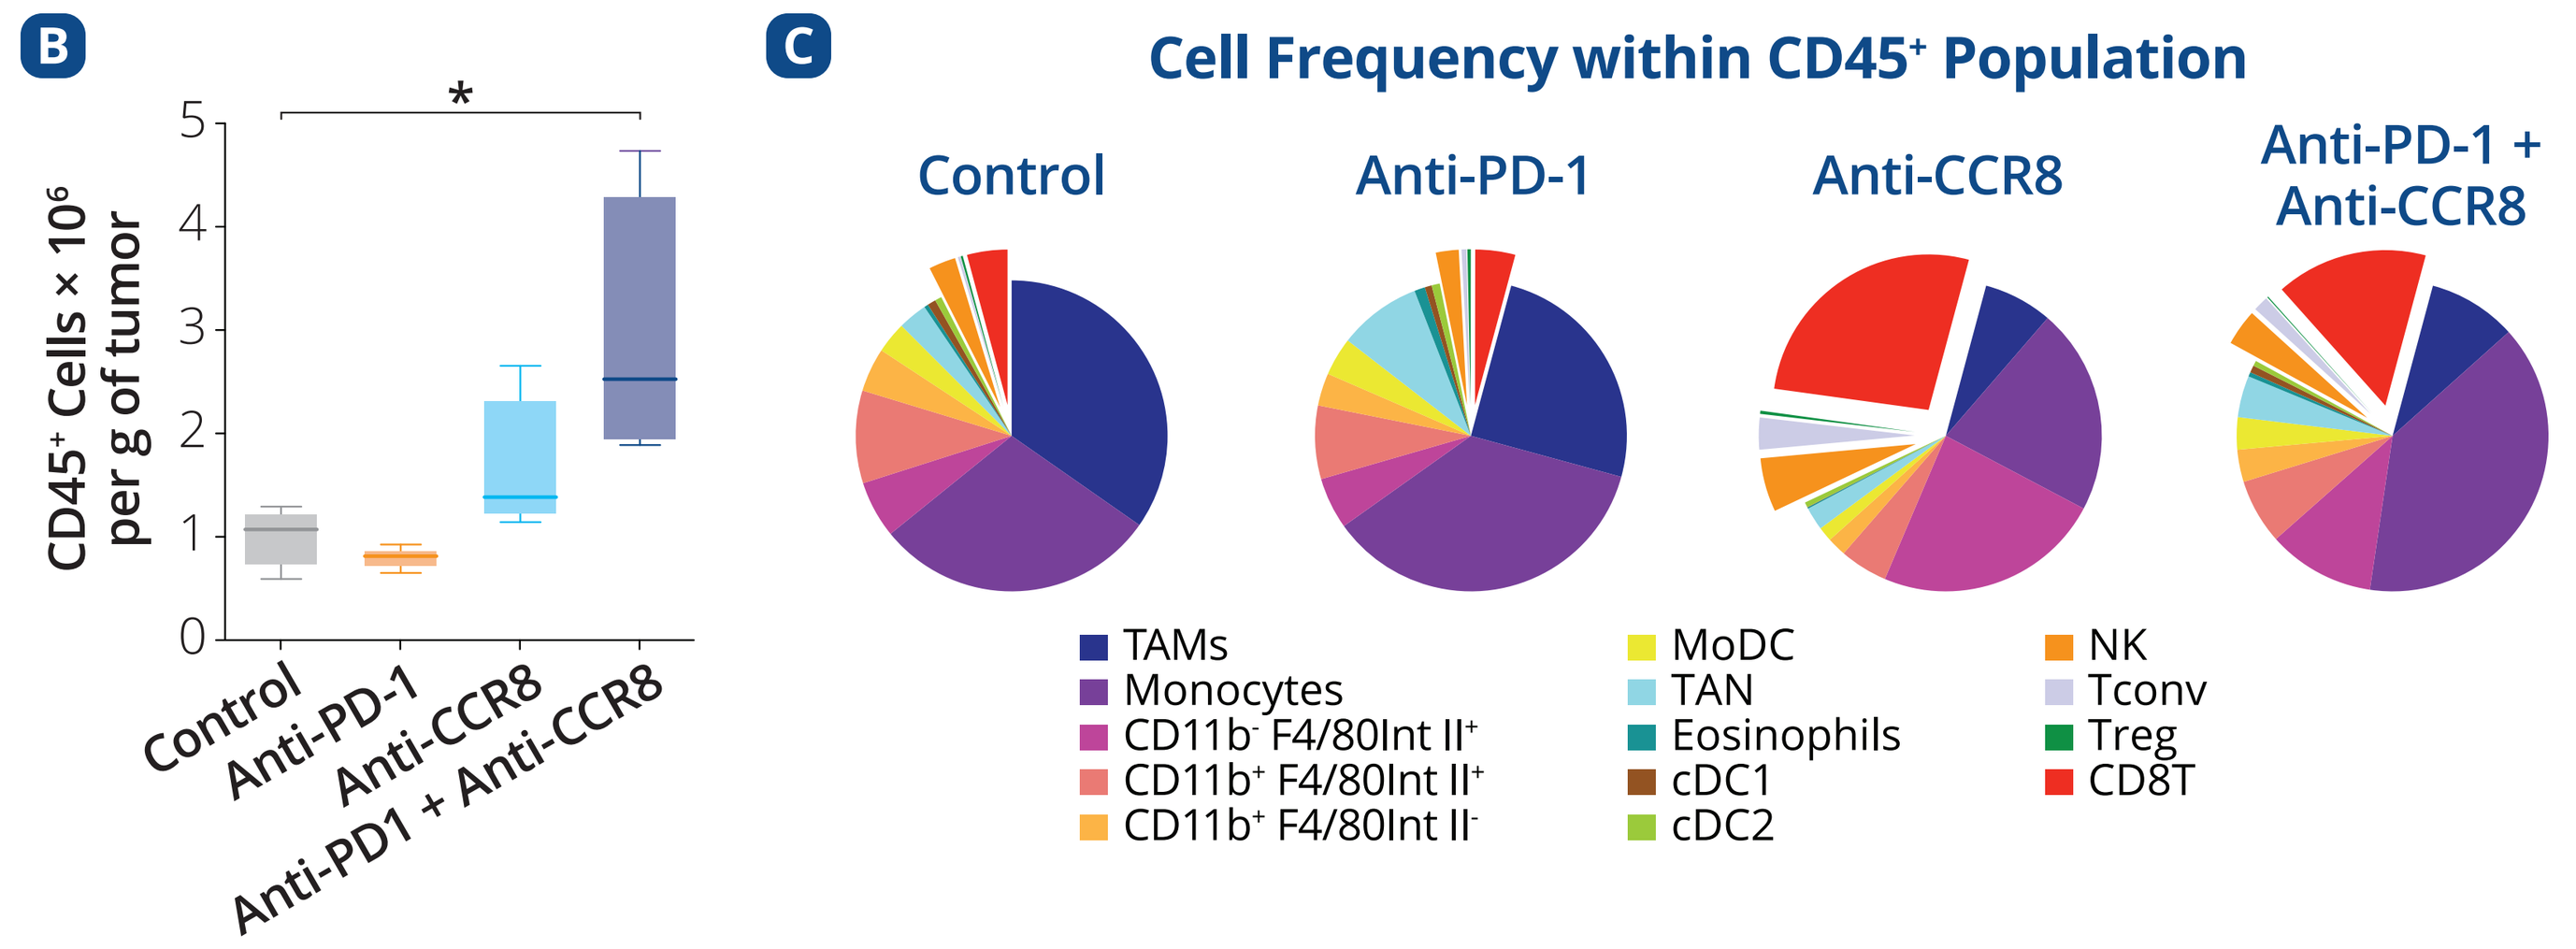

B
CD45⁺ Cells × 10⁶ per g of tumor
Control
Anti-PD-1
Anti-CCR8
Anti-PD1 + Anti-CCR8
C
Cell Frequency within CD45⁺ Population
Control
Anti-PD-1
Anti-CCR8
Anti-PD-1 + Anti-CCR8
TAMs
Monocytes
CD11b⁻ F4/80Int II⁺
CD11b⁺ F4/80Int II⁺
CD11b⁺ F4/80Int II⁻
MoDC
TAN
Eosinophils
cDC1
cDC2
NK
Tconv
Treg
CD8T

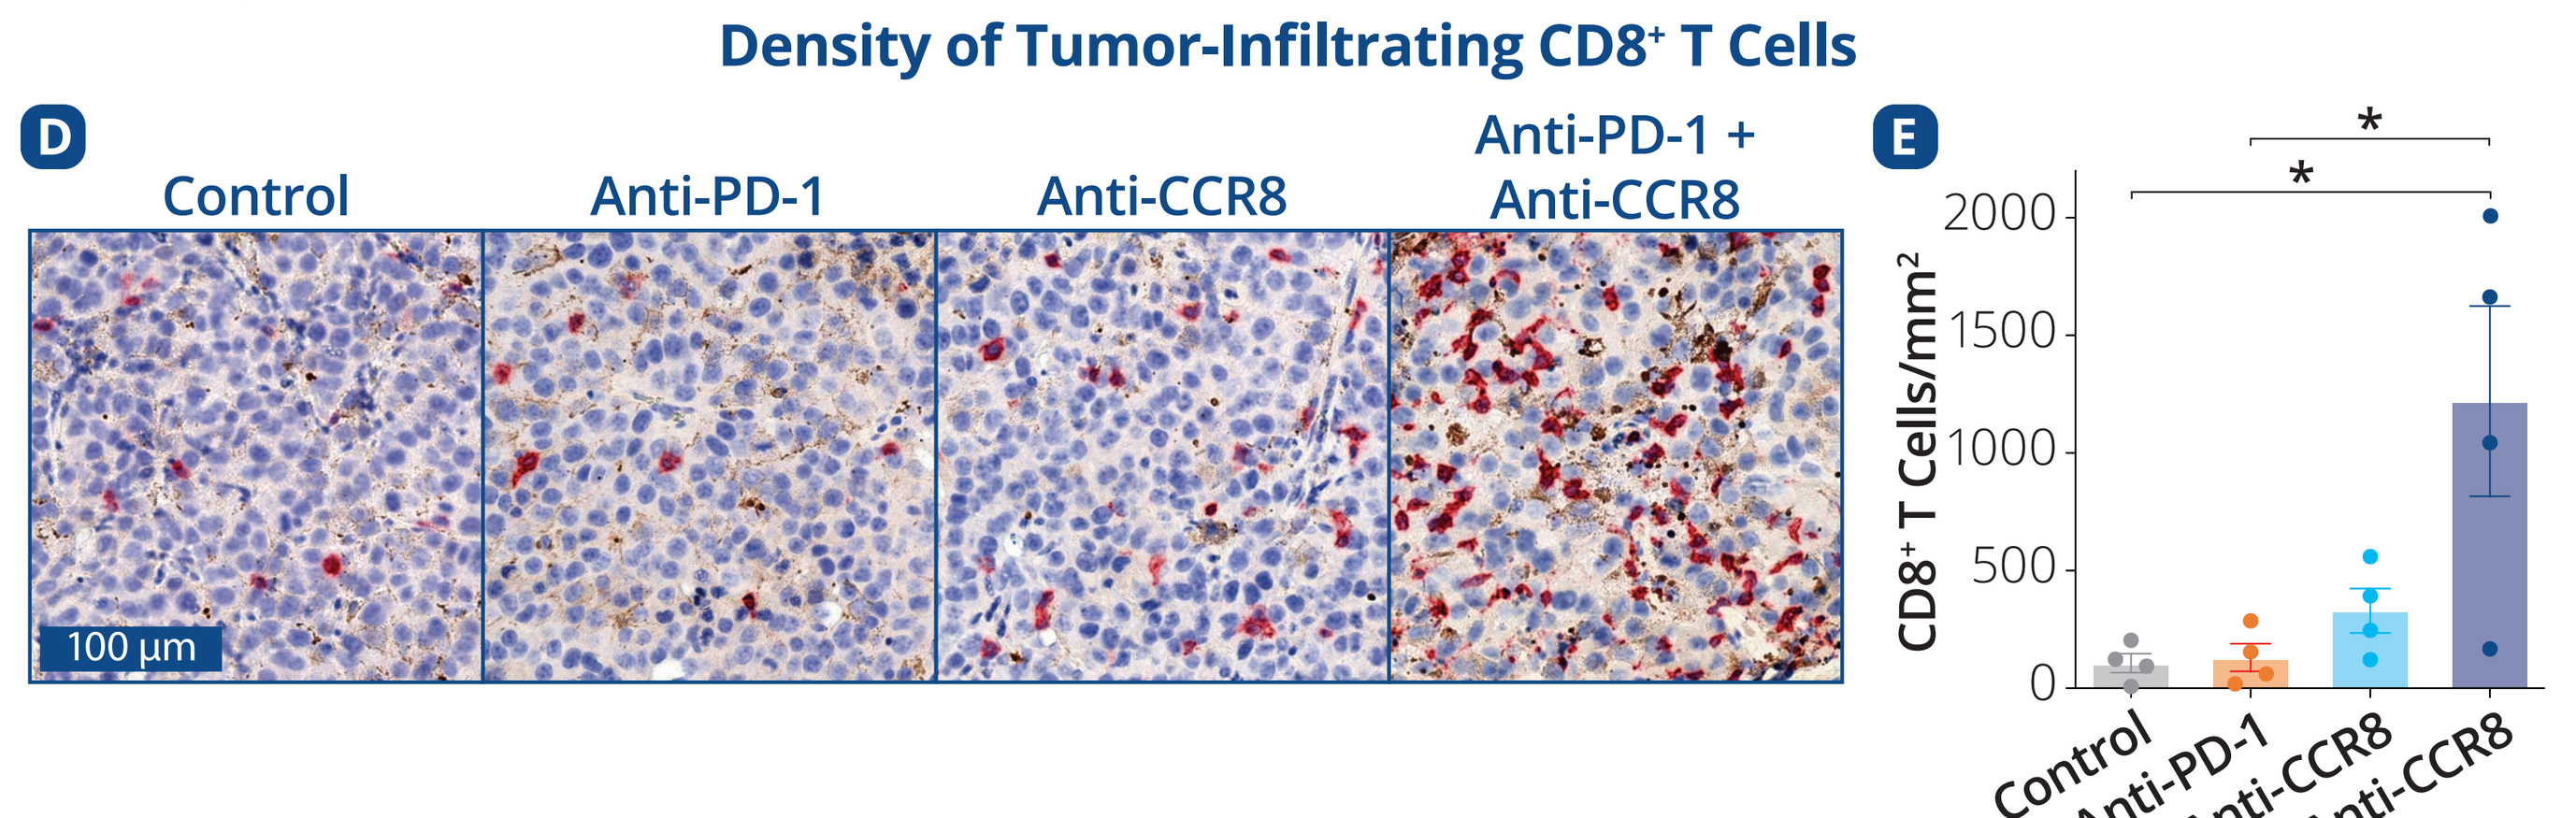

Density of Tumor-Infiltrating CD8⁺ T Cells
D
Control
Anti-PD-1
Anti-CCR8
Anti-PD-1 + Anti-CCR8
100 µm
E
CD8⁺ T Cells/mm²

A) Survival study in B16F10 melanoma model. **B-C)** Cytometric analyses and average frequency, respectively, of TILs on Day 9 of study. **D)** Representative IHC images of infiltrating CD8⁺ T cells in B16F10 tumors on Day 9 of study. **E)** Quantification of density of infiltrating CD8⁺ T cells.

A) Mice bearing subcutaneous B16F10 tumors were treated with anti-CCR8, anti-PD-1, or control antibodies (n = 10). B) Total number of CD45⁺ cells per gram of tumor was measured by normalizing the total CD45⁺ cell count to tumor weight (n = 4). C) Frequencies of defined TILs were averaged per group and normalized to their total sum. D) IHC for CD8 using a red chromogen was performed on FFPE samples of tumors collected from the study. E) Infiltrating CD8⁺ T cells were quantified using image analysis on digitally scanned slides (n = 4).

Anti-CCR8 + Anti-PD-1 Combination Treatment Enhances Production of Pro-inflammatory Cytokines and Chemokines in Melanoma Tumors

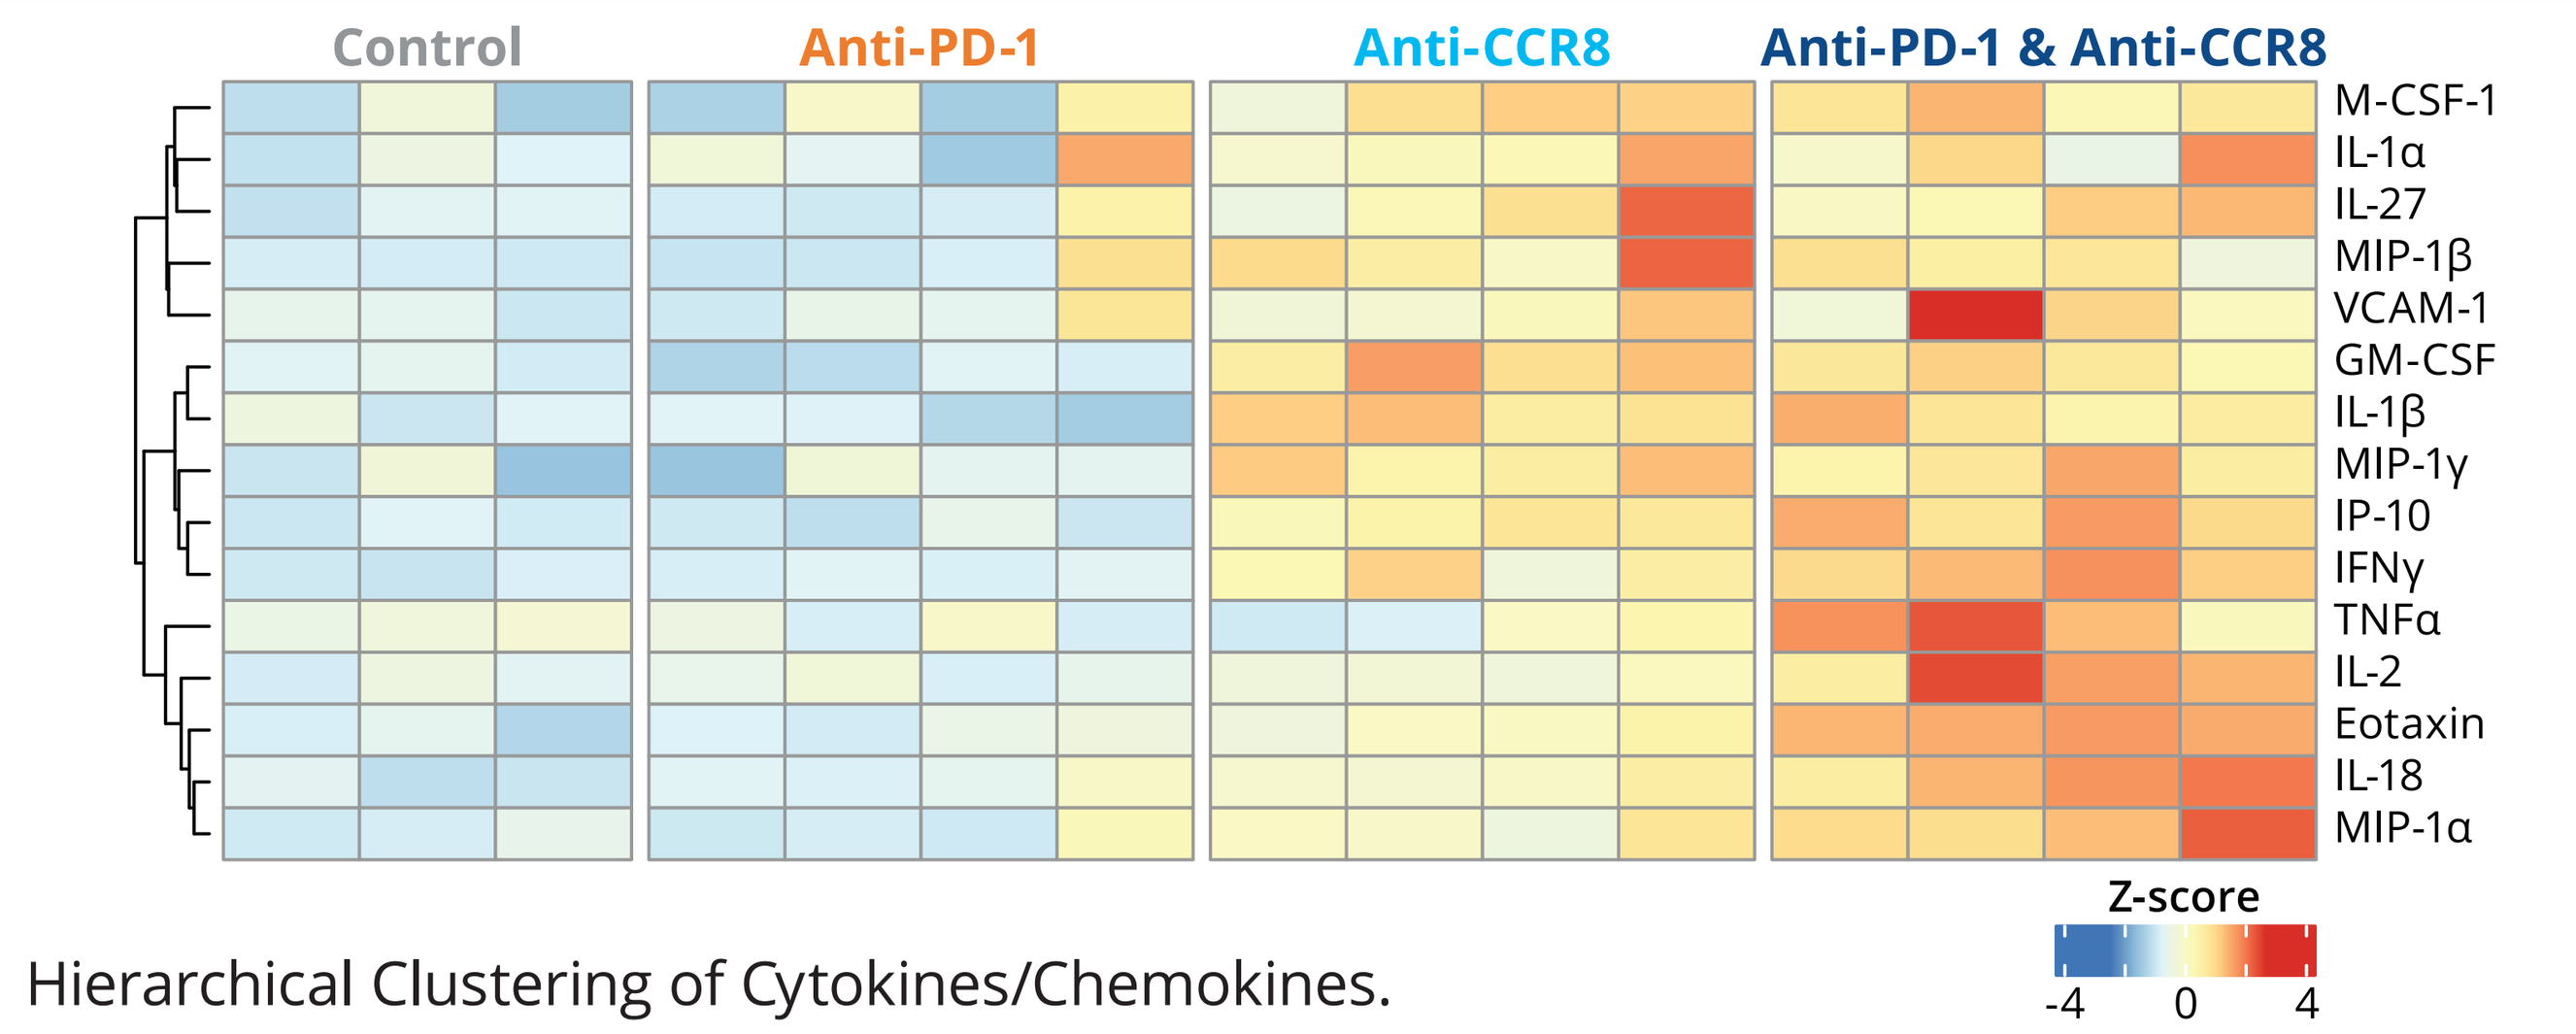

Control
Anti-PD-1
Anti-CCR8
Anti-PD-1 & Anti-CCR8
M-CSF-1
IL-1α
IL-27
MIP-1β
VCAM-1
GM-CSF
IL-1β
MIP-1γ
IP-10
IFNγ
TNFα
IL-2
Eotaxin
IL-18
MIP-1α
Z-score

Hierarchical Clustering of Cytokines/Chemokines.

Detection by Luminex assay in snap-frozen and lysed bulk B16F10 tumors 8 days after treatment initiation. Tumor-bearing mice were dosed with anti-CCR8, anti-PD-1, a combination of both, or control antibodies, (n = 3-4).

Treatment with SRF114, a Fully Human Afucosylated IgG1 Antibody, Results in Robust Anti-Tumor Activity in Human CCR8 Knock-In Mice

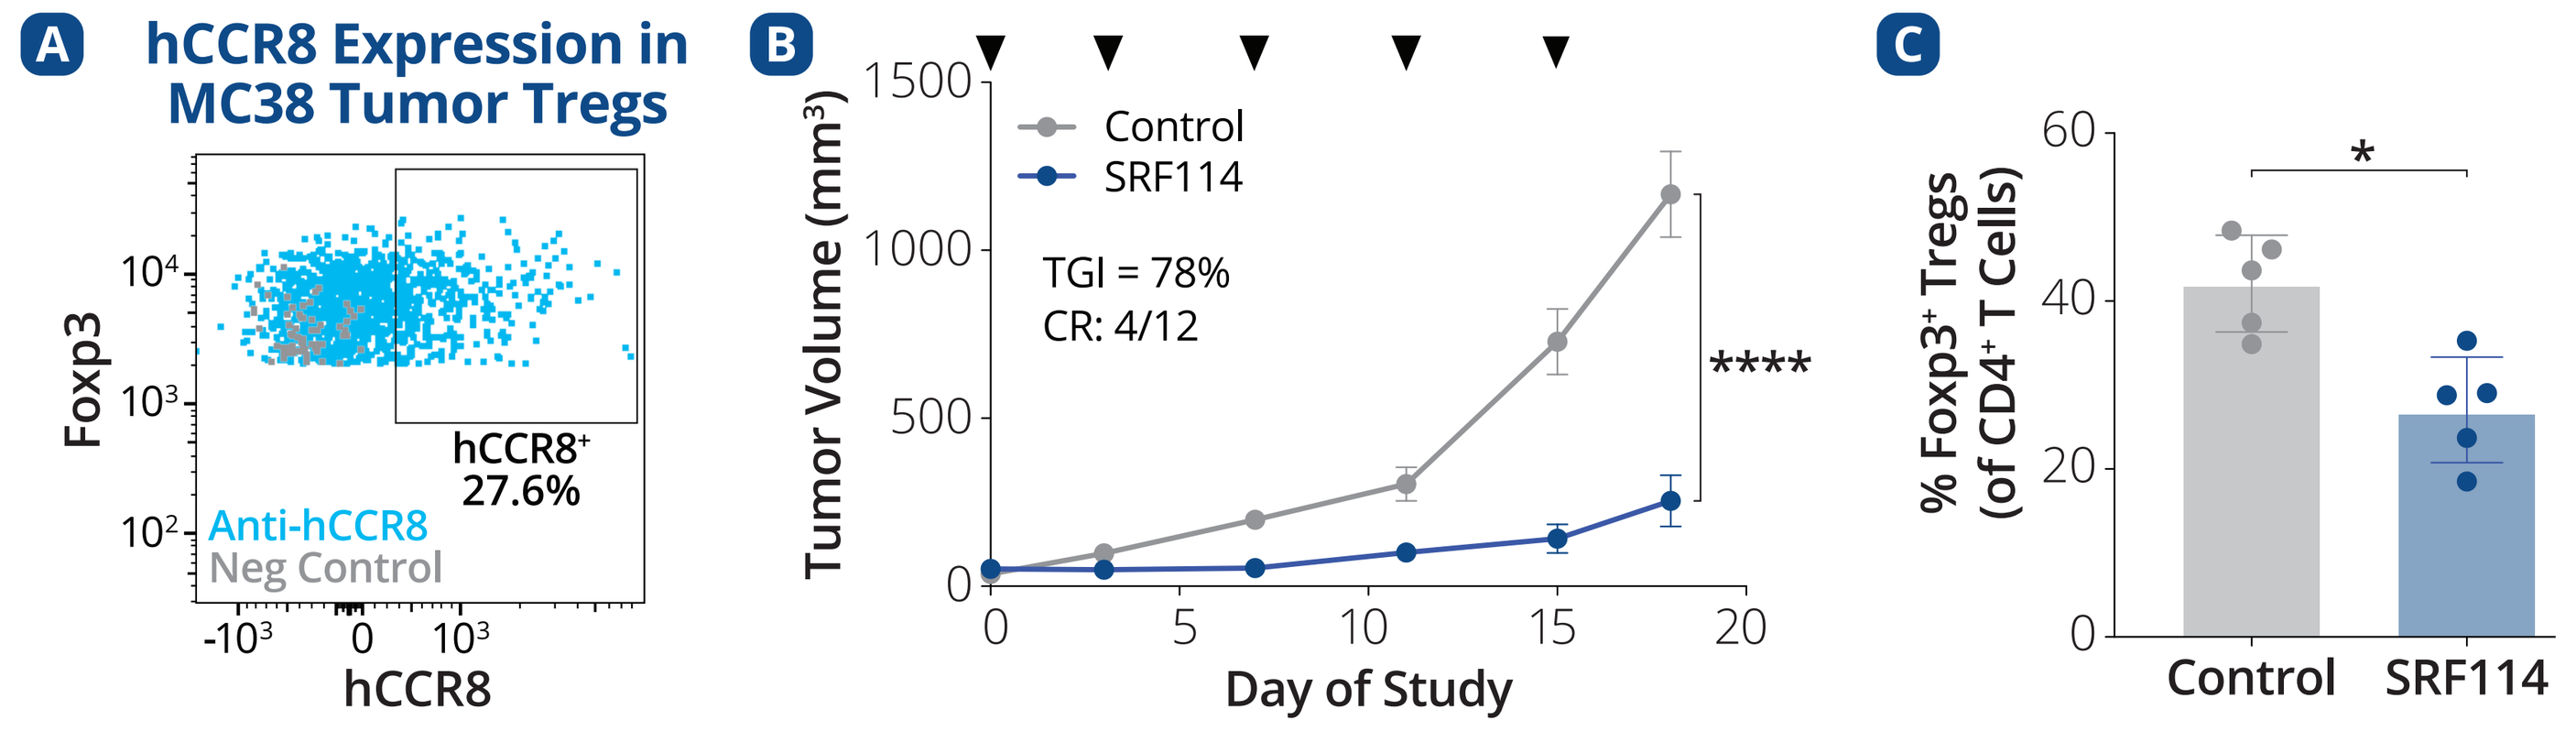

A
hCCR8 Expression in MC38 Tumor Tregs
Foxp3
hCCR8
hCCR8⁺ 27.6%
Anti-hCCR8
Neg Control
B
Tumor Volume (mm³)
Control
SRF114
TGI = 78%
CR: 4/12
Day of Study
C
% Foxp3⁺ Tregs (of CD4⁺ T Cells)
Control
SRF114

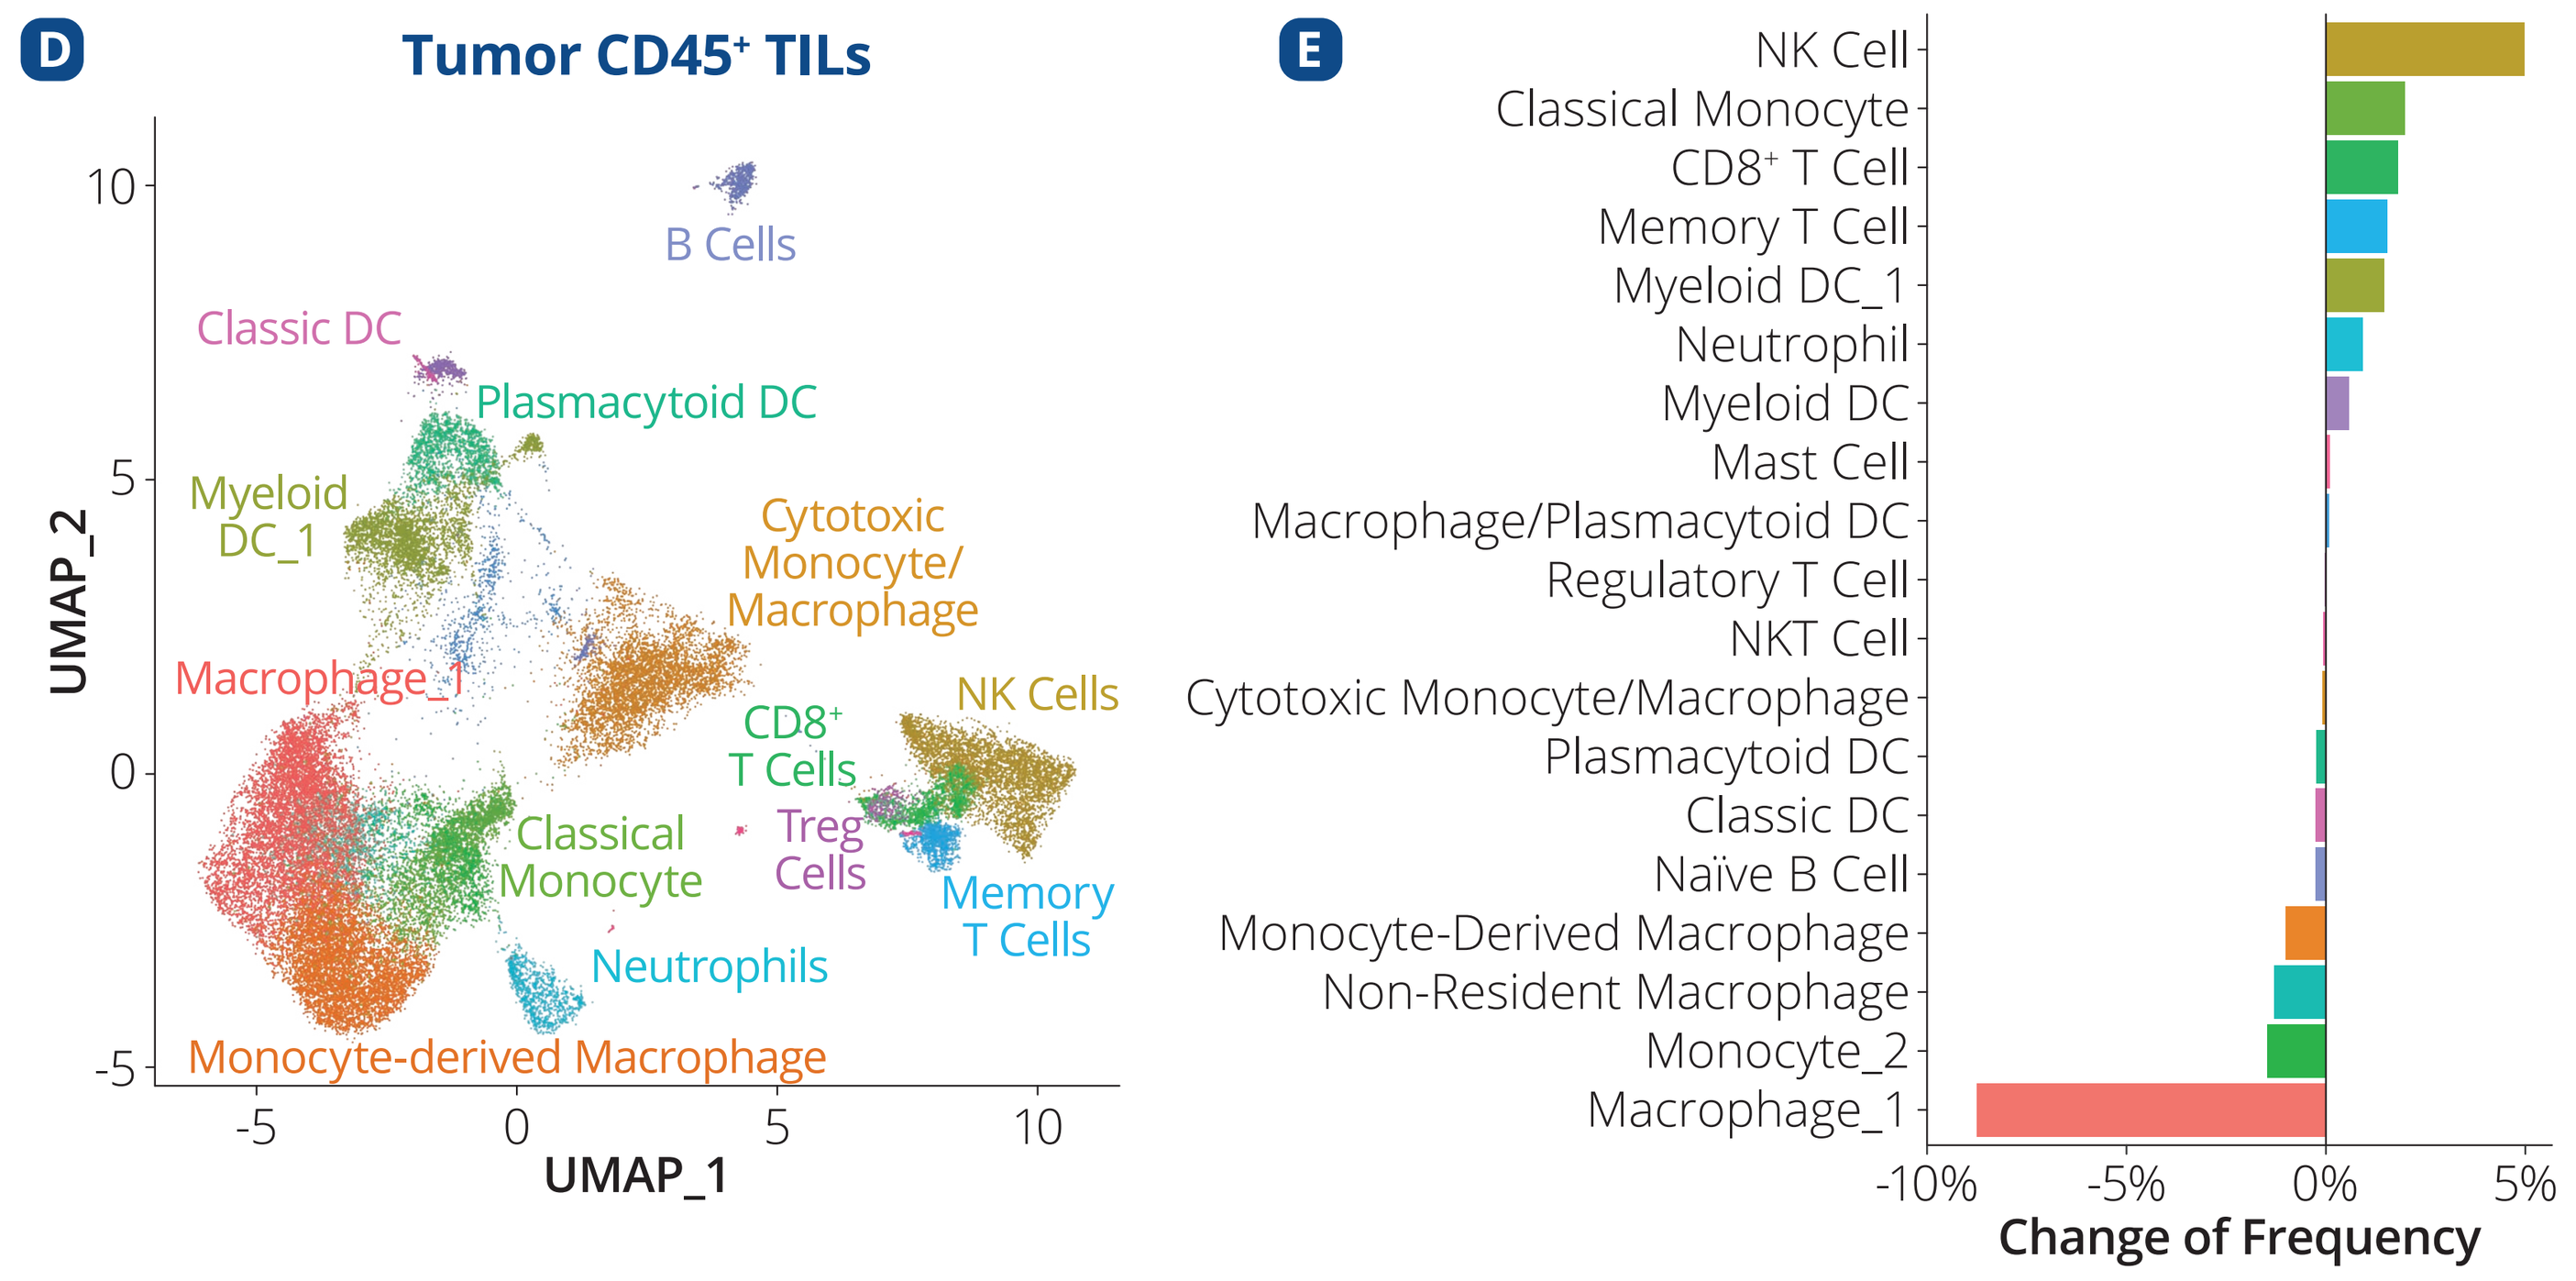

D
Tumor CD45⁺ TILs
UMAP_1
UMAP_2
B Cells
Classic DC
Plasmacytoid DC
Myeloid DC_1
Cytotoxic Monocyte/ Macrophage
Macrophage_1
CD8⁺ T Cells
NK Cells
Classical Monocyte
Treg Cells
Memory T Cells
Neutrophils
Monocyte-derived Macrophage
E
NK Cell
Classical Monocyte
CD8⁺ T Cell
Memory T Cell
Myeloid DC_1
Neutrophil
Myeloid DC
Mast Cell
Macrophage/Plasmacytoid DC
Regulatory T Cell
NKT Cell
Cytotoxic Monocyte/Macrophage
Plasmacytoid DC
Classic DC
Naïve B Cell
Monocyte-Derived Macrophage
Non-Resident Macrophage
Monocyte_2
Macrophage_1
Change of Frequency

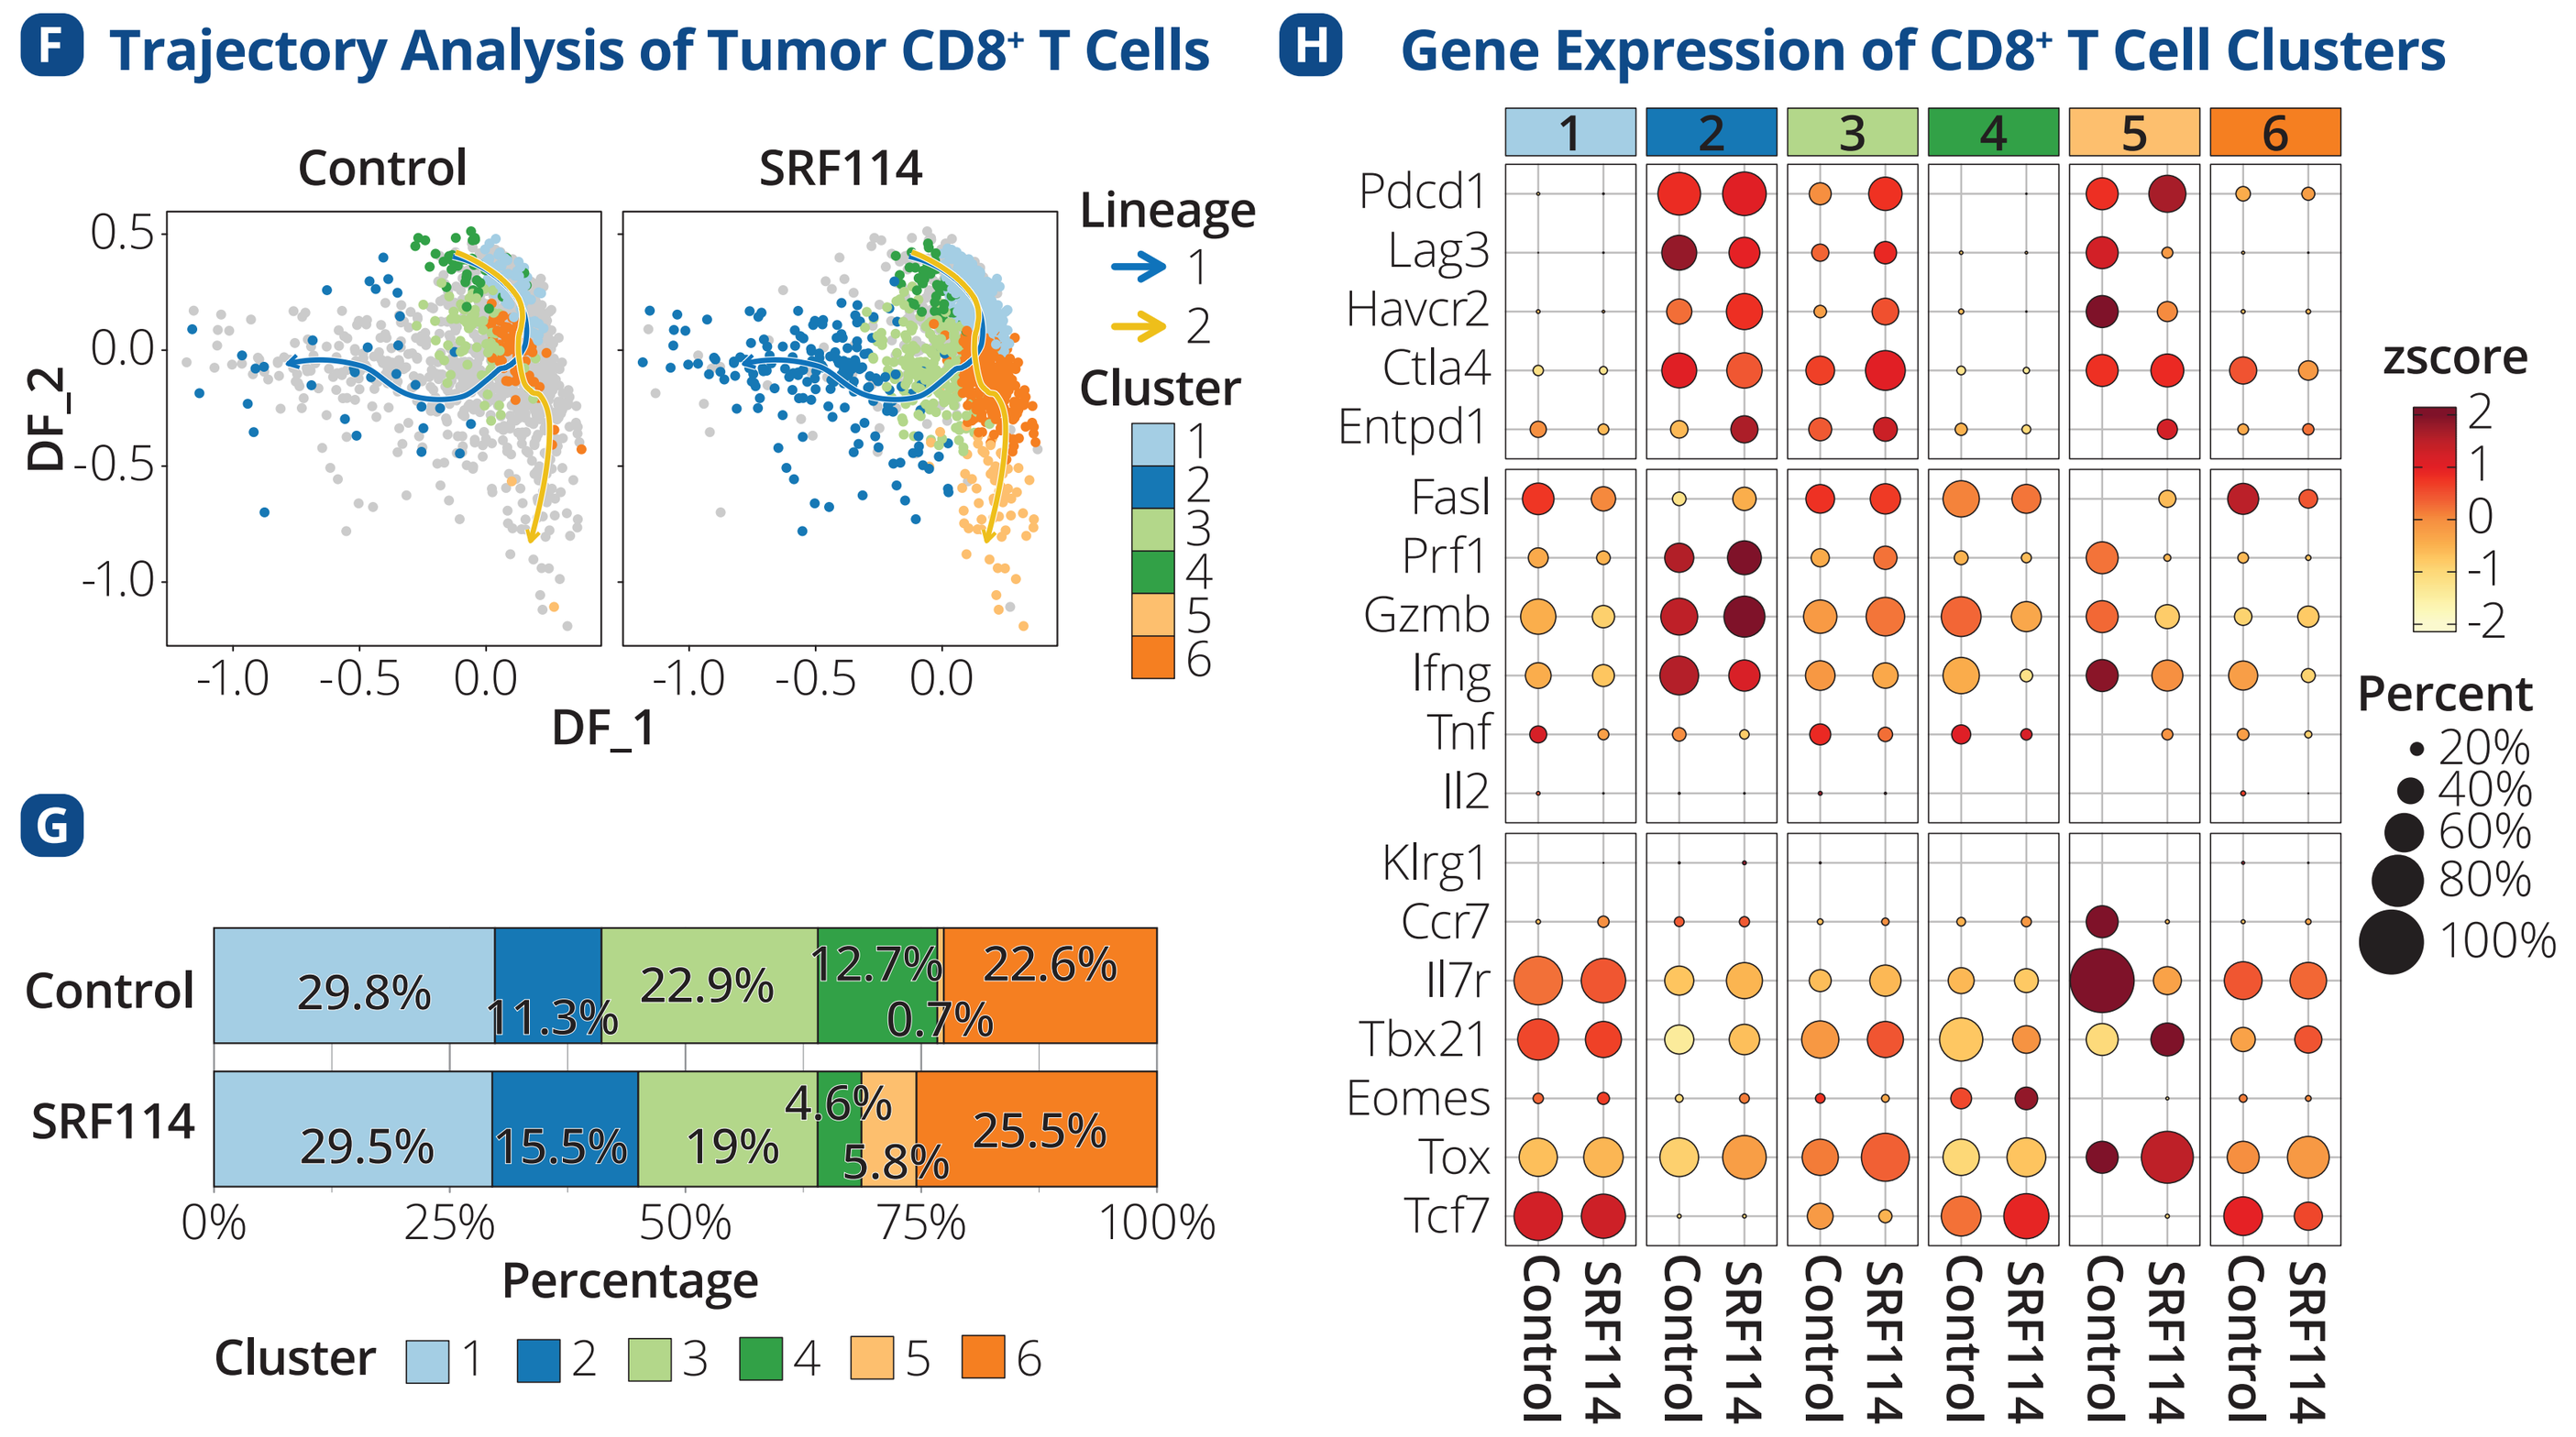

F Trajectory Analysis of Tumor CD8⁺ T Cells
Control
SRF114
DF_1
DF_2
Lineage
Cluster
G
Control
SRF114
29.8%
11.3%
22.9%
12.7%
0.7%
22.6%
29.5%
15.5%
19%
4.6%
5.8%
25.5%
Percentage
Cluster
H Gene Expression of CD8⁺ T Cell Clusters
Pdcd1
Lag3
Havcr2
Ctla4
Entpd1
Fasl
Prf1
Gzmb
Ifng
Tnf
Il2
Klrg1
Ccr7
Il7r
Tbx21
Eomes
Tox
Tcf7
zscore
Percent

A) Representative graph of hCCR8 expression in tumor Tregs of MC38-bearing human CCR8 knock-in (hCCR8KI) mice. **B)** Efficacy study evaluating anti-tumor activity of SRF114 in MC38-bearing hCCR8KI mice. **C)** Frequency of tumor Tregs on Day 4 of study. **D-H)** Single-cell RNA-seq analysis of MC38 TILs. **D)** UMAP clustering of 35,016 cells defined by 19 immune cell populations. **E)** Change of relative frequency of immune cells after treatment with SRF114. **F)** Pseudo-time trajectory analysis of six clusters of all tumor CD8⁺ T cells (gray) and highlighting cells of control (292 cells) and SRF114 (1098 cells) treatment conditions. **G)** Percent frequency of CD8⁺ T cell clusters after each treatment condition. **H)** Dot heatmap showing activation-induced inhibitory receptors, effector function, and transcription factor genes within each CD8⁺ T cell cluster per treatment condition.

A) MC38 tumors were collected on Day 10 after implantation and hCCR8 expression was detected on Foxp3⁺ Treg cells by flow cytometry. B) Tumor size was measured over time after mice were dosed with SRF114 or control antibodies (n = 10). C) MC38 tumors were dissociated and Treg cell frequency was quantified by flow cytometry (n = 5). D-H) CD45⁺ TILs were isolated on Day 6 after treatment with SRF114 or control antibodies and transcriptome analysis was performed using 10X Genomics (n = 2-3 replicates, 5 pooled tumors per replicate).

Conclusions

Mechanism of Action: SRF114 or Anti-CCR8 therapies Promote Depletion of Tumor CCR8⁺ Tregs and Reshape the TME Towards a Pro-inflammatory phenotype

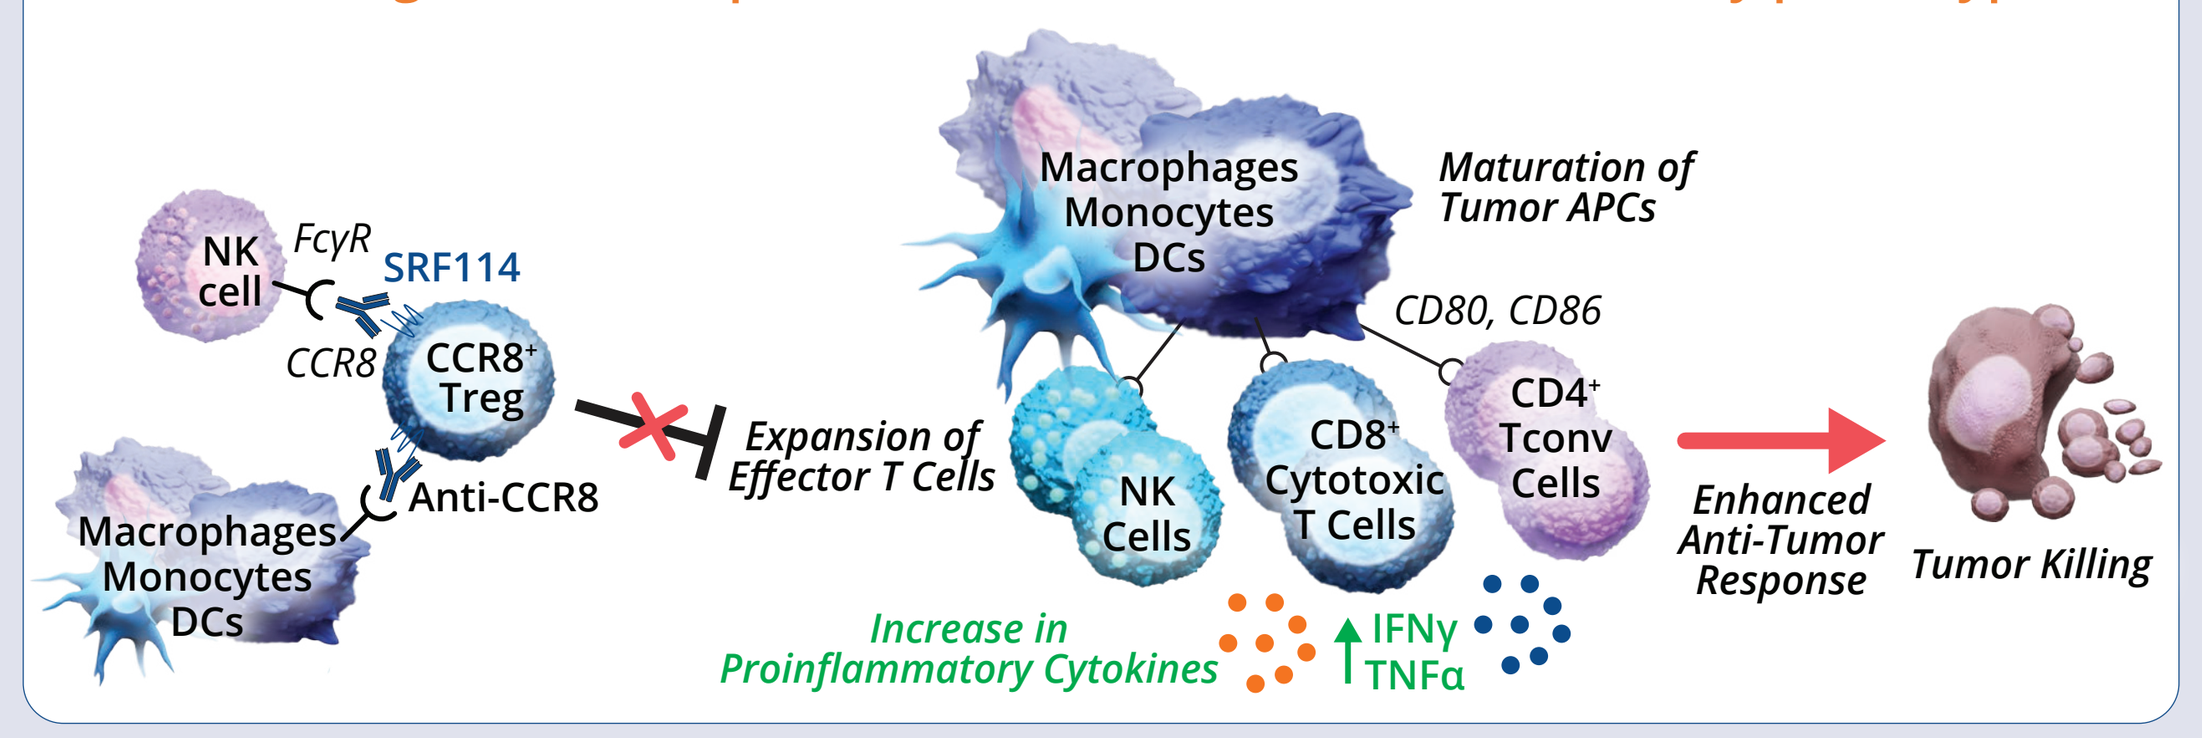

NK cell
FcγR
SRF114
CCR8
CCR8⁺ Treg
Anti-CCR8
Macrophages Monocytes DCs
Expansion of Effector T Cells
Maturation of Tumor APCs
CD80, CD86
NK Cells
CD8⁺ Cytotoxic T Cells
CD4⁺ Tconv Cells
Increase in Proinflammatory Cytokines
IFNγ
TNFα
Enhanced Anti-Tumor Response
Tumor Killing

- Anti-CCR8 therapy depletes a subset of suppressive tumor Tregs, resulting in robust anti-tumor activity in multiple tumor models, including checkpoint inhibitor-resistant tumors.
- Anti-CCR8 therapy promotes expansion of tumor cytotoxic T cells and NK cells, and remodeling and maturation of the myeloid compartment.
- Anti-CCR8 and anti-PD-1 combination therapy improves overall survival in a checkpoint inhibitor-resistant melanoma model by promoting expansion of effector T cells and increasing pro-inflammatory cytokines and chemokines in the TME.
- SRF114, a fully human antibody targeting hCCR8, confers robust anti-tumor activity and reshapes the TME towards a proinflammatory state in hCCR8KI mice. SRF114 is currently being evaluated in Phase 1 clinical trials (NCT05635643).

Presented at AACR 2023