Toripalimab, an Anti-PD-1 Antibody Demonstrating Potent T-Cell Activation and Enhanced Clinical Efficacy Irrespective of PD-L1 Status

Xiaoguang Wang[1†], Sruthi Ravindranathan[1†], Daniel Chin[1], Su-Yi Tseng[1], Scott Klakamp[1], Kate Widmann[1], Varun Kapoor[1], Vladimir Vexler[1], Patricia Keegan[2], Sheng Yao[2,3], Sanjay D. Khare[1], Theresa LaVallee[1], and Narendiran Rajasekaran[1*†]

[1]Coherus BioSciences, Redwood City, CA, USA; [2]TopAlliance Biosciences, Menlo Park, CA, USA; [3]Shanghai Junshi Biosciences, Shanghai, China

Background

Immune checkpoint inhibitors targeting PD-1 have revolutionized cancer treatment, resulting in clinical benefit to a broad range of cancer patients. PD-1 is an inhibitory cell surface receptor that is upregulated upon T-cell activation. Upon binding to its ligands, PD-L1 or PD-L2 expressed on APCs and/or tumor cells, PD-1 recruits SHP1 and SHP2 phosphatases, which suppress T-cell activation and thus function. Toripalimab is a PD-1-targeting humanized IgG4 mAb designed to bind the FG loop of PD-1, thereby blocking the interaction of PD-1 to its ligands PD-L1 and PD-L2.

In clinical trials, toripalimab (tori), in combination with chemotherapy, demonstrated significant efficacy in several cancer indications (JUPITER-02 [NPC], JUPITER-06 [ESCC], and CHOICE-01 [NSCLC]) irrespective of the PD-L1 expression status. Tori is currently under review by the FDA for metastatic or recurrent nasopharyngeal carcinoma (NPC) as first-line treatment in combination with chemotherapy or as a second- or greater-line monotherapy treatment.

Here we report on the molecular and functional characteristics of tori compared to pembrolizumab (pembro), a PD-1 mAb that is approved in most indications in the PD-1 mAb class. Tori demonstrated

- A different PD-1 binding epitope
- A higher binding affinity to PD-1
- Higher Th1 and myeloid-derived inflammatory cytokine responses in human PBMC
- Enhanced expression of several unique genes in IFN-γ and immune cell pathways
- Diminished recruitment of SHP1 and SHP2, upon binding to PD-1-expressing Jurkat T cells

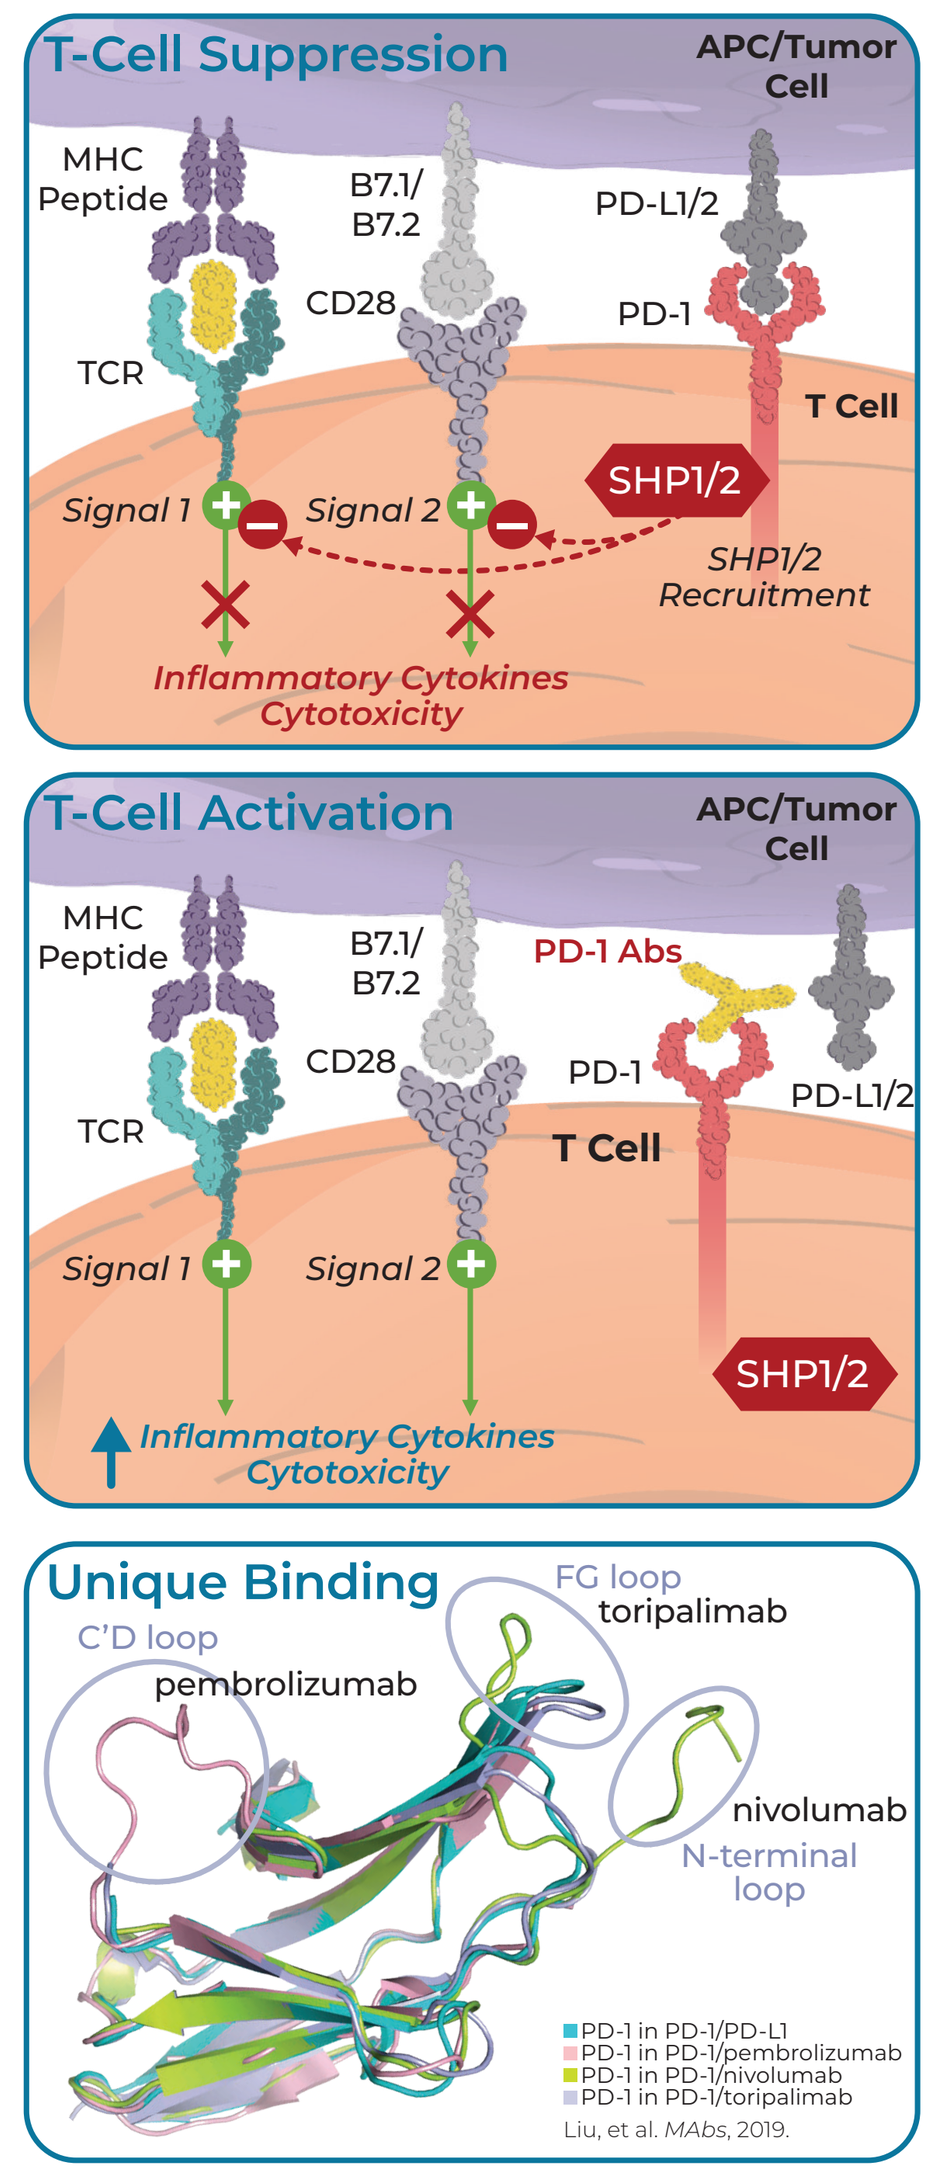

T-Cell Suppression
APC/Tumor Cell
MHC Peptide
B7.1/ B7.2
PD-L1/2
CD28
PD-1
TCR
T Cell
Signal 1
Signal 2
SHP1/2
SHP1/2 Recruitment
Inflammatory Cytokines Cytotoxicity
T-Cell Activation
APC/Tumor Cell
MHC Peptide
B7.1/ B7.2
PD-1 Abs
CD28
PD-1
PD-L1/2
TCR
T Cell
Signal 1
Signal 2
SHP1/2
Inflammatory Cytokines Cytotoxicity
Unique Binding
FG loop
toripalimab
C'D loop
pembrolizumab
nivolumab
N-terminal loop
PD-1 in PD-1/PD-L1
PD-1 in PD-1/pembrolizumab
PD-1 in PD-1/nivolumab
PD-1 in PD-1/toripalimab
Liu, et al. MAbs, 2019.

Overall Survival of Patients With NPC, NSCLC, and ESCC Receiving Combination Toripalimab and Chemotherapy

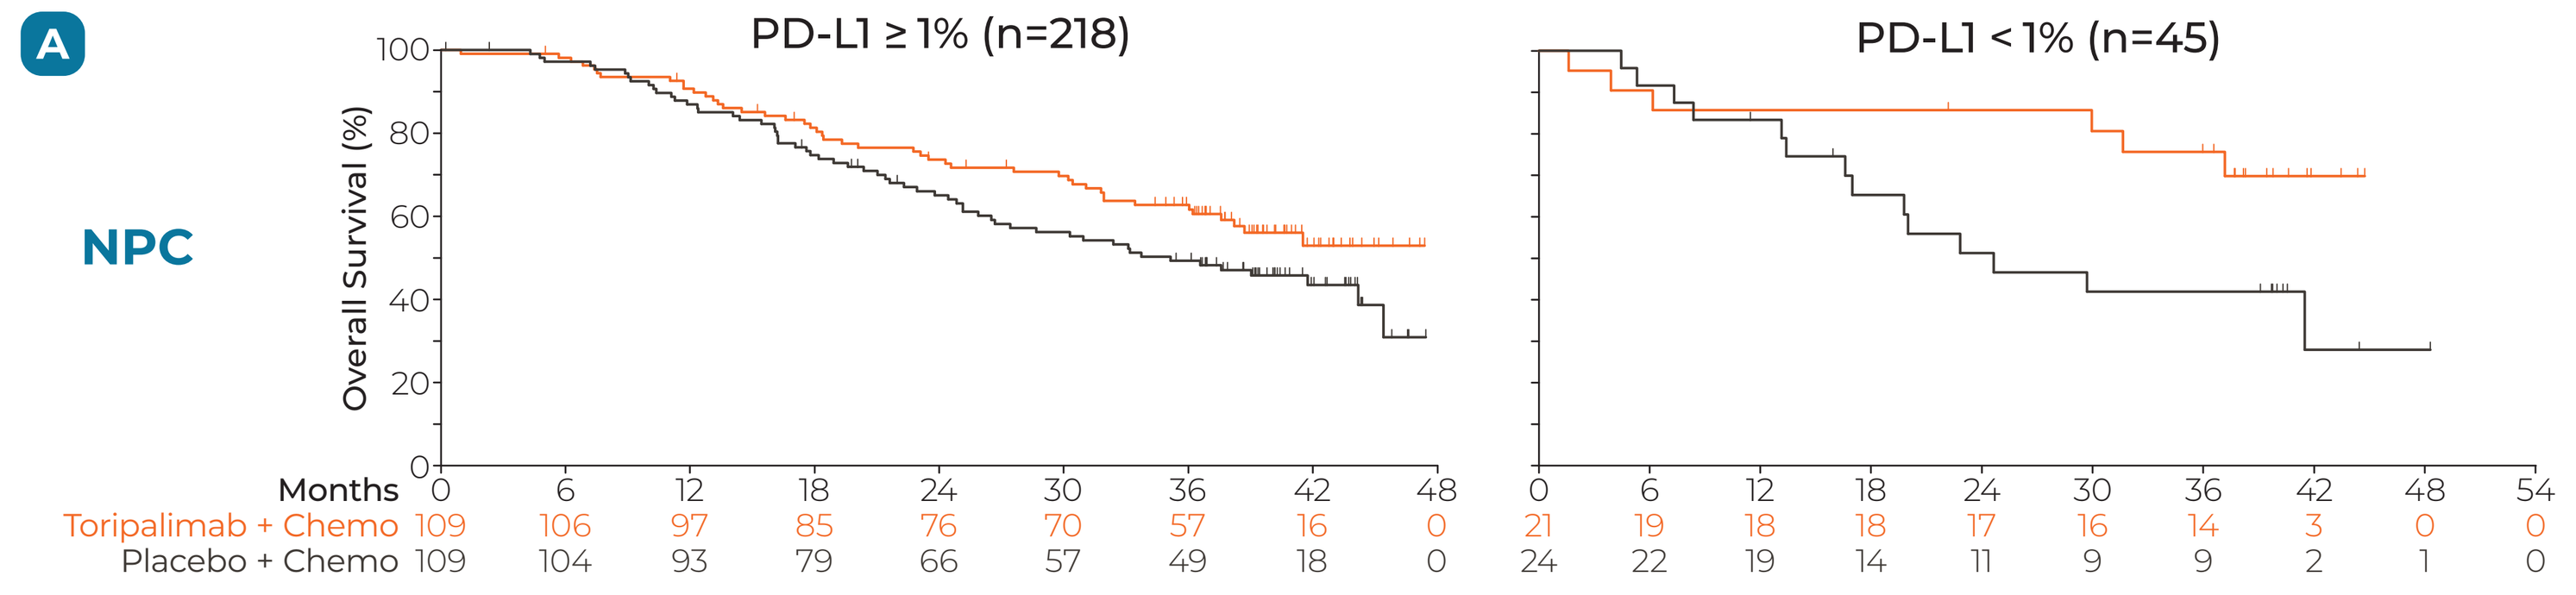

A
NPC
PD-L1 ≥ 1% (n=218)
PD-L1 < 1% (n=45)
Overall Survival (%)
Months 0 6 12 18 24 30 36 42 48
Toripalimab + Chemo 109 106 97 85 76 70 57 16 0
Placebo + Chemo 109 104 93 79 66 57 49 18 0
0 6 12 18 24 30 36 42 48 54
21 19 18 18 17 16 14 3 0 0
24 22 19 14 11 9 9 2 1 0

	#Patients / #Events	Median OS, mos (95% CI)	HR (95% CI)	Nominal p value
Tori + Chemo	45/109	NE (37.6, NE)	0.71 (0.48, 1.04)	0.0786
Placebo + Chemo	59/109	35.2 (26.5, 45.4)		

	#Patients / #Events	Median OS, mos (95% CI)	HR (95% CI)	Nominal p value
Tori + Chemo	6/21	NE (37.2, NE)	0.36 (0.14, 0.95)	0.0322
Placebo + Chemo	14/24	24.6 (16.6, NE)		

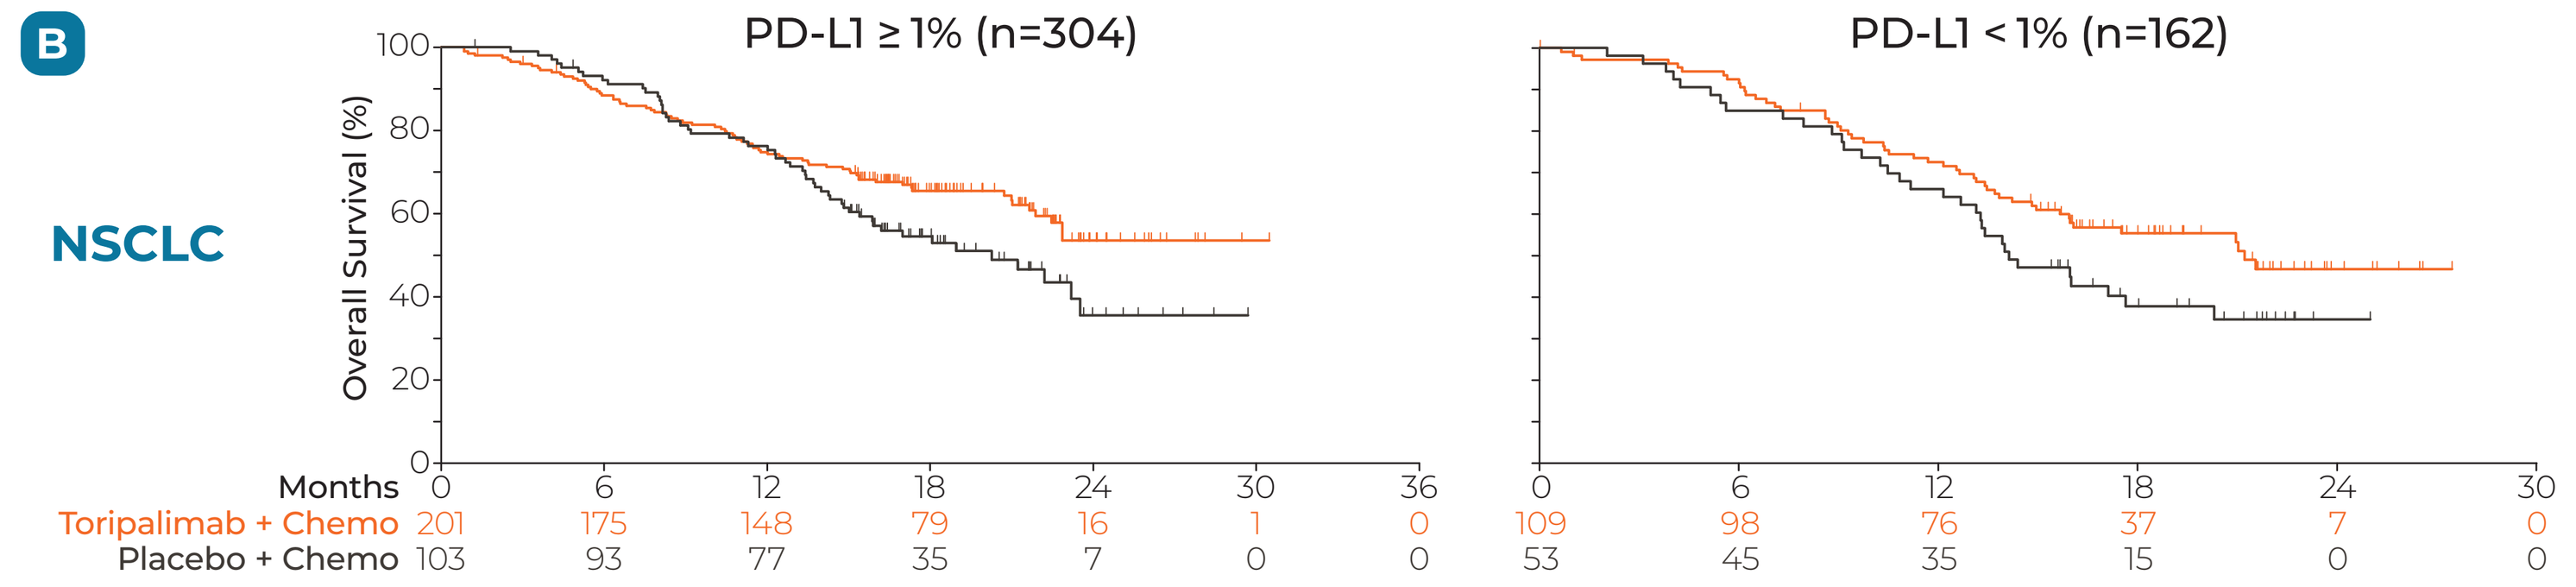

B
NSCLC
PD-L1 ≥ 1% (n=304)
PD-L1 < 1% (n=162)
Overall Survival (%)
Months 0 6 12 18 24 30 36
Toripalimab + Chemo 201 175 148 79 16 1 0
Placebo + Chemo 103 93 77 35 7 0 0
0 6 12 18 24 30
109 98 76 37 7 0
53 45 35 15 0 0

	#Patients / #Events	Median OS, mos (95% CI)	HR (95% CI)	Nominal p value
Tori + Chemo	75/201	NE (22.5, NE)	0.69 (0.49, 1.00)	0.0435
Placebo + Chemo	52/103	20.3 (15.4, 23.5)		

	#Patients / #Events	Median OS, mos (95% CI)	HR (95% CI)	Nominal p value
Tori + Chemo	50/109	21.2 (15.7, NE)	0.70 (0.45, 1.10)	0.1097
Placebo + Chemo	33/53	14.1 (12.2, 20.3)		

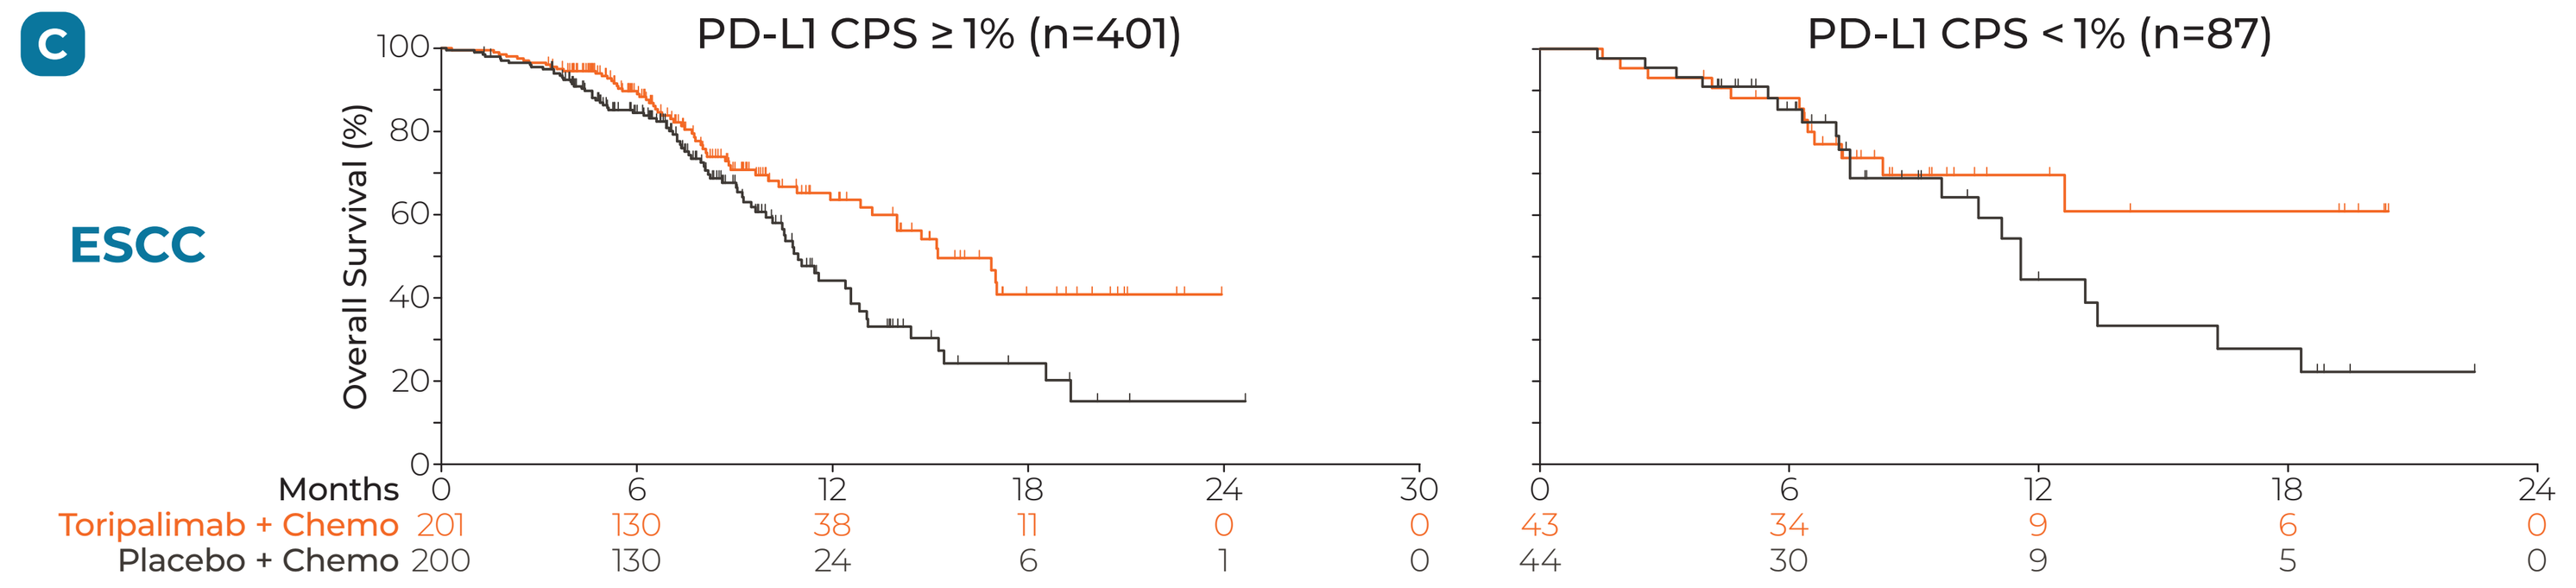

C
ESCC
PD-L1 CPS ≥ 1% (n=401)
PD-L1 CPS < 1% (n=87)
Overall Survival (%)
Months 0 6 12 18 24 30
Toripalimab + Chemo 201 130 38 11 0 0
Placebo + Chemo 200 130 24 6 1 0
0 6 12 18 24
43 34 9 6 0
44 30 9 5 0

	#Patients / #Events	Median OS, mos (95% CI)	HR (95% CI)	Nominal p value
Tori + Chemo	56/201	15.2 (13.2, NE)	0.61 (0.44, 0.87)	0.0056
Placebo + Chemo	77/200	10.9 (10.0, 12.6)		

	#Patients / #Events	Median OS, mos (95% CI)	HR (95% CI)	Nominal p value
Tori + Chemo	12/43	NE (12.6, NE)	0.61 (0.30, 1.25)	0.1737
Placebo + Chemo	20/44	11.6 (9.7, 16.3)		

Toripalimab in combination with chemotherapy significantly improved overall survival (OS) irrespective of PD-L1 status in post hoc analyses of 3 randomized controlled trials, including NPC (A), NSCLC (B), and ESCC (C).

Binding Affinity of Toripalimab Versus Pembrolizumab for PD-1

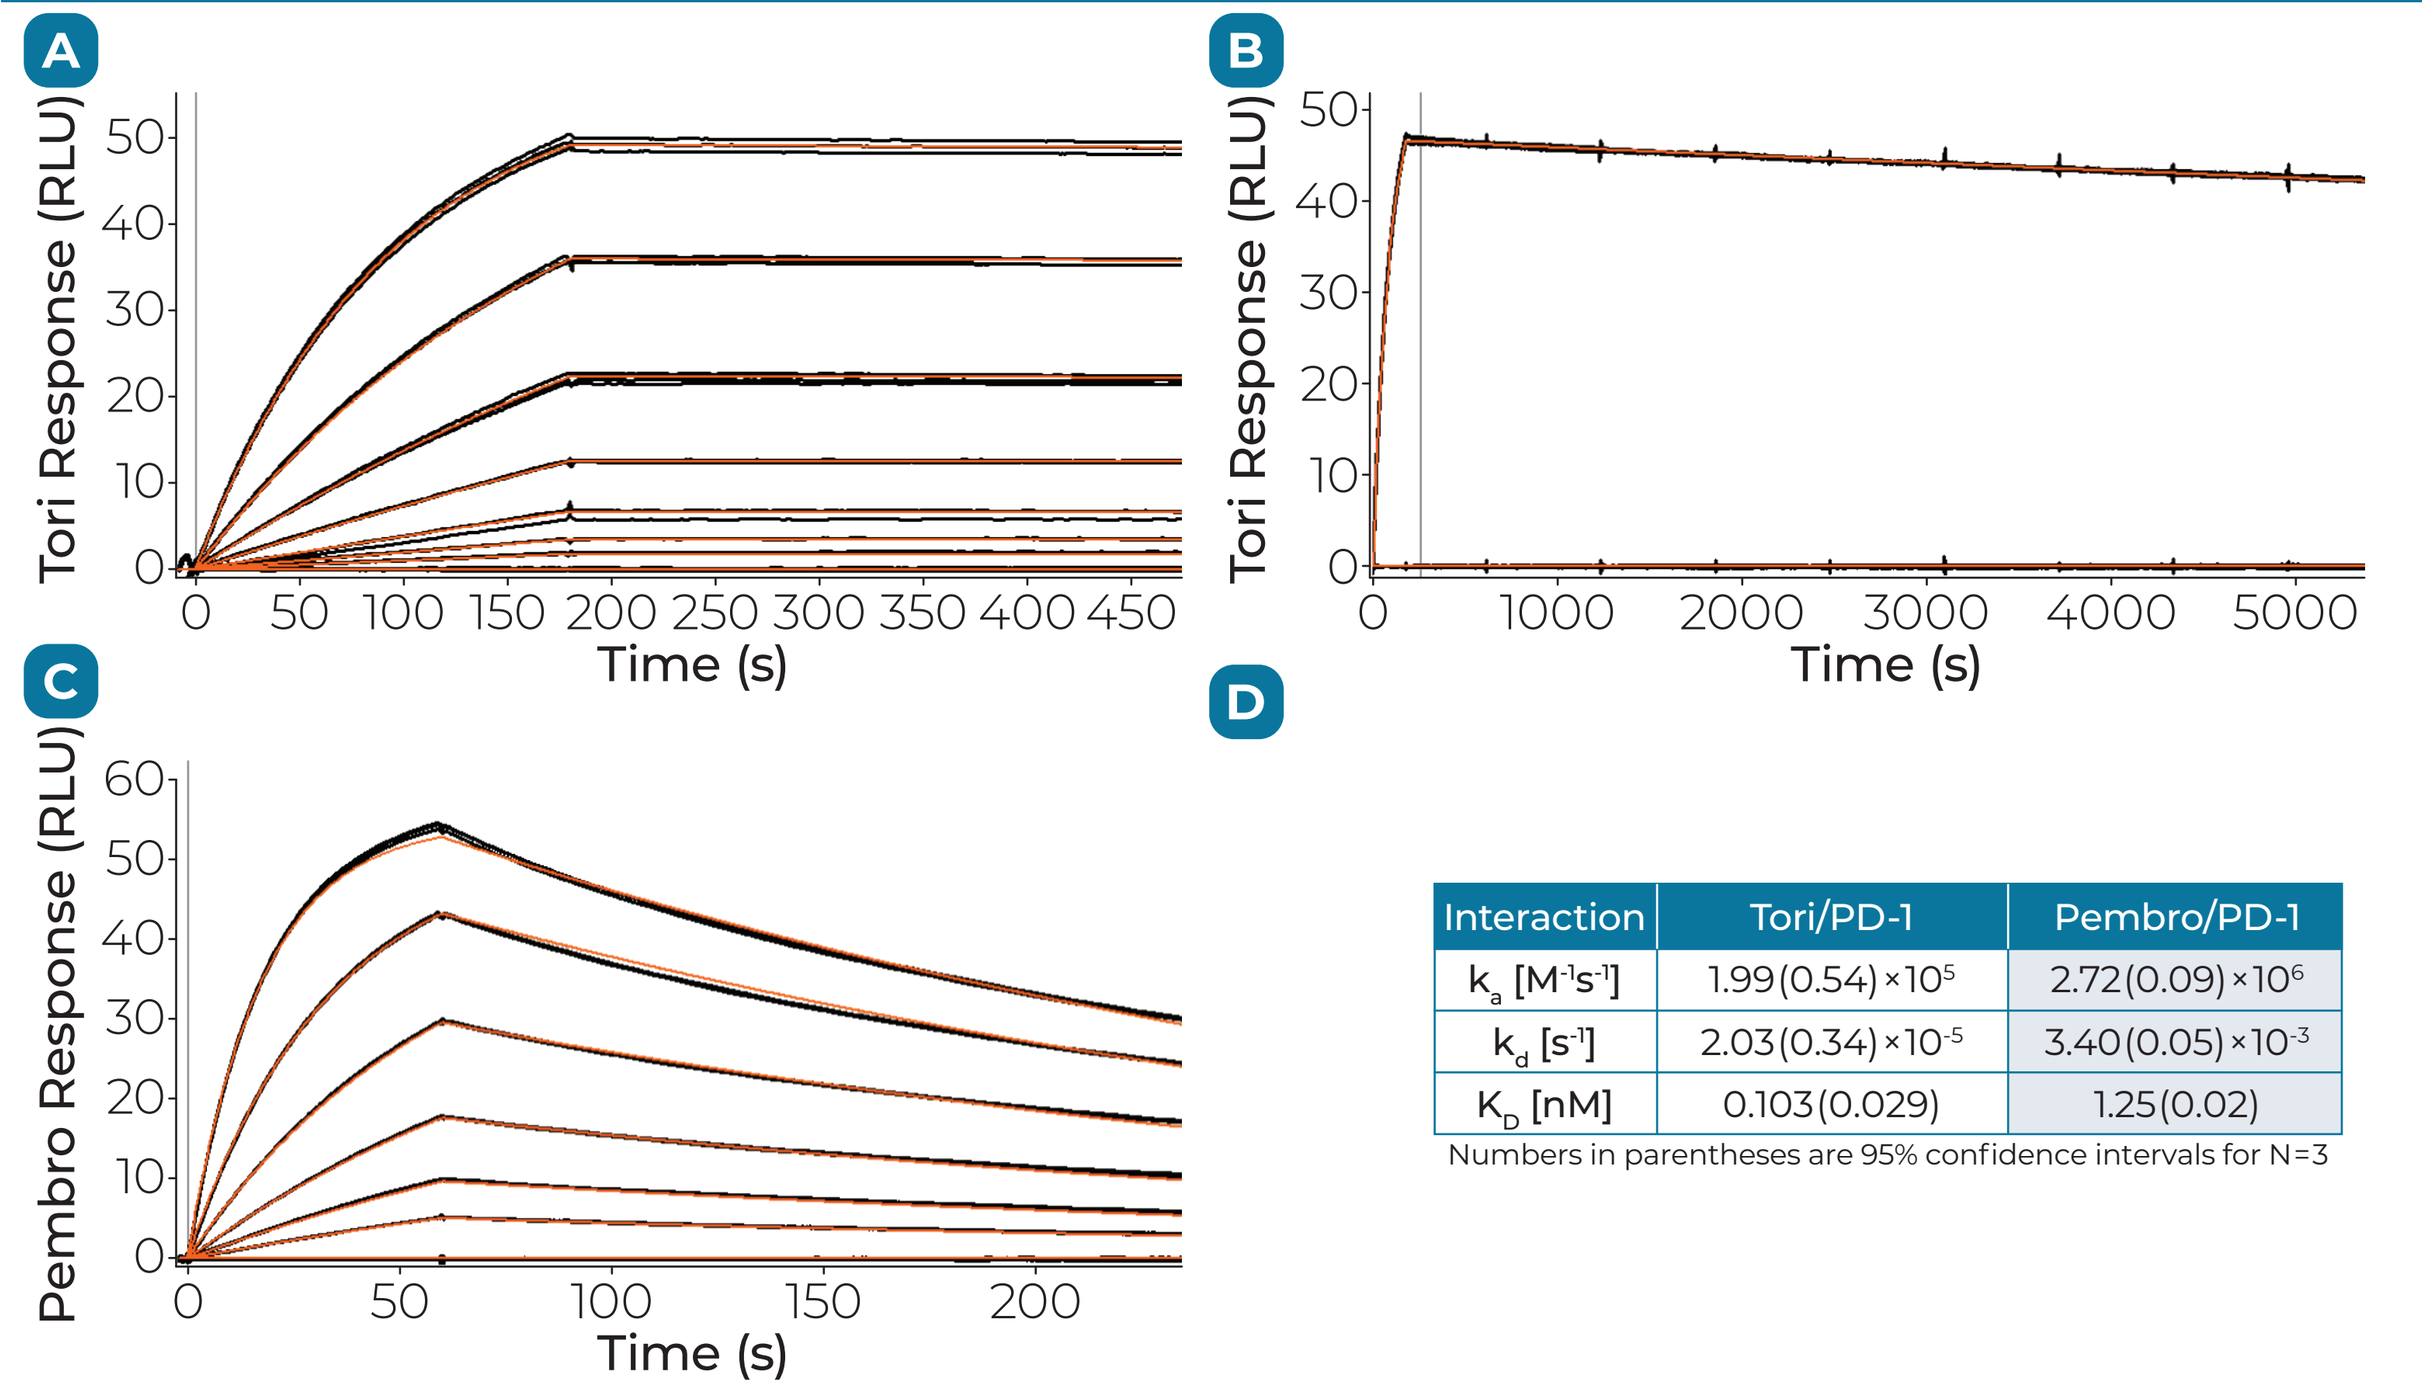

A
Tori Response (RLU)
Time (s)
B
Tori Response (RLU)
Time (s)
C
Pembro Response (RLU)
Time (s)
D

Interaction	Tori/PD-1	Pembro/PD-1
k_a [$M^{-1}s^{-1}$]	1.99(0.54)×10^5	2.72(0.09)×10^6
k_d [s^{-1}]	2.03(0.34)×10^{-5}	3.40(0.05)×10^{-3}
K_D [nM]	0.103(0.029)	1.25(0.02)

Numbers in parentheses are 95% confidence intervals for N=3

Tori exhibits a 12-fold higher binding affinity for PD-1 compared to pembro. Biacore sensorgrams of PD-1 binding to covalently immobilized tori (A, B) or pembro (C). (D) Average values for k_a, k_d, and K_D from 3 replicate experiments for PD-1 binding to tori and pembro.

Induced Cytokine Expression in Human PBMC

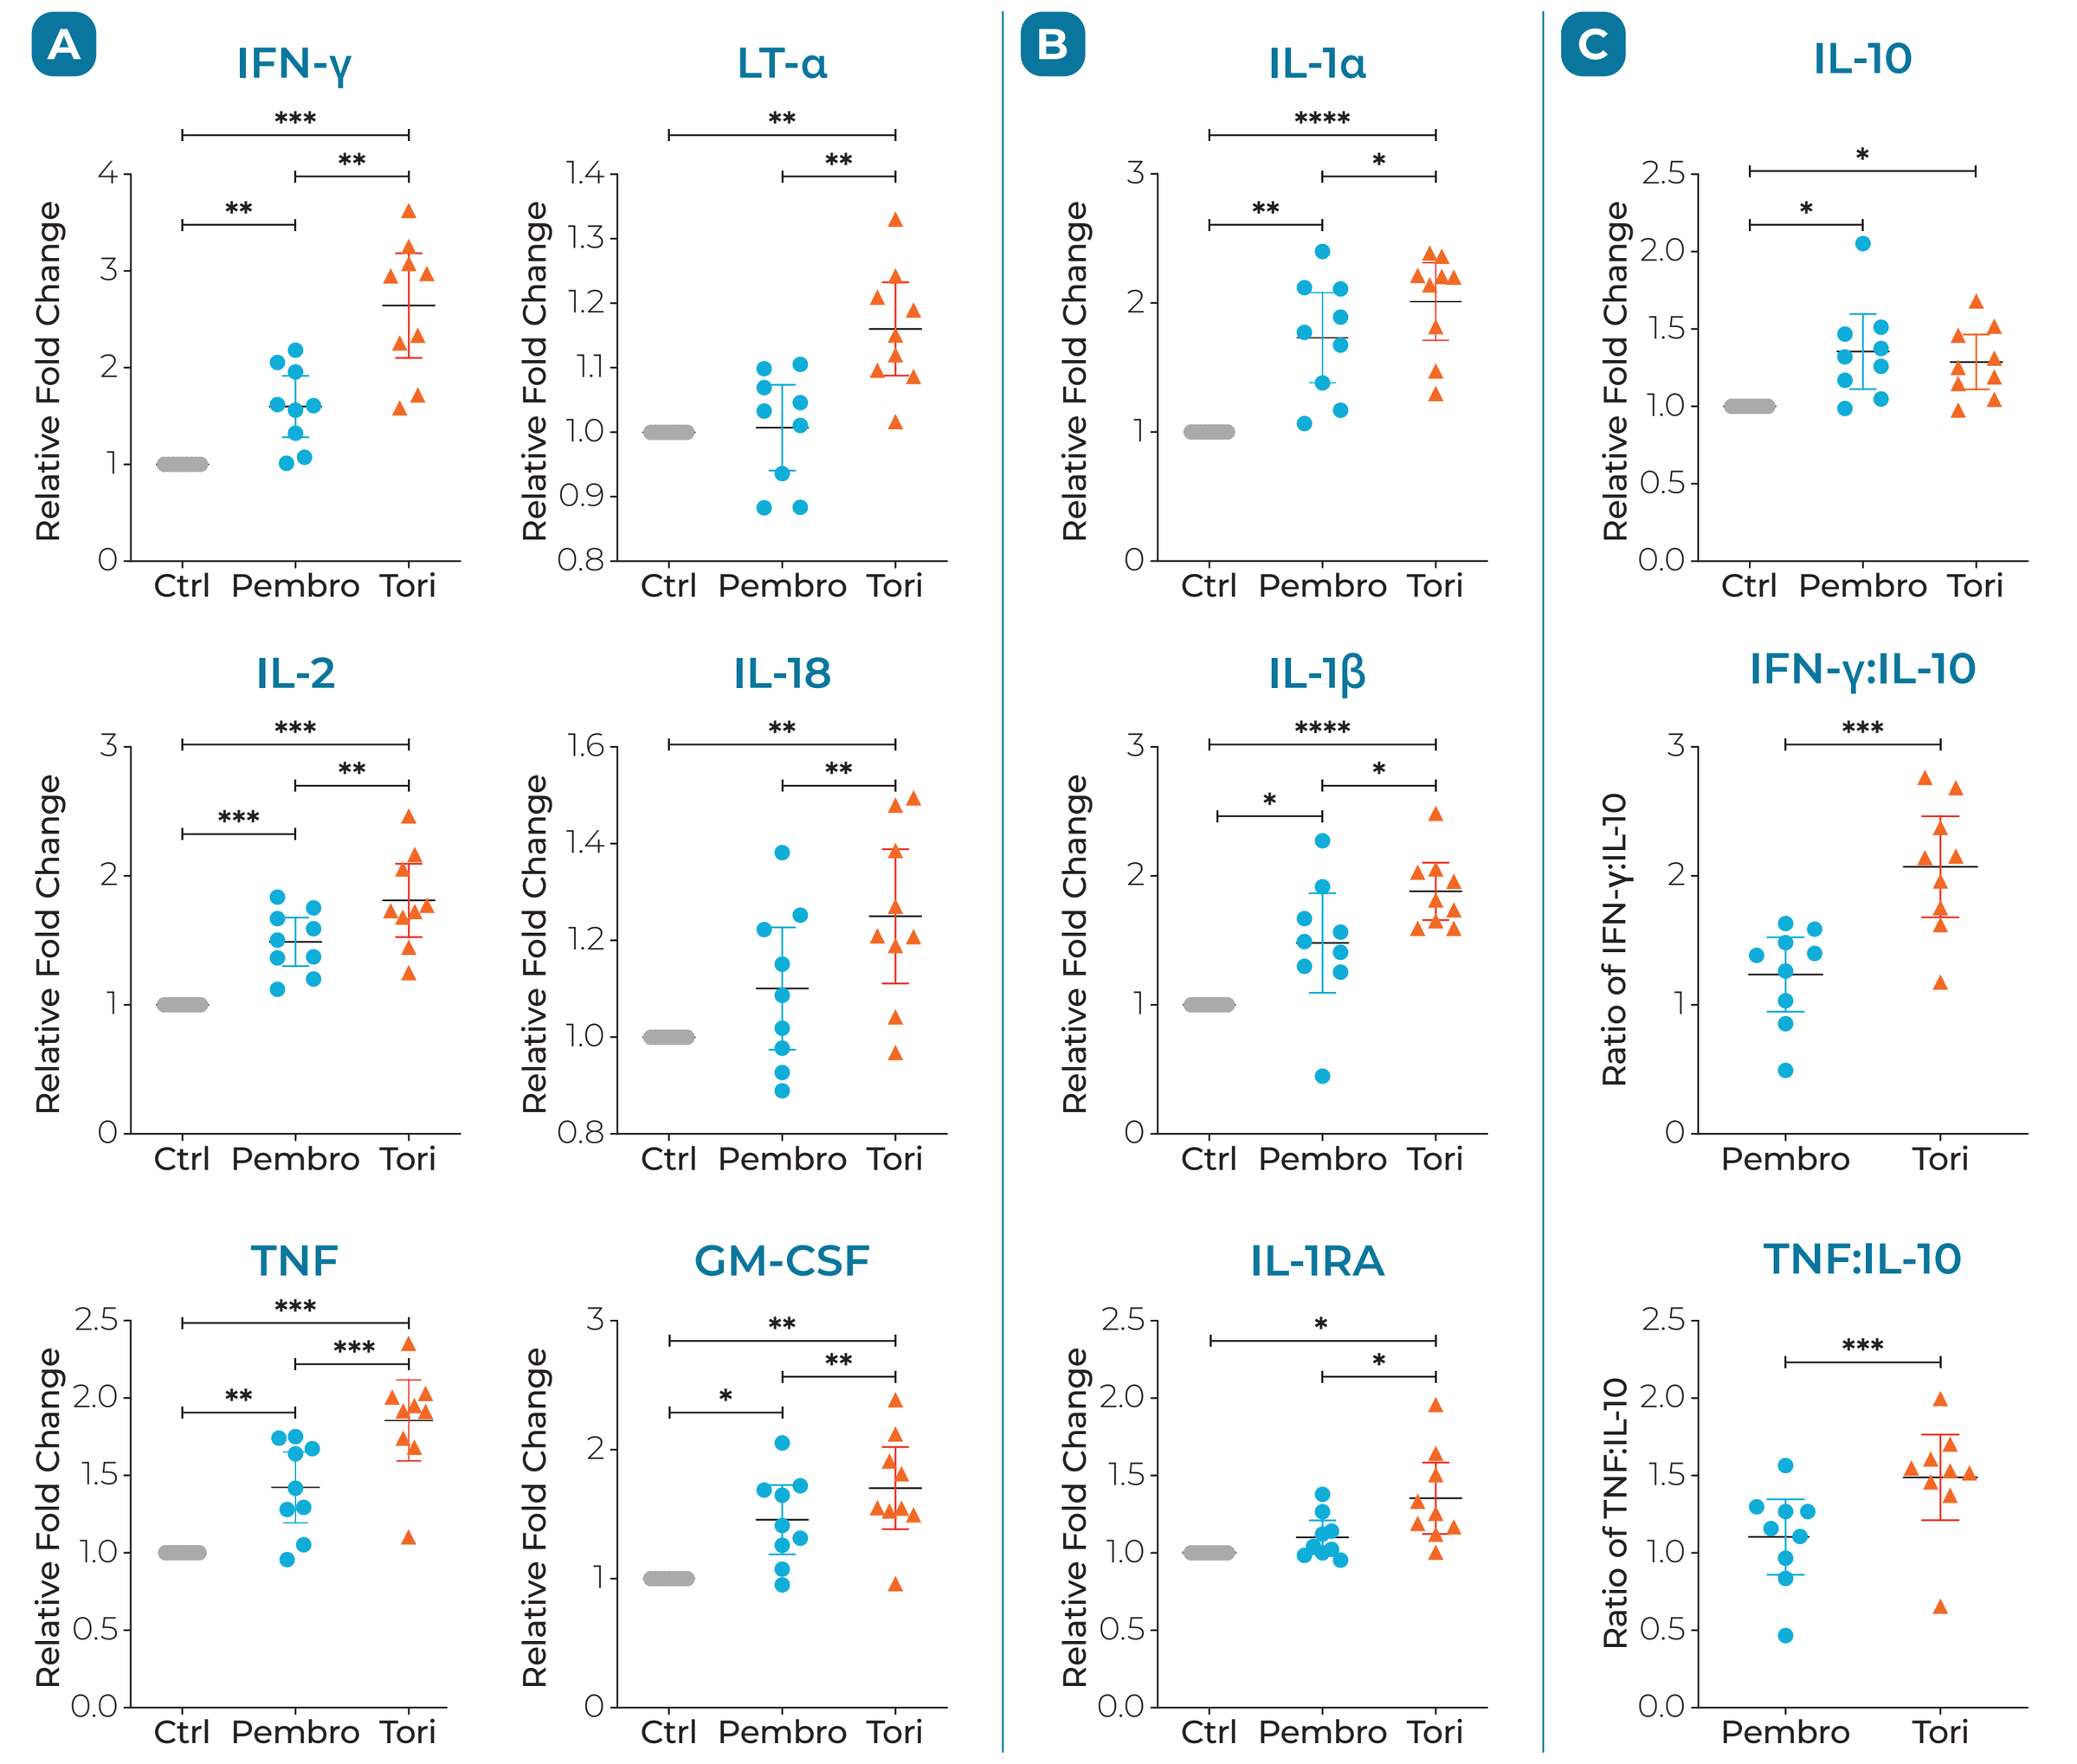

A
IFN-γ
LT-α
IL-2
IL-18
TNF
GM-CSF
B
IL-1α
IL-1β
IL-1RA
C
IL-10
IFN-γ:IL-10
TNF:IL-10
Relative Fold Change
Ratio of IFN-γ:IL-10
Ratio of TNF:IL-10
Ctrl Pembro Tori

Tori is more potent than pembro in enhancing levels of Th1 and myeloid-derived cytokines in human PBMC (n=9 healthy donors). Supernatants from PBMC cultured with superantigen SEB in the presence of anti-PD-1 antibody: pembro or tori or isotype Ab ctrl were tested by performing Luminex assays to examine Th1 cytokines (A), myeloid-derived cytokines (B), and immunosuppressive cytokine IL-10 (C).

IFN-γ Secretion Levels of Human CD8+ Cells

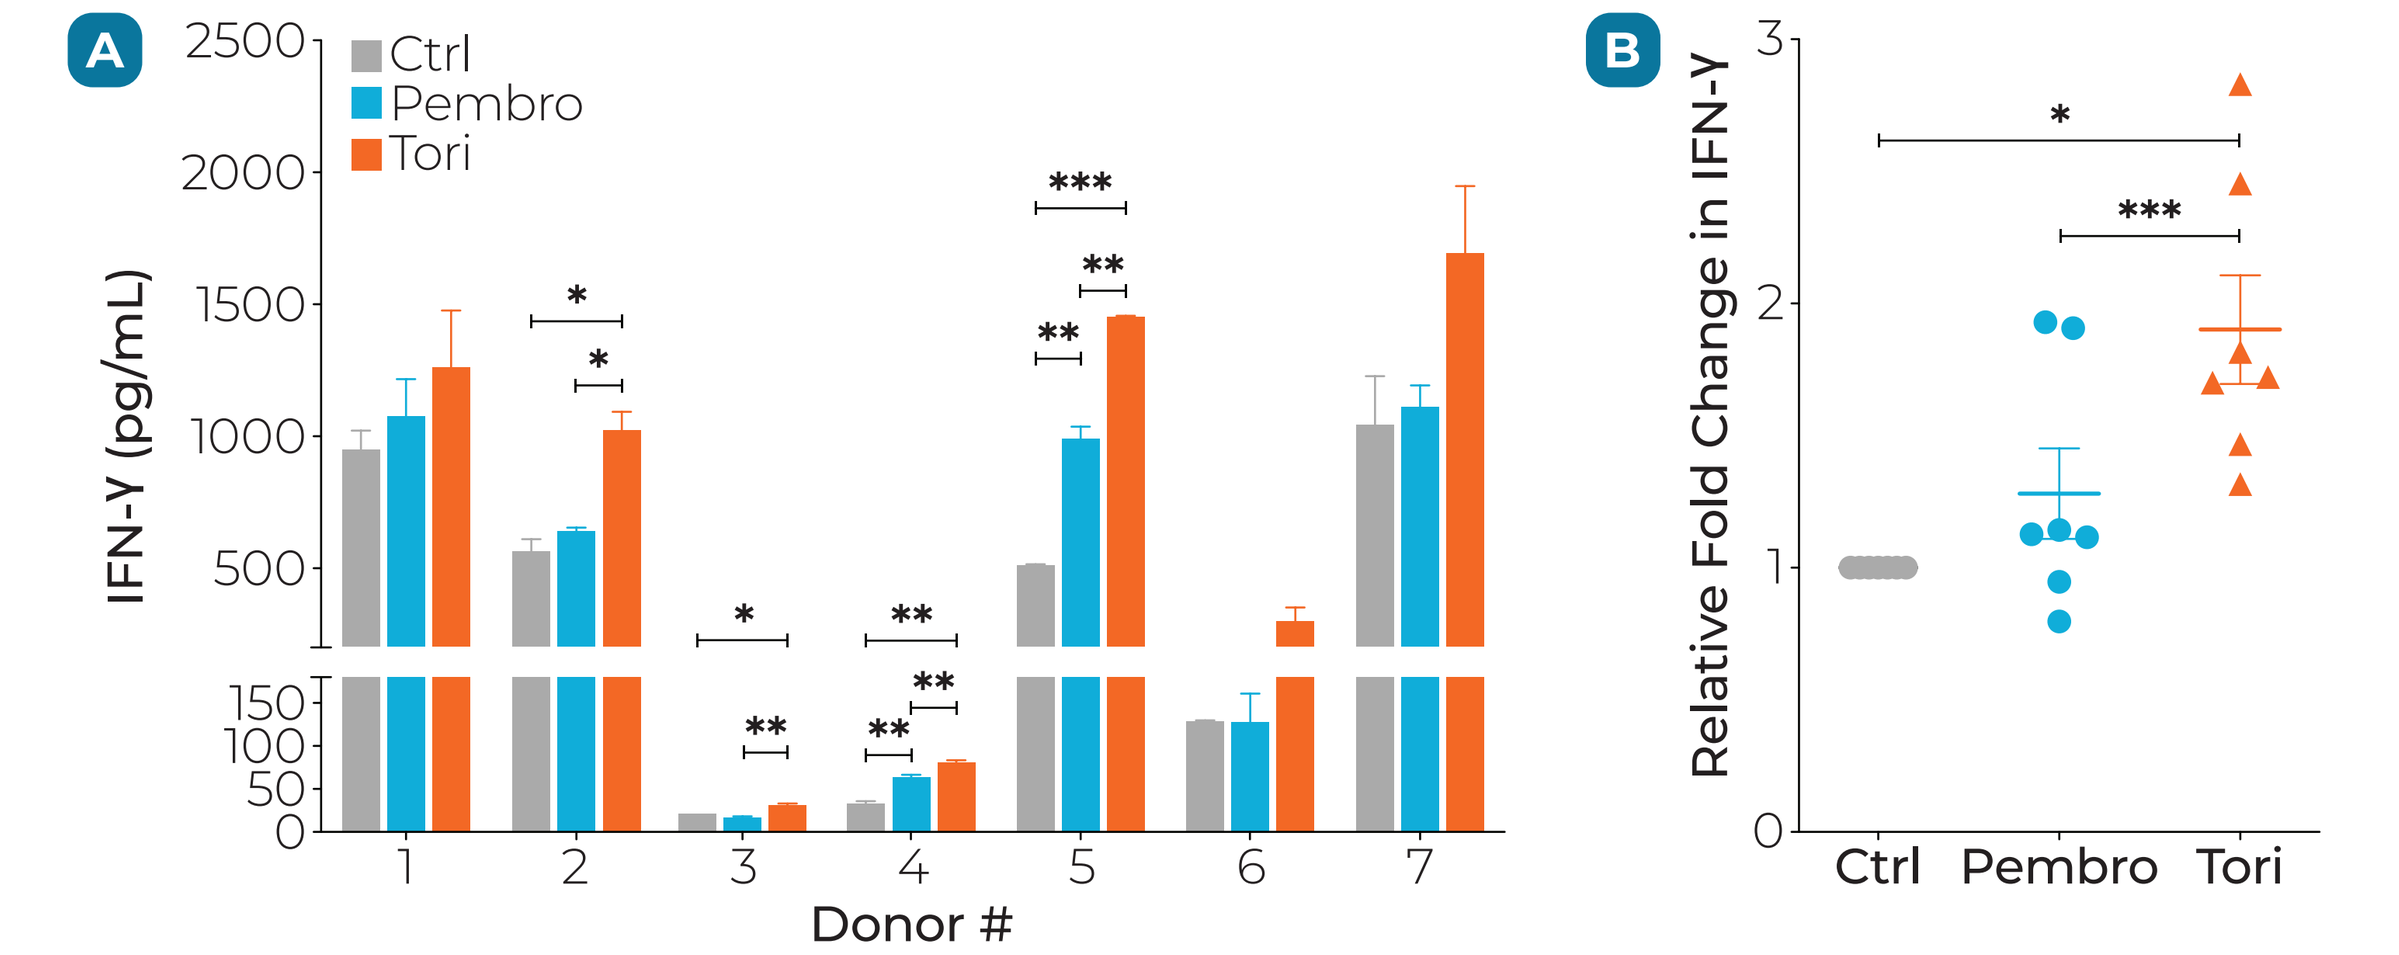

A
Ctrl
Pembro
Tori
IFN-γ (pg/mL)
Donor #
B
Relative Fold Change in IFN-γ
Ctrl Pembro Tori

Tori is more potent than pembro in enhancing IFN-γ secretion in CD3/CD28 activated naive human CD8+ T cells. (A) Absolute concentrations of IFN-γ secreted by antibody-induced CD8+ T cells from 7 donors as measured by ELISA. (B) Fold-change in concentration of IFN-γ relative to the Ctrl.

Tumor Cell Cytotoxicity of Activated PBMC Cultured With Toripalimab or Pembrolizumab

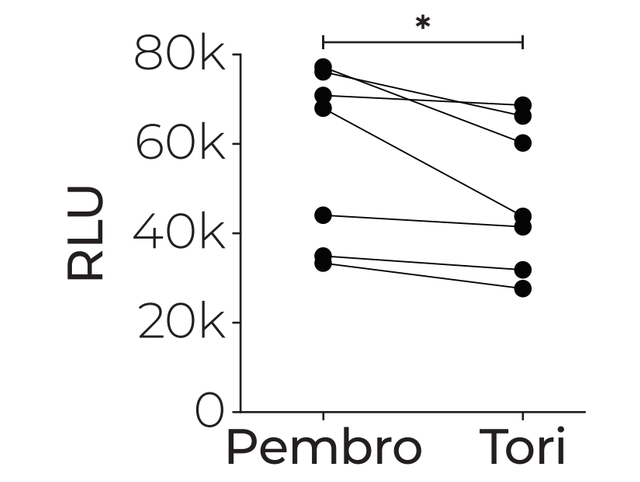

RLU
Pembro
Tori

Following activation and treatment with PD-1 mAbs, PBMC treated with tori exhibit significantly higher cytotoxicity to tumor cells compared to PBMC that were treated with pembro. PBMC isolated from healthy donors were cultured with IL2, IL7, IL15, and anti-CD3 Ab overnight. Target cells, SK-OV3, were co-cultured with PBMC in the presence of PD-1 Ab for 3 days. The intensity of chemiluminescence signal, detected as relative luminescent units (RLU), correlates to the number of live tumor cells.

Antibody-Mediated SHP1 and SHP2 Recruitment

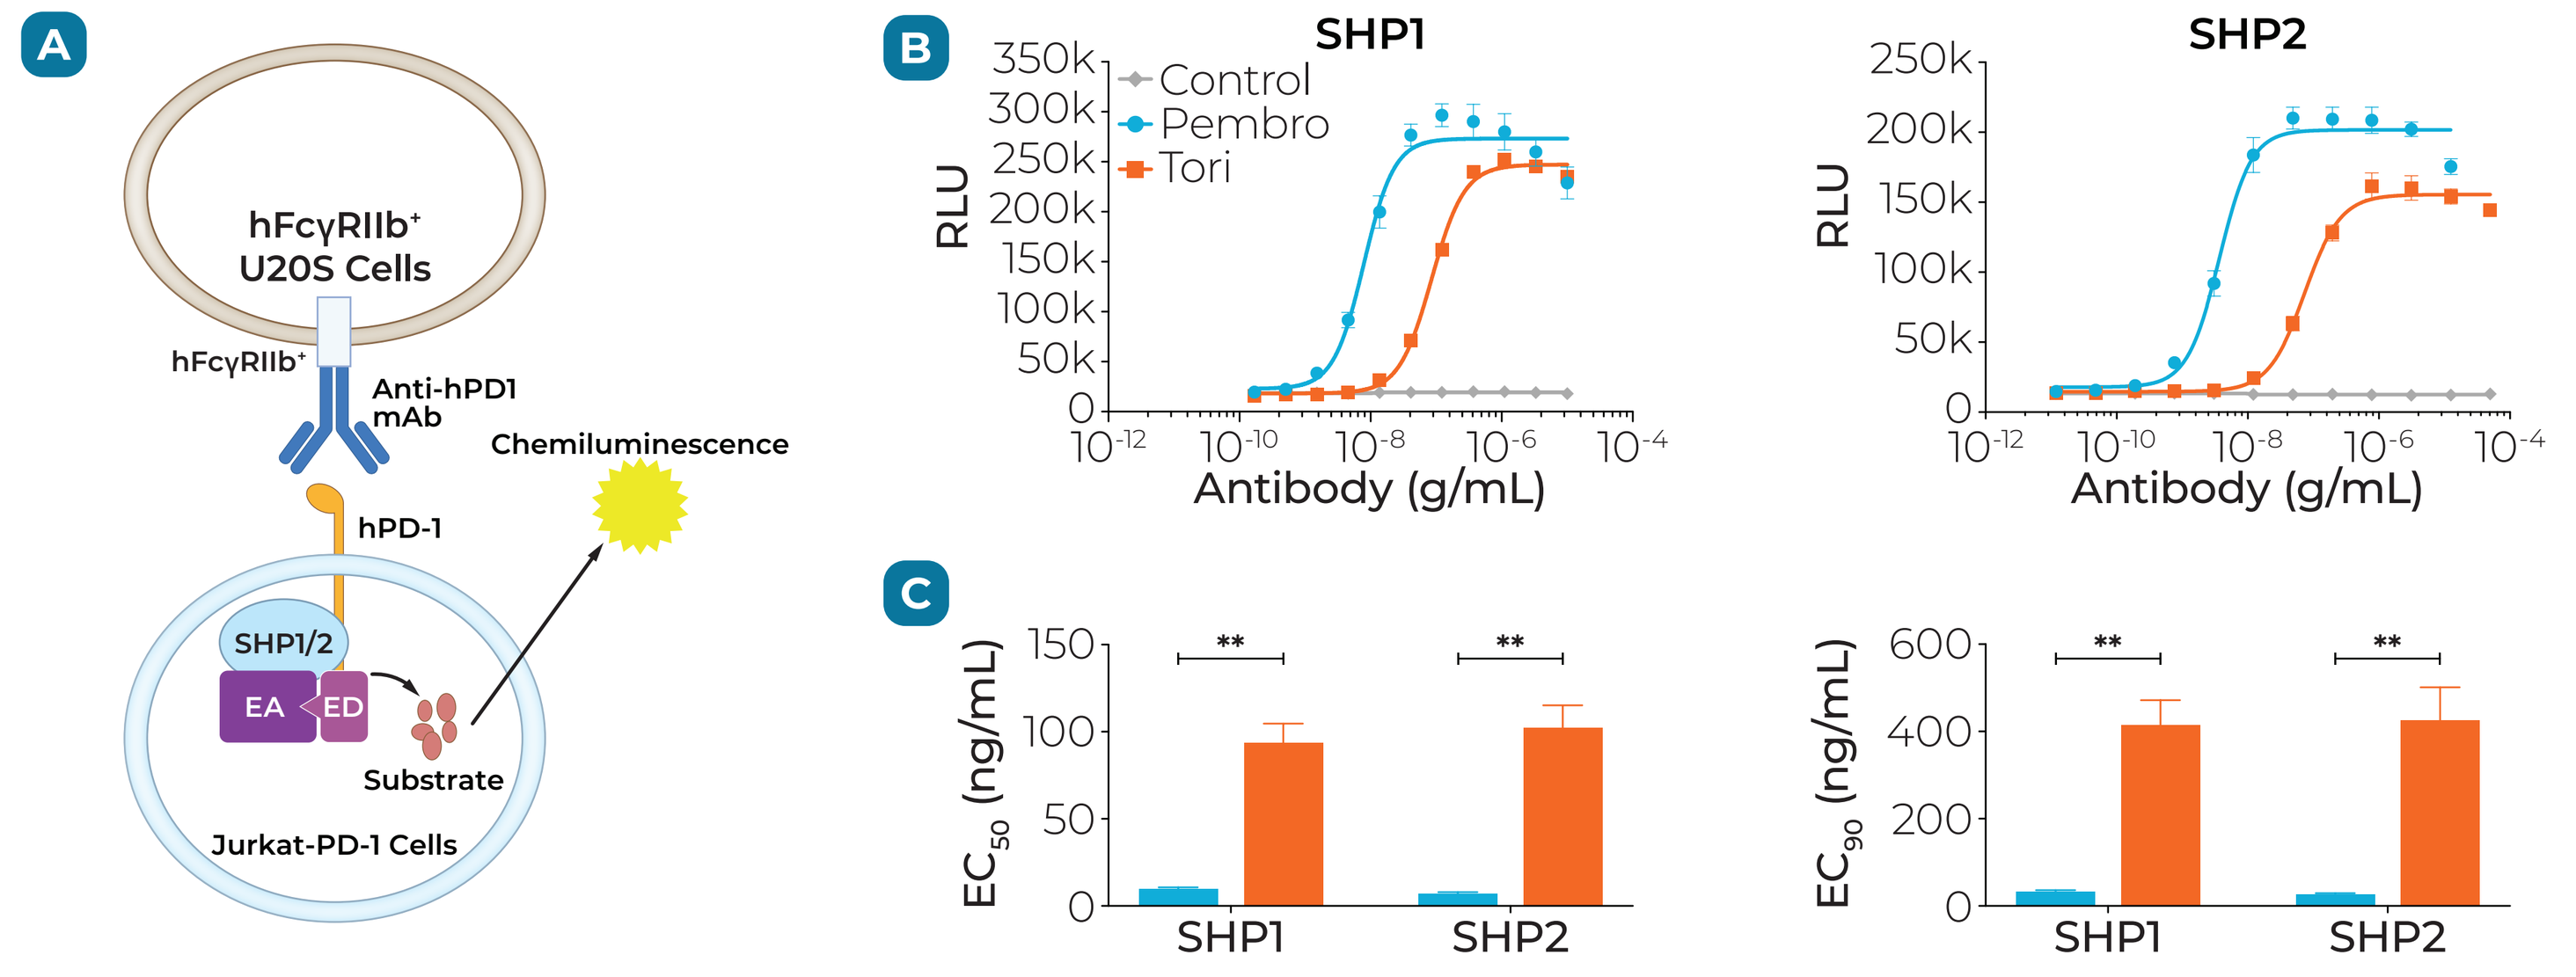

A
hFcγRIIb+ U20S Cells
hFcγRIIb+
Anti-hPD1 mAb
Chemiluminescence
hPD-1
SHP1/2
EA
ED
Substrate
Jurkat-PD-1 Cells
B
SHP1
SHP2
Control
Pembro
Tori
RLU
Antibody (g/mL)
C
EC50 (ng/mL)
EC90 (ng/mL)
SHP1
SHP2

Tori recruits lower levels of SHP1 or SHP2, the negative regulators of T-cell activation, compared to pembro in Jurkat PD-1 cells. (A) Schematic of the experimental system. (B) Dose response curve for SHP1 and SHP2 recruitment. (C) The EC_{50} and EC_{90} values calculated from dose response curves.

Genes Associated With IFN-γ Production in Dissociated NSCLC Tumor Cells

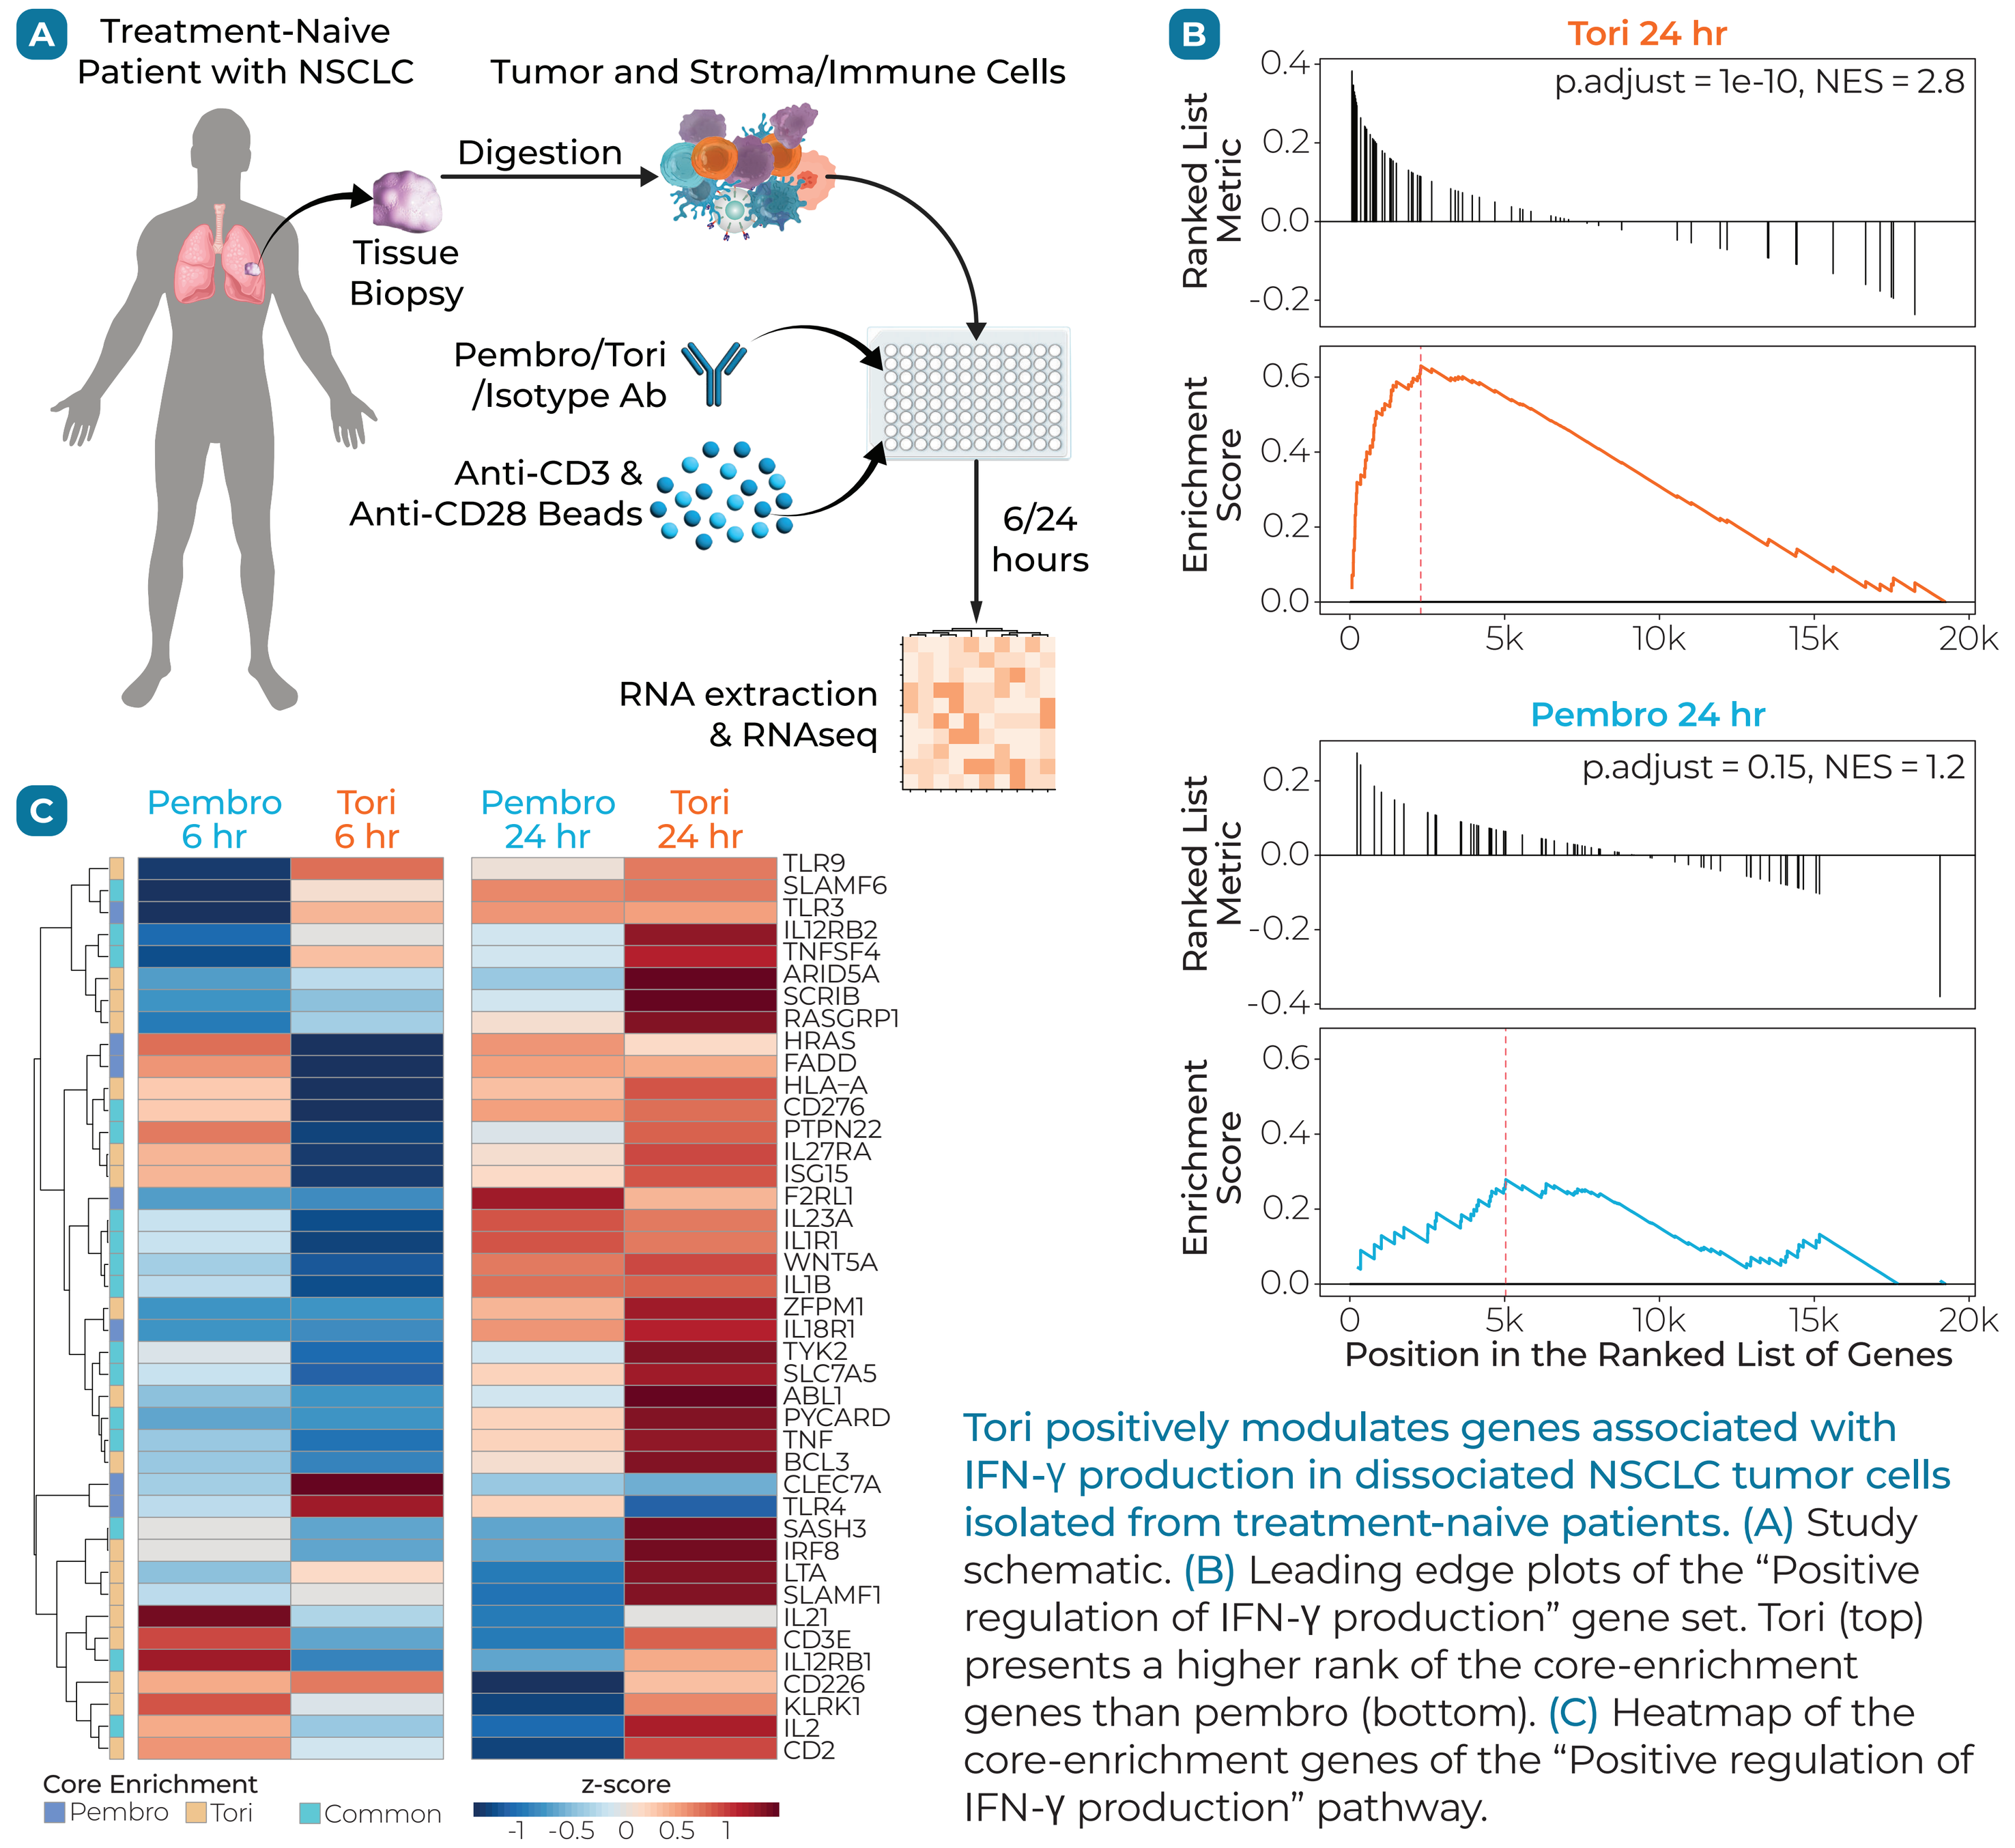

A
Treatment-Naive Patient with NSCLC
Tumor and Stroma/Immune Cells
Digestion
Tissue Biopsy
Pembro/Tori /Isotype Ab
Anti-CD3 & Anti-CD28 Beads
6/24 hours
RNA extraction & RNAseq
B
Tori 24 hr
p.adjust = 1e-10, NES = 2.8
Pembro 24 hr
p.adjust = 0.15, NES = 1.2
Ranked List Metric
Enrichment Score
Position in the Ranked List of Genes
C
Pembro 6 hr
Tori 6 hr
Pembro 24 hr
Tori 24 hr
TLR9
SLAMF6
TLR3
IL12RB2
TNFSF4
ARID5A
SCRIB
RASGRP1
HRAS
FADD
HLA-A
CD276
PTPN22
IL27RA
ISG15
F2RL1
IL23A
IL1R1
WNT5A
IL1B
ZFPM1
IL18R1
TYK2
SLC7A5
ABL1
PYCARD
TNF
BCL3
CLEC7A
TLR4
SASH3
IRF8
LTA
SLAMF1
IL21
CD3E
IL12RB1
CD226
KLRK1
IL2
CD2
Core Enrichment
Pembro
Tori
Common
z-score

Tori positively modulates genes associated with IFN-γ production in dissociated NSCLC tumor cells isolated from treatment-naive patients. (A) Study schematic. (B) Leading edge plots of the "Positive regulation of IFN-γ production" gene set. Tori (top) presents a higher rank of the core-enrichment genes than pembro (bottom). (C) Heatmap of the core-enrichment genes of the "Positive regulation of IFN-γ production" pathway.

Acknowledgment & Contacts: We thank the patients who volunteered to participate in the toripalimab clinical studies and their families, all the investigators, and study site personnel. We thank Marc Pondel, Rosh Dias, Nathalie Vandenkoornhuyse, and the Coherus Scientific Advisory Board members for thoughtful review and editorial input into the manuscript. We thank Dr. Karen Miller for the research support. We also thank members in the Legal Department of Coherus Biosciences for review. Please contact **swang@coherus.com** for any questions.

Abbreviations: Ab = antibody; APC = antigen presenting cell; CPS = combined positive score; ctrl = control; EC_{50} = half-maximal effective concentration; ESCC = esophageal squamous cell carcinoma; GM-CSF = Granulocyte-macrophage colony-stimulating factor; HR = hazard ratio; IFN-γ = interferon gamma; IL = interleukin; k_a = association rate constant; k_d = dissociation rate constant; K_D = dissociation equilibrium constant; LT-α = lymphotoxin alpha; mAb = monoclonal antibody; MHC = major histocompatibility complex; NE = median OS not reached; NES = normalized enrichment score; NPC = nasopharyngeal carcinoma; NSCLC = non-small cell lung cancer; OS = overall survival; PBMC = peripheral blood mononuclear cell; PD-1/2 = programmed cell death protein 1/2; PD-L1/L2 = programmed cell death protein ligand 1/2; pembro = pembrolizumab; RLU = relative luminescent unit; RNA = ribonucleic acid; SHP1/2 = Src homology region 2 domain-containing phosphatase-1/2; TCR = T-cell receptor; TNF = tumor necrosis factor; tori = toripalimab.

Statistics: * = $p < 0.05$; ** = $p < 0.01$; *** = $p < 0.001$; **** = $p < 0.0001$.

IFN-γ Gene Response Signature in Dissociated NSCLC Tumor Cells Following PD-1 Antibody Treatment

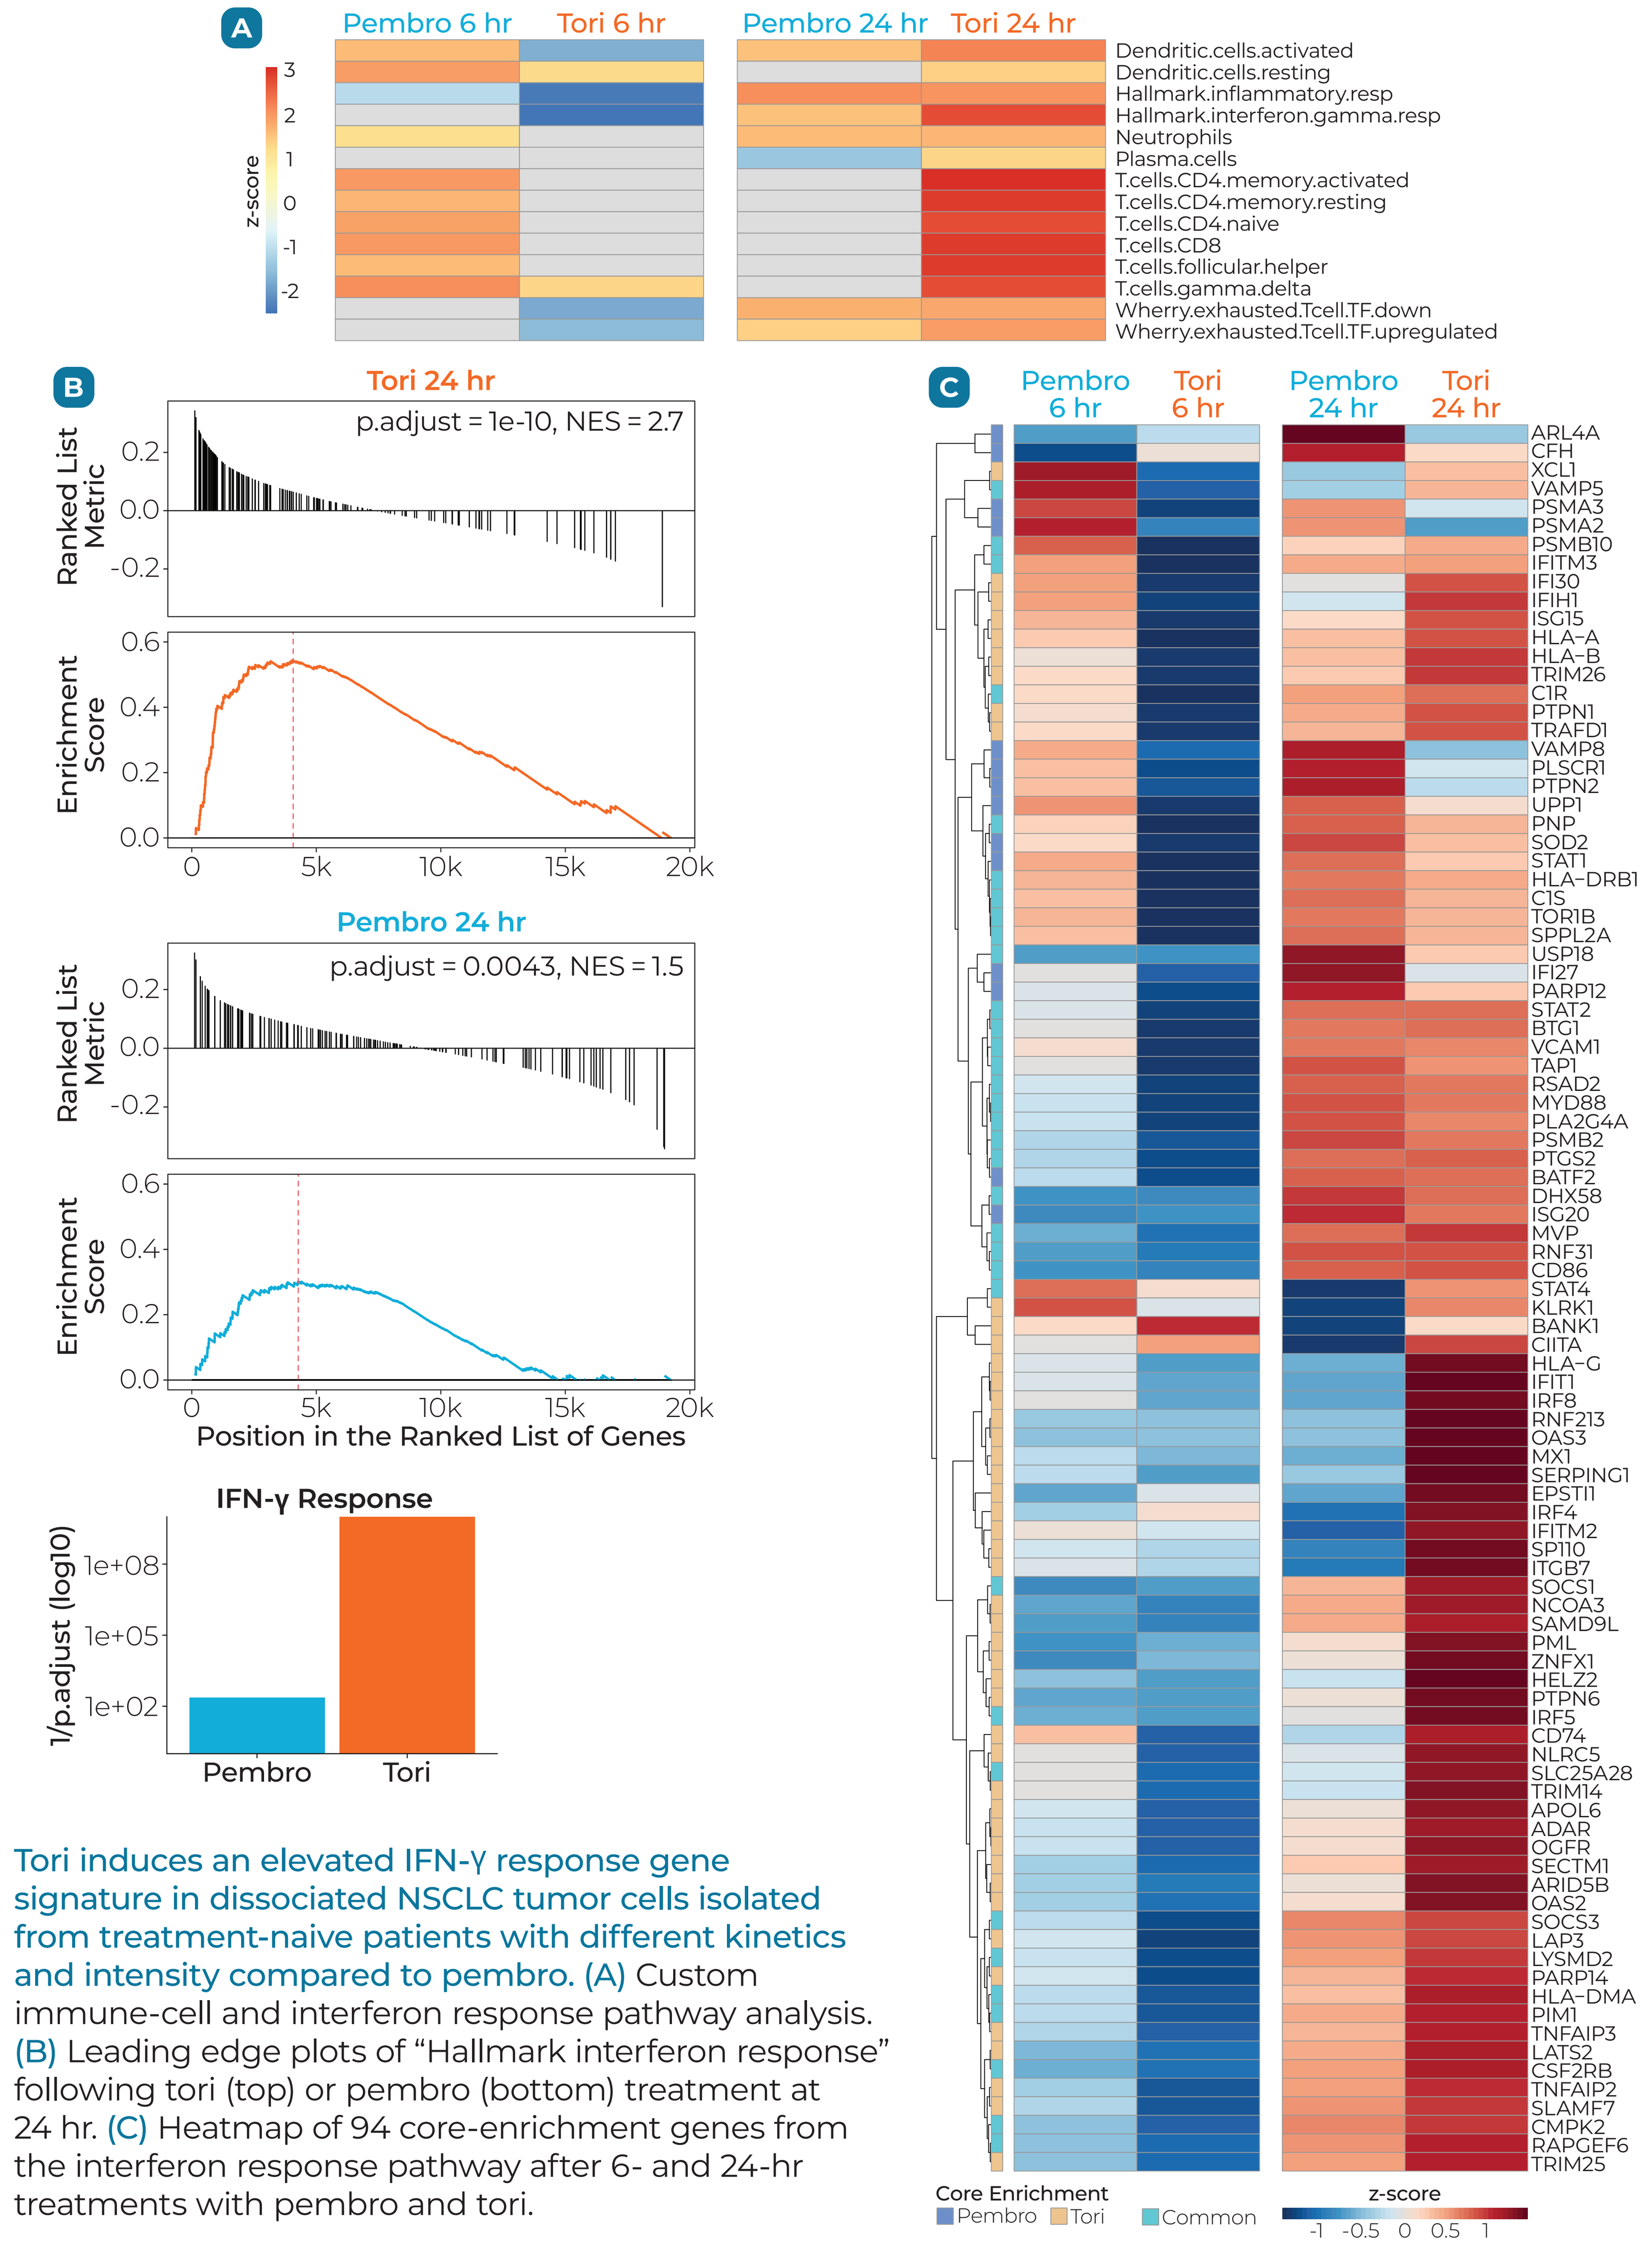

A
Pembro 6 hr
Tori 6 hr
Pembro 24 hr
Tori 24 hr
z-score
Dendritic.cells.activated
Dendritic.cells.resting
Hallmark.inflammatory.resp
Hallmark.interferon.gamma.resp
Neutrophils
Plasma.cells
T.cells.CD4.memory.activated
T.cells.CD4.memory.resting
T.cells.CD4.naive
T.cells.CD8
T.cells.follicular.helper
T.cells.gamma.delta
Wherry.exhausted.Tcell.TF.down
Wherry.exhausted.Tcell.TF.upregulated
B
Tori 24 hr
p.adjust = 1e-10, NES = 2.7
Pembro 24 hr
p.adjust = 0.0043, NES = 1.5
Ranked List Metric
Enrichment Score
Position in the Ranked List of Genes
IFN-γ Response
1/p.adjust (log10)
Pembro
Tori
C
Pembro 6 hr
Tori 6 hr
Pembro 24 hr
Tori 24 hr
ARL4A
CFH
XCL1
VAMP5
PSMA3
PSMA2
PSMB10
IFITM3
IFI30
IFIH1
ISG15
HLA-A
HLA-B
TRIM26
C1R
PTPN1
TRAFD1
VAMP8
PLSCR1
PTPN2
UPP1
PNP
SOD2
STAT1
HLA-DRB1
C1S
TOR1B
SPPL2A
USP18
IFI27
PARP12
STAT2
BTG1
VCAM1
TAP1
RSAD2
MYD88
PLA2G4A
PSMB2
PTGS2
BATF2
DHX58
ISG20
MVP
RNF31
CD86
STAT4
KLRK1
BANK1
CIITA
HLA-G
IFIT1
IRF8
RNF213
OAS3
MX1
SERPING1
EPSTI1
IRF4
IFITM2
SP110
ITGB7
SOCS1
NCOA3
SAMD9L
PML
ZNFX1
HELZ2
PTPN6
IRF5
CD74
NLRC5
SLC25A28
TRIM14
APOL6
ADAR
OGFR
SECTM1
ARID5B
OAS2
SOCS3
LAP3
LYSMD2
PARP14
HLA-DMA
PIM1
TNFAIP3
LATS2
CSF2RB
TNFAIP2
SLAMF7
CMPK2
RAPGEF6
TRIM25
Core Enrichment
Pembro
Tori
Common
z-score

Tori induces an elevated IFN-γ response gene signature in dissociated NSCLC tumor cells isolated from treatment-naive patients with different kinetics and intensity compared to pembro. (A) Custom immune-cell and interferon response pathway analysis. (B) Leading edge plots of "Hallmark interferon response" following tori (top) or pembro (bottom) treatment at 24 hr. (C) Heatmap of 94 core-enrichment genes from the interferon response pathway after 6- and 24-hr treatments with pembro and tori.

Conclusions

- Toripalimab in combination with chemotherapy significantly improves overall survival irrespective of PD-L1 status.
- Toripalimab binds a unique epitope (FG loop) on PD-1 compared to pembrolizumab (C'D loop). (Liu, et al. *MAbs*, 2019)
- Toripalimab exhibits a 12-fold higher binding affinity for PD-1 compared to pembrolizumab.
- Toripalimab is more potent than pembrolizumab in enhancing levels of Th1 cytokines and cytotoxicity.
- In comparison to pembrolizumab, binding of toripalimab to PD-1 induces lower levels of SHP1 and SHP2 recruitment.
- Toripalimab induces an elevated IFN-γ response gene signature in NSCLC dissociated tumor cells compared to pembrolizumab.

Data demonstrate toripalimab as a next-generation anti-PD-1 checkpoint inhibitor that warrants future multiregional clinical trials to evaluate its efficacy in combination with chemotherapy and other combination treatments in multiple cancer types.

C069 • Presented at AACR-NCI-EORTC 2023