Characterization of CHS-1000, a Humanized Anti-ILT4 Monoclonal Antibody for Reprogramming Suppressive Myeloid Cells in Solid Tumors

Poster 1364

Narendiran Rajasekaran, Xiaoguang Wang, Scott Klakamp, Sruthi Ravindranathan, Suzanna Tatarewicz, Julia Du, Brian Nguyen, Kate Widmann, Nessa Hawkins, Daniel J. Chin, Theresa LaVallee, Varun N. Kapoor

Coherus Biosciences, Redwood City, CA

Background

- Myeloid cell-mediated immunosuppression in the tumor microenvironment (TME) contributes to tumor immune evasion and PD-1 resistance.
- ILT4 (LILRB2) is an immunoreceptor tyrosine-based inhibitory motif (ITIM)–containing receptor that is widely expressed on immunosuppressive myeloid cells in the TME.
- Interaction of ILT4 with its primary ligands, HLA-A and HLA-G, promotes development of tolerogenic dendritic cells (DCs), immunosuppressive M2 macrophages, and inhibits pro-inflammatory cytokine production.
- CHS-1000 is a novel, humanized, modified IgG1 monoclonal antibody (mAb) that potently and specifically targets ILT4 and blocks its interaction with HLA-A and HLA-G.
- CHS-1000 promotes repolarization of suppressive M2 macrophages to a pro-inflammatory M1 phenotype and enhances activation of DCs and T cells *in vitro*.
- Combination of CHS-1000 with toripalimab-tpzi (PD-1 inhibitor) or other immunotherapy agents holds promise for improved antitumor therapies. The CHS-1000 IND to support FIH studies in cancer patients is planned for 2024.

CHS-1000 Mechanism of Action

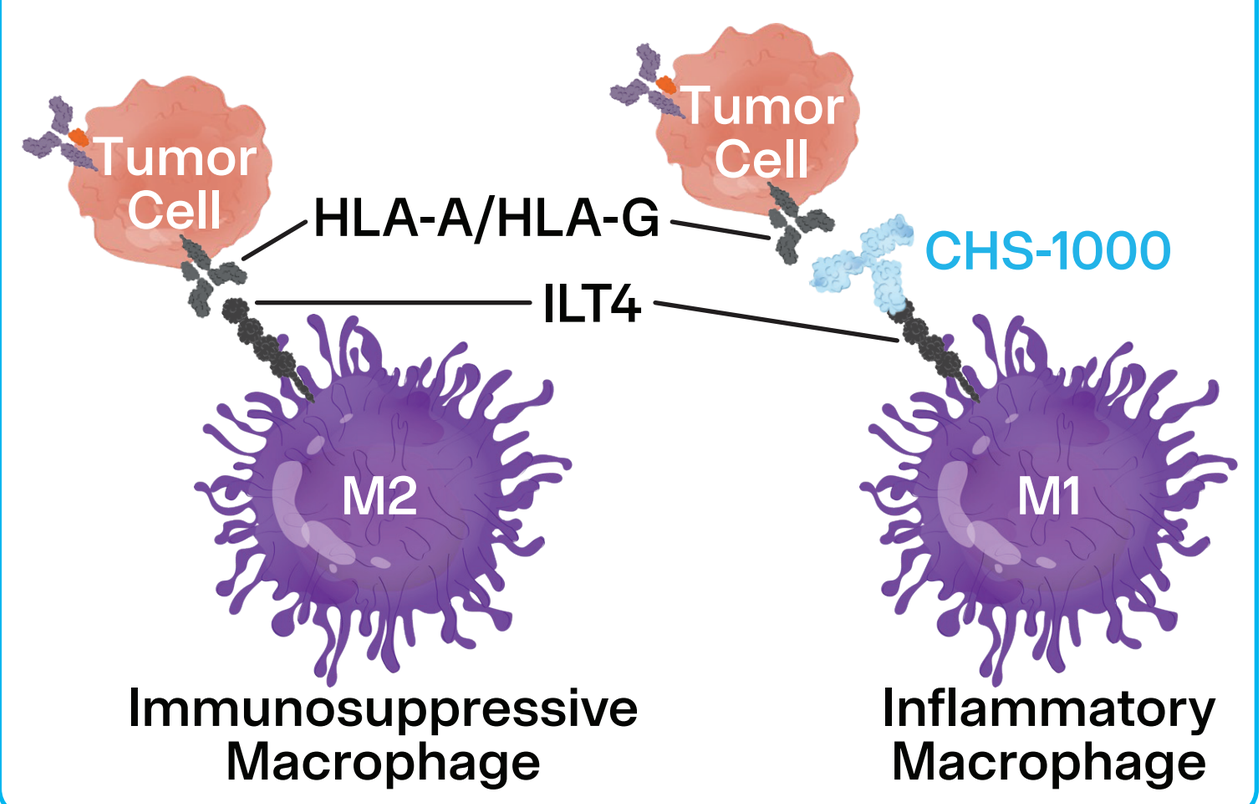
Tumor Cell
HLA-A/HLA-G
Tumor Cell
CHS-1000
ILT4
M2
M1
Immunosuppressive Macrophage
Inflammatory Macrophage

CHS-1000 is Specific and Selective for Human ILT4

A

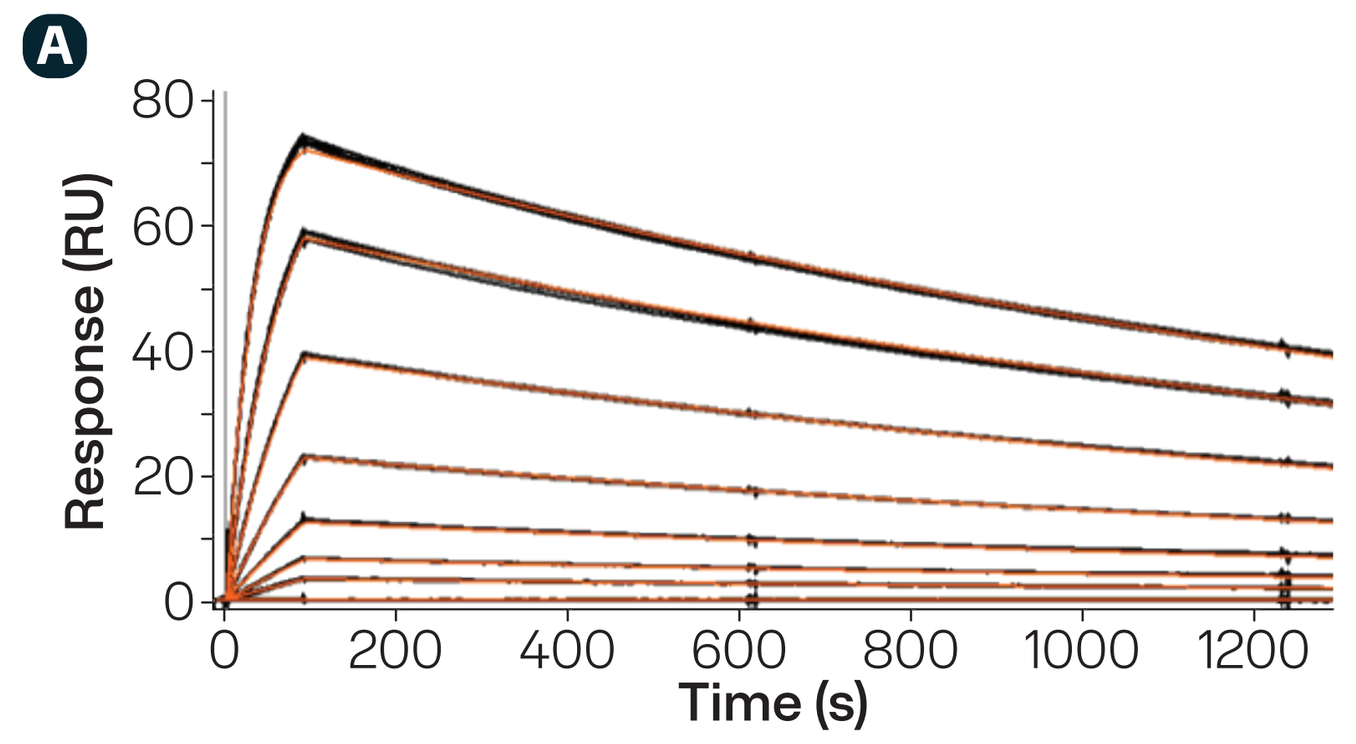
Response (RU)
Time (s)

	k_a ($M^{-1}s^{-1}$)	k_d (s^{-1})	K_D (pM)
Average* (95% CI)	$3.45 (0.39) \times 10^6$	$5.31 (0.28) \times 10^{-4}$	154 (9)

*Average of 3 experiments

B

LILRA and LILRB Family Members	CHS-1000 Binding
LILRA1	–
LILRA2	–
LILRA3	–
LILRA4	–
LILRA5	–
LILRA6	–
LILRB1	–
LILRB2 (ILT4)	+
LILRB3	–
LILRB4	–

SPR assays were carried out to confirm CHS-1000 binding to human ILT4 and to assess binding to human LILR family members. **(A)** Representative sensorgrams showing binding of ILT4 to CHS-1000 by SPR. Sensorgrams are shown in black and fits are shown in red. **(B)** CHS-1000 binds to LILRB2 and shows no binding to other LILRA or LILRB family members.

CHS-1000 Demonstrates Attenuated Binding to C1q and Fcγ Receptors but Retains Binding to Neonatal FcRn

Receptor	IgG1 Control (nM)	CHS-1000 (nM)
C1q	36 ± 7	229 ± 49
FcγRI	0.0667 ± 0.0010	136 ± 8
FcγRIIa	760 ± 140	4530 ± 1070
FcγRIIb/c	1370 ± 140	Negligible binding
FcγRIIIa (158V)	440 ± 90	2170 ± 190
FcγRIIIa (158F)	2610 ± 340	7780 ± 1410
FcγRIIIb	3620 ± 340	Negligible binding
FcRn	4780 ± 80	4910 ± 60

CHS-1000 binding affinity to human C1q, FcγRs, and FcRn was assessed by SPR and compared with an IgG1 control antibody (anti-human ILT4 IgG1 without LALA modification). CHS-1000 is engineered with substitutions in specific leucine residues to alanine (L240A and L241A [LALA]) that attenuates its binding to complement component C1q and FcγRs, particularly the high-affinity FcγRI. However, CHS-1000 preserves binding to the FcRn maintaining IgG-like PK parameters including half-life. Values shown are average K_D values with standard deviations from 2 or more measurements.

Abbreviations: 1E1 = Anti-ILT4 mAb based on U.S. Patent Application Publication No. 2018/0298096; ADCC = antibody-dependent cell-mediated cytotoxicity; C1q = complement component 1q; CDC = complement-mediated cytotoxicity; CHO = Chinese hamster ovary cells; DC = dendritic cell; ELISA = enzyme linked immunosorbent assay; FcγR = Fc gamma receptor; FcRn = neonatal Fc receptor; FIH = first in human; GM-CSF = granulocyte-macrophage colony stimulating factor; HLA = human leukocyte antigen; ILT4 = immunoglobulin-like transcript 4; IV = intravenous; k_a = association rate constant; k_d = dissociation rate constant; K_D = equilibrium dissociation constant; KLH = keyhole limpet hemocyanin; LDH = lactate dehydrogenase; LPS = lipopolysaccharide; M-CSF = macrophage colony-stimulating factor; mAb = monoclonal antibody; MFI = mean fluorescence intensity; NK = natural killer cell; OV = Ovarian; PBMC = peripheral blood mononuclear cells; PD-1 = programmed death-1 immunoglobulin superfamily member; PE = phycoerythrin; PK = pharmacokinetics; RLU = relative light units; RU = resonance units; TME = tumor microenvironment.

Reference: George, et al. *Nature*. 2015. Aug 6; 524(7563); 47-53.

Acknowledgments: The authors would like to thank Claire Willey and Kurt Koziol for thoughtful review and comments. Medical writing, editorial assistance, and poster production support were provided by Acumen Medical Communications, funded by Coherus Biosciences.

Statistics: * = p < 0.05; ** = p < 0.01; *** = p < 0.001; **** = p < 0.0001; when not shown, comparisons are not statistically significant.

CHS-1000 Lacks Effector Functions

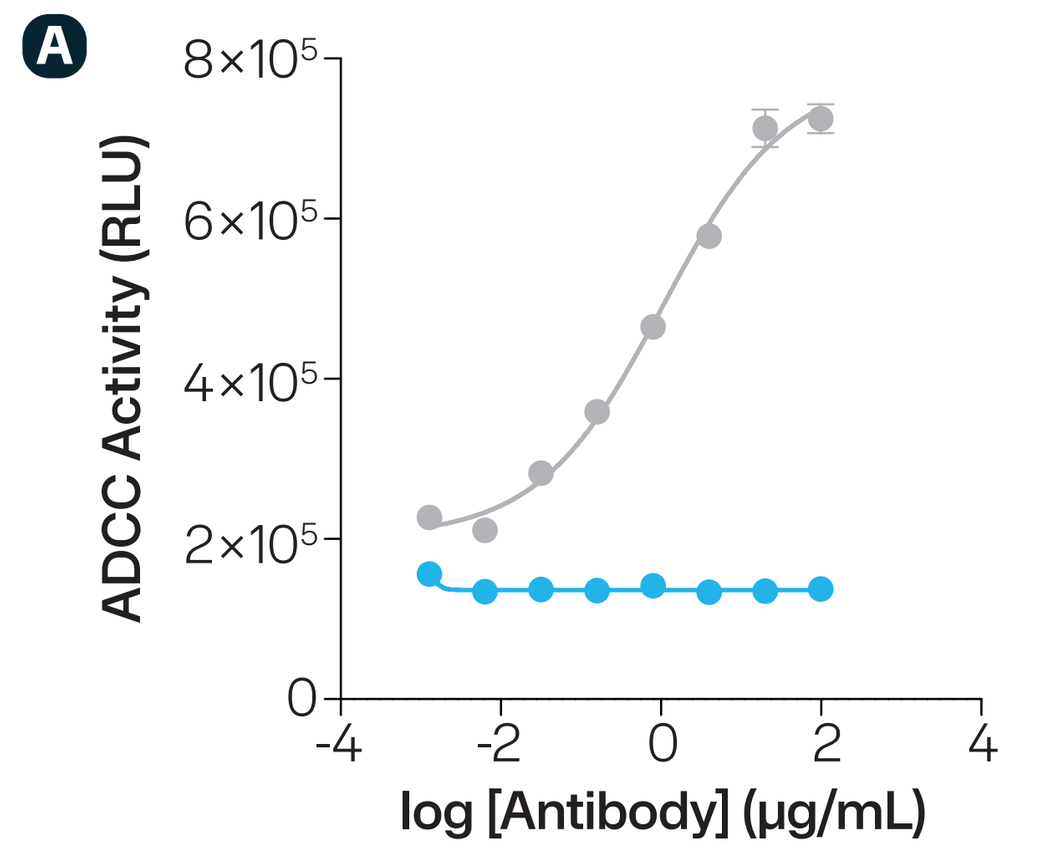
A
ADCC Activity (RLU)
log [Antibody] (μg/mL)
Rituximab
CHS-1000

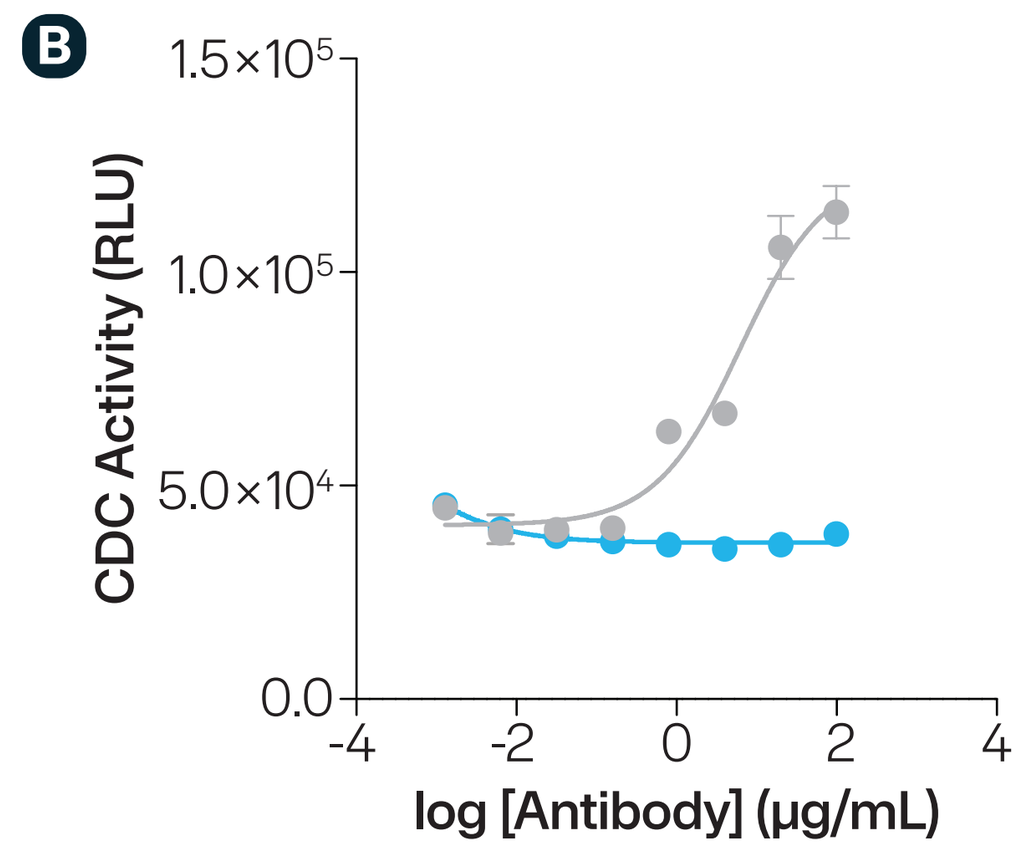
B
CDC Activity (RLU)
log [Antibody] (μg/mL)

Lack of ADCC- and CDC-inducing properties of CHS-1000 was confirmed in *in vitro* cytotoxicity assays. **(A)** Activated human PBMC from healthy donors were co-incubated with ILT4-expressing CHO cells at varying concentrations of CHS-1000. **(B)** Human ILT4-expressing CHO cells were incubated with human complement serum at varying CHS-1000 concentrations. Rituximab, an anti-CD20 IgG1 mAb, and its target, Raji cells, were used as assay controls. Cytotoxicity-related cell death was quantified using an LDH-Glo™ cytotoxicity assay.

CHS-1000 PK Profile Is Similar to Bevacizumab (IgG1) in a Human FcRn Transgenic Mouse Model

A

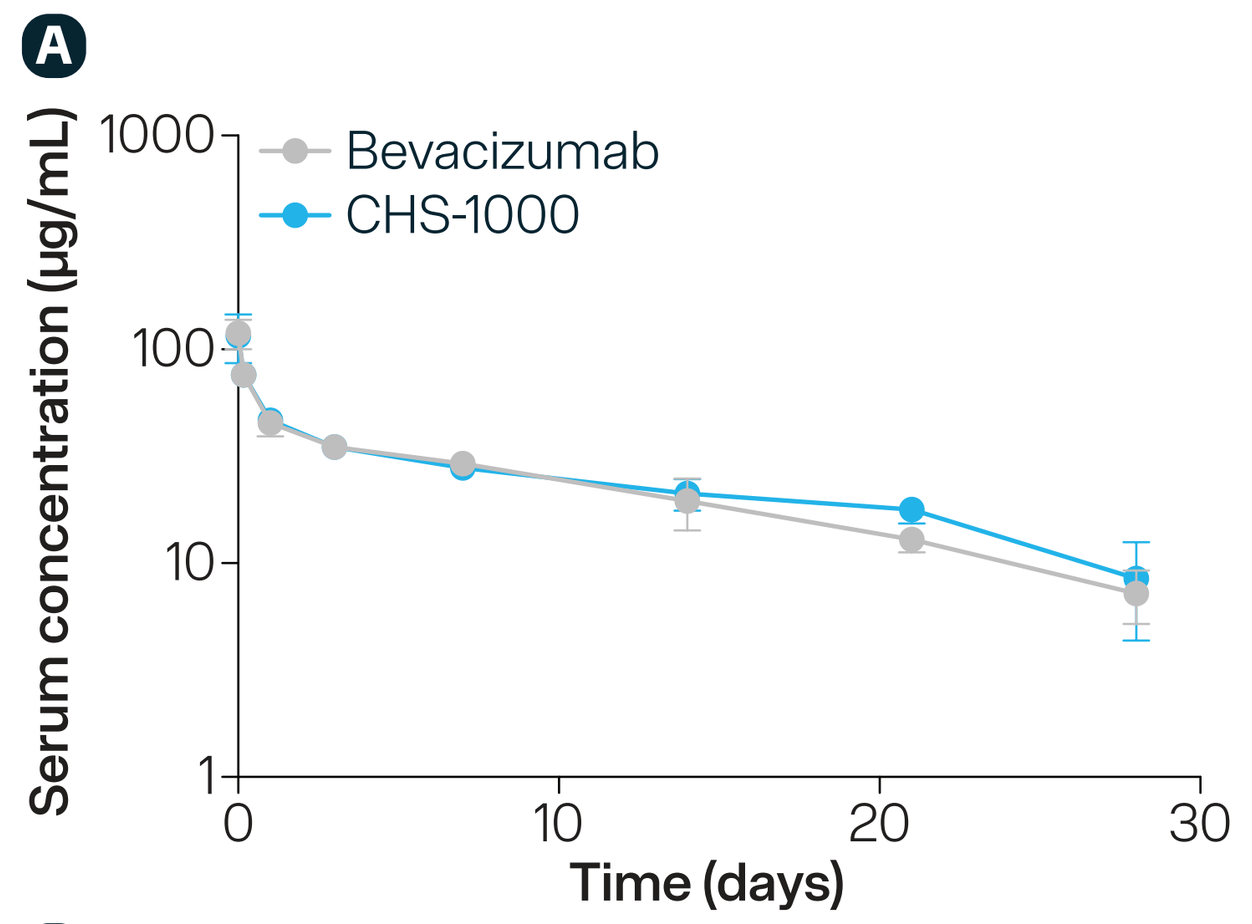
Serum concentration (μg/mL)
Time (days)
Bevacizumab
CHS-1000

Due to the lack of a pharmacologically relevant species, we performed *in vivo* PK evaluation of CHS-1000 in the transgenic huFcRn Tg32 mice expressing human FcRn. Homozygous male huFcRn Tg32 mice received a single 5 mg/kg IV dose of CHS-1000 (IgG1-LALA) or bevacizumab (IgG1) (n=6 per group). Blood samples were collected via serial bleeds at 5 minutes; 4 hours; and 1, 2, 7, 14, 21, and 28-days post-dose. **(A)** Time-concentration profiles and **(B)** summarized PK parameters for CHS-1000 and bevacizumab.

B

Group	$T_{1/2}$_range* (day)	$T_{1/2}$ (day)	C_{max} (μg/mL)	AUC_{0-t} (day·μg/mL)	$AUC_{0-\infty}$ (day·μg/mL)	CL_obs (mL/day/kg)	Vss_obs (mL/kg)
Bevacizumab	1-28	10.8	119	631	744	6.72	99.2
CHS-1000	1-28	12.7	116	675	830	6.02	103

*Time points used for calculating $T_{1/2}$

CHS-1000 Binds Selectively to Human Granulocytes, Monocytes, and DCs

A

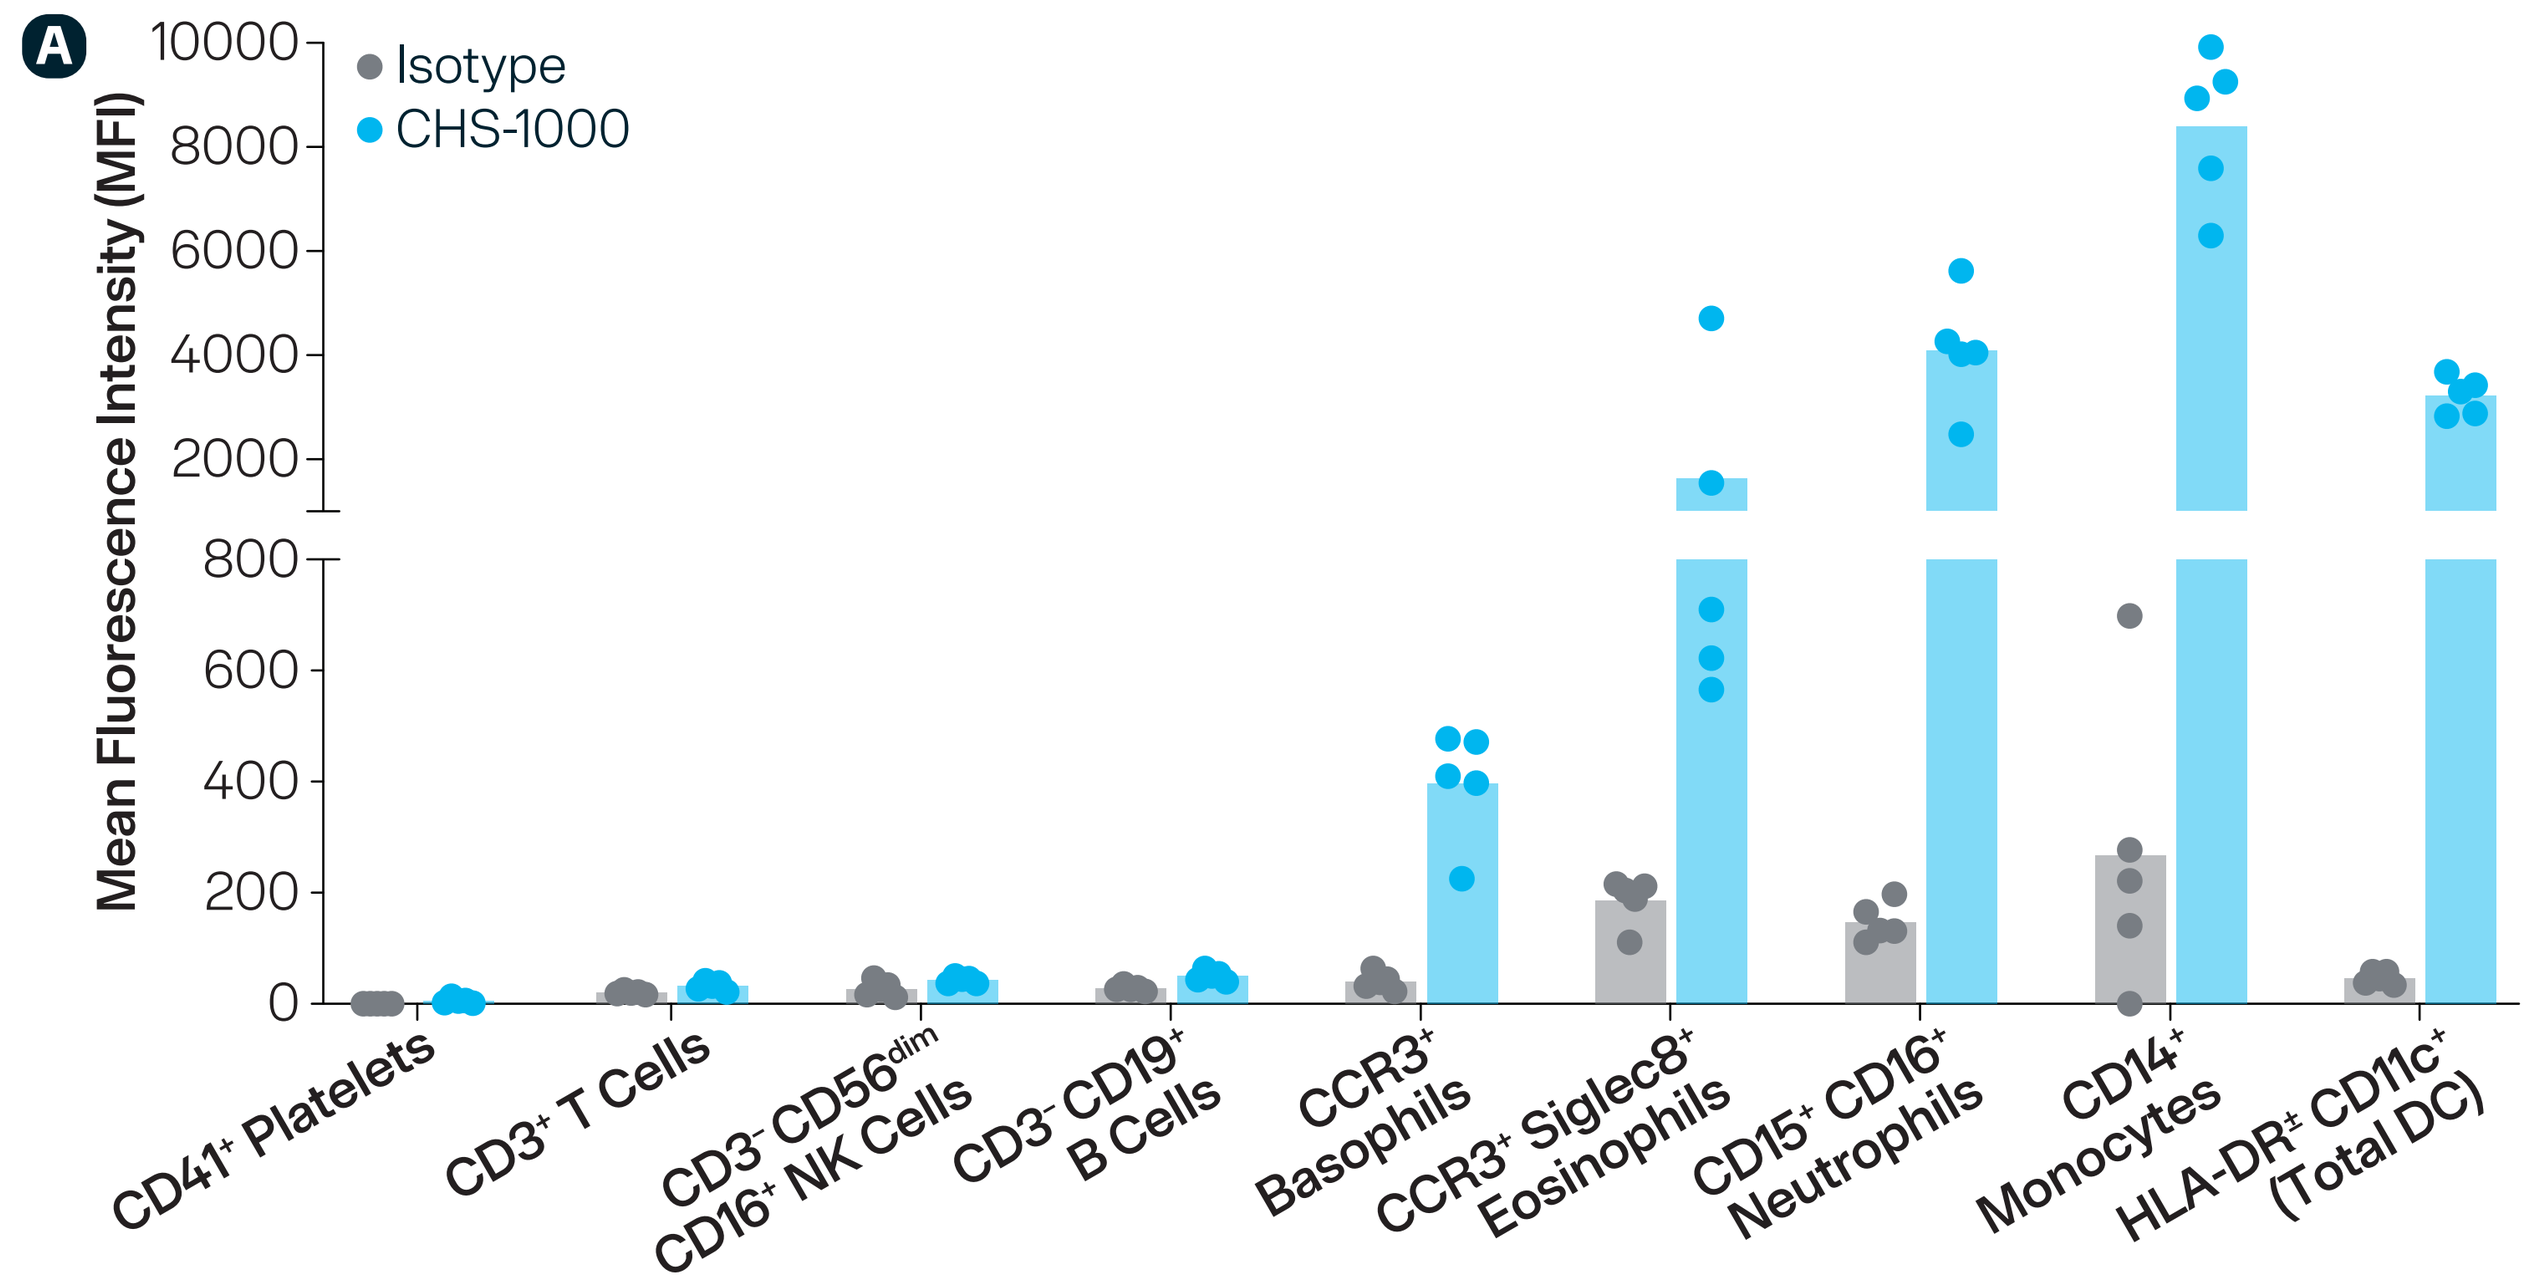
Mean Fluorescence Intensity (MFI)
Isotype
CHS-1000
CD41+ Platelets
CD3+ T Cells
CD3- CD56dim CD16+ NK Cells
CD3- CD19+ B Cells
CCR3+ Basophils
CCR3+ Siglec8+ Eosinophils
CD15+ CD16+ Neutrophils
CD14+ Monocytes
HLA-DR+ CD11c+ (Total DC)

B

	Cynomolgus	Rhesus	Canine	Mouse	Rat	Rabbit	Pig	Human
Platelet	–	–	NT	–	NT	NT	NT	–
T Cells	–	–	–	–	–	–	–	–
B Cells	–	–	–	–	–	–	–	–
NK Cells	–	–	NT	–	–	NT	–	–
Granulocytes	–	–	–	–	–	–	–	+
Monocytes	–	–	–	–	–	–	–	+
DC	–	–	NT	–	NT	NT	–	+

Whole blood from healthy human donors or various animal species was stained with fluorochrome-labeled CHS-1000 and an antibody cocktail identifying immune cell subsets, including cells of lymphoid and myeloid origin. **(A)** MFI of CHS-1000 for 5 replicates of each human cell subset. **(B)** Binding of CHS-1000 in immune cell subsets of all species tested is listed as either + (positive), – (negative) or NT (Not Tested). Granulocytes = eosinophils, basophils, and neutrophils. Isotype control: anti-KLH-IgG1-LALA.

CHS-1000 Blocks Human HLA-A and HLA-G Ligand Binding to ILT4

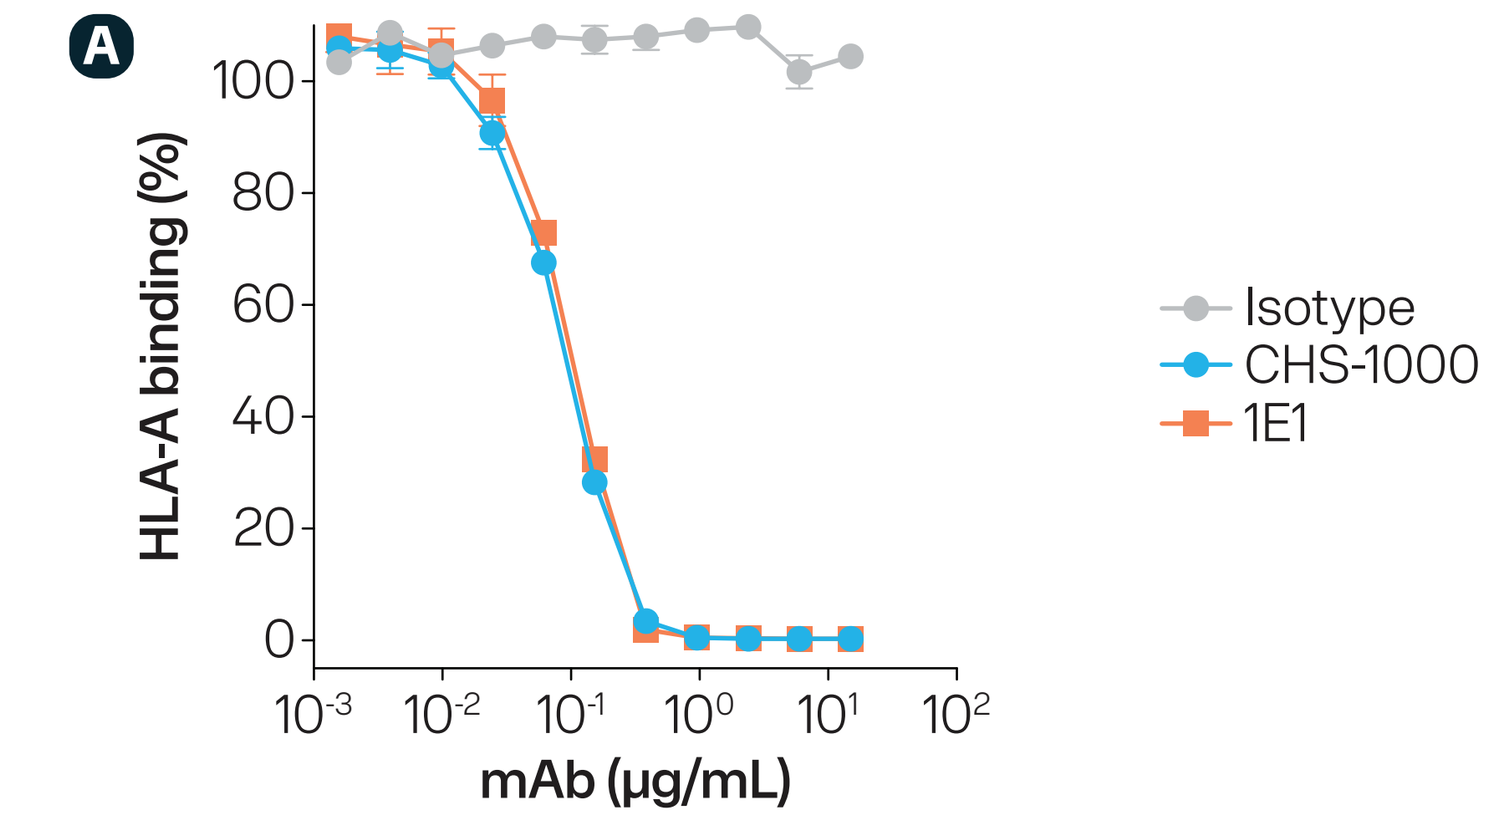
A
HLA-A binding (%)
mAb (μg/mL)
Isotype
CHS-1000
1E1

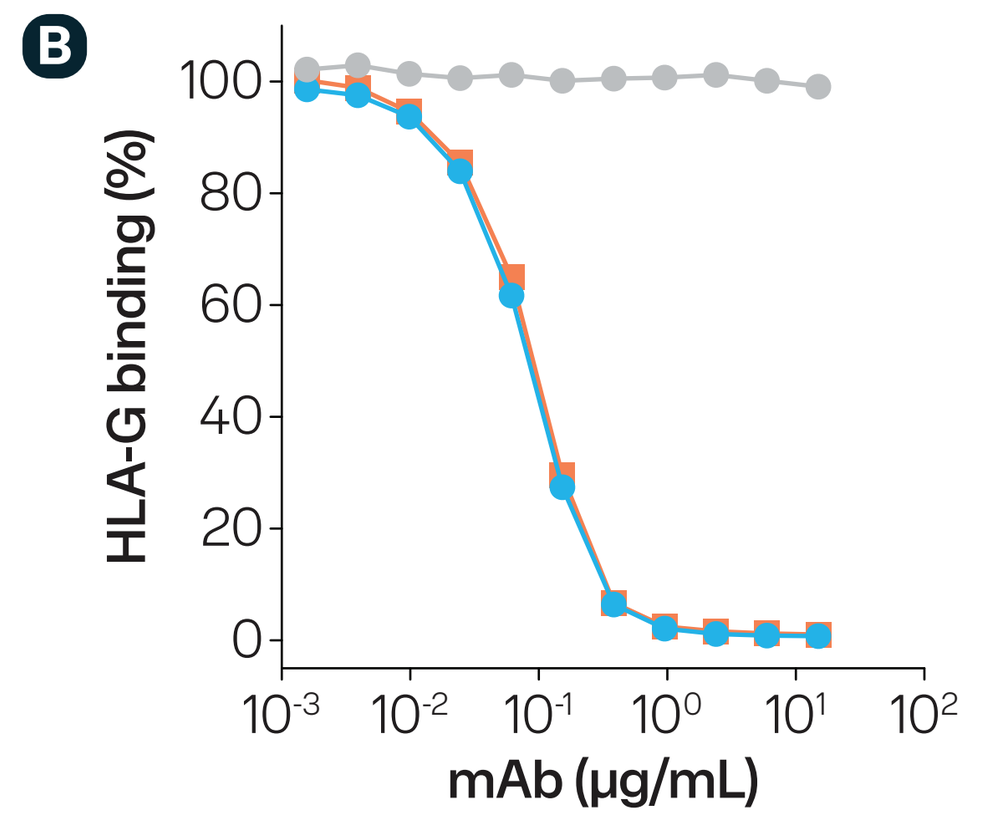
B
HLA-G binding (%)
mAb (μg/mL)

C

	HLA-A Binding		HLA-G Binding	
	CHS-1000	1E1	CHS-1000	1E1
IC_{50} (μg/mL), mean ± SD*	0.0862 ± 0.0073	0.0972 ± 0.0004	0.0825 ± 0.0040	0.0878 ± 0.0004

*Average of 2 experiments

CHS-1000 at various concentrations (0-15 μg/mL) was incubated with human ILT4-expressing CHO (ILT4-CHO) cells. Cells were washed and stained with 5 μg/mL **(A)** PE-HLA-A or **(B)** PE-HLA-G tetramer and analyzed by flow cytometry. Representative image from 3 independent experiments is shown with the data normalized to the binding in the absence of CHS-1000. Each data point is the mean of triplicates (± SD). **(C)** Average IC_{50} values for HLA-A and HLA-G binding from 2 independent experiments.

CHS-1000 Treatment Leads to Activation of Human Dendritic Cells

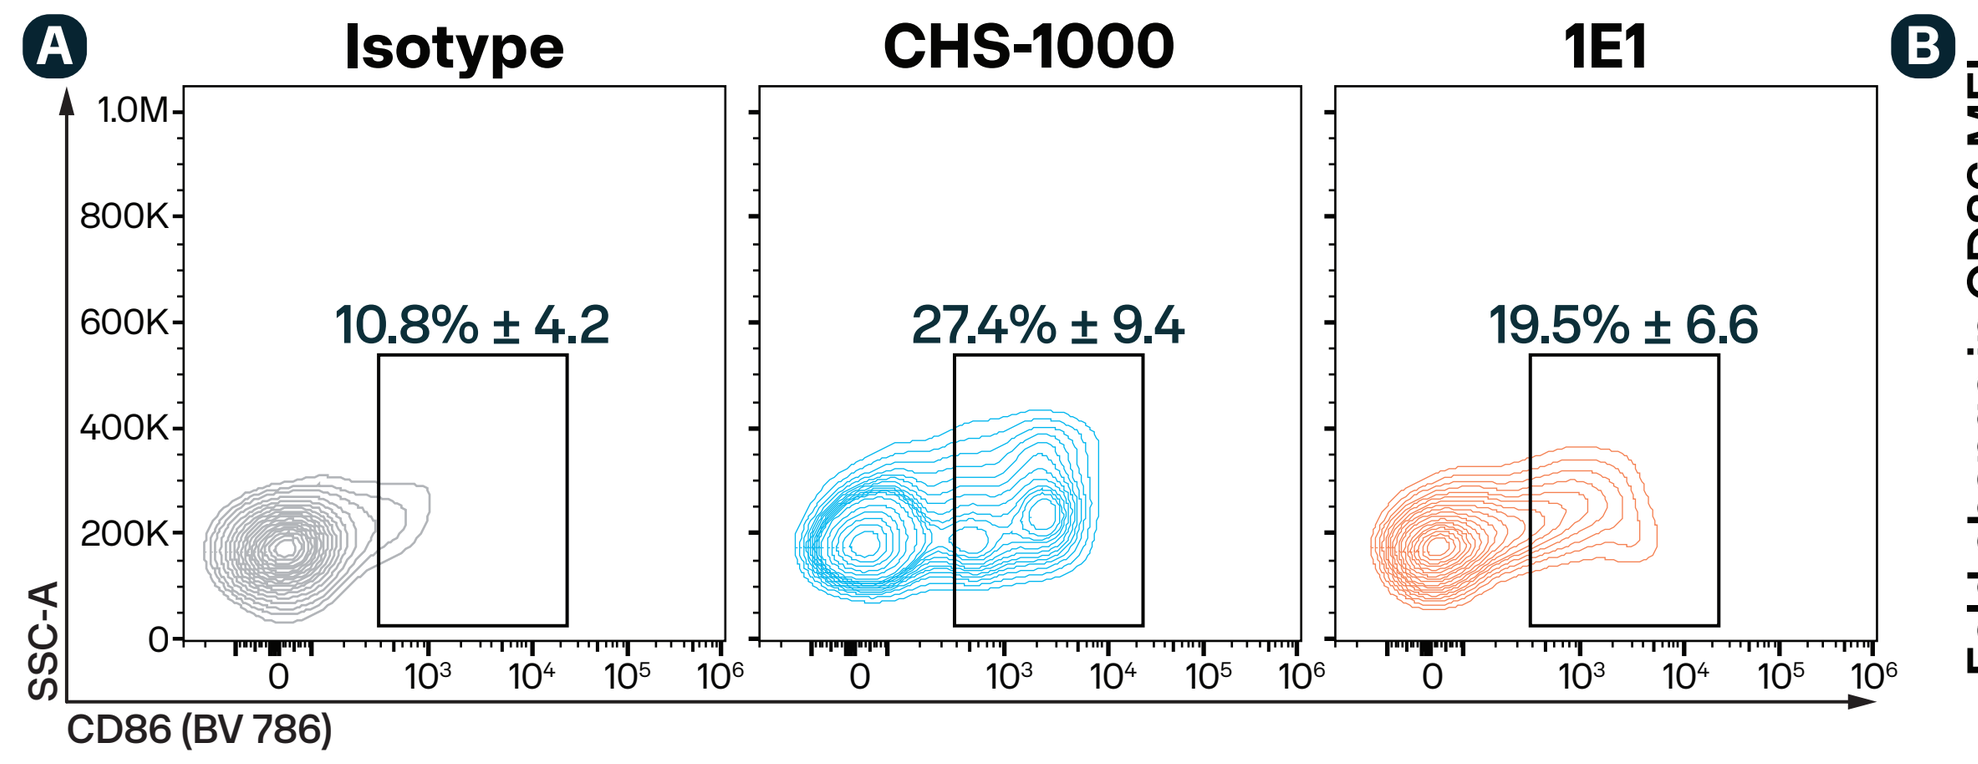
A
Isotype
CHS-1000
1E1
10.8% ± 4.2
27.4% ± 9.4
19.5% ± 6.6
SSC-A
CD86 (BV 786)

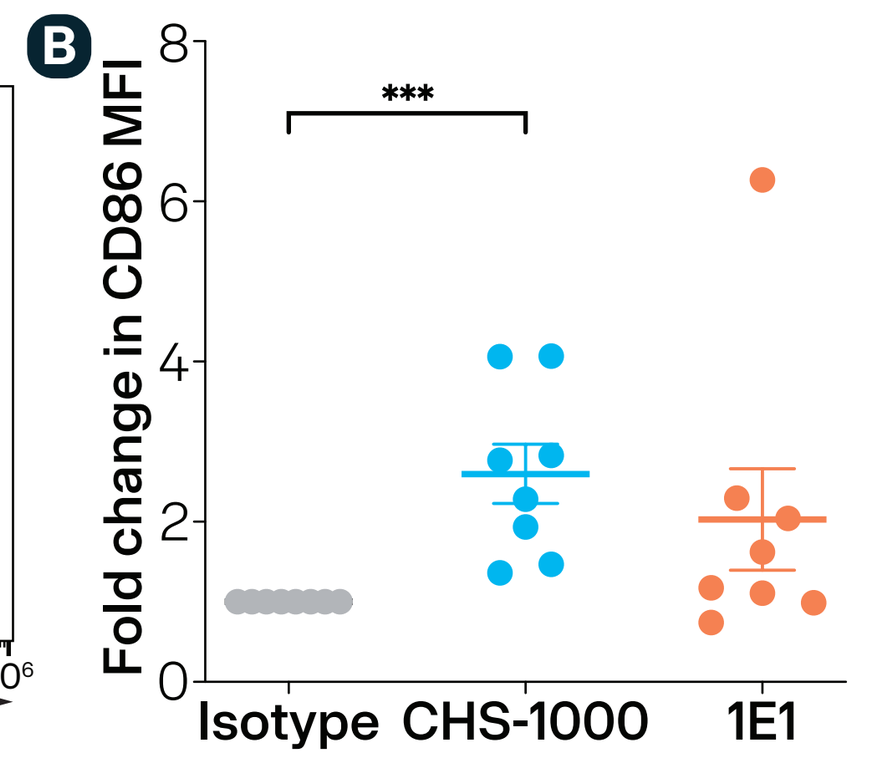
B
Fold change in CD86 MFI

Isotype
CHS-1000
1E1

Human monocytes isolated from peripheral blood of healthy donors were differentiated into DCs in the presence of GM-CSF and IL-4. DCs were treated with 1 μg/mL of CHS-1000, 1E1, or isotype control for 7 days and expression of activation marker CD86 quantified by flow cytometry. **(A)** Representative plots showing CD86 expression (gated population) with mean % expression ± SEM for each treatment. **(B)** Fold change in MFI for CD86 expression with respect to isotype control. Statistical analysis was performed using unpaired t test. n=8.

CHS-1000 Treatment Promotes Polarization of Macrophages to an Inflammatory M1 Phenotype

CHS-1000 Treatment Reduces Expression of M2 Macrophage Marker CD163

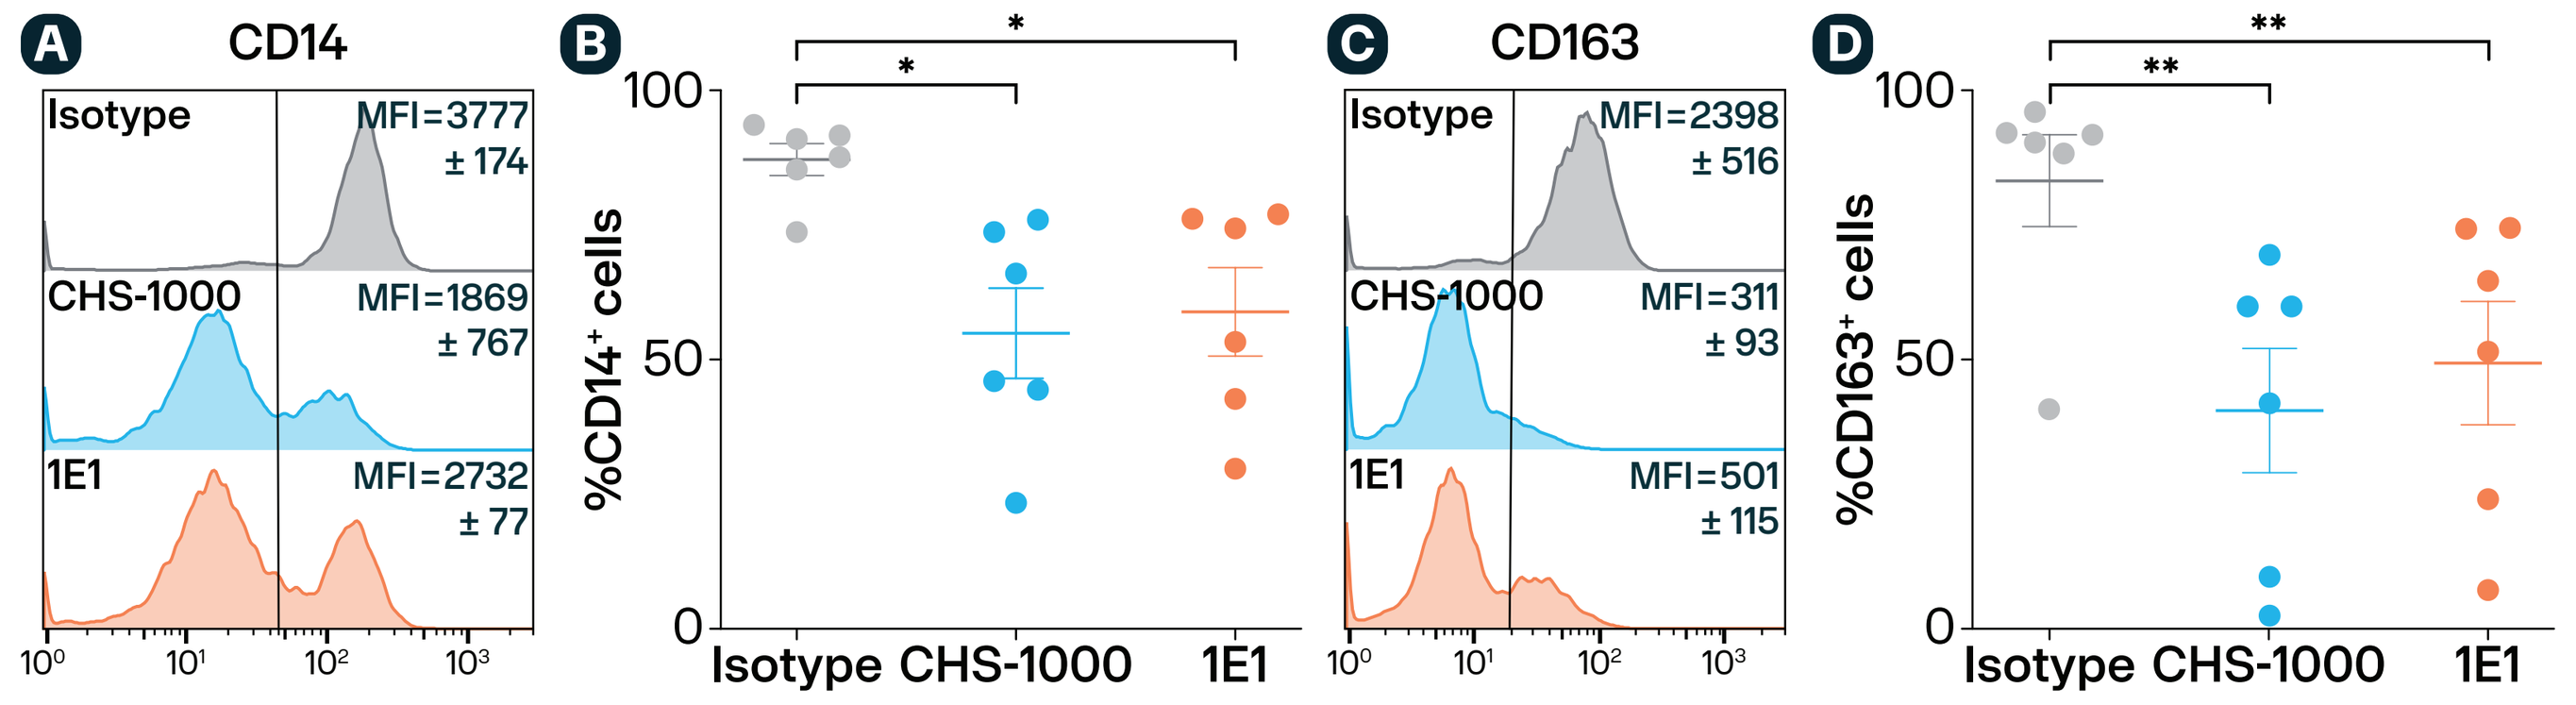
A
CD14
Isotype
MFI=3777 ± 174
CHS-1000
MFI=1869 ± 767
1E1
MFI=2732 ± 77
B
%CD14+ cells
C
CD163
Isotype
MFI=2398 ± 516
CHS-1000
MFI=311 ± 93
1E1
MFI=501 ± 115
D
%CD163+ cells
Isotype
CHS-1000
1E1

CHS-1000 Treatment Increases Expression of Inflammatory Cytokines

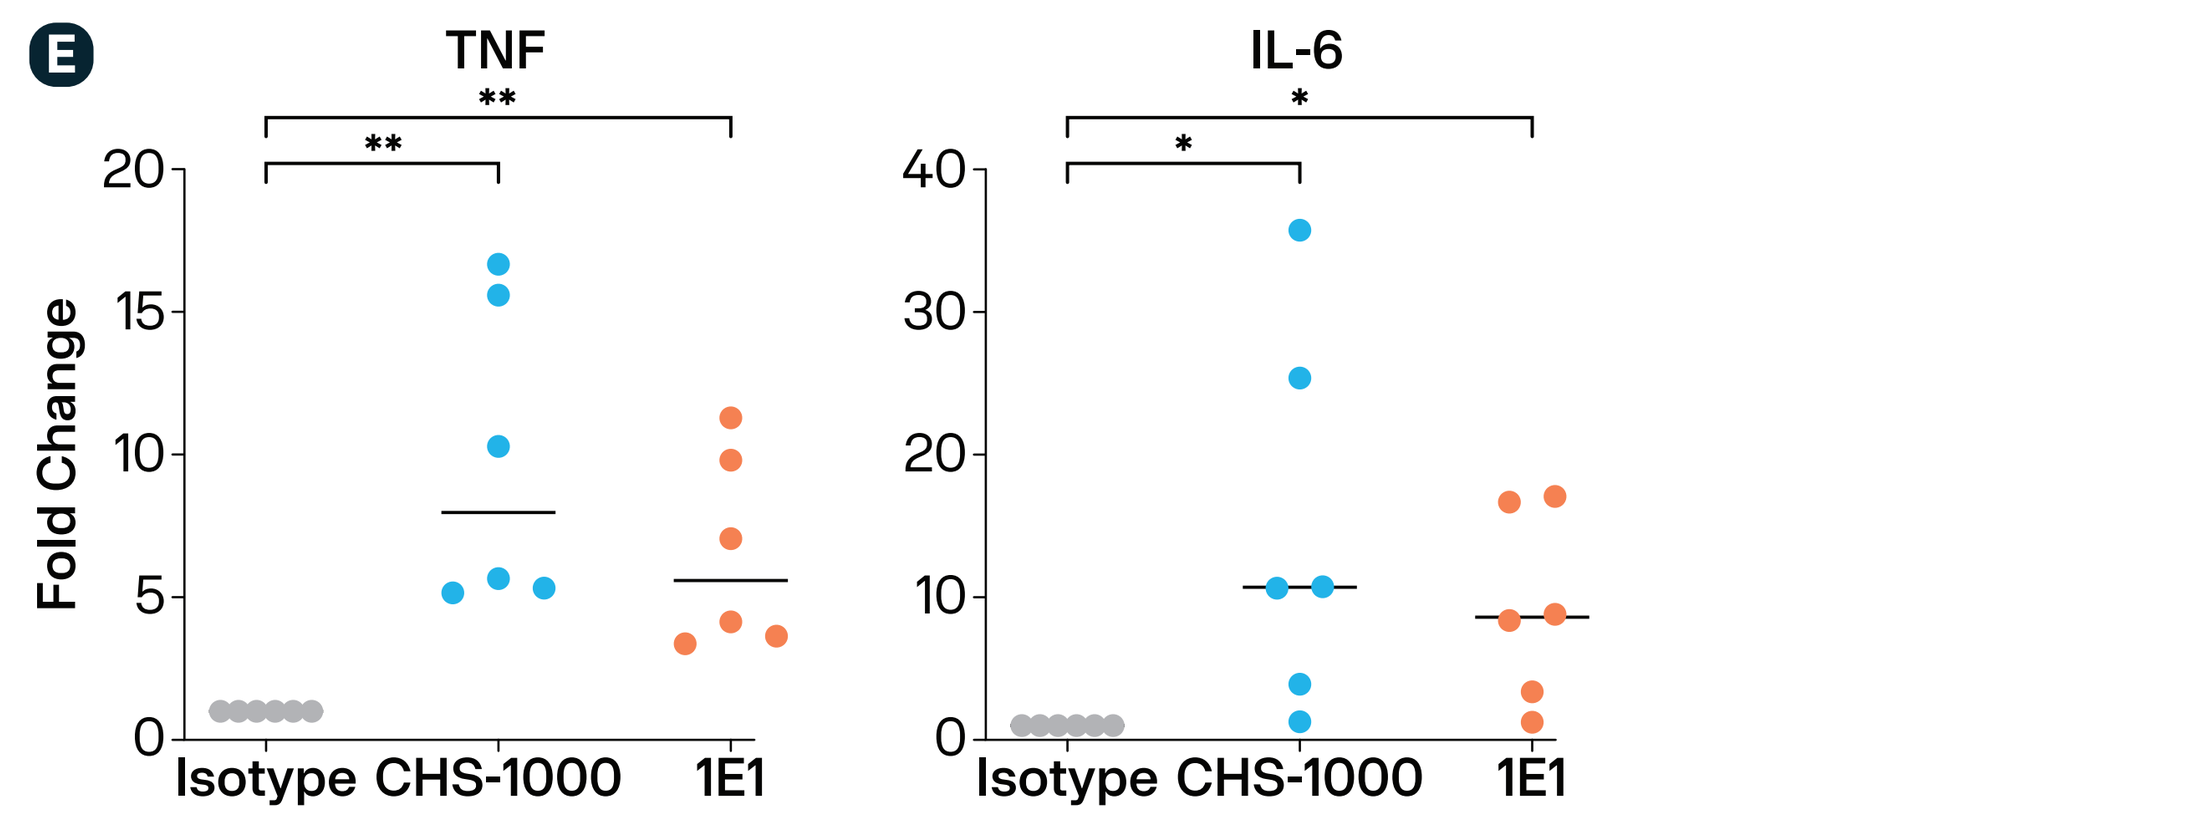
E
TNF
IL-6
Fold Change
Isotype
CHS-1000
1E1

Human monocyte derived macrophages cultured in the presence of LPS were treated with 4 μg/mL of CHS-1000, 1E1, or isotype control antibody for 24 hours. Treated cells were then tested for expression of surface markers CD14 (associated with immunosuppressive function of myeloid cells) and CD163 (marker for M2 macrophages). **(A)** & **(C)** are representative flow cytometry plots showing CD14 and CD163 expression with average MFI ± SEM for each treatment. **(B)** & **(D)** show the average frequency of cells expressing CD14 and CD163, respectively. Cell culture supernatants were collected and analyzed for levels of TNF and IL-6. **(E)** Fold change of TNF and IL-6 compared with isotype control. Statistical analysis was performed using unpaired t test for **(B)** & **(D)** and paired t test for **(E)**. n=6.

CHS-1000 Treatment Results in T Cell Activation in a Mixed Lymphocyte Reaction Assay

A CHS-1000 promotes upregulation of IFN-γ expression by T cells

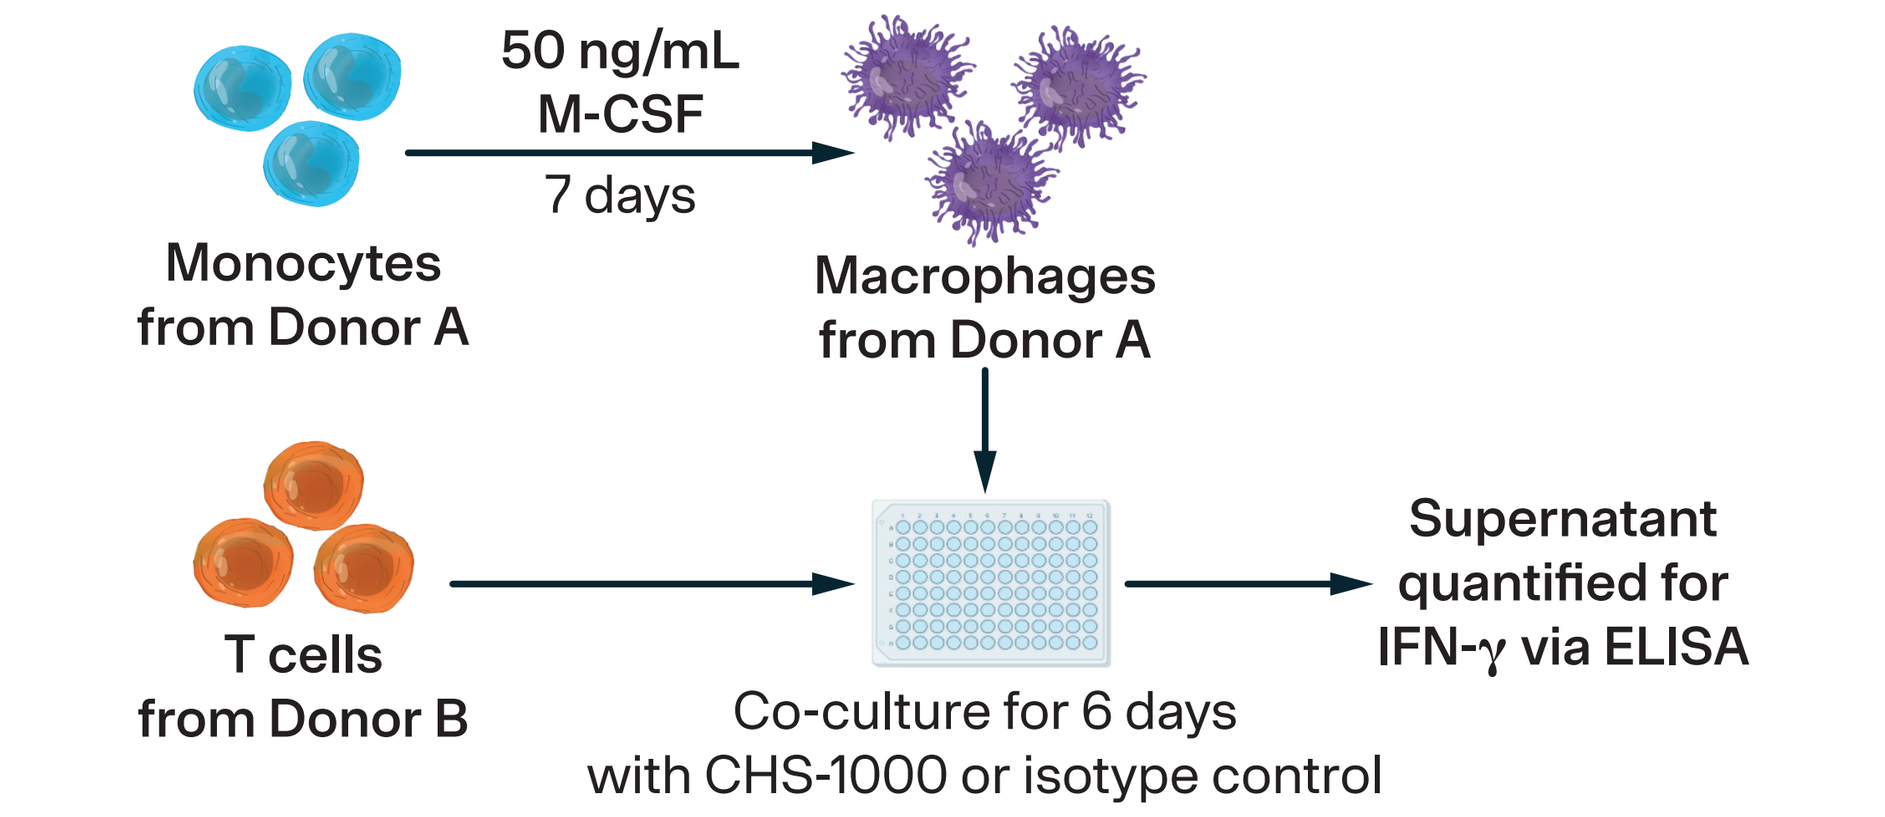
Monocytes from Donor A
50 ng/mL M-CSF
7 days
Macrophages from Donor A
T cells from Donor B
Co-culture for 6 days with CHS-1000 or isotype control
Supernatant quantified for IFN-γ via ELISA

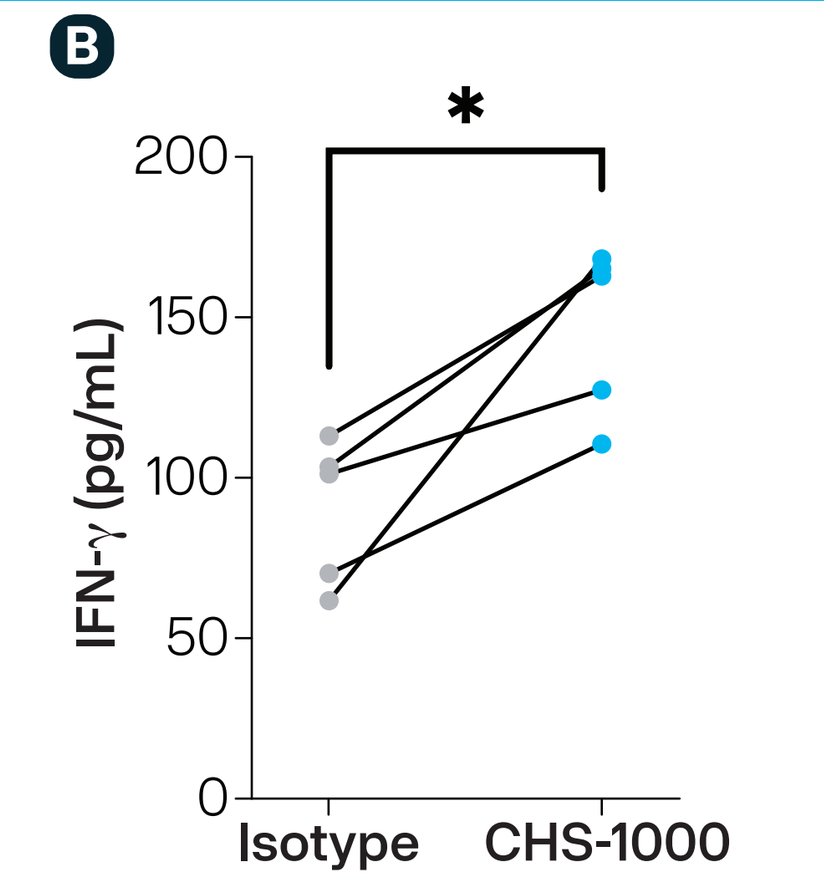
B
*
IFN-γ (pg/mL)
Isotype
CHS-1000

(A) Monocytes isolated from peripheral blood of a healthy human donor were differentiated to M2 macrophages and co-cultured with T cells from mismatched donors at a ratio of 1:10 in the presence of 4 μg/mL CHS-1000 or isotype control. **(B)** IFN-γ levels in culture supernatants collected on Day 6 were quantified by ELISA. Each data point represents an individual donor. Statistical analysis was performed using paired t test. n=5.

Disease Linkage Studies to Identify Solid Tumors Enriched in ILT4 and CD163 (M2 Macrophage) Expression

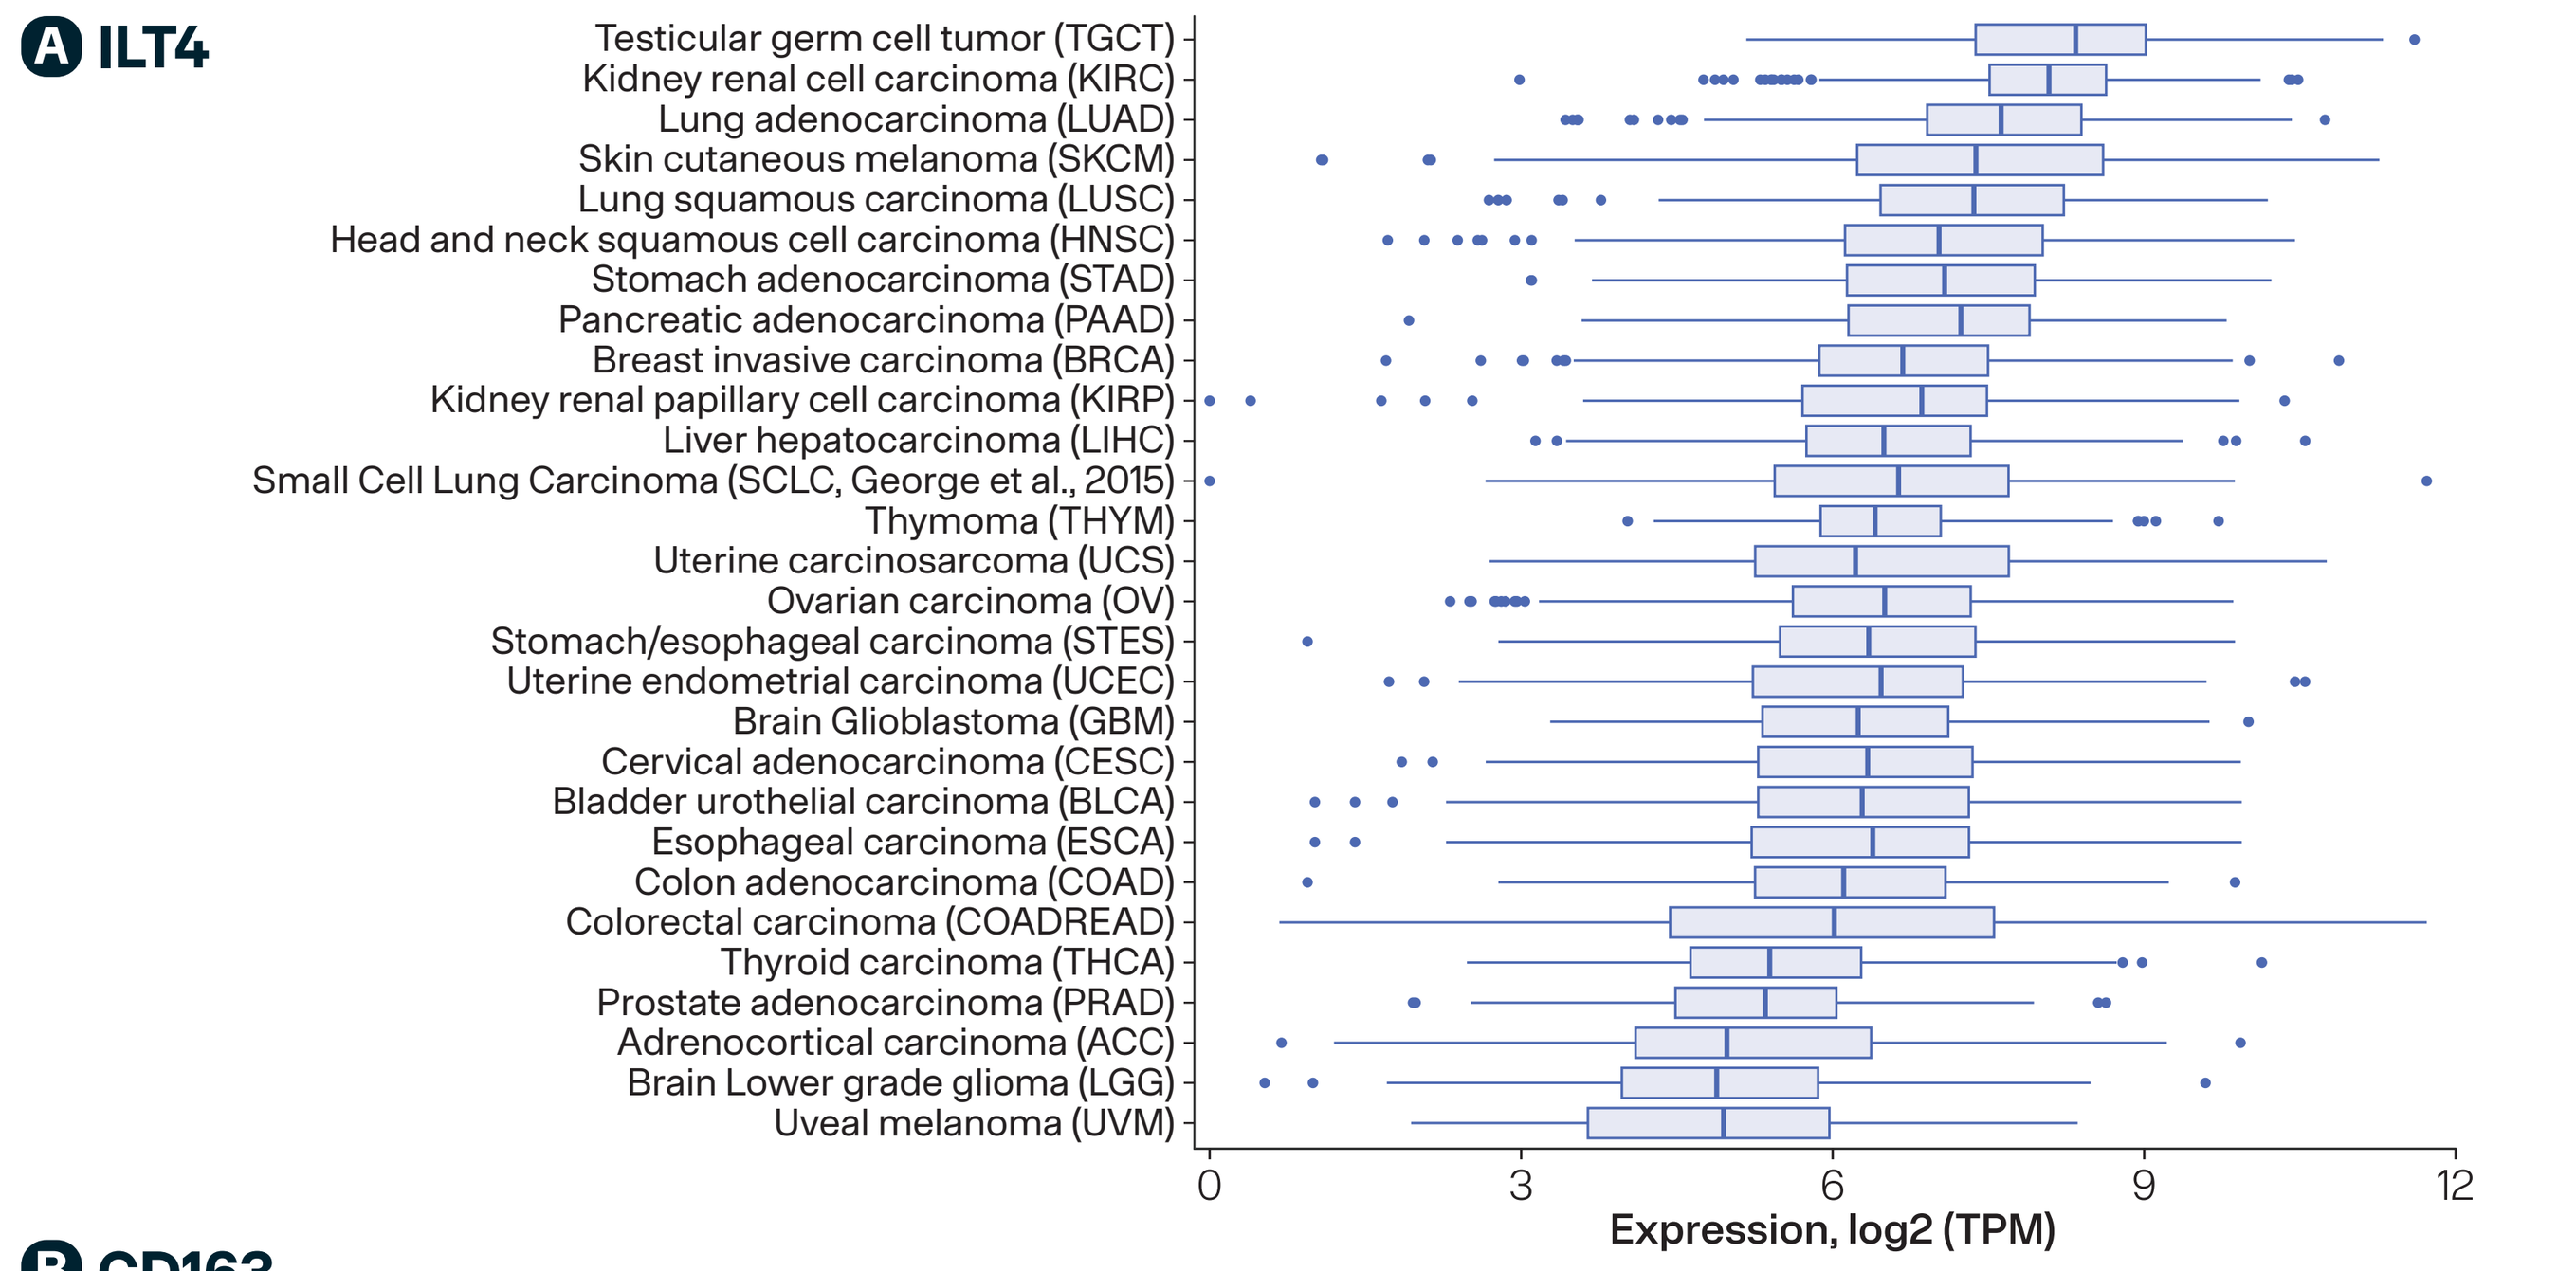
A ILT4
Testicular germ cell tumor (TGCT)
Kidney renal cell carcinoma (KIRC)
Lung adenocarcinoma (LUAD)
Skin cutaneous melanoma (SKCM)
Lung squamous carcinoma (LUSC)
Head and neck squamous cell carcinoma (HNSC)
Stomach adenocarcinoma (STAD)
Pancreatic adenocarcinoma (PAAD)
Breast invasive carcinoma (BRCA)
Kidney renal papillary cell carcinoma (KIRP)
Liver hepatocarcinoma (LIHC)
Small Cell Lung Carcinoma (SCLC, George et al., 2015)
Thymoma (THYM)
Uterine carcinosarcoma (UCS)
Ovarian carcinoma (OV)
Stomach/esophageal carcinoma (STES)
Uterine endometrial carcinoma (UCEC)
Brain Glioblastoma (GBM)
Cervical adenocarcinoma (CESC)
Bladder urothelial carcinoma (BLCA)
Esophageal carcinoma (ESCA)
Colon adenocarcinoma (COAD)
Colorectal carcinoma (COADREAD)
Thyroid carcinoma (THCA)
Prostate adenocarcinoma (PRAD)
Adrenocortical carcinoma (ACC)
Brain Lower grade glioma (LGG)
Uveal melanoma (UVM)
Expression, log2 (TPM)

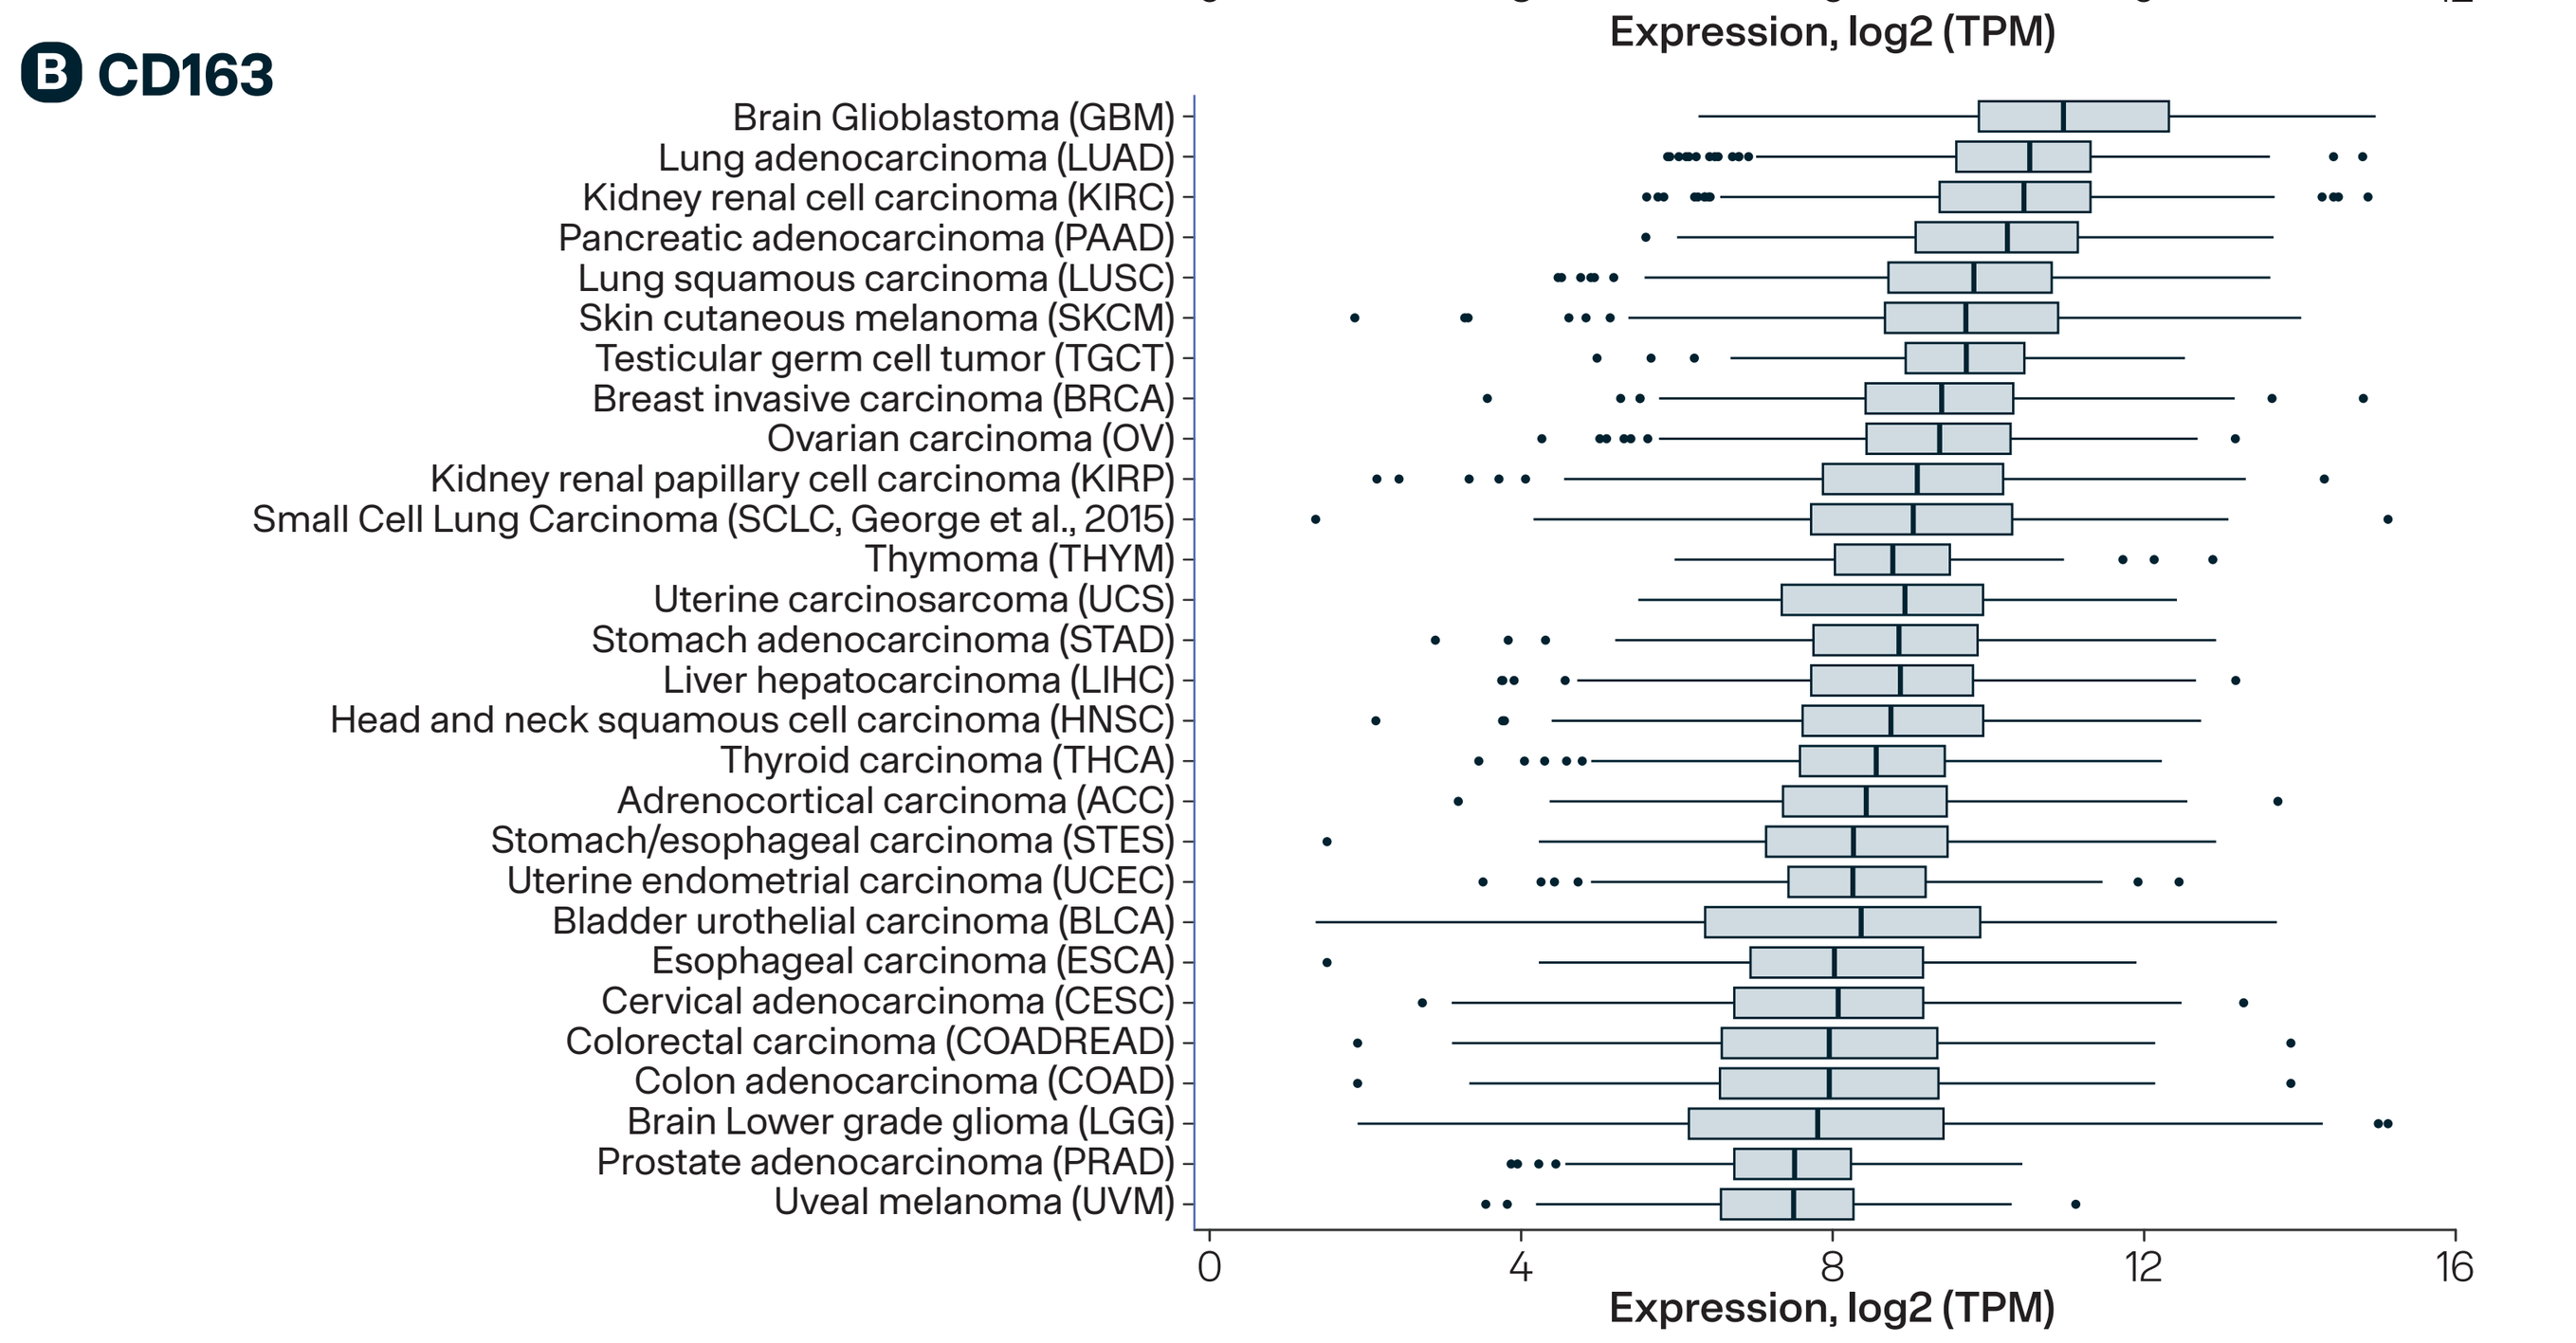
B CD163
Brain Glioblastoma (GBM)
Lung adenocarcinoma (LUAD)
Kidney renal cell carcinoma (KIRC)
Pancreatic adenocarcinoma (PAAD)
Lung squamous carcinoma (LUSC)
Skin cutaneous melanoma (SKCM)
Testicular germ cell tumor (TGCT)
Breast invasive carcinoma (BRCA)
Ovarian carcinoma (OV)
Kidney renal papillary cell carcinoma (KIRP)
Small Cell Lung Carcinoma (SCLC, George et al., 2015)
Thymoma (THYM)
Uterine carcinosarcoma (UCS)
Stomach adenocarcinoma (STAD)
Liver hepatocarcinoma (LIHC)
Head and neck squamous cell carcinoma (HNSC)
Thyroid carcinoma (THCA)
Adrenocortical carcinoma (ACC)
Stomach/esophageal carcinoma (STES)
Uterine endometrial carcinoma (UCEC)
Bladder urothelial carcinoma (BLCA)
Esophageal carcinoma (ESCA)
Cervical adenocarcinoma (CESC)
Colorectal carcinoma (COADREAD)
Colon adenocarcinoma (COAD)
Brain Lower grade glioma (LGG)
Prostate adenocarcinoma (PRAD)
Uveal melanoma (UVM)
Expression, log2 (TPM)

Data from The Cancer Genome Atlas (supplemented with published SCLC data, George et al., 2015) were used to analyze the expression and enrichment of **(A)** ILT4 (target) and **(B)** CD163 (marker for M2 macrophages) in solid tumor indications for disease linkage studies.

Conclusions

- CHS-1000 is a novel mAb targeting human ILT4 that has favorable pharmacodynamic properties that warrants evaluation in cancer patients in combination with toripalimab-tpzi (PD-1 inhibitor) with the aim of improving antitumor immunity and clinical benefit.
- CHS-1000 binds specifically and selectively to human ILT4 with high affinity and showed no cross reactivity to other LILRB family members.
- CHS-1000 efficiently blocks the interaction of ILT4 to its ligands, HLA-A and HLA-G, and reverses ILT4-mediated immunosuppressive functions leading to activation of M1 macrophage, DC and T cell activation, and increases in pro-inflammatory cytokine secretion in *in vitro* assays.
- CHS-1000 lacks ADCC or CDC activity consistent with the LALA substitution and has IgG1-like linear PK parameters in human FcRn transgenic mice.
- Combinations of CHS-1000 with toripalimab-tpzi (PD-1 inhibitor) or other immunotherapy agents hold promise for enhanced anti-tumor activity and clinical benefit in multiple solid tumors.
- CHS-1000 IND filing to enable FIH studies in adult patients with solid tumors is planned for 2024.