CHS-114, an Anti-CCR8 Cytolytic Monoclonal Antibody Demonstrates Selective Intratumoral Treg Depletion and Favorable Immune Remodeling in Participants With Advanced Solid Tumors

Xiaoguang Wang, Xiao Wang, Daniel J. Chin, Federico Baruffaldi, Hong Tang, Koho Iizuka, Rosh Dias, Theresa LaVallee, Narendiran Rajasekaran, Varun N. Kapoor

Coherus Oncology, Inc., Redwood City, CA

Coherus

Poster 640

INTRODUCTION

- Intratumoral regulatory T cells (itTregs) are associated with resistance to cancer therapy, including programmed cell death protein 1 (PD-1) inhibitors.
- C-C motif chemokine receptor 8 (CCR8) is a G-protein-coupled receptor that is preferentially expressed on highly immunosuppressive tumor-infiltrating Tregs.[1,2]
- Selective depletion of CCR8⁺ itTregs aims to improve cancer therapy and avoid autoimmunity associated with broad Treg depletion.[3,4]
- CCR8⁺ itTregs are prevalent in a broad range of solid tumors including head and neck squamous cell carcinoma (HNSCC), gastric cancer (GC), and colorectal cancer (CRC).
- CHS-114 is a selective, cytolytic anti-CCR8 monoclonal antibody with enhanced effector function (afucosylated) and no off-target binding.[5]
- CHS-114 has the potential to overcome immune suppression within the tumor microenvironment (TME) by eliminating the suppressive Treg cells and recruiting effector T cells, turning cold tumors to hot, and enhancing antitumor immunity when combined with other immuno-oncology agents such as the anti-PD-1 antibody toripalimab.[6]
- An ongoing Phase 1 (NCT05635643), single-agent and toripalimab (tori) combination dose escalation/expansion study is evaluating CHS-114 in advanced solid tumors including HNSCC. Additionally, a study evaluating CHS-114 in upper gastrointestinal cancers (GC, gastroesophageal junction [GEJ], esophageal adenocarcinoma [EAC]), esophageal squamous cell carcinoma (ESCC), and CRC recently began enrolling study participants (NCT06657144).
- CHS-114 has demonstrated an acceptable safety profile to date and antitumor activity in HNSCC.

CCR8⁺ Tregs Are Enriched in Several Solid Tumors

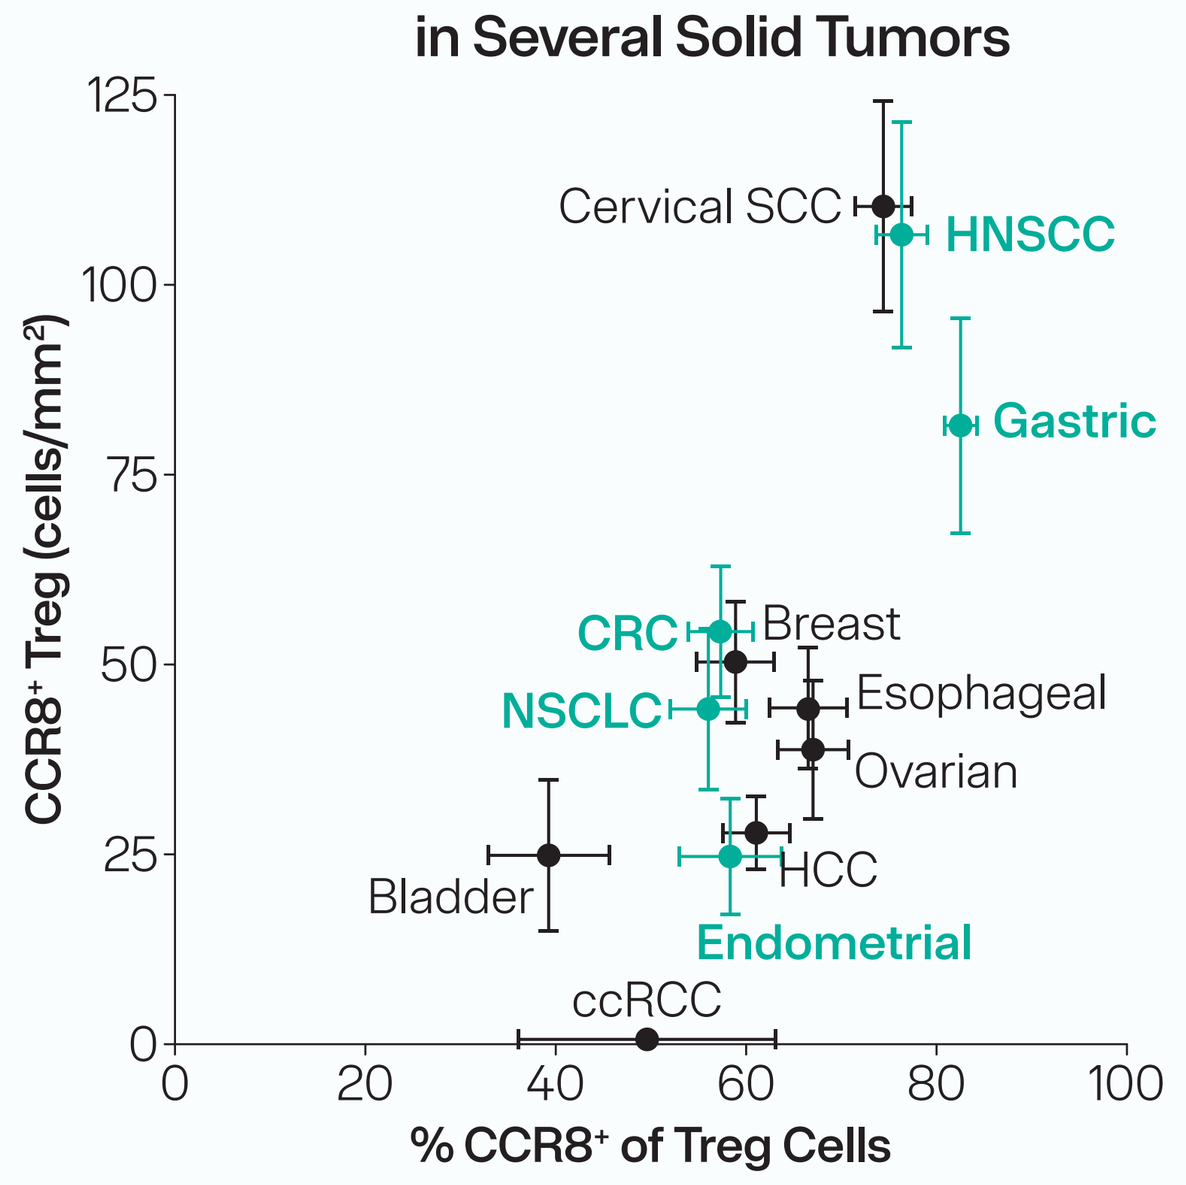
CCR8⁺ Treg (cells/mm²)
% CCR8⁺ of Treg Cells
Cervical SCC
HNSCC
Gastric
CRC
Breast
NSCLC
Esophageal
Ovarian
HCC
Bladder
Endometrial
ccRCC

Density (y-axis) and frequency (x-axis) of CCR8⁺ Tregs (of total Tregs) in tumor microarrays from 12 types of solid tumors evaluated using a multiplex immunofluorescence (mIF) assay. Tumor types in which clinical benefit has been observed with anti-CCR8 antibodies (CHS-114-101 study or competitor trials) are bolded in green. For evaluating CCR8⁺ Tregs, CCR8 and FOXP3 staining was performed.

CHS-114 Is a Selective, Cytolytic Anti-CCR8 Monoclonal Antibody

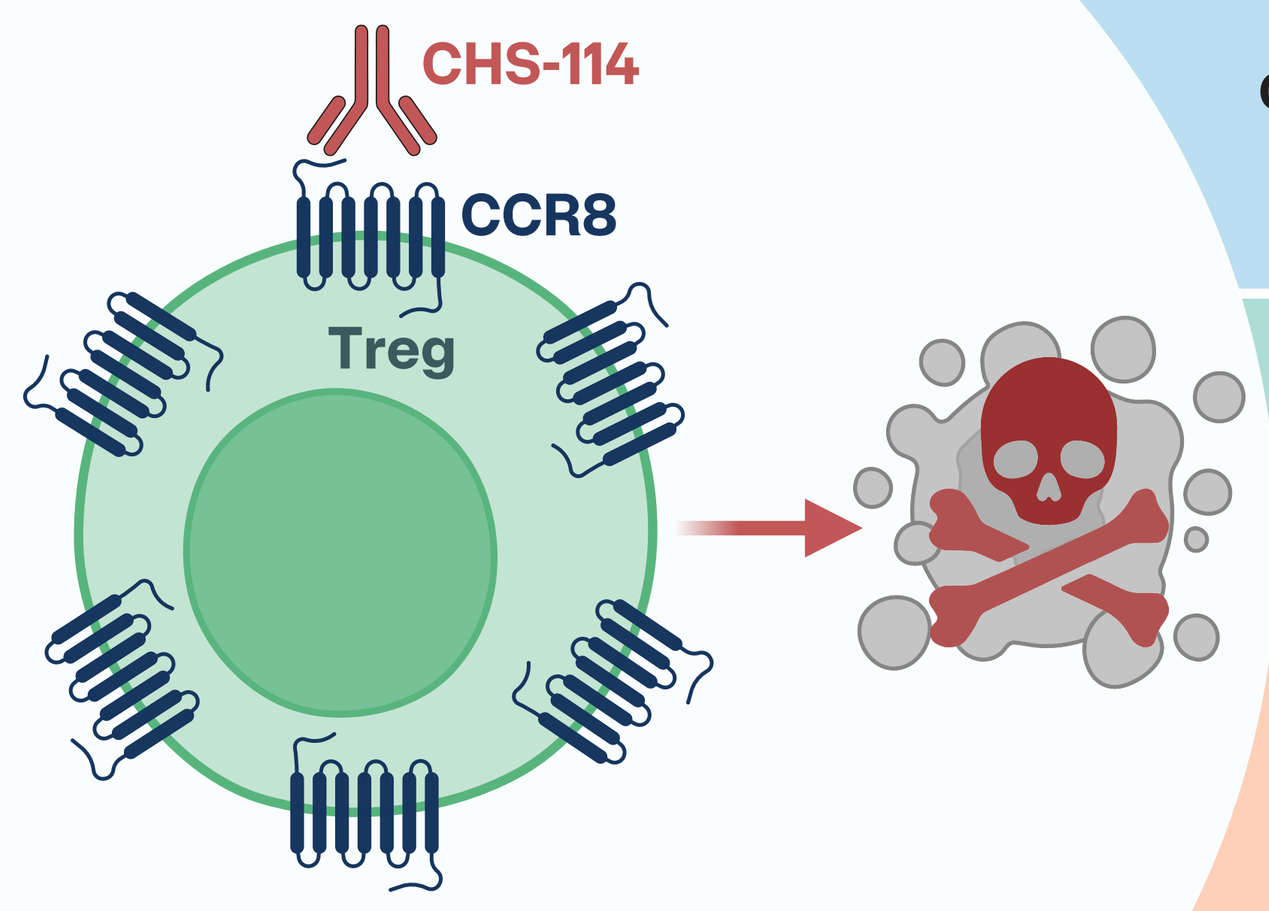
CHS-114
CCR8
Treg

Antibody Characteristics
- High affinity to CCR8 (K_D = 502 pm)
- Effector function enhanced: afucosylated for potent ADCC and ADCP activity*
- Highly selective with no off-target binding**
- Targeted therapy: "Bind and Kill" via CCR8 to deplete intratumoral Tregs

Non-Clinical Studies
- Robust CCR8⁺ Treg depletion in in vitro and in vivo studies[5]
- Tumor growth inhibition in hCCR8 knock-in murine model[5]
- Disease linkage: high CCR8⁺ Treg density observed in a majority of solid tumors

Clinical Studies
- CCR8⁺ Tregs are associated with poor prognosis and cancer therapy resistance (including PD-1 inhibitors [PD-1i]).
- Partial response in 4L PD-1 refractory HNSCC in combination with toripalimab[7]
- No dose-limiting toxicities (DLTs) during dose escalation with monotherapy or in combination treatment with toripalimab
- Robust Treg depletion and increased CD8⁺ T cells in the TME (on-treatment) as monotherapy and in combination with toripalimab

*ADCC = antibody-dependent cellular cytotoxicity; ADCP = antibody-dependent cellular phagocytosis. **Off-target binding was evaluated on a retrogenix screen against ~5500 extracellular/secreted proteins. Illustration created with BioRender.com.

METHODS

- This is a Phase 1, first-in-human, open-label single-agent and combination dose trial to evaluate CHS-114 in patients with advanced solid tumors and HNSCC (NCT05635643).
- Here we present updated interim clinical biomarker data from the HNSCC expansion cohort for CHS-114 monotherapy (Arm 1b) and the toripalimab combination (Arm 2) study; enrollment for Arm 3 is ongoing.

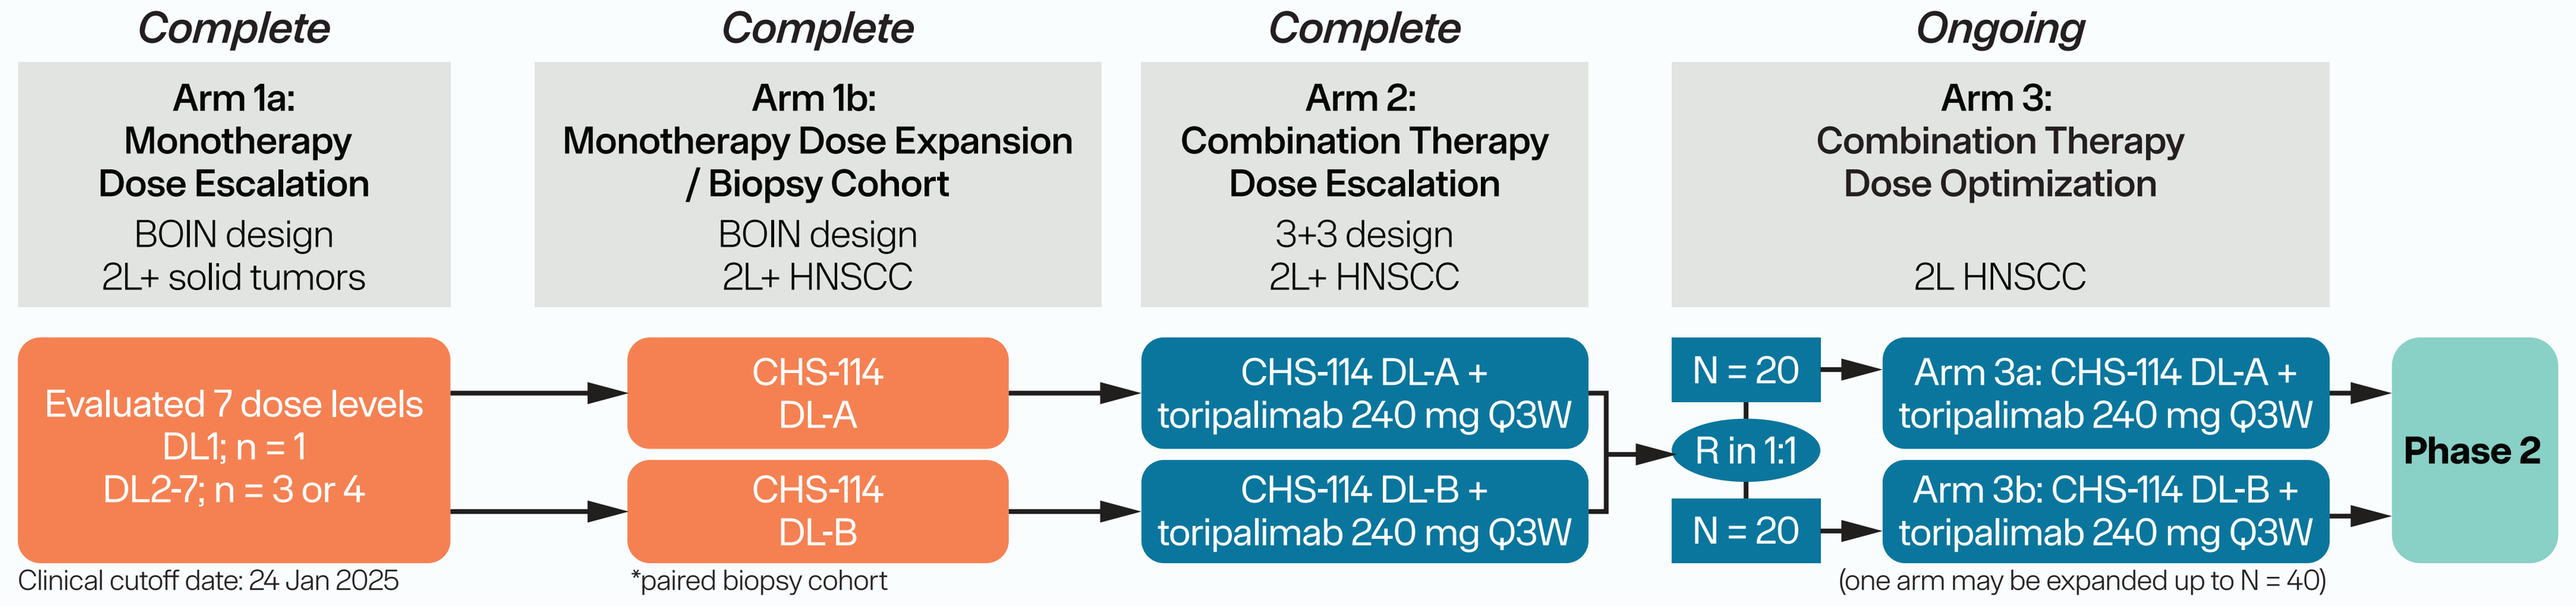
Complete
Arm 1a: Monotherapy Dose Escalation
BOIN design
2L+ solid tumors
Complete
Arm 1b: Monotherapy Dose Expansion / Biopsy Cohort
BOIN design
2L+ HNSCC
Complete
Arm 2: Combination Therapy Dose Escalation
3+3 design
2L+ HNSCC
Ongoing
Arm 3: Combination Therapy Dose Optimization
2L HNSCC
Evaluated 7 dose levels
DL1; n = 1
DL2-7; n = 3 or 4
CHS-114 DL-A
CHS-114 DL-B
CHS-114 DL-A + toripalimab 240 mg Q3W
CHS-114 DL-B + toripalimab 240 mg Q3W
N = 20
R in 1:1
N = 20
Arm 3a: CHS-114 DL-A + toripalimab 240 mg Q3W
Arm 3b: CHS-114 DL-B + toripalimab 240 mg Q3W
Phase 2
Clinical cutoff date: 24 Jan 2025
*paired biopsy cohort
(one arm may be expanded up to N = 40)

- 20 heavily pre-treated participants
- No DLTs reported up to dose level (DL) 7 (1200 mg)
- Stable disease observed in NSCLC, OC, PDAC, EC (~ 1 yr), and CRC
- Disease control rate: 47%
- Pharmacologically active doses identified based on safety and immune biomarkers (≥ DL5)

- 20 evaluable HNSCC participants (13 monotherapy, 7 in combination)
- Acceptable safety profile and no DLTs with and without toripalimab
- Intratumoral biomarker data (Arm1b): Robust CCR8 Treg cell depletion and increase in CD8 T cell numbers
- 1 partial response in CHS-114/toripalimab combo (4L; PD-1i refractory)

Additional Platform Study
- Upper gastrointestinal tumors (GC, GEJ, EAC), ESCC, and CRC (NCT06657144)
- Toripalimab and other combinations

RESULTS

CHS-114-Mediated Robust and Selective Peripheral CCR8⁺ Treg Depletion in HNSCC Participants Treated With and Without Toripalimab

Robust Target Depletion (Proof of Mechanism) — **CHS-114 Spares Non-CCR8⁺ Tregs, CD4⁺ T, and CD8⁺ T Cells (Proof of Selectivity)**

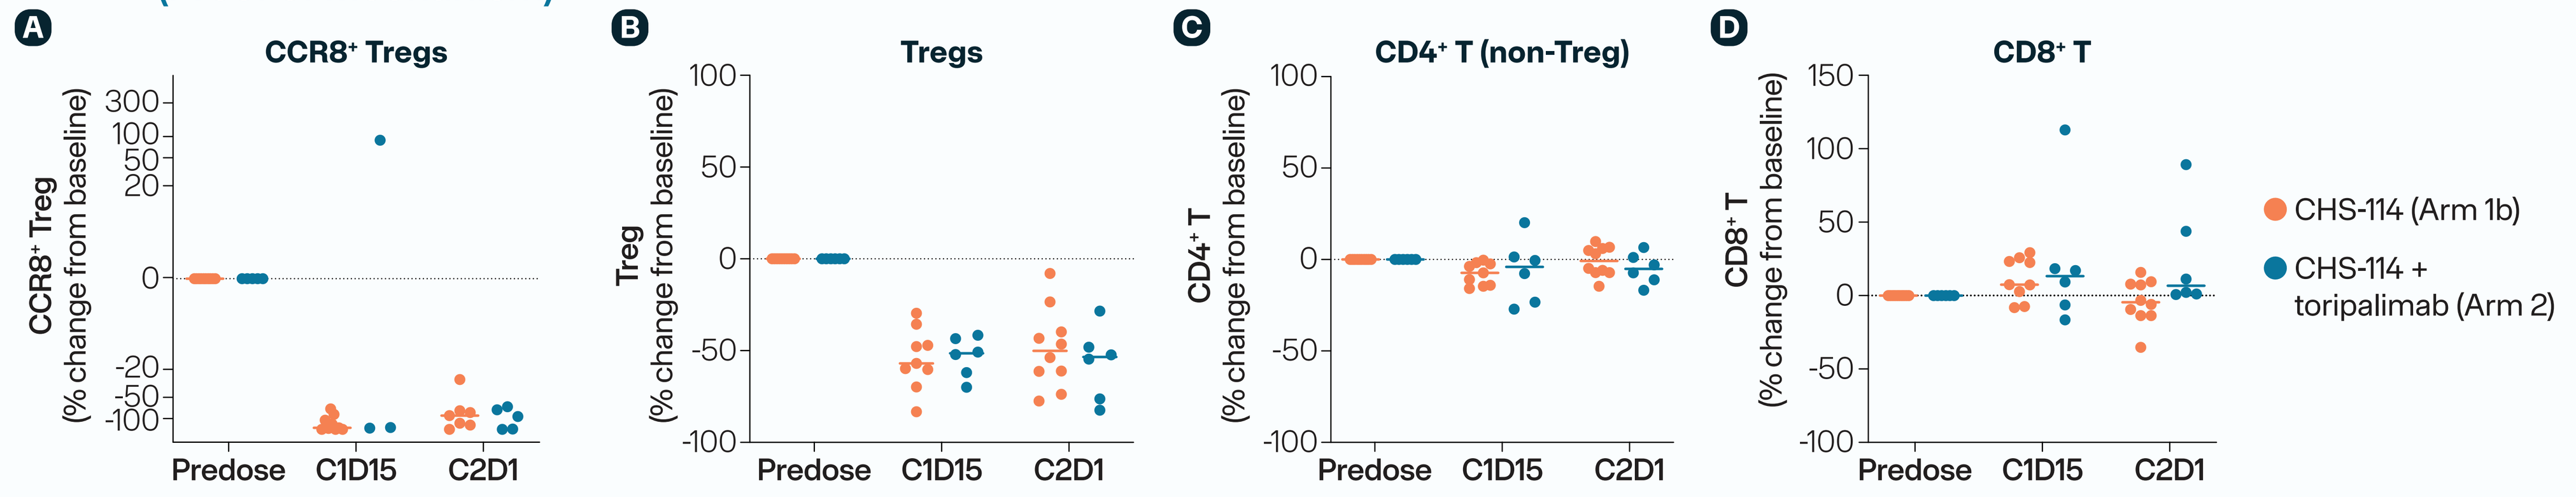
A
CCR8⁺ Tregs
CCR8⁺ Treg (% change from baseline)
B
Tregs
Treg (% change from baseline)
C
CD4⁺ T (non-Treg)
CD4⁺ T (% change from baseline)
D
CD8⁺ T
CD8⁺ T (% change from baseline)
Predose
C1D15
C2D1
CHS-114 (Arm 1b)
CHS-114 + toripalimab (Arm 2)

CCR8⁺ Tregs and other immune cells in PBMCs of HNSCC participants were evaluated by Cytek flow cytometry at indicated time points in CHS-114-101 monotherapy dose expansion and toripalimab combination dose escalation cohorts. **(A)** Robust CCR8⁺ Treg depletion in the periphery was observed through Cycle 1. CHS-114 treatment with and without toripalimab led to a decrease in the subset of Tregs, while preserving broader Treg **(B)**, non-Treg CD4⁺ **(C)**, and CD8⁺ **(D)** T cell populations, confirming the specificity of CHS-114 for CCR8⁺ Tregs. Tregs: CD127^{lo} CD25^{hi} within CD3⁺ CD4⁺ T cells; non-Treg CD4 and CD8⁺ T: CD56⁻ FOXP3⁻ CD3⁺. n = 10 (CHS-114) and n = 6 (CHS-114 + toripalimab) at baseline.

CHS-114 Administration Promotes Peripheral Immune Activation in HNSCC Participants

A — CHS-114 Promotes Peripheral CD8⁺ T Activation — **B — CHS-114 Increases Inflammatory Serum Cytokine Levels**

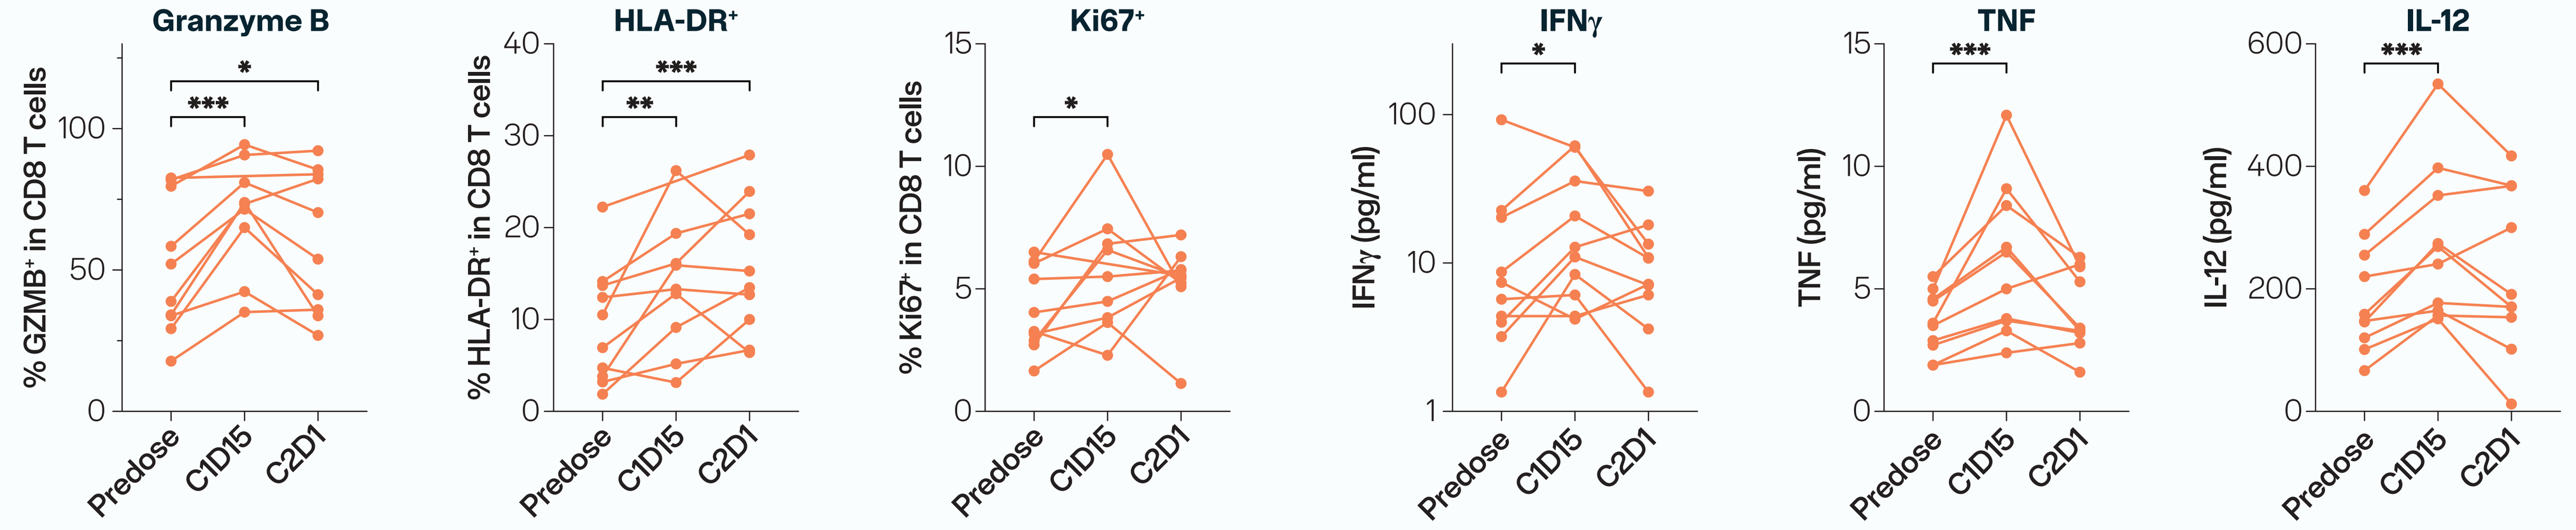
Granzyme B
% GZMB⁺ in CD8 T cells
HLA-DR⁺
% HLA-DR⁺ in CD8 T cells
Ki67⁺
% Ki67⁺ in CD8 T cells
IFNγ
IFNγ (pg/ml)
TNF
TNF (pg/ml)
IL-12
IL-12 (pg/ml)
Predose
C1D15
C2D1

Immune profiling of PBMC using Cytek flow cytometry **(A)** and serum cytokine analysis by MSD **(B)** at indicated time points for participants enrolled in CHS-114-101 HNSCC monotherapy dose expansion. **(A)** Increases in CD8⁺ T cell cytotoxicity (granzyme B [GZMB]), activation (HLA-DR), and proliferation (Ki67) compared with pretreatment levels were observed. **(B)** Increases in serum inflammatory and Th1 cytokines (IFNγ, TNF, IL-12) were also observed. The data are consistent with CHS-114 promoting peripheral immune activation in HNSCC participants. Participants with longitudinal samples are plotted (C1D1 [predose], C1D15, and C2D1); n = 10 at baseline. Statistical tests: Mixed-effect analysis (A) and paired t-test (B).

Synergistic Activity of CHS-114 and Toripalimab on Peripheral Immune Activation in HNSCC Participants

CHS-114-Mediated Peripheral Immune Activation Is Further Augmented by Toripalimab

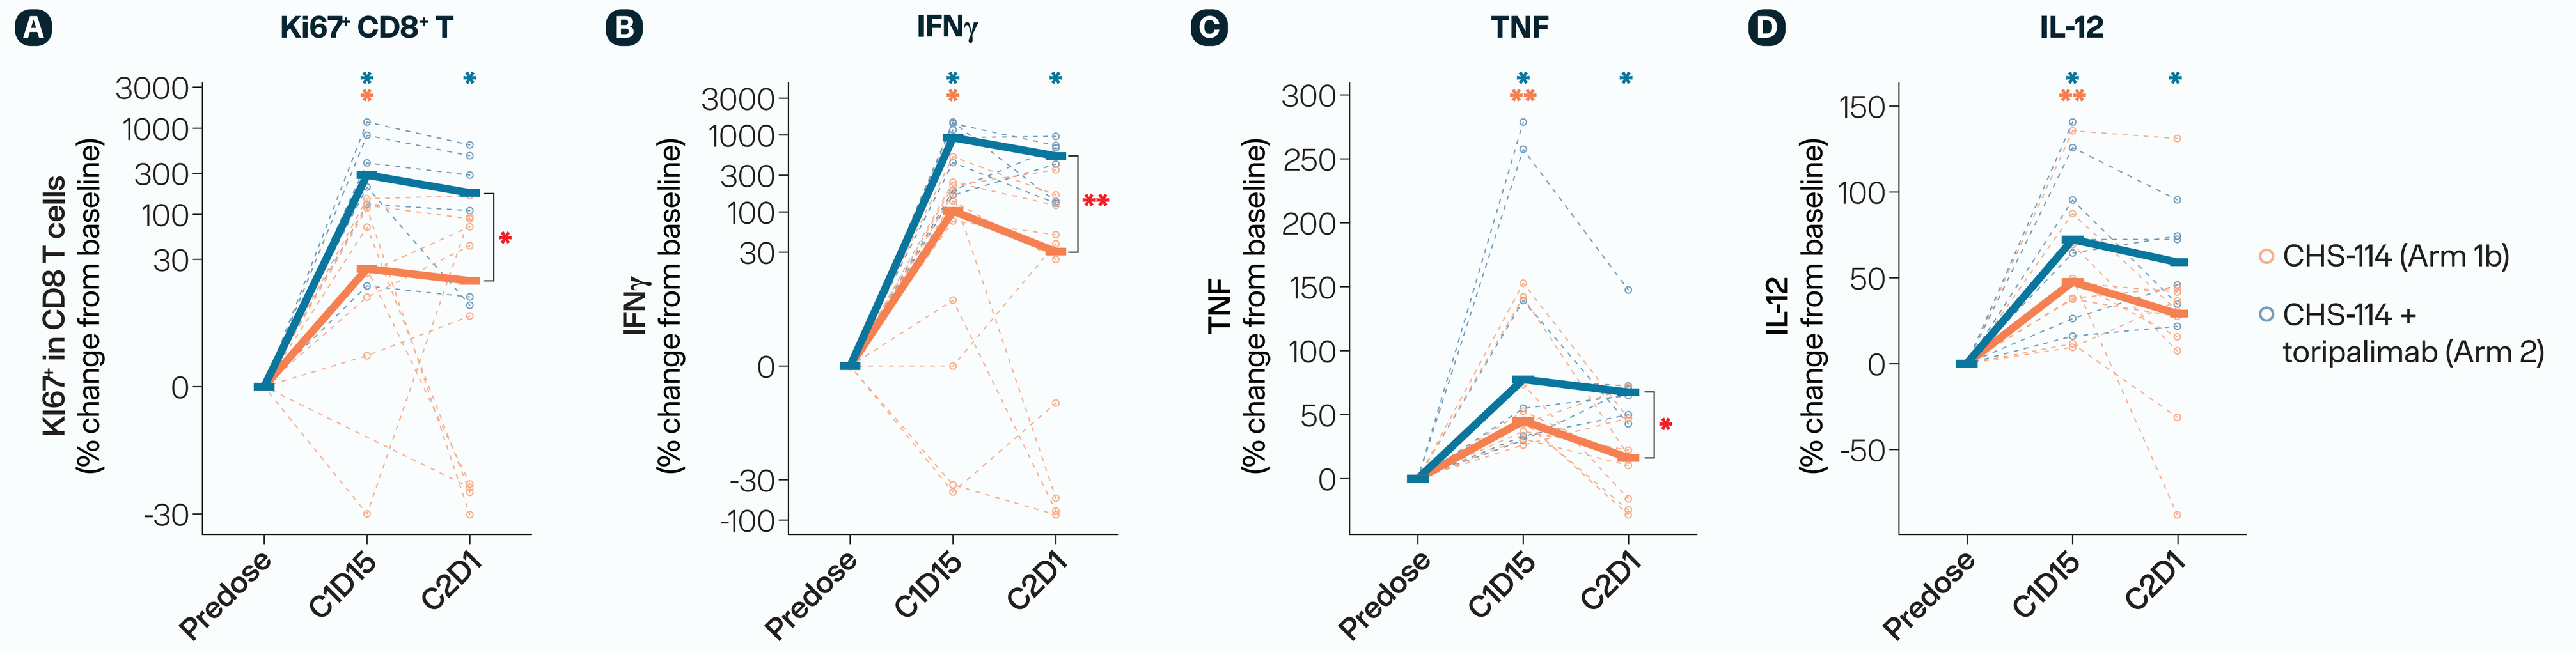
A
Ki67⁺ CD8⁺ T
Ki67⁺ in CD8 T cells (% change from baseline)
B
IFNγ
IFNγ (% change from baseline)
C
TNF
TNF (% change from baseline)
D
IL-12
IL-12 (% change from baseline)
Predose
C1D15
C2D1
CHS-114 (Arm 1b)
CHS-114 + toripalimab (Arm 2)

Ki67⁺ CD8⁺ T	C1D15	C2D1
CHS-114	23%	25%
CHS-114 + tori	302%	198%

IFNγ	C1D15	C2D1
CHS-114	108%	31%
CHS-114 + tori	922%	549%

TNF	C1D15	C2D1
CHS-114	45%	16%
CHS-114 + tori	78%	68%

IL-12	C1D15	C2D1
CHS-114	48%	29%
CHS-114 + tori	72%	59%

Immune profiling of PBMC using Cytek flow cytometry **(A)** and serum cytokine analysis by MSD **(B, C, D)** at indicated time points for participants enrolled in CHS-114-101 HNSCC monotherapy dose expansion (orange) or toripalimab (tori) combination dose escalation cohorts (blue). CHS-114 with toripalimab mediated a robust increase in **(A)** CD8⁺ T cell proliferation (Ki67) and **(B-D)** Th1 inflammatory cytokines that was sustained through the dosing cycle. The data support the combination of CHS-114 with toripalimab to enhance immune responses (orange vs blue). Participants with longitudinal samples are plotted (C1D1 [predose], C1D15, and C2D1); thick solid lines (median values); thin dashed lines (individual participants). Asterisks (orange [CHS-114]; blue: [CHS-114 + tori]) denote significant percent changes from C1D1 (one-sample Wilcoxon). Red asterisks denote significant differences between arms at C2D1 (Wilcoxon test). Tables show median percent change from baseline (n = 10 for CHS-114 and n = 6 for CHS-114 + tori at baseline).

Abbreviations: 2L/4L = second/fourth line; BOIN = Bayesian optimal interval design; C1D1 = Cycle 1 Day 1, predose; C1D15 = Cycle 1 Day 15; C2D1 = Cycle 2 Day 1, predose; CCR8 = chemokine receptor 8; ccRCC = clear cell renal cell carcinoma; CRC = colorectal cancer; DL = dose level; DLT = dose-limiting toxicity; EC = endometrial cancer; GC = gastric cancer; HCC = hepatic cell carcinoma; HNSCC = head and neck squamous cell carcinoma; itTreg = intratumoral regulatory T cell; mIF = multiplex immunofluorescence; MSD = Meso Scale Discovery; NSCLC = non-small cell lung cancer; OC ovarian cancer; PBMC = peripheral blood mononuclear cells; PD-1 = programmed cell death protein 1; PD-1i = PD-1 inhibitor; PDAC = pancreatic ductal adenocarcinoma; Q3W = every 3 weeks; SCC = squamous cell carcinoma; TME = tumor micro-environment; tori = toripalimab; TNF = tumor necrosis factor; Treg = regulatory T cell.

References: **1)** De Simone et al. *Immunity* 2016;45(5):1135-47. **2)** Plitas et al. *Immunity* 2016;45(5):1122-34. **3)** Kidani Y, et al. *Proc Natl Acad Sci USA* 2022;119(7):e2114282119. **4)** Haruna M, et al. *Sci Rep* 2022;12(1):5377. **5)** Wang X, et al. *J Immunother Cancer* 2024;12(Suppl 2): A1–A1683. **6)** Rajasekaran N, et al. *Int J Radiat Oncol Biol Phys* 2024;118(5):e89. **7)** Worden F et al. *Cancer Res* 2025;85 (8_Supplement_2): CT038.

Statistics: * = p < 0.05; ** = p < 0.01; *** = p < 0.001; **** = p < 0.0001; when not shown, comparisons are not statistically significant.

CHS-114 Treatment Depletes CCR8⁺ itTregs and Increases CD8⁺ T Cells in HNSCC Tumors — Turns Tumors "Hot"

A — CHS-114 Treatment Remodels the Tumor Microenvironment (TME)

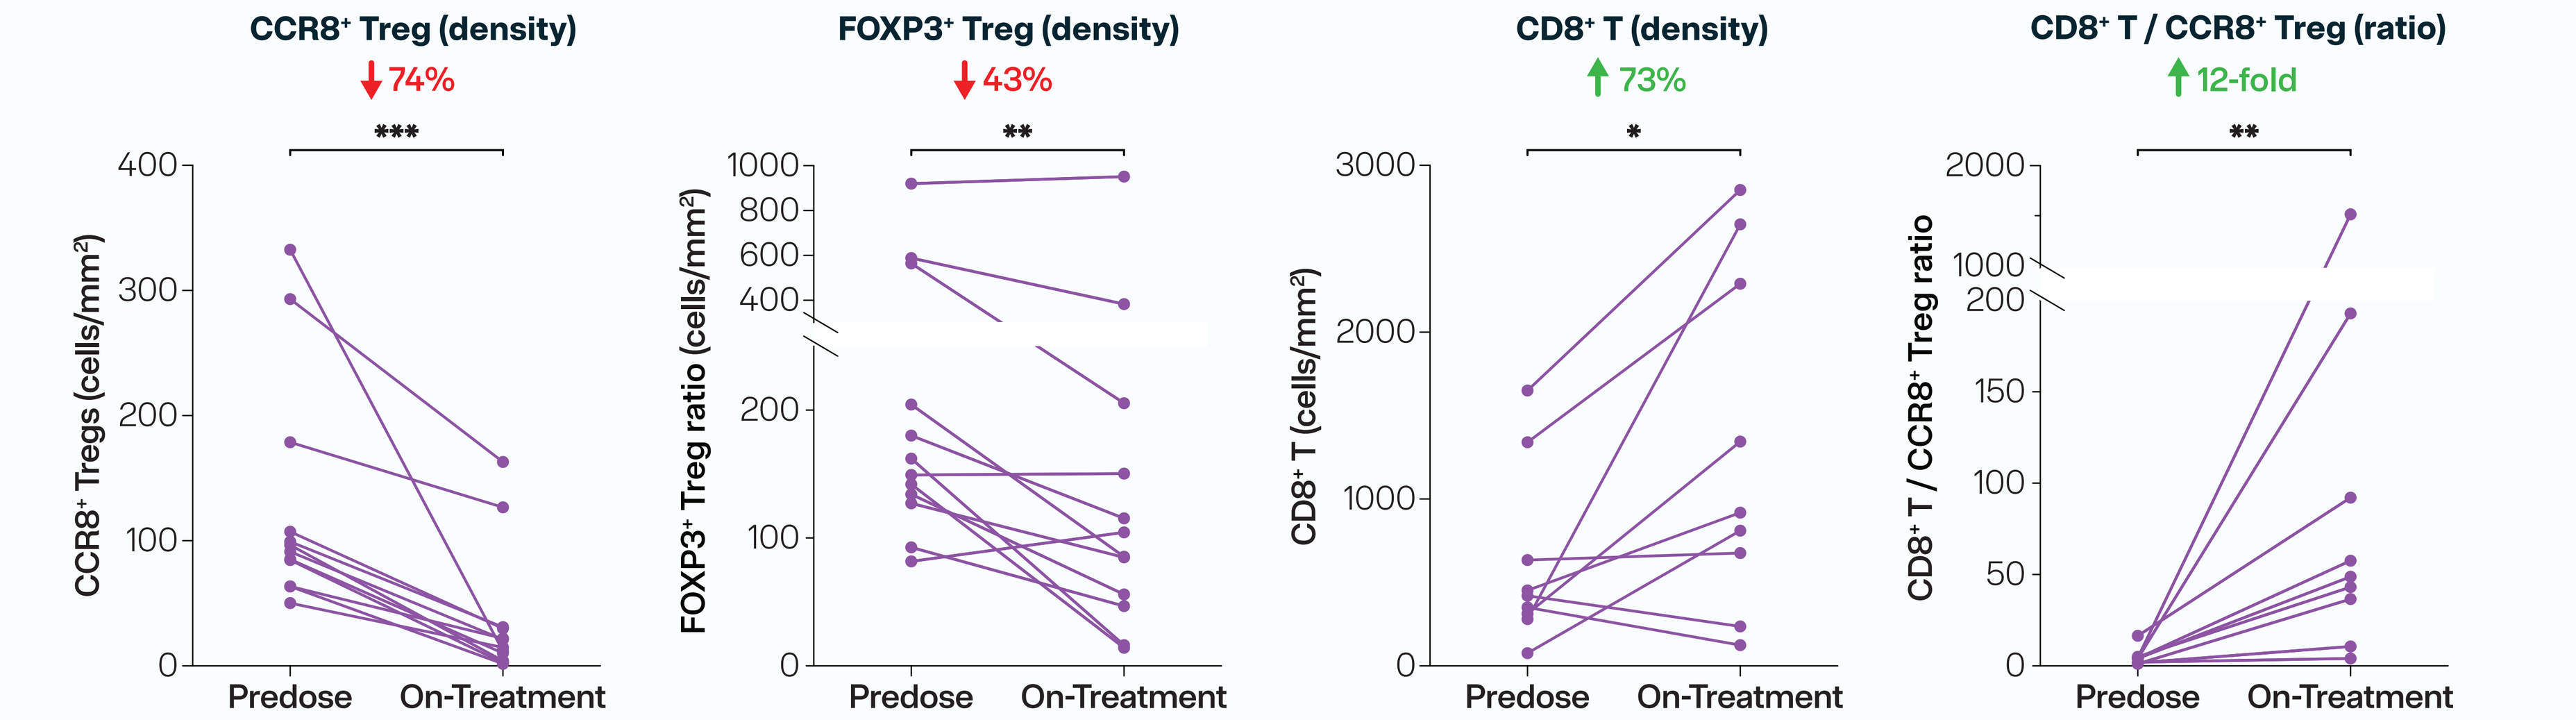
CCR8⁺ Treg (density)
↓74%
CCR8⁺ Tregs (cells/mm²)
FOXP3⁺ Treg (density)
↓43%
FOXP3⁺ Treg ratio (cells/mm²)
CD8⁺ T (density)
↑73%
CD8⁺ T (cells/mm²)
CD8⁺ T / CCR8⁺ Treg (ratio)
↑12-fold
CD8⁺ T / CCR8⁺ Treg ratio
Predose
On-Treatment

B — Remodeled TME Following CHS-114 Treatment

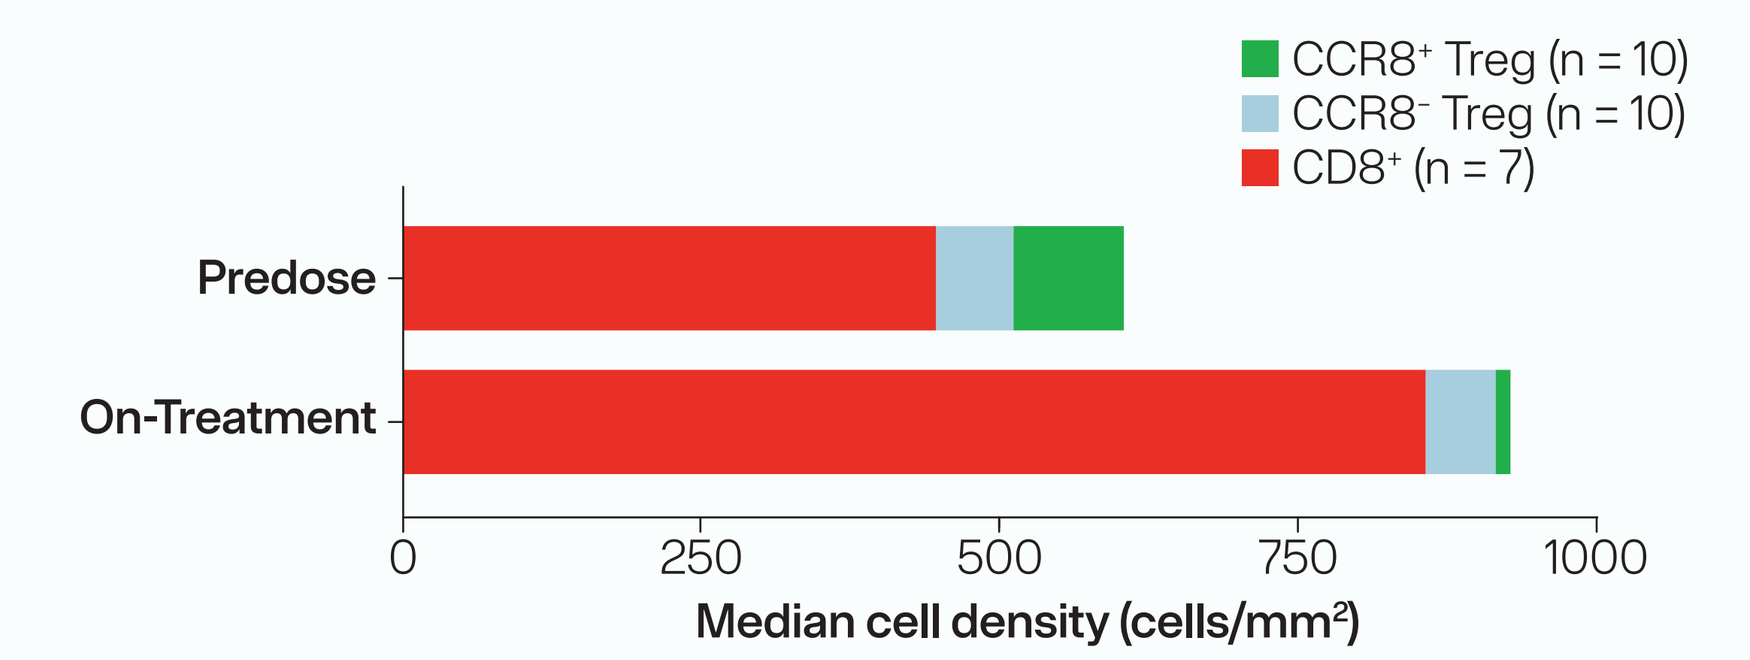
CCR8⁺ Treg (n = 10)
CCR8⁻ Treg (n = 10)
CD8⁺ (n = 7)
Predose
On-Treatment
Median cell density (cells/mm²)

C — CHS-114 selectively depletes CCR8⁺ Tregs in the TME

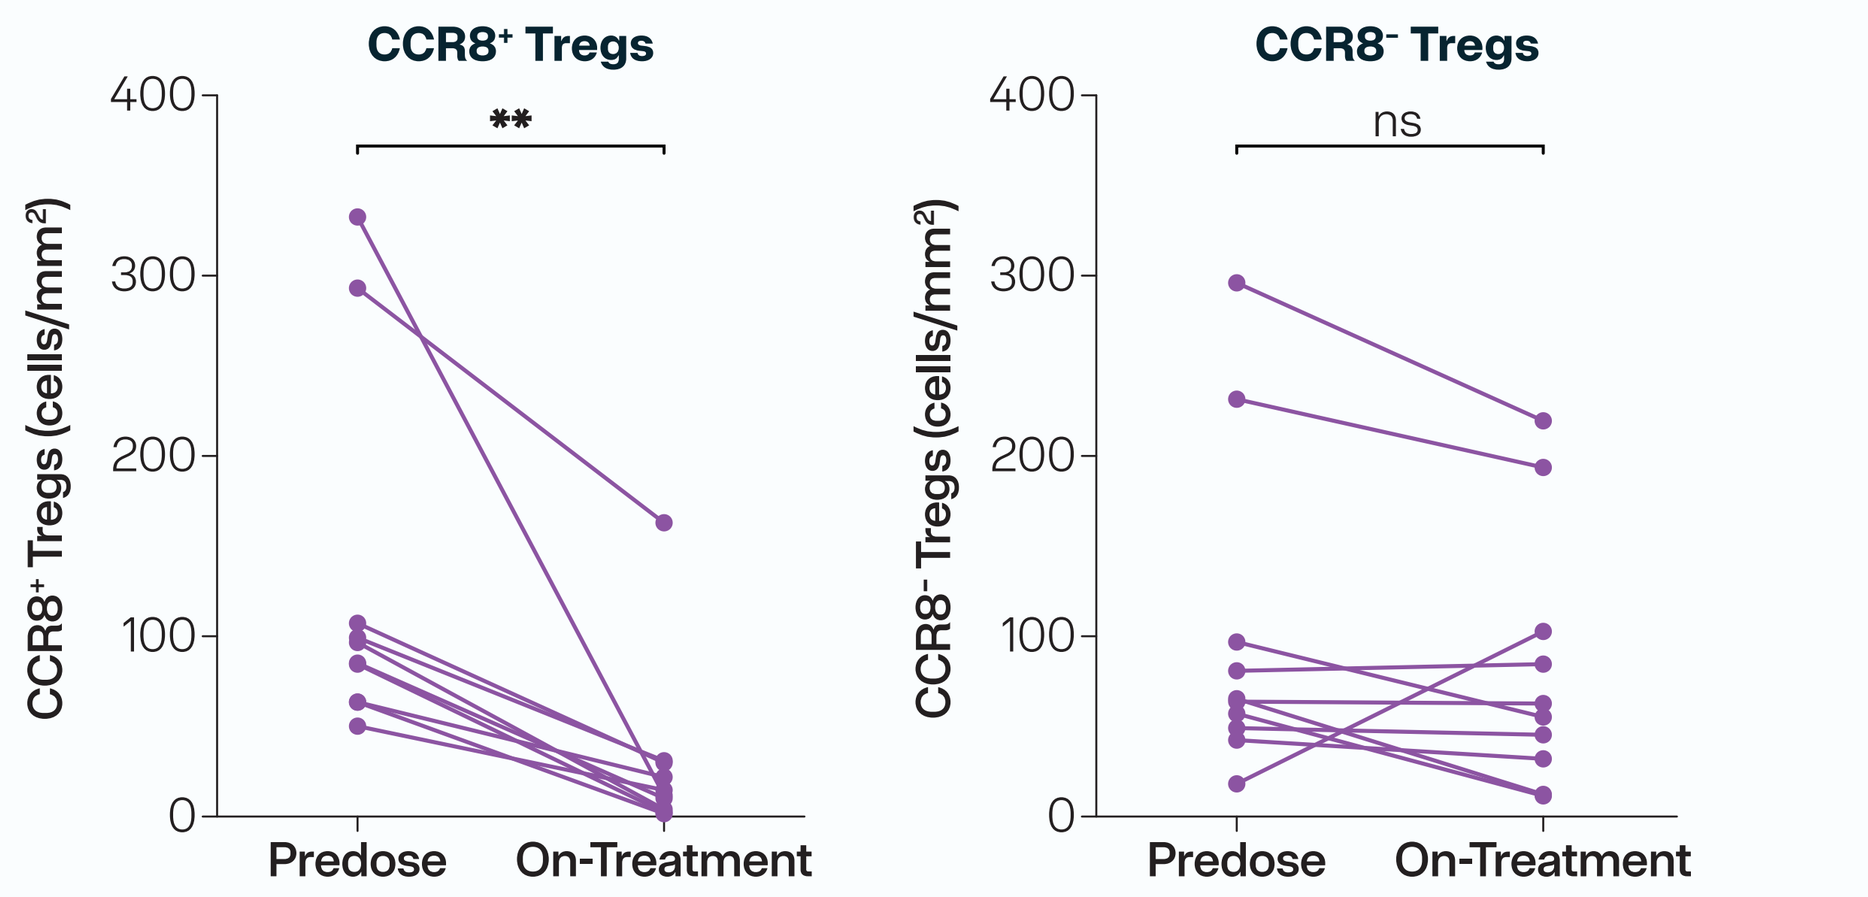
CCR8⁺ Tregs
CCR8⁺ Tregs (cells/mm²)
CCR8⁻ Tregs
CCR8⁻ Tregs (cells/mm²)
ns
Predose
On-Treatment

D — CHS-114 Monotherapy Treatment Promotes An Inflamed TME

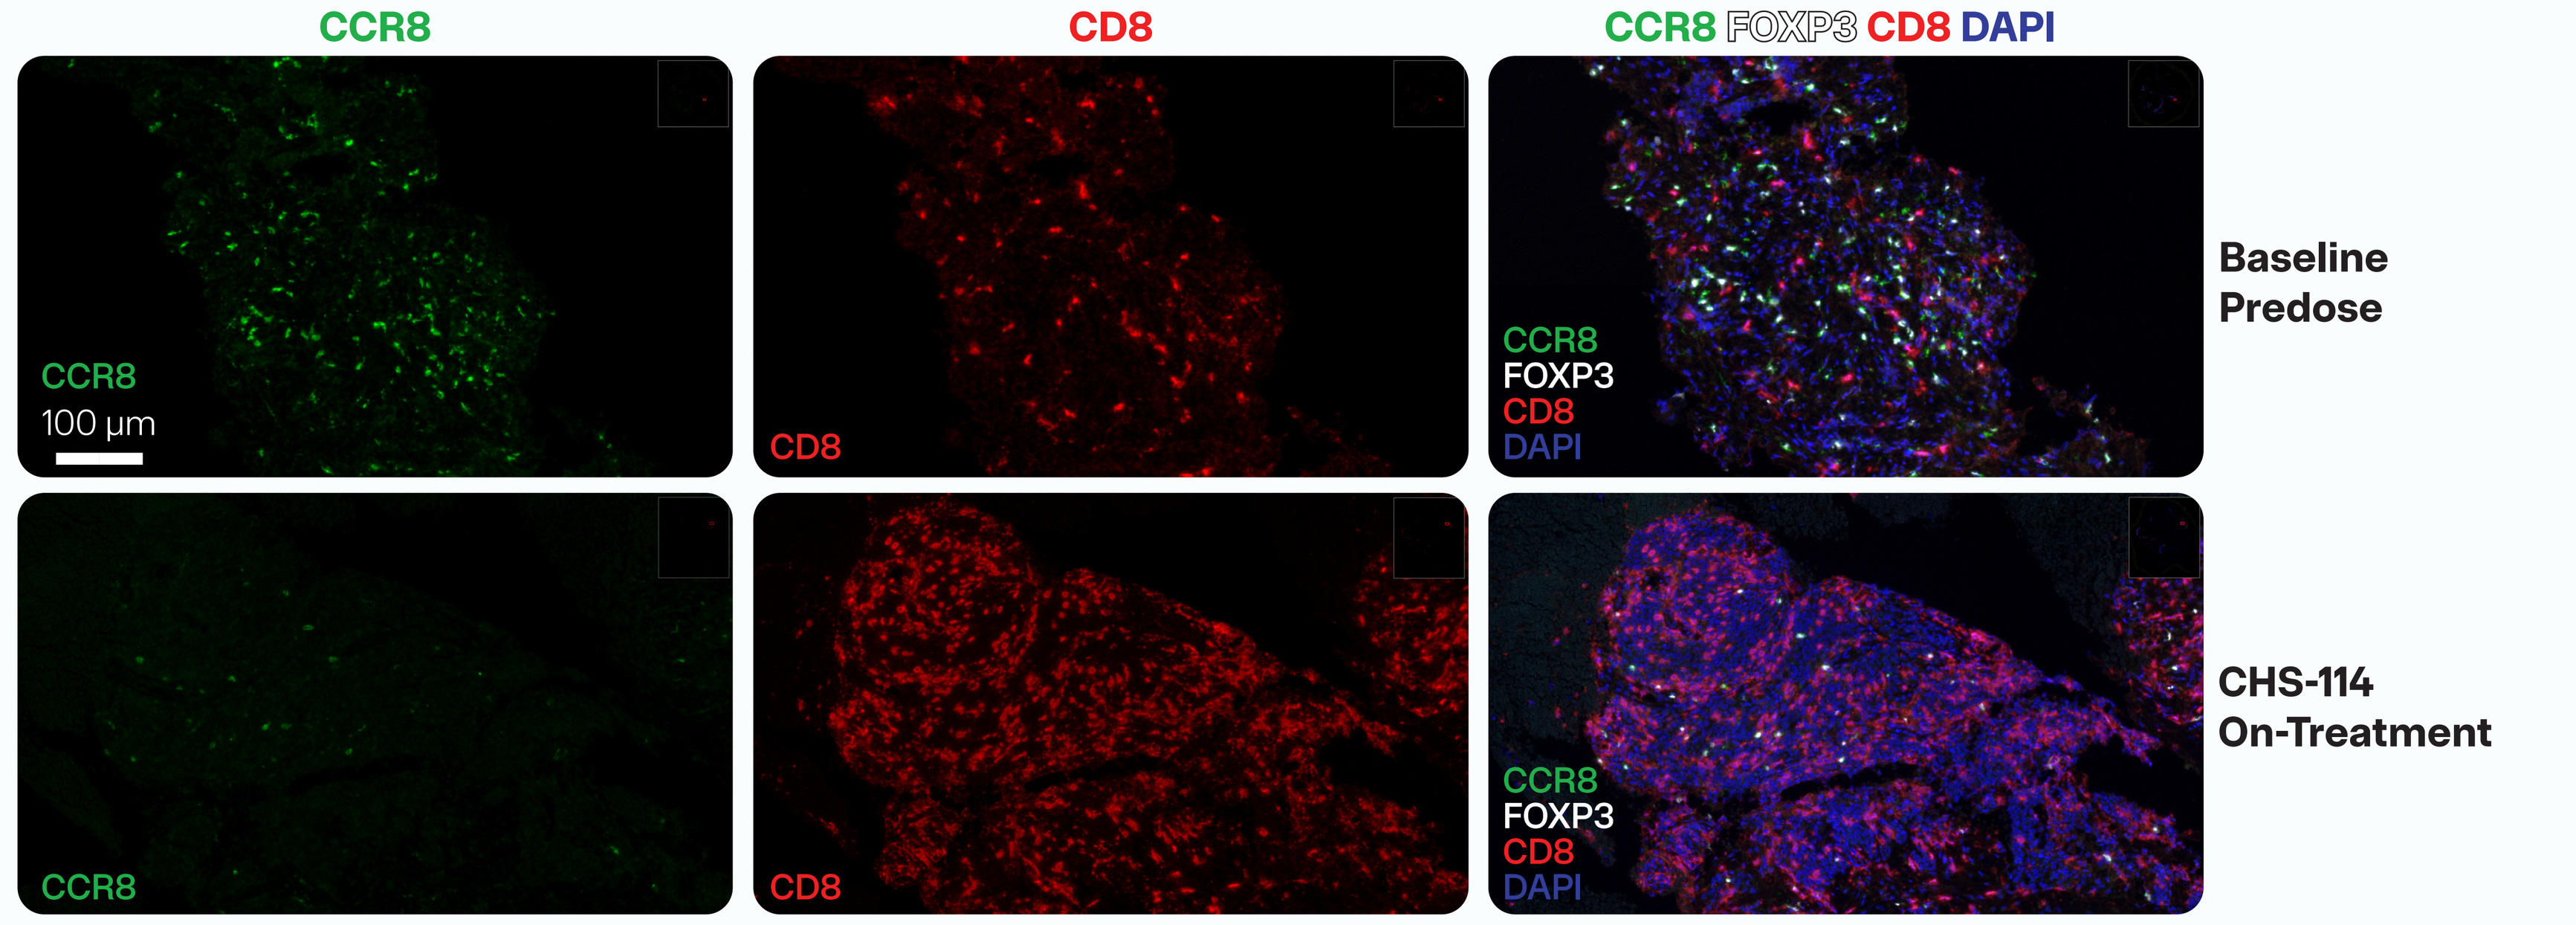
CCR8
CD8
CCR8 FOXP3 CD8 DAPI
100 μm
Baseline Predose
CHS-114 On-Treatment

A validated mIF assay was performed on predose and on-treatment (post-Cycle 2 dosing) tumor tissue samples from HNSCC participants in the CHS-114-101 monotherapy dose expansion (n = 10) and toripalimab combination dose escalation studies (n = 2). **(A)** Density of CCR8⁺ Tregs (CCR8⁺ FOXP3⁺ CD8⁻; n = 12), FOXP3⁺ Tregs (FOXP3⁺ CD8⁻; n = 12), and CD8⁺ T cells (CD8⁺ CCR8⁻ FOXP3⁻; n = 9) and fold change of ratio of CD8⁺ cells to CCR8⁺ Tregs (n = 9) are depicted. **(B)** Median cell density of indicated cell populations is shown predose and on CHS-114 monotherapy treatment. **(C)** Comparing density of CCR8⁺ (pos) and CCR8⁻ (neg) Tregs within the TME predose and on CHS-114 monotherapy treatment (n = 10). CHS-114-mediated depletion of CCR8⁺ Tregs, but not CCR8⁻ Treg cells, in the TME, confirming CHS-114 specificity within the TME. **(D)** Representative images from the mIF assay show that CHS-114 remodeled the TME in favor of antitumor immunity by increasing the ratio of CD8⁺ T cells (cytotoxic T lymphocytes) to CCR8⁺ Tregs, confirming the mechanism of action.

CONCLUSIONS

- CHS-114 mediated robust and selective depletion of peripheral CCR8⁺ Tregs, accompanied by immune activation in HNSCC study participants. These findings further support the two doses selected for the dose expansion studies.
- CHS-114-mediated immune activation is significantly enhanced and sustained with toripalimab in HNSCC study participants.
- In on-treatment tumor biopsies, CHS-114 depleted CCR8⁺ Tregs and increased CD8⁺ T cells in the TME, indicating favorable TME remodeling and establishing proof of mechanism.
- To date, CHS-114 with and without toripalimab has a manageable safety profile and promising early antitumor activity (with toripalimab) in patients with HNSCC.
- Taken together, these data support further evaluation of CHS-114 in combination with other drugs, including toripalimab; a 2L PD-1i-experienced HNSCC CHS-114 study with toripalimab dose optimization is ongoing to support recommended Phase 2 dose selection and to evaluate whether CCR8⁺ Tregs are the primary mechanism of PD-1i resistance in HNSCC.