Casdozokitug (casdozo, CHS-388), a first-in-class IL-27 targeting antibody, as monotherapy or in combination with pembrolizumab (pembro) in treatment-refractory non-small cell lung cancer (NSCLC)

FPN 221

Thomas U. Marron,[1] Aung Naing,[2] Charlene Mantia,[3] Nathan A. Pennell,[4] Hye Ryun Kim,[5] Liza Villaruz,[6] Anthony B. El-Khoueiry,[7] David E. Gerber,[8] Angel Qin,[9] Tae-Yong Kim,[10] Mehmet Altan,[2] Leena Gandhi,[3] Leonard J. Appleman,[6] Shinkyo Yoon,[11] Jonathan Hill,[12,13] Vienna Reichert,[12,13] Ricard Masia,[12,13] Lauren C. Harshman,[12] Amita Patnaik,[14] Daniel Morgensztern[15]

[1]The Mount Sinai Hospital, Icahn School of Medicine at Mount Sinai, New York, NY, USA; [2]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [3]Dana-Farber Cancer Institute, Boston, MA, USA; [4]Cleveland Clinic Taussig Cancer Institute, Cleveland, Ohio, USA; [5]Yonsei Cancer Center, Yonsei University Health System, Seoul, Republic of Korea; [6]University of Pittsburgh Medical Center, Pittsburgh, PA, USA; [7]Norris Comprehensive Cancer Center, University of Southern California, Los Angeles, CA, USA; [8]Harold C. Simmons Comprehensive Cancer Center, University of Texas Southwestern Medical Center, Dallas, TX, USA; [9]University of Michigan Rogel Cancer Center, Ann Arbor, MI, USA; [10]Seoul National University College of Medicine, Seoul National University Hospital, Seoul, Republic of Korea; [11]Asan Medical Center, Republic of Korea; [12]Surface Oncology (acquired by Coherus Biosciences), Cambridge, MA, USA; [13]Coherus Biosciences, Redwood City, CA, USA; [14]START San Antonio, San Antonio, TX, USA; [15]Washington University School of Medicine, St. Louis, MO, USA

Copies of this poster obtained through QR codes are for personal use only and may not be reproduced without written permission of the authors.

BACKGROUND

- IL-27 is a heterodimeric cytokine expressed by myeloid cells, including macrophages and dendritic cells, which plays a role in modulating immune responses during infection and tumor immune surveillance
- IL-27 regulates the activity of several immune cell types through upregulation of immune suppressive receptors (PD-L1, TIGIT, LAG3) and inhibition of inflammatory cytokines
- Blocking IL-27 with CHS-388 (casdozokitug or casdozo; formerly SRF388), an anti-IL27 antibody, in clinical trials has led to monotherapy tumor growth inhibition and partial responses in patients with non-small cell lung cancer (NSCLC) and renal cell carcinoma (RCC) (NCT04374877) and ongoing trials are studying combinations with PD-1/PD-L1 pathway blockade in patients with NSCLC and hepatocellular carcinoma (HCC)
- Immune activation as evidenced by increases in serum IFNγ and NK cell gene activation in peripheral blood mononuclear cells (PBMCs) after CHS-388 administration in patients establishes proof of mechanism in patients with cancer
- SRF388-101 is a Phase 1b/2 dose escalation and expansion study of casdozo in advanced solid tumors
- As monotherapy, casdozo demonstrated favorable safety and antitumor activity[1,2]
- Expansion cohorts in NSCLC have evaluated casdozo 10 mg/kg IV q4w as monotherapy and q3w in combination with pembrolizumab in treatment-refractory NSCLC

IL-27 is a Cytokine Involved in Resolving Inflammation and Inhibiting Anti-Tumor Immune Responses

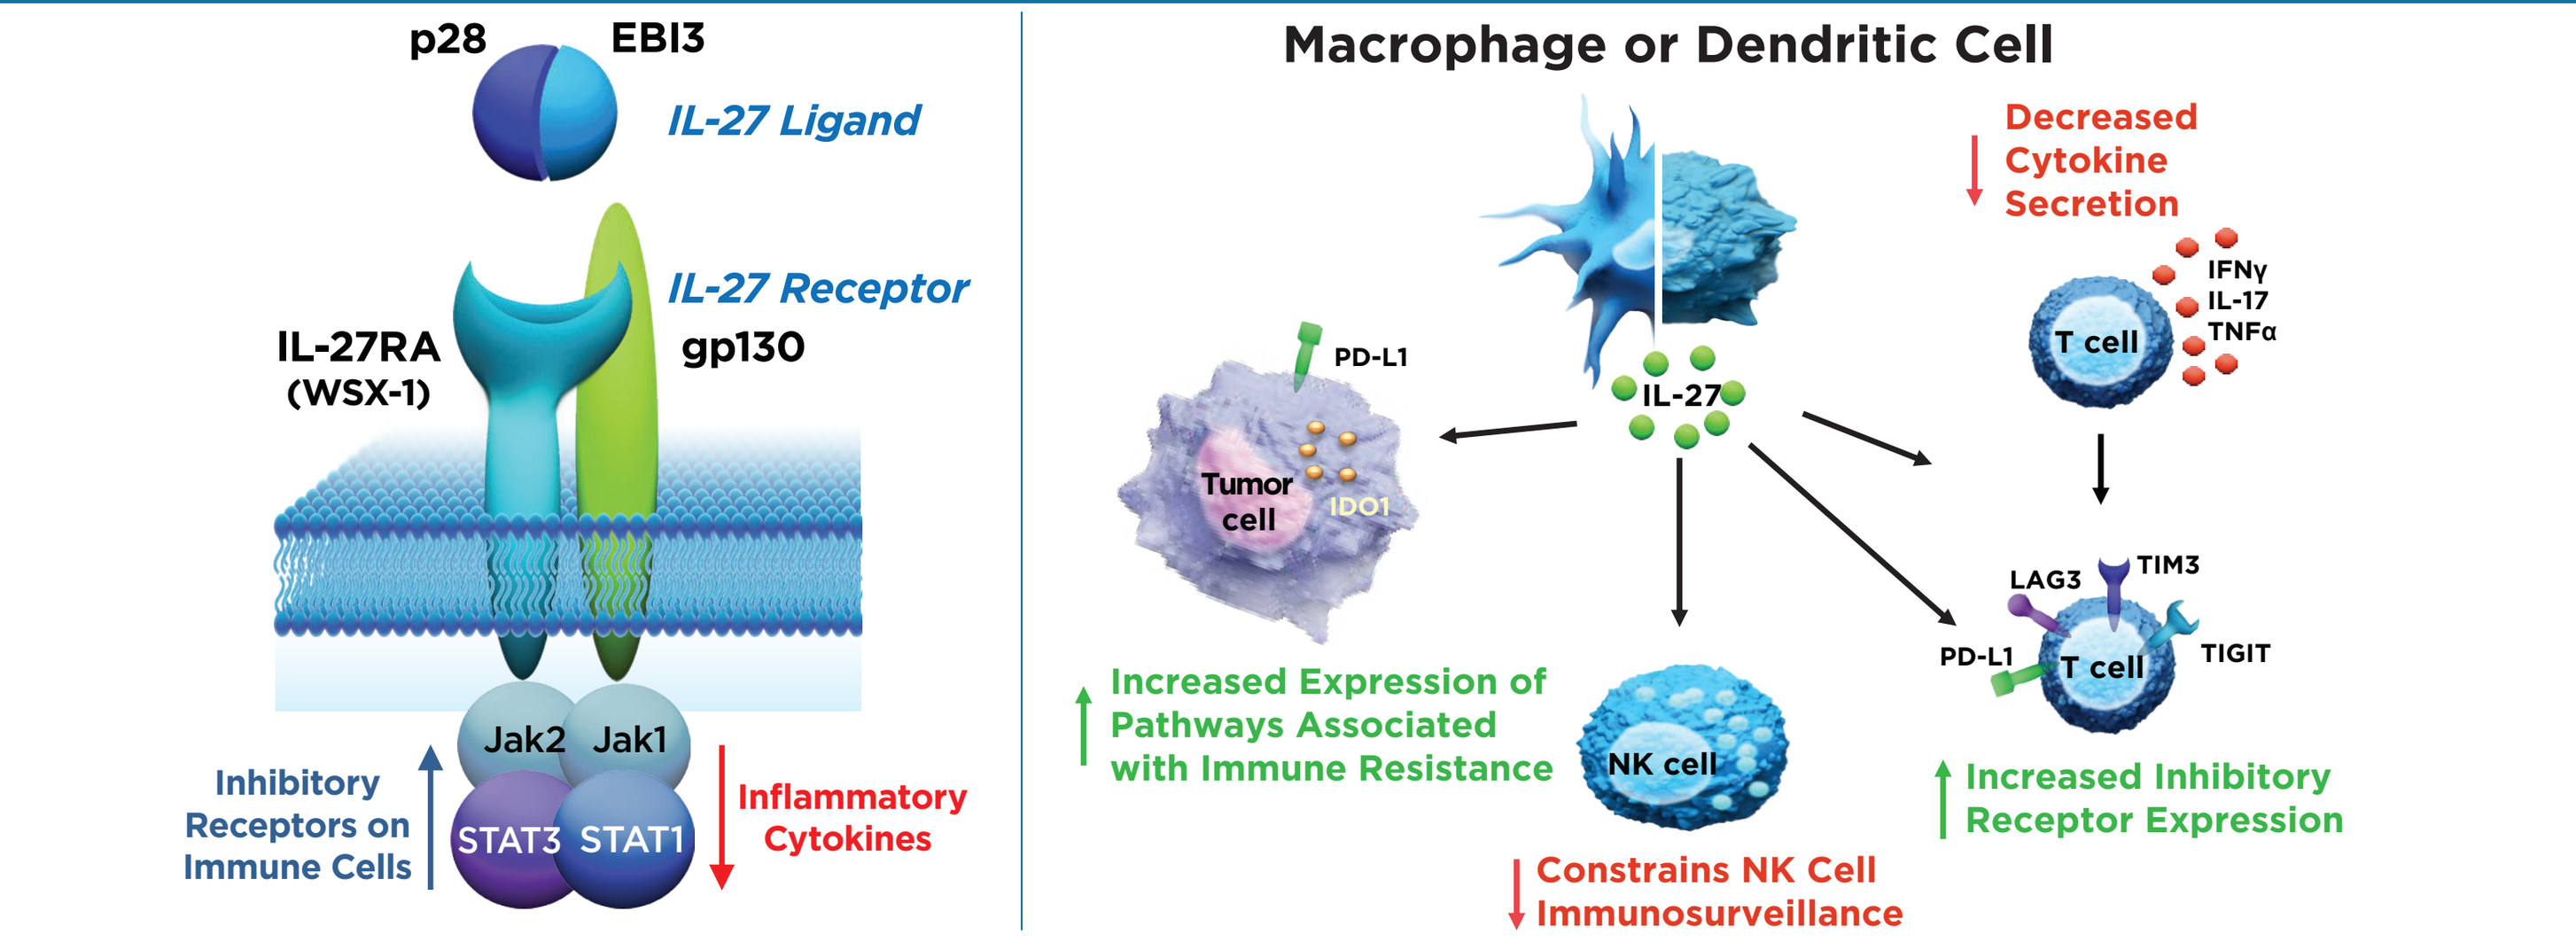
p28
EBI3
IL-27 Ligand
IL-27 Receptor
IL-27RA (WSX-1)
gp130
Jak2 Jak1
STAT3 STAT1
Inhibitory Receptors on Immune Cells
Inflammatory Cytokines
Macrophage or Dendritic Cell
Decreased Cytokine Secretion
IFNγ IL-17 TNFα
T cell
PD-L1
IL-27
Tumor cell
IDO1
Increased Expression of Pathways Associated with Immune Resistance
NK cell
Constrains NK Cell Immunosurveillance
LAG3
TIM3
TIGIT
Increased Inhibitory Receptor Expression

Villarino, et al. *Immunity*. 2003; Saur, et al. *J Immunol*. 2008; Carbotti, et al. *Oncotarget*. 2015; Chihara, et al. *Nature*. 2018; DeLong, et al. *Immunohorizons*. 2019; Aghayev, et al. *Cancer Discovery*. 2022

Biomarkers of Response to Casdozo in the Phase 1 Trial

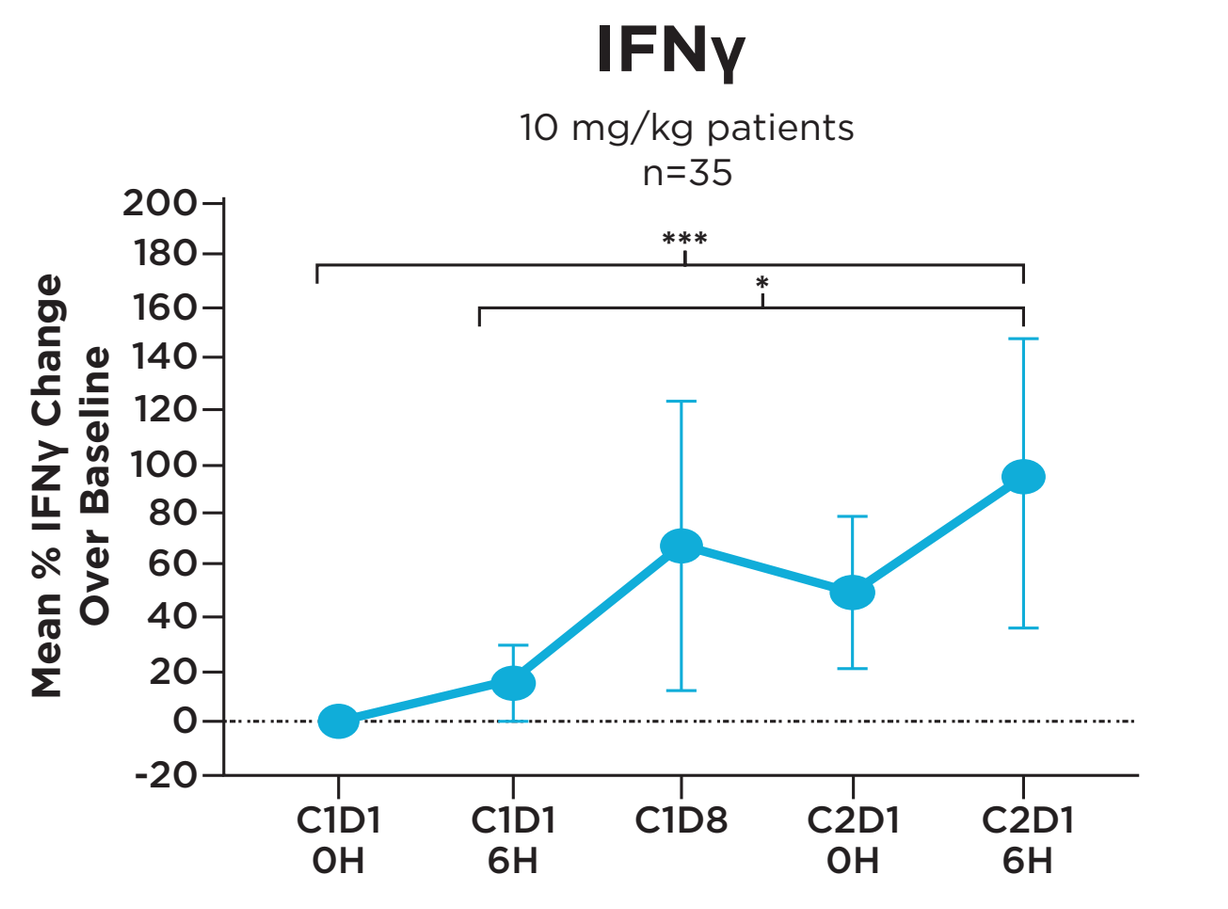
IFNγ
10 mg/kg patients
n=35
Mean % IFNγ Change Over Baseline
C1D1 0H
C1D1 6H
C1D8
C2D1 0H
C2D1 6H

*

***p<0.0001; *p<0.05

Relative change in IFNγ concentrations in serum relative to pre-dose levels. Serum IFNγ concentrations were determined by MSD.[2]

Increased Expression of NK Cell Activation Genes After Casdozo

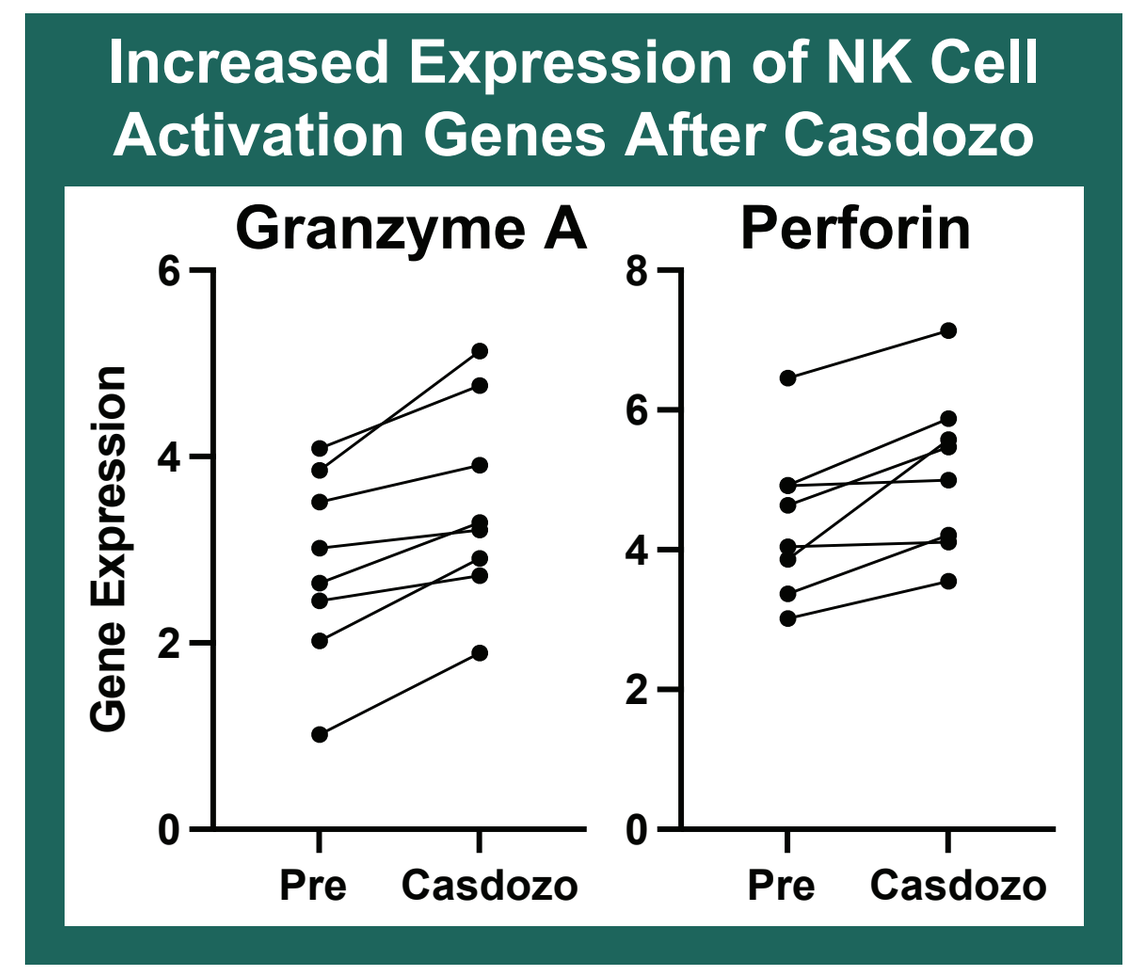
Granzyme A
Perforin
Gene Expression
Pre
Casdozo

PBMCs from pts that received casdozo at dose levels ranging from 3-20 mg/kg q4w were collected before (C1D1) or after (C1D8) treatment and processed for bulk RNA sequencing. Gene expression data highlight increased expression of Granzyme A and Perforin after treatment with casdozo.

METHODS

SRF388-101: Study Design – NSCLC

First in-human, open-label, monotherapy, dose escalation and dose expansion phase 1b/2 trial (NCT04374877)
Part A: Monotherapy dose escalation in advanced solid tumors (q4w): N=29
Part B: Monotherapy dose expansion in HCC, RCC, and NSCLC (10 mg/kg q4w): N=84
Part C: Casdozo + Pembro dose expansion in HCC, RCC, and NSCLC (10 mg/kg q3w) + Pembro (200 mg q3w): N=21

NSCLC Cohorts: Monotherapy (Dose Escalation and Expansion) and PD-1 Combo Expansion

Part A: Dose Escalation (N=29; NSCLC N=5)
Monotherapy in patients with advanced solid tumors
- Accelerated titration and standard 3+3 design
- Explored 8 dose levels ranging from 0.003 - 20 mg/kg q4w
- **NSCLC dose levels**: 1 mg/kg (N=1); 10 mg/kg (N=1); 20 mg/kg (N=3)

→ RP2D 10 mg/kg →

Part B: 2-5L NSCLC: Casdozo Monotherapy Expansion
Single Arm Simon 2-Stage Phase 2 (N=40)
Stage 1: n=17/17; Stage 2 n=23/23

Part C: 2-4L aPD-(L)1 R/R NSCLC: Casdozo + Pembro*
Single Arm Simon 2-Stage Phase 2 (N=6)
Stage 1: n=6/15; Stage 2 n=0/25

Part D: 2-4L aPD-(L)1 R/R NSCLC: Casdozo + Toripalimab (aPD-1 mAb) *(enrolling)*
Single Arm Simon 2-Stage Phase 2 (N=40)

- Primary endpoint: objective response rate (ORR) based on investigator review per RECIST v1.1
- Key secondary endpoints:
 - Safety and tolerability of casdozo as monotherapy or in combination with an anti-PD1 antibody based on rate of AEs and safety laboratory values
 - Additional measures of efficacy including duration of response (DoR) and disease control rate (DCR) based on investigator review per RECIST v1.1

*The casdozo + pembro cohort was halted early due to changes in company objectives. Casdozo is currently being studied with other checkpoint inhibitors.

RESULTS

SRF388-101 NSCLC Dose Escalation, Expansion and Combination: Time on Therapy

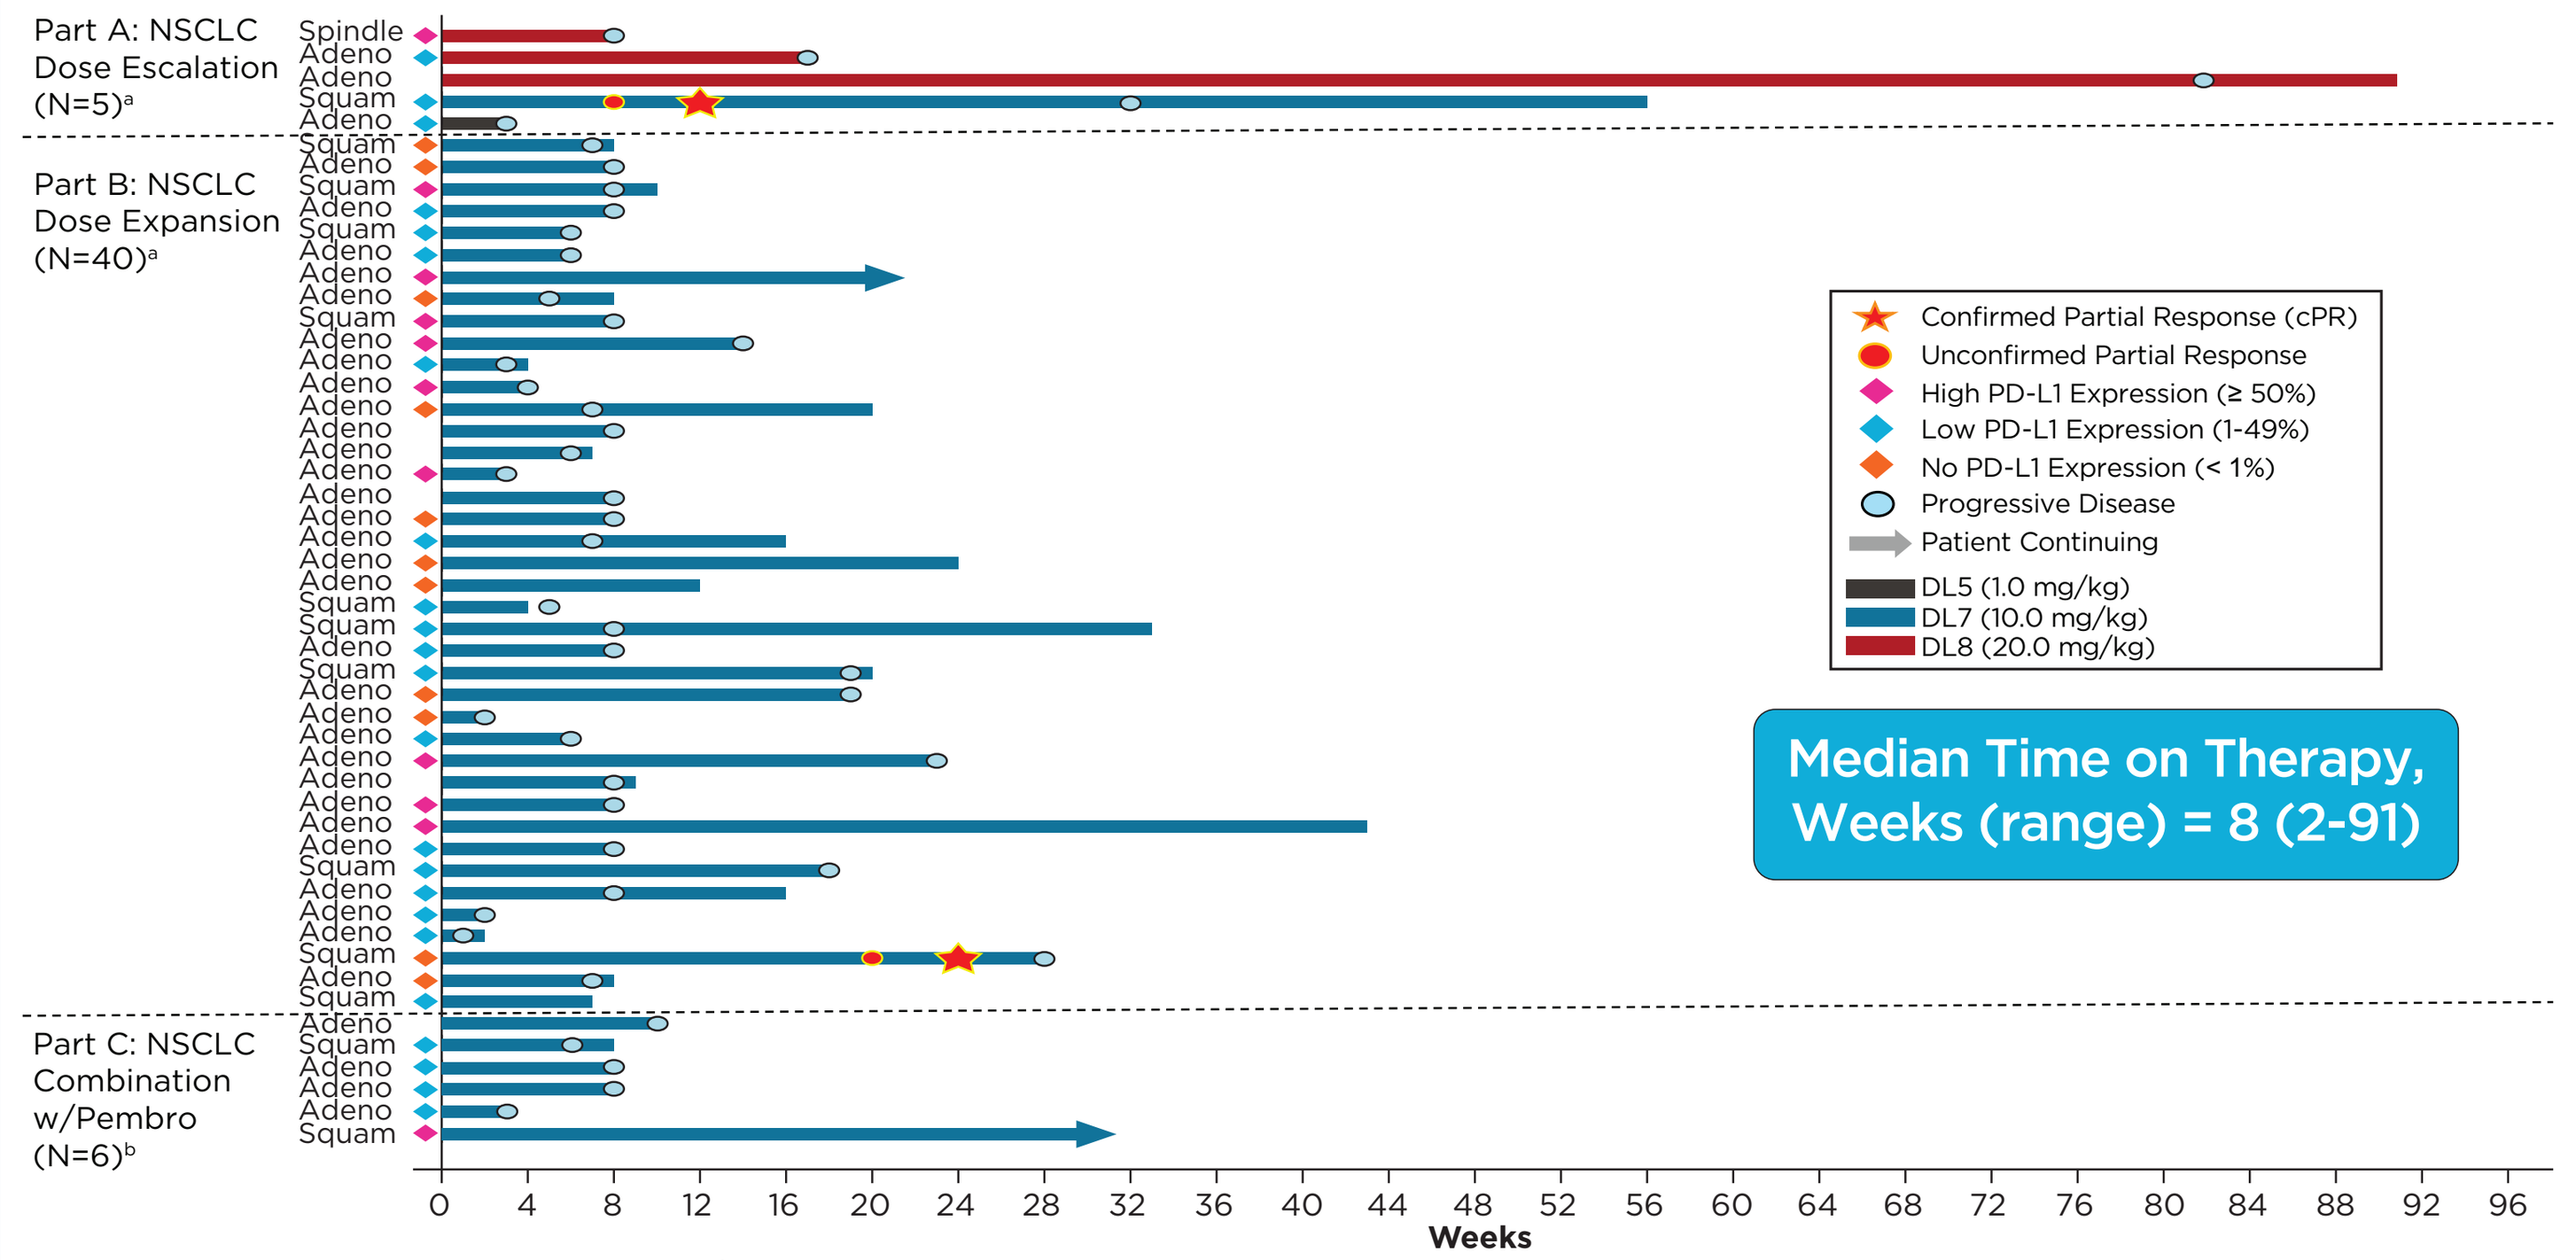
Part A: NSCLC Dose Escalation (N=5)a
Part B: NSCLC Dose Expansion (N=40)a
Part C: NSCLC Combination w/Pembro (N=6)b
Weeks
Confirmed Partial Response (cPR)
Unconfirmed Partial Response
High PD-L1 Expression (≥ 50%)
Low PD-L1 Expression (1-49%)
No PD-L1 Expression (< 1%)
Progressive Disease
Patient Continuing
DL5 (1.0 mg/kg)
DL7 (10.0 mg/kg)
DL8 (20.0 mg/kg)
Median Time on Therapy, Weeks (range) = 8 (2-91)

[a]Parts A and B patients received casdozo 10 mg/kg q4w; disease assessment performed at 8 weeks and then every 12 weeks; [b]Part C patients received casdozo 10 mg/kg and pembrolizumab 200 mg IV q3w; disease assessment performed at C4D1 and then every 9 weeks thereafter

Data cut as of 21 Sep 2023, subject to change

NSCLC Partial Responder Displayed Histological Features of an Immune Excluded Tumor Microenvironment and Prominent IL-27 Staining

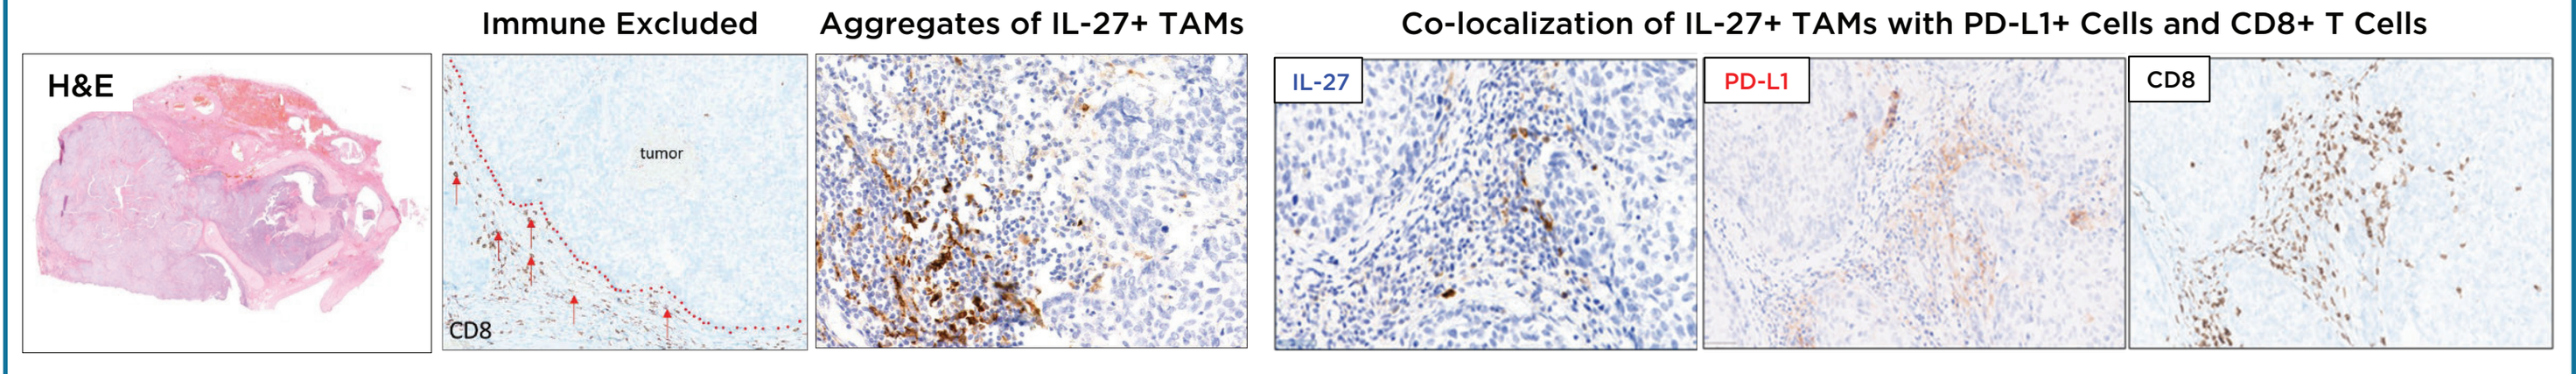
H&E
Immune Excluded
Aggregates of IL-27+ TAMs
Co-localization of IL-27+ TAMs with PD-L1+ Cells and CD8+ T Cells
tumor
CD8
IL-27
PD-L1
CD8

Archival tissue (pneumonectomy) from a patient with Stage IIA squamous cell carcinoma (PD-L1 low) who experienced a partial response showed an immune excluded phenotype by CD8 staining and a high density of peritumoral IL-27+ macrophages.

RESULTS

Monotherapy Anti-Tumor Activity in NSCLC (Dose Escalation & Expansion)

Two confirmed monotherapy PRs in αPD-(L)1-experienced patients with squamous cell histology

Best Percent Change from Baseline in Sum of Target Lesions (n=38)

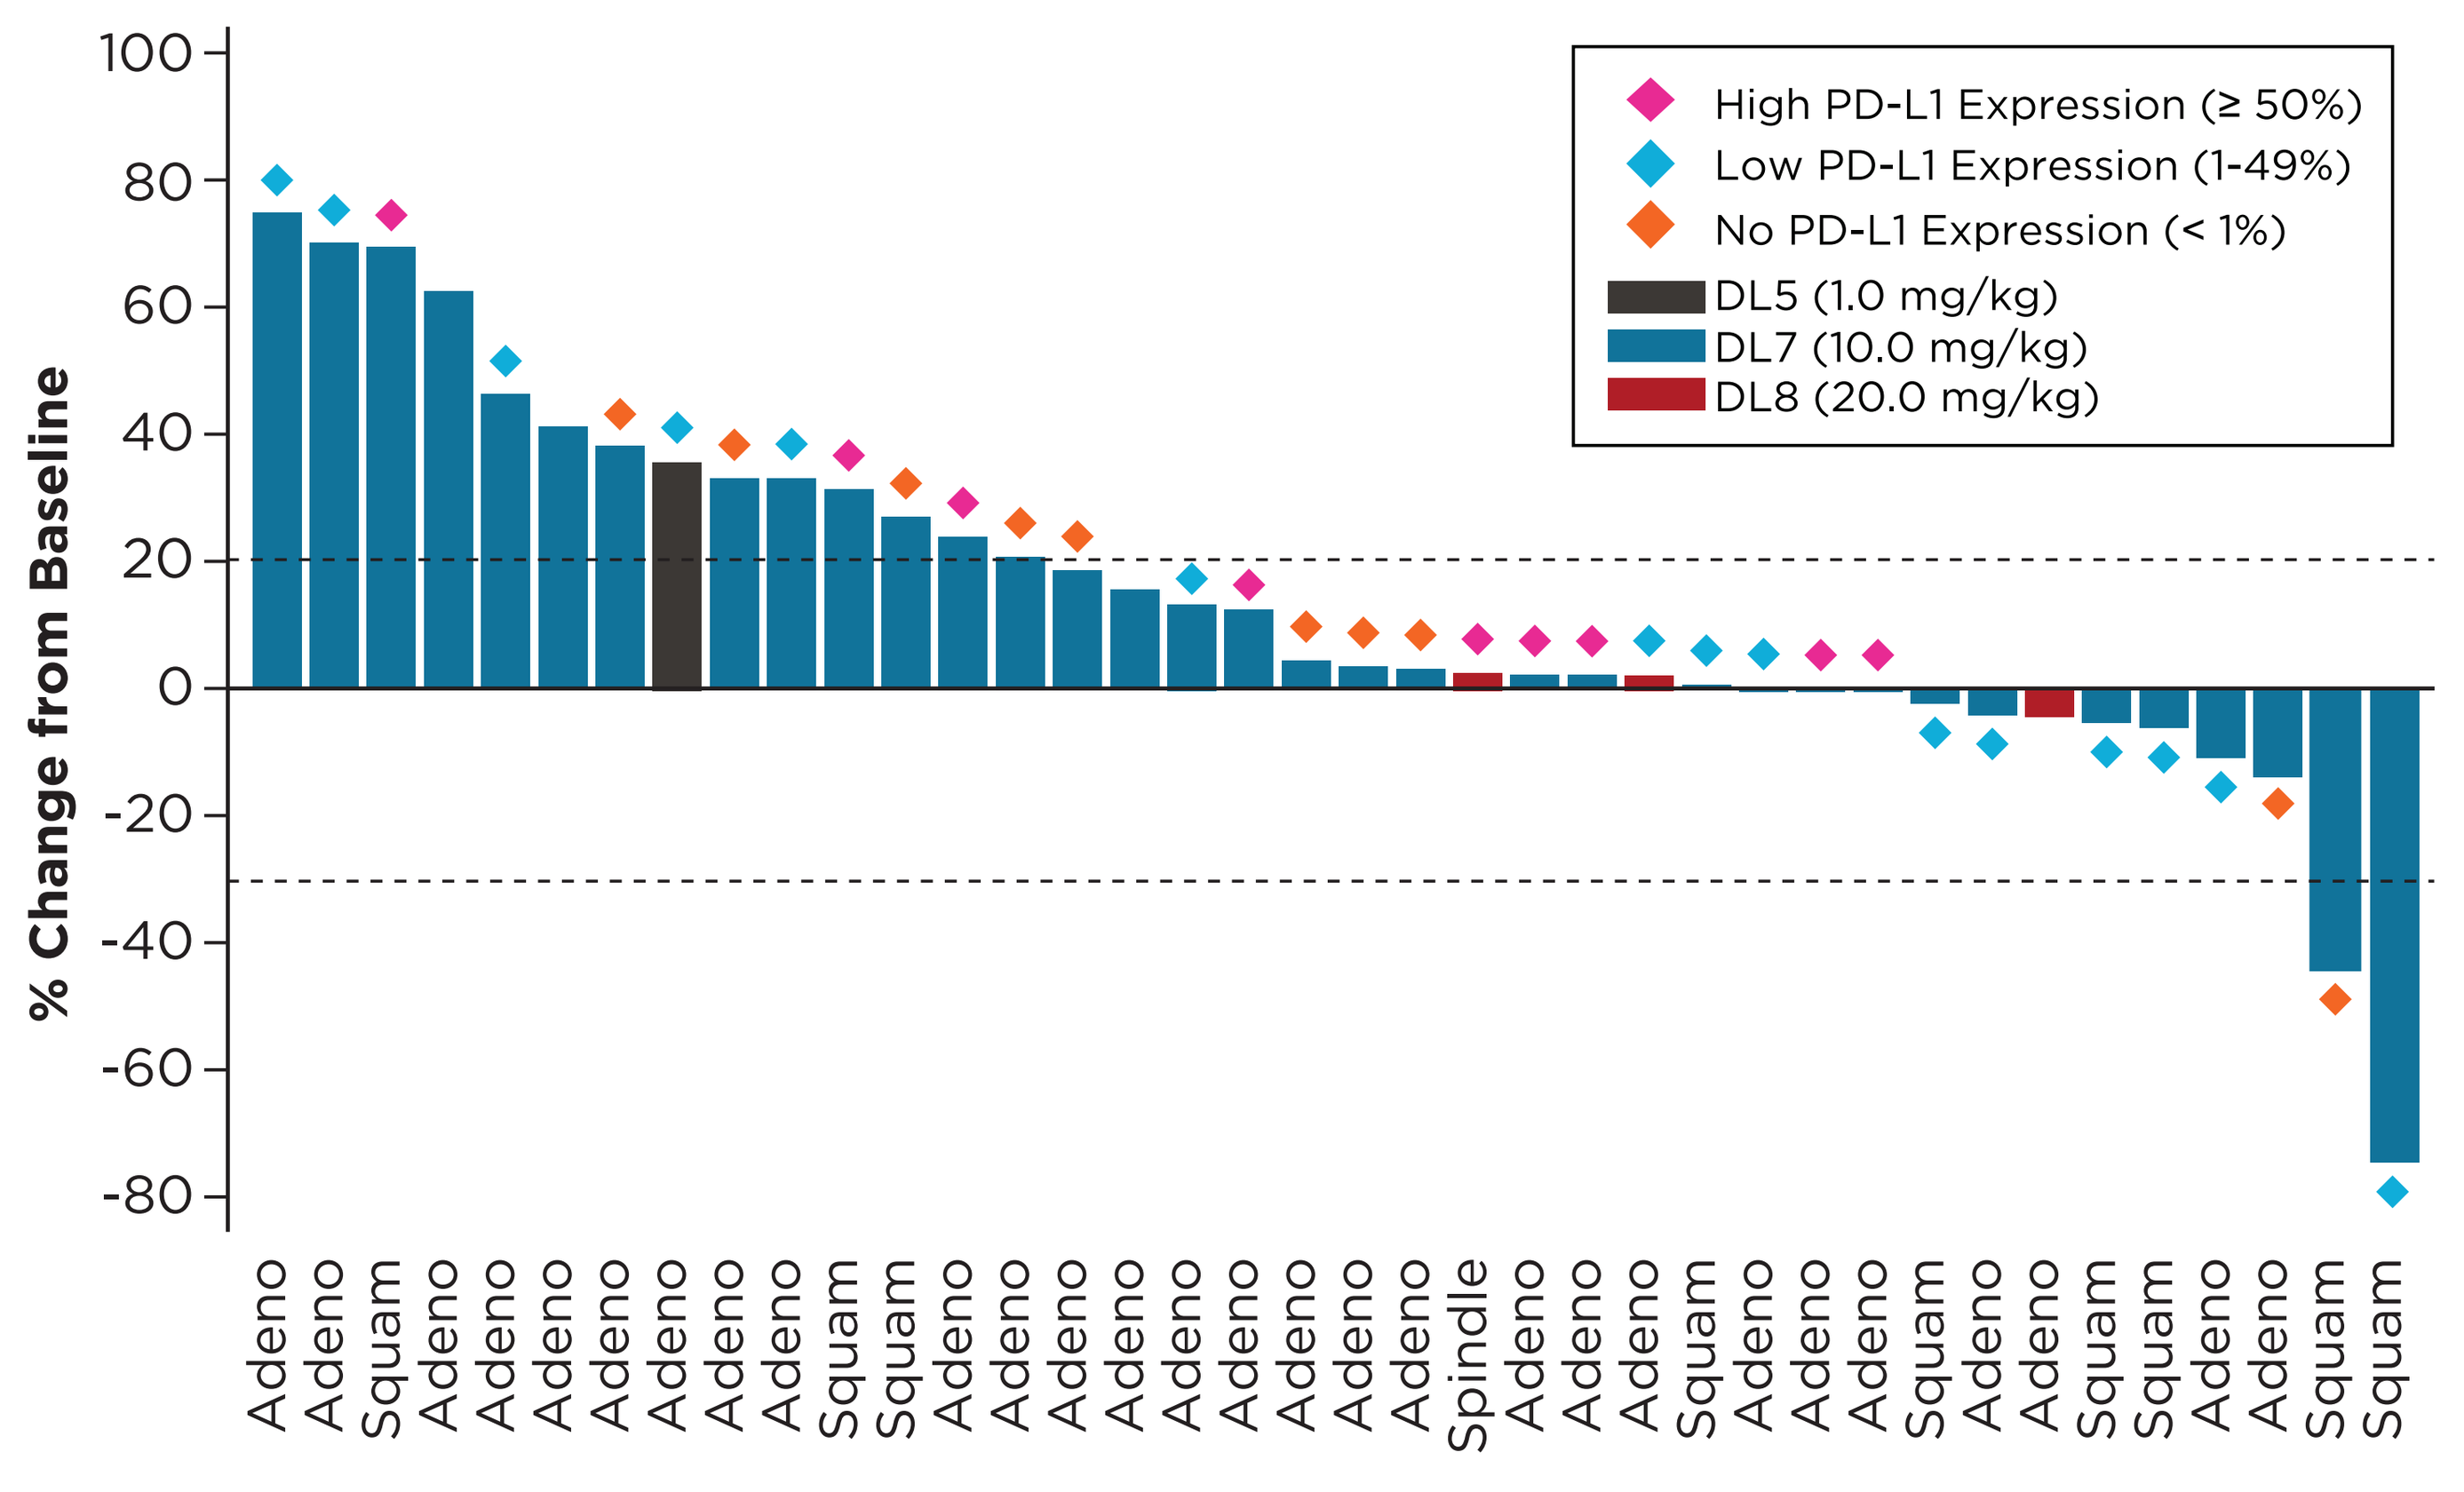
% Change from Baseline
High PD-L1 Expression (≥ 50%)
Low PD-L1 Expression (1-49%)
No PD-L1 Expression (< 1%)
DL5 (1.0 mg/kg)
DL7 (10.0 mg/kg)
DL8 (20.0 mg/kg)

Target Lesion Change Over Time (n=38)

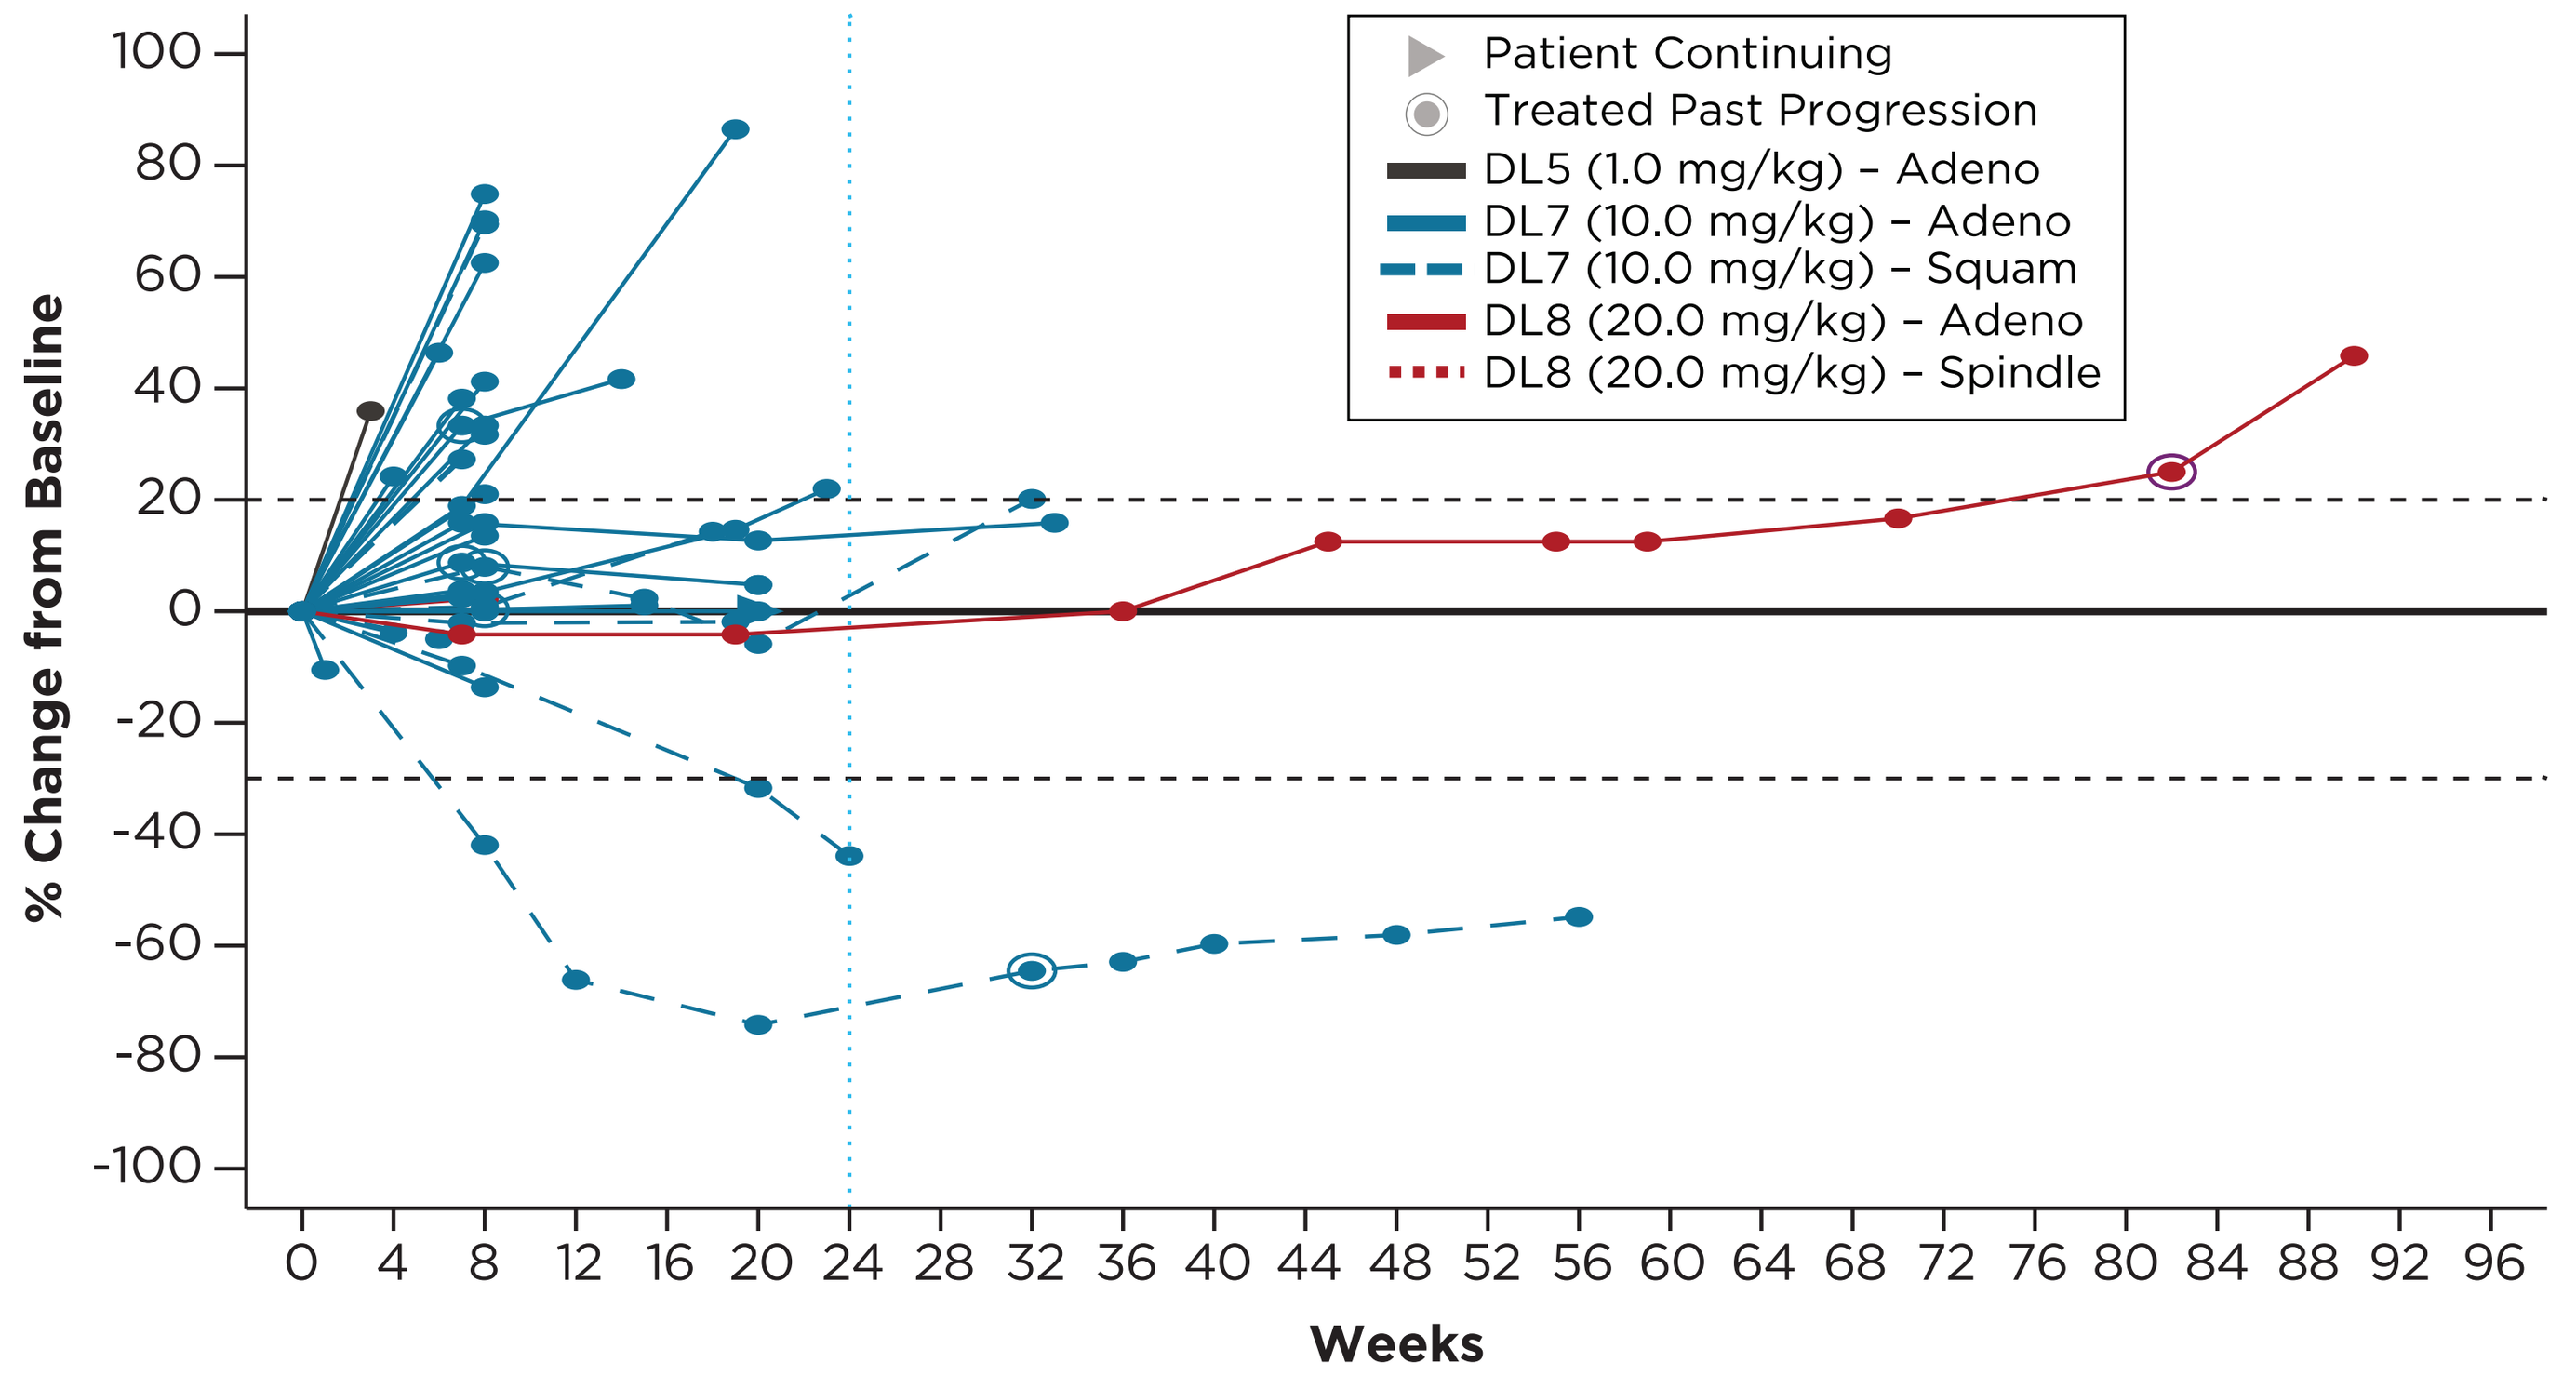
% Change from Baseline
Weeks
Patient Continuing
Treated Past Progression
DL5 (1.0 mg/kg) – Adeno
DL7 (10.0 mg/kg) – Adeno
DL7 (10.0 mg/kg) – Squam
DL8 (20.0 mg/kg) – Adeno
DL8 (20.0 mg/kg) – Spindle

- 2 confirmed PRs in PD-L1 negative or low, squamous NSCLC and 1 durable disease stabilization in adenocarcinoma; all 3 previously treated with aPD-(L)1 antibodies
- 5% ORR in all evaluable patients
- 22% ORR in RECIST evaluable squamous subset (n=2/9)

Best Overall Response, n (%)	Response Evaluable (n=43)
Complete response	0
Confirmed partial response	2 (4.7)
Stable disease	13 (30.2)
Progressive disease	28 (65.1)

Data cut as of 21 Sep 2023, subject to change

SRF388-101 Safety Summary for All NSCLC Monotherapy and Combo Therapy

AE Summary	Casdozo NSCLC Monotherapy (N=45)	Casdozo + Pembro NSCLC Combo (N=6)
Treatment Emergent Adverse Event (TEAE), n (%)	41 (91.1)	5 (83.3)
Treatment-related AE, n (%)	19 (42.2)	5 (83.3)
Grade ≥3 TEAE, n (%)	21 (46.7)	2 (33.3)
Grade ≥3 Treatment-related AE, n (%)	5 (11.1)	0
Serious Treatment Emergent Adverse Event (TESAE), n (%)	19 (42.2)	1 (16.7)
Treatment-related SAE, n (%)	3 (6.7)	0
TEAE Leading to Discontinuation, n (%)	8 (17.8)	0
Treatment-related AE leading to discontinuation, n (%)	2 (4.4)	0
TEAE Leading to Death, n (%)	6 (13.3)	0
Treatment-related AE Leading to Death, n (%)	1 (2.2)	0

SRF388-101 NSCLC TEAEs: Casdozo (N=45) and Combination with PD-1 Inhibitor (N=6)

Majority of TEAEs consistent with advanced disease; low rate of grade ≥ 3 TEAEs

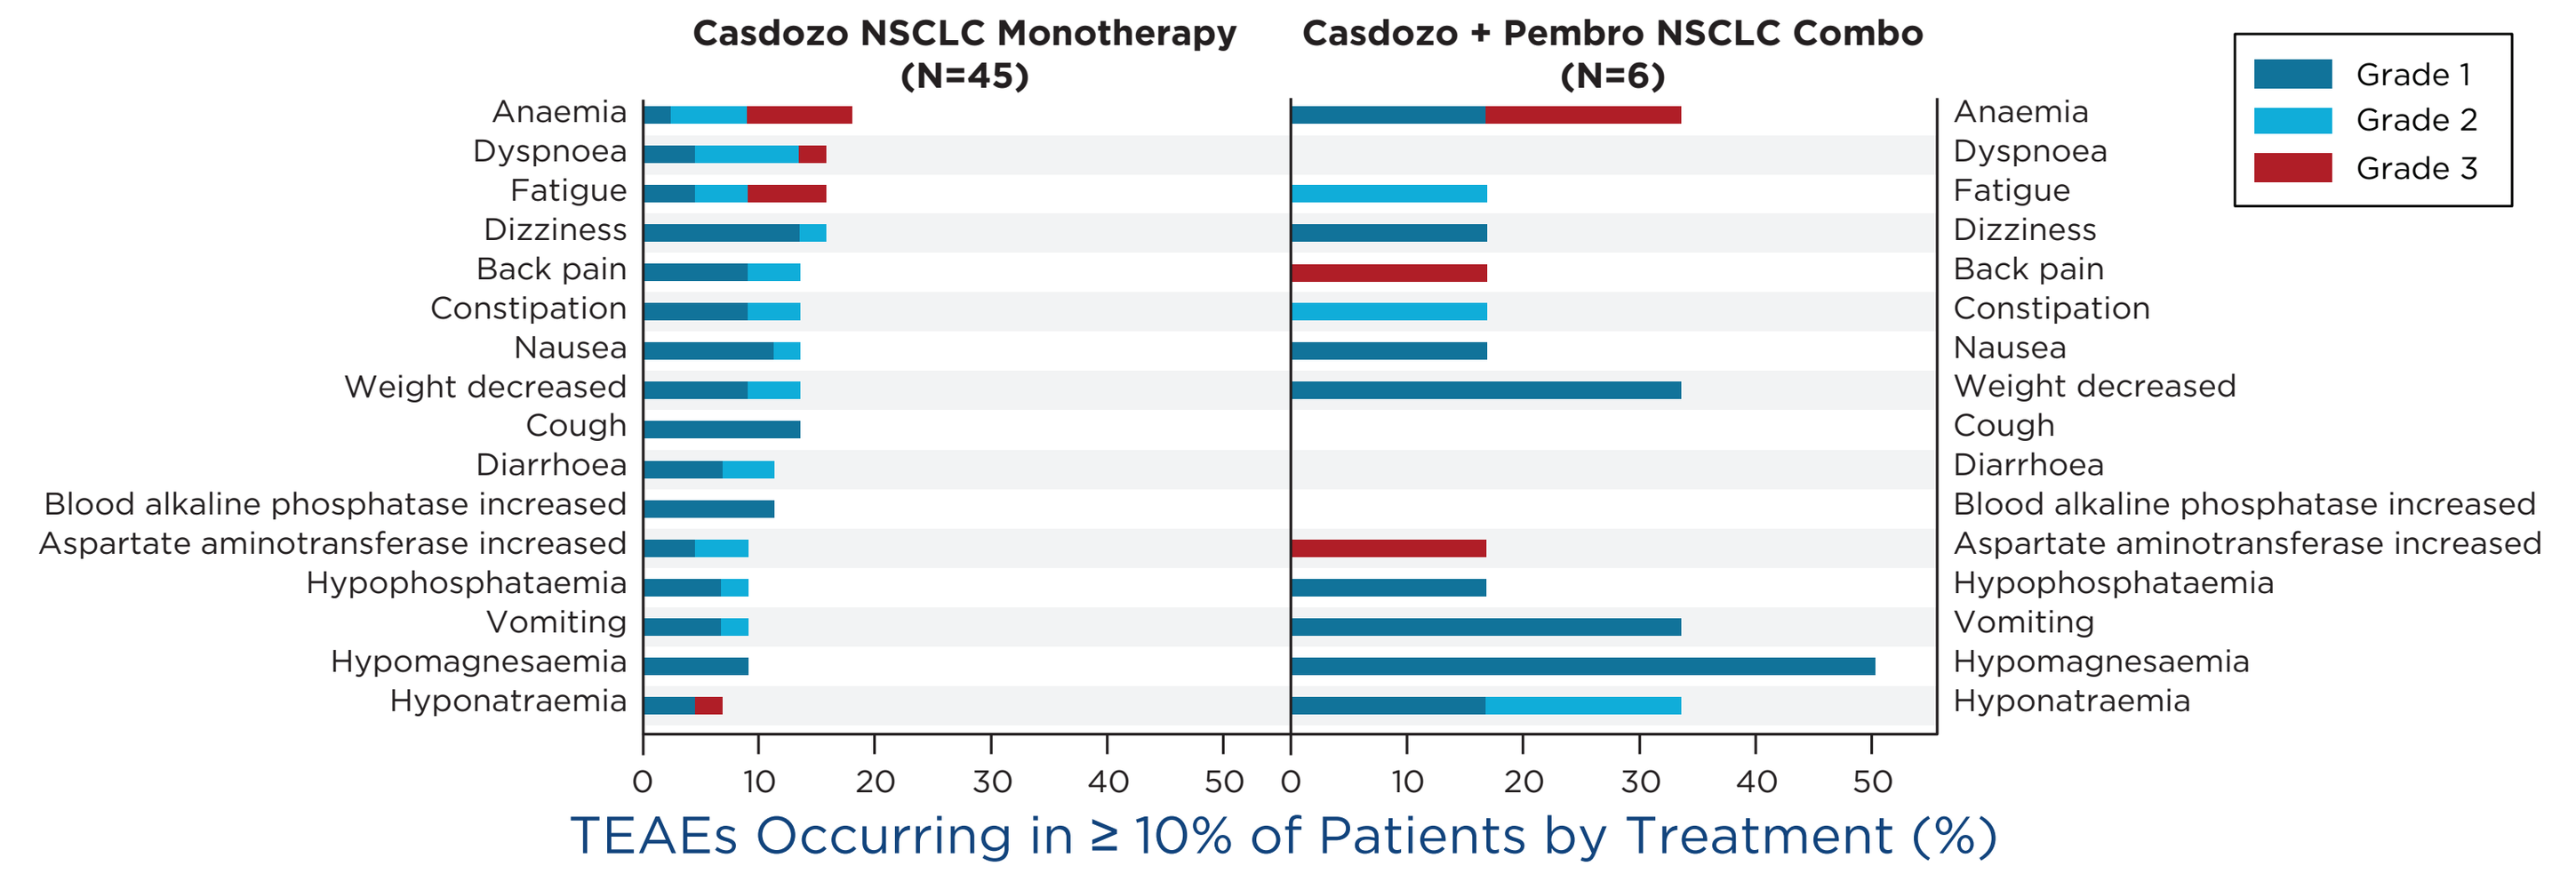
Casdozo NSCLC Monotherapy (N=45)
Casdozo + Pembro NSCLC Combo (N=6)
Grade 1
Grade 2
Grade 3
Anaemia
Dyspnoea
Fatigue
Dizziness
Back pain
Constipation
Nausea
Weight decreased
Cough
Diarrhoea
Blood alkaline phosphatase increased
Aspartate aminotransferase increased
Hypophosphataemia
Vomiting
Hypomagnesaemia
Hyponatraemia
TEAEs Occurring in ≥ 10% of Patients by Treatment (%)

TEAE defined as an AE that emerges or worsens in the period from the first dose of casdozo to 30 days after the last dose.

Data cut as of 21 Sep 2023, subject to change

CONCLUSIONS

Casdozo is a promising novel immunomodulatory anti-IL-27 antibody with evidence of antitumor activity and tolerable safety profile alone and in combination with anti-PD-1

- IL-27 is an immunoregulatory cytokine that can suppress the antitumor response
- Casdozo (CHS-388) is a first-in-class, well-tolerated immunomodulatory cytokine antagonist targeting IL-27
- Casdozo has demonstrated immune activation and single agent responses in PD-(L)1 experienced, PD-L1 low NSCLC patients, with an acceptable safety profile alone and in combination with a PD-1 inhibitor (pembrolizumab)
 - In squamous NSCLC, 2/9 patients had confirmed PRs
- The combination of casdozo + pembrolizumab was well tolerated with a best response of SD in the first 5 response evaluable NSCLC patients treated
- Results support continued evaluation of casdozo to relieve tumor immune suppression in combination with PD-1 inhibitors and other novel agents in NSCLC
- Phase 2 study of the aPD-1 antibody, toripalimab, and casdozo is currently enrolling (NCT04374877)

REFERENCES: **1.** Patnaik A, et al. *J Clin Oncol.* 2021;39 (suppl 15):2551; **2.** Naing A, et al. *J Clin Oncol*. 2022;40 (suppl 16):2501.

DISCLOSURE: Thomas Marron discloses consulting/advisory role for Regeneron, Boehringer Ingelheim, AstraZeneca, DBV Technologies, Celldex, Surface Oncology, NGM Biopharmaceuticals, Glenmark, Abbvie; travel/accommodations/expenses support from Abbvie and Genentech; research funding from Regeneron, Bristol-Myers Squibb, Merck, Boehringer Ingelheim. Use of any of this material requires permission from thomas.marron@mssm.edu or medinfo@coherus.com.

FUNDING: Study was funded by Surface Oncology, which was acquired by Coherus BioSciences in September 2023.
This study is in collaboration with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.